4206

Healthcare Resource Use and Costs in Patients With Idiopathic Pulmonary Fibrosis (IPF) in the US Medicare Population

Sheila R. Reddy,[1] Eunice Chang,[1] Michael S. Broder,[1] Sohum Gokhale,[1] Mitral Corral[2]

[1]Partnership for Health Analytic Research, LLC, Beverly Hills, CA; [2]Genentech, Inc., South San Francisco, CA

BACKGROUND

- Idiopathic pulmonary fibrosis (IPF) is an irreversible, progressive, fibrotic lung disease with a poor prognosis[1,2]
- The antifibrotic therapies pirfenidone and nintedanib were approved by the US Food and Drug Administration on October 15, 2014, for the treatment of IPF[3-4]
 - Both therapies slow disease progression as measured by changes in forced vital capacity[5-7]
- Respiratory-related hospitalizations are associated with increased morbidity, cost and risk of death in patients with IPF[8-10]
- Previous analyses of US Medicare beneficiaries have shown that the annual IPF-attributable medical cost to the US healthcare system, excluding medication costs, is approximately $2 billion[11]
- Limited real-world data are available about the healthcare utilization and costs associated with antifibrotic use in patients with IPF

OBJECTIVE

- To examine healthcare utilization and costs among Medicare beneficiaries with IPF who initiated antifibrotic therapy

METHODS

Patients

- A retrospective observational study of Medicare beneficiaries using the 100% Medicare Research Identifiable File was conducted using data from October 2014 through December 2015
- Patients were included if they met each of the following criteria:
 - Initiated antifibrotic therapy (≥ 1 fill of pirfenidone or nintedanib) during the identification period, with the first fill date defined as the index date
 - No use of antifibrotics in the 2 years prior to the index date (baseline period); extended baseline period allowed for the identification of patients with newly diagnosed disease
 - ≥ 1 inpatient or outpatient claim with IPF listed as a diagnosis (ICD-9-CM: 516.3, 516.30 and 516.31; ICD-10-CM: J84.111 and J84.112) occurring on or before the index date
 - Aged ≥ 67 years on index date
- Key exclusion criteria were lung transplant prior to index and lack of eligibility for, or lack of continuous enrollment in, fee-for-service Medicare Parts A and B or Medicare Part D (and those enrolled in Medicare Advantage) during baseline
- The cohort was divided into 2 study groups: pirfenidone or nintedanib initiators
- Patients who received a lung transplant after index, switched or ended treatment, disenrolled or died were censored at the time of occurrence (for end of treatment, censoring occurred 60 days later); thus, follow-up ended at censoring or study end, whichever occurred first

Outcomes

- The following outcomes were measured:
 - Healthcare utilization
 - Proportion of patients with and length of stay for hospitalization (all cause and respiratory related)
 - Proportion of patients with emergency department (ED) visits
 - Mean number of office visits, ED visits and inpatient hospitalizations
 - Healthcare costs
 - Inpatient services (excluding those associated with lung transplant)
 - Outpatient services (including ED)
 - Outpatient pharmacy claims
- Claims with a primary diagnosis of respiratory disease (ICD-9-CM: 460.xx–519.xx; ICD-10-CM: J00.xx–J99.xx) or a diagnosis of respiratory disease in any field were defined as respiratory related for inpatient and outpatient services, respectively

Statistical Analyses

- Demographics and clinical characteristics were characterized using descriptive statistics
- Healthcare utilization, except hospitalization, and costs were measured descriptively as monthly outcomes (per patient month) and weighted by follow-up length
 - The treatment period was defined from the index date until either 60 days after end of index treatment or end of follow-up, whichever occurred first
 - All costs were inflation-adjusted to 2015 United States Dollars
- Stabilized inverse probability of treatment weighting (IPTW) using the propensity score was used to adjust for baseline confounding factors
 - Propensity scores were estimated using the following covariates: age, sex, region, Charlson comorbidity index (excluding chronic obstructive pulmonary disease [COPD]), COPD, stroke, newly diagnosed IPF, pneumonia 3 months prior to initiation, quartile of median income of patient residential area and distance from patient residential area to interstitial lung disease (ILD) specialty center
- Risk of hospitalization, based on time to first hospitalization, was compared between study groups using Kaplan-Meier estimation and Cox proportional hazard modeling
- Rate of hospitalization, based on the number of hospitalizations per patient year, was compared between study groups using negative binomial modeling
- All models included the same baseline covariates as the IPTW procedure to adjust for potential residual confounding and to assess individual main effects

RESULTS

Study Population

- In total, 3546 patients who received a diagnosis of IPF initiated antifibrotic therapy and met the study criteria (**Table 1**)
 - 2082 patients received pirfenidone
 - 1464 patients received nintedanib
- The majority of patients in both groups were male and white, with mean age > 75 years
- Significantly fewer patients who initiated pirfenidone than those who initiated nintedanib had newly diagnosed IPF within 1 year prior to index (50.9% vs. 57.5%; $P < 0.001$)
- Patients treated with pirfenidone had a longer duration of follow-up than patients treated with nintedanib
 - The mean (SD) duration of follow-up for the pirfenidone and nintedanib groups was 204.1 (97.7) and 188.6 (113.6) days, respectively (*t* test, $P < 0.001$)
- After PS weighting, age, sex, region, newly diagnosed status and distance from ILD specialty center were nearly identical between antifibrotic study groups

Table 1. Baseline Demographics and Clinical Characteristics Among Medicare Beneficiaries With IPF Receiving Antifibrotics*

Characteristic	Pirfenidone (n = 2082)	Nintedanib (n = 1464)	*P* Value
Age, mean (SD), years	75.6 (5.5)	76.3 (5.9)	< 0.001†
Female, n (%)	694 (33.3)	593 (40.5)	< 0.001‡
White, n (%)	1985 (95.3)	1397 (95.4)	0.908‡
Region, n (%)			< 0.001‡
Midwest	507 (24.4)	389 (26.6)	–
Northeast	384 (18.4)	207 (14.1)	–
South	798 (38.3)	670 (45.8)	–
West	393 (18.9)	198 (13.5)	–
Newly diagnosed IPF, n (%)§	1059 (50.9)	842 (57.5)	< 0.001‡
CT scan in previous year, n (%)	1536 (73.8)	1133 (77.4)	0.014‡
Distance from patient residential area to an ILD center, mean (SD), miles	100 (175)	98 (138)	0.661†

CT, computed tomography; ILD, interstitial lung disease; IPF, idiopathic pulmonary fibrosis; IPTW, inverse probability of treatment weight.
* Several of these baseline variables were included in the IPTW procedure.
† *t* test.
‡ χ^2 test.
§ ≥ 1 year disease free in pre-index period.

Healthcare Utilization by Patients With IPF in a US Medicare Population

- Of patients who received pirfenidone and nintedanib, there was no significant difference observed in those who had ≥ 1 all-cause inpatient hospitalization and in mean length of stay ($P = 0.066$ and 0.101, respectively) (**Table 2**)
- No statistically significant differences were observed in the percentages of any respiratory-related inpatient hospitalizations in patients receiving pirfenidone or nintedanib ($P = 0.257$)
 - The mean length of stay for any respiratory-related inpatient hospitalization was significantly shorter in patients receiving pirfenidone vs. nintedanib ($P = 0.043$)
- The number of monthly respiratory-related utilizations, including ED visits and hospitalizations, was significantly lower in patients receiving pirfenidone than in those receiving nintedanib ($P = 0.017$ and 0.004, respectively) (**Table 2**)
 - Similar significant results were observed in all-cause ED visits and hospitalizations ($P = 0.010$ and $P < 0.001$, respectively)
- However, there was no significant difference in either number of all-cause or respiratory-related office visits ($P = 0.425$ and 0.727, respectively)

Table 2. Post-Index Healthcare Utilization After Initiation of Antifibrotic Therapy Among Medicare Beneficiaries With IPF, Weighted

Outcome	Pirfenidone (n = 2082)	Nintedanib (n = 1464)	*P* Value
Utilization*			
All cause utilization			
Inpatient hospitalization, n (%)	417.3 (20.0)	330.6 (22.6)	0.066§
Length of stay, mean (SD), days†	8.1 (8.5)	9.4 (12.2)	0.101‖
ED visit, %	18.2	18.4	0.927§
Respiratory-related utilization‡			
Inpatient hospitalization, n (%)	231.7 (11.1)	181.0 (12.4)	0.257§
Length of stay, mean (SD), days†	7.2 (7.6)	9.3 (23.3)	0.043‖
ED visit, (%)	11.2	12.2	0.353§
Utilization per patient month			
All-cause, mean (SD)			
Office visits	1.7 (1.5)	1.7 (1.4)	0.425‖
ED visits	0.055 (0.33)	0.060 (0.22)	0.010¶
Inpatient hospitalizations	0.080 (0.28)	0.106 (0.38)	< 0.001¶
Respiratory related, mean (SD)‡			
Office visits	0.73 (1.03)	0.72 (0.77)	0.727‖
ED visits	0.028 (0.11)	0.038 (0.19)	0.017¶
Inpatient hospitalizations	0.041 (0.20)	0.052 (0.25)	0.004¶

ED, emergency department; IPF, idiopathic pulmonary fibrosis; IPTW, inverse probability of treatment weight.
* Non-integer values are due to weighting.
† Length of stay reported among a subset of beneficiaries with a hospitalization.
‡ Inpatient claims with primary diagnosis of respiratory disease (ICD-9-CM: 460.xx–519.xx; ICD-10-CM: J00.xx–J99.xx) or outpatient claims with any diagnosis of respiratory disease.
§ χ^2 test, weighted by stabilized IPTW.
‖ *t* test, weighted by stabilized IPTW.
¶ Wald χ^2 test based on negative binomial model, weighted by stabilized IPTW.

Risk and Rate of Hospitalization in Patients With IPF in a US Medicare Population

- Both the risk and rate of all-cause and respiratory-related hospitalizations were statistically significantly lower in patients receiving pirfenidone than in those receiving nintedanib (**Table 3**)

Table 3. Comparative Risk and Rate of Hospitalization After Initiation of Antifibrotic Therapy Among Medicare Beneficiaries, Weighted*

Outcome (Pirfenidone vs. Nintedanib)	Risk of Hospitalization		Rate of Hospitalization†	
	HR (95% CI)	*P* Value‡	IRR (95% CI)	*P* Value§
All cause	0.79 (0.68, 0.91)	0.001	0.69 (0.59, 0.82)	< 0.001
Respiratory related	0.80 (0.65, 0.97)	0.022	0.71 (0.57, 0.90)	0.004

HR, hazard ratio; IPF, idiopathic pulmonary fibrosis; IPTW, inverse probability of treatment weight; IRR, incidence rate ratio.
* Weighted by stabilized IPTW.
† Per patient per year.
‡ Cox proportional hazards regression.
§ Negative binomial regression.

- The risk of respiratory-related hospitalization was significantly lower in patients receiving pirfenidone vs. nintedanib (weighted log rank $P = 0.016$) (**Figure 1**)

Figure 1. Risk of Respiratory-Related Hospitalization After Initiation of Antifibrotic Therapy Among Medicare Beneficiaries With IPF, Weighted*

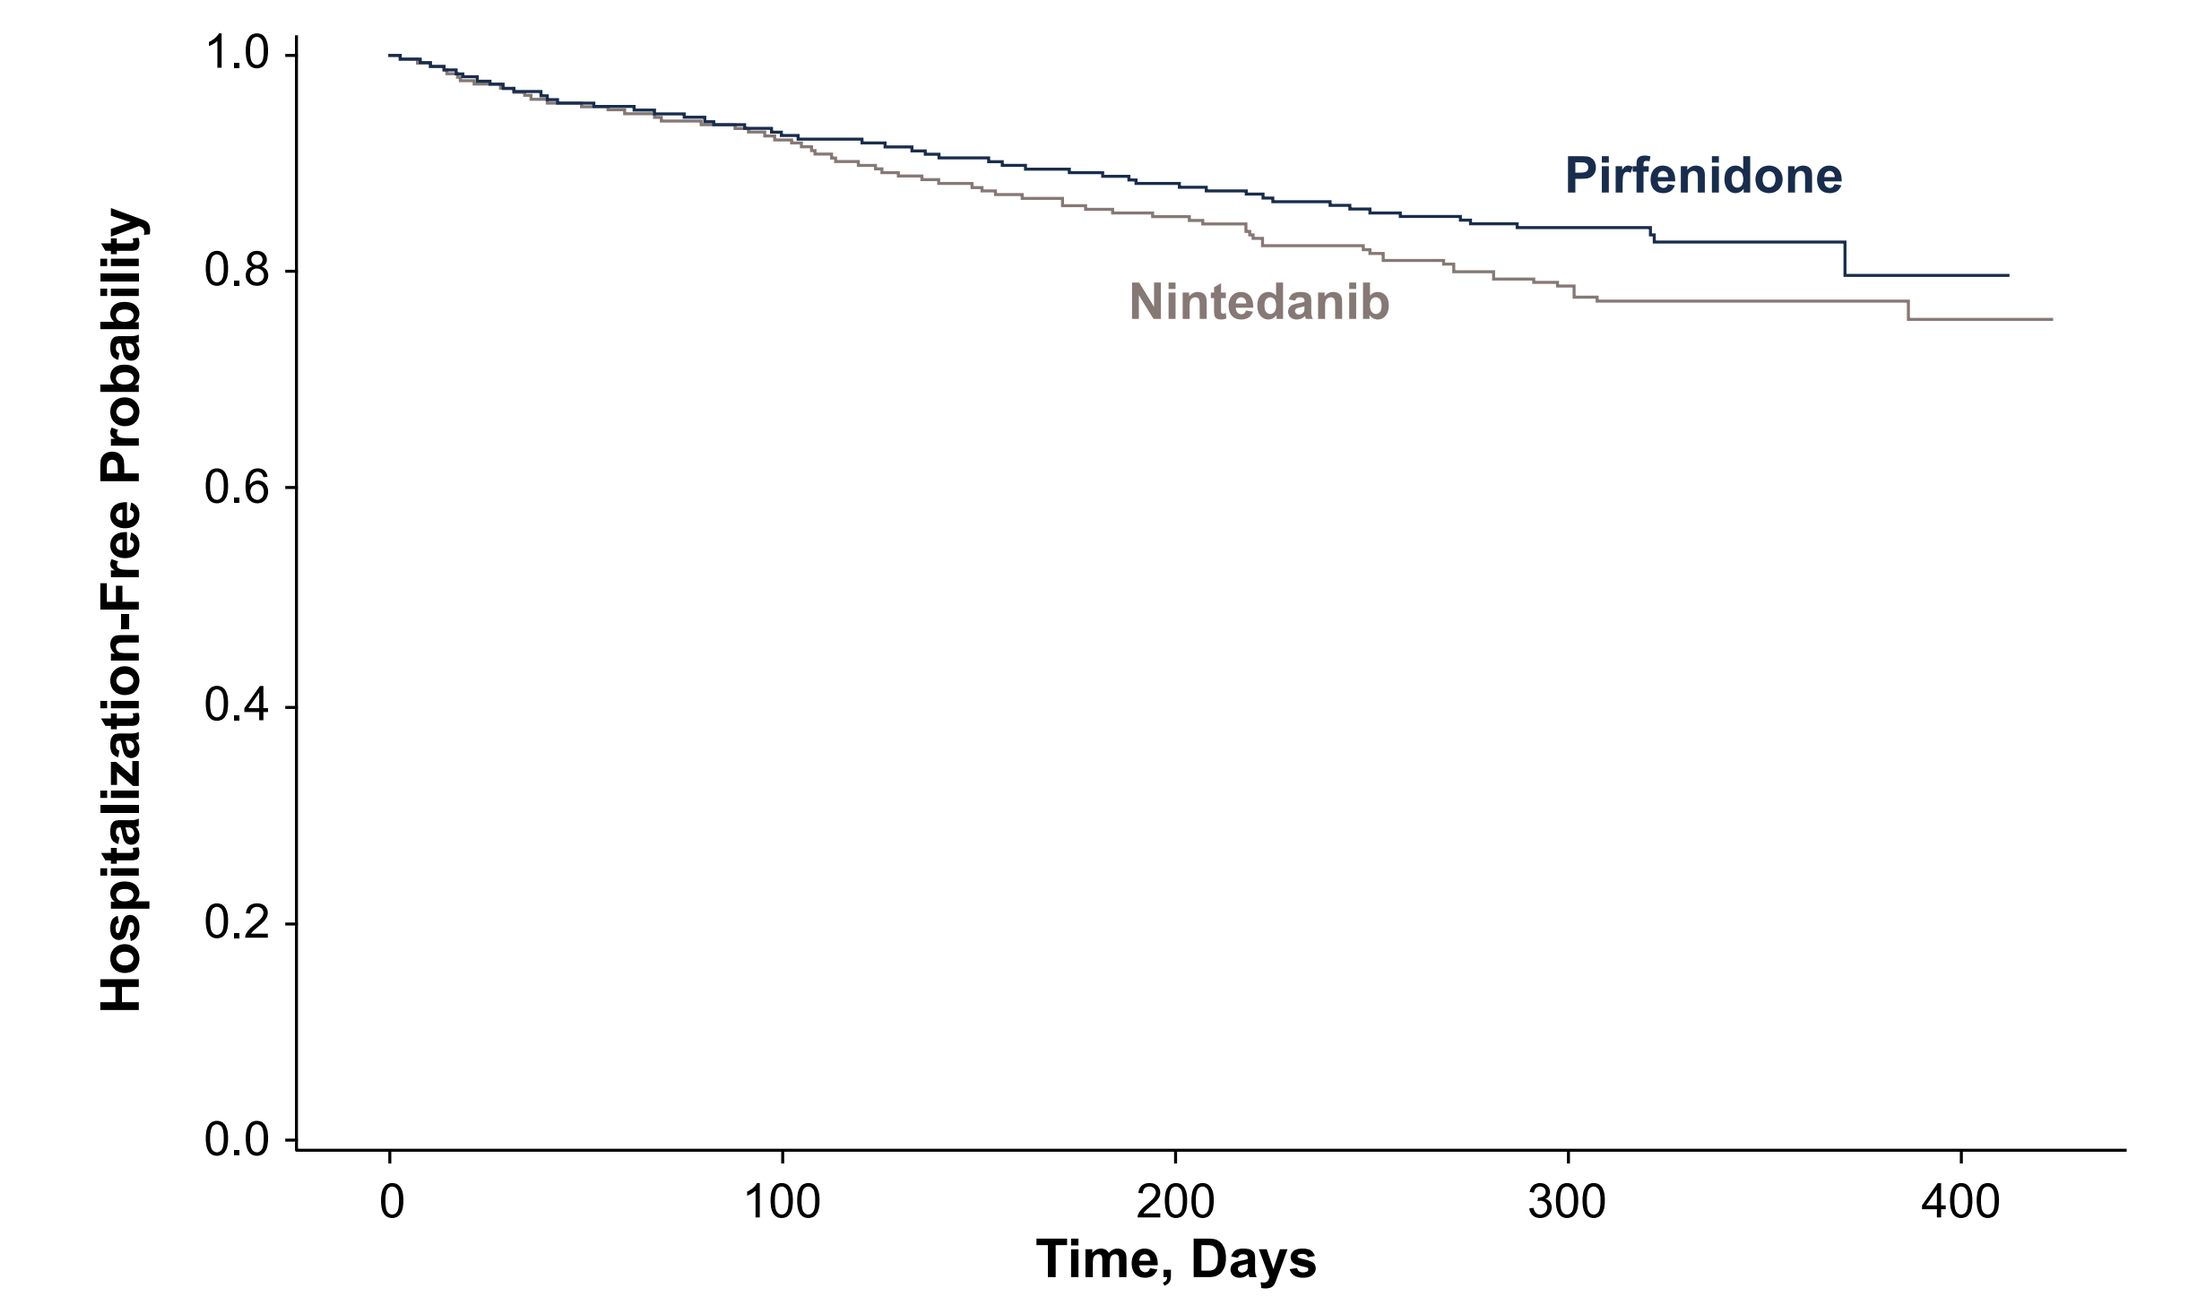

Hospitalization-Free Probability
1.0
0.8
0.6
0.4
0.2
0.0
0
100
200
300
400
Time, Days
Pirfenidone
Nintedanib

No. at Risk	Day 90	Day 180	Day 365
Pirfenidone	1482	1003	26
Nintedanib	920	483	54

IPF, idiopathic pulmonary fibrosis; IPTW, inverse probability of treatment weight.
* Weighted by stabilized IPTW.

Healthcare Costs of Patients With IPF in a US Medicare Population

- Mean all-cause and respiratory-related inpatient costs were statistically significantly lower in patients receiving pirfenidone than in those receiving nintedanib (*t* test weighted $P = 0.026$ and 0.041, respectively)
- Group differences for other categories (e.g., outpatient, pharmacy) were not statistically significant
- For both treatment groups, pharmacy services (treatment costs) accounted for the largest portion of total costs per month, followed by inpatient services and then outpatient services (**Figure 2**)
 - This pattern was similar for respiratory-related costs
- There was no statistically significant difference in antifibrotic medication costs between the 2 groups ($P = 0.171$)

Figure 2. Healthcare Costs Per Patient Month After Initiation of Antifibrotic Therapy Among Medicare Beneficiaries With IPF, Weighted

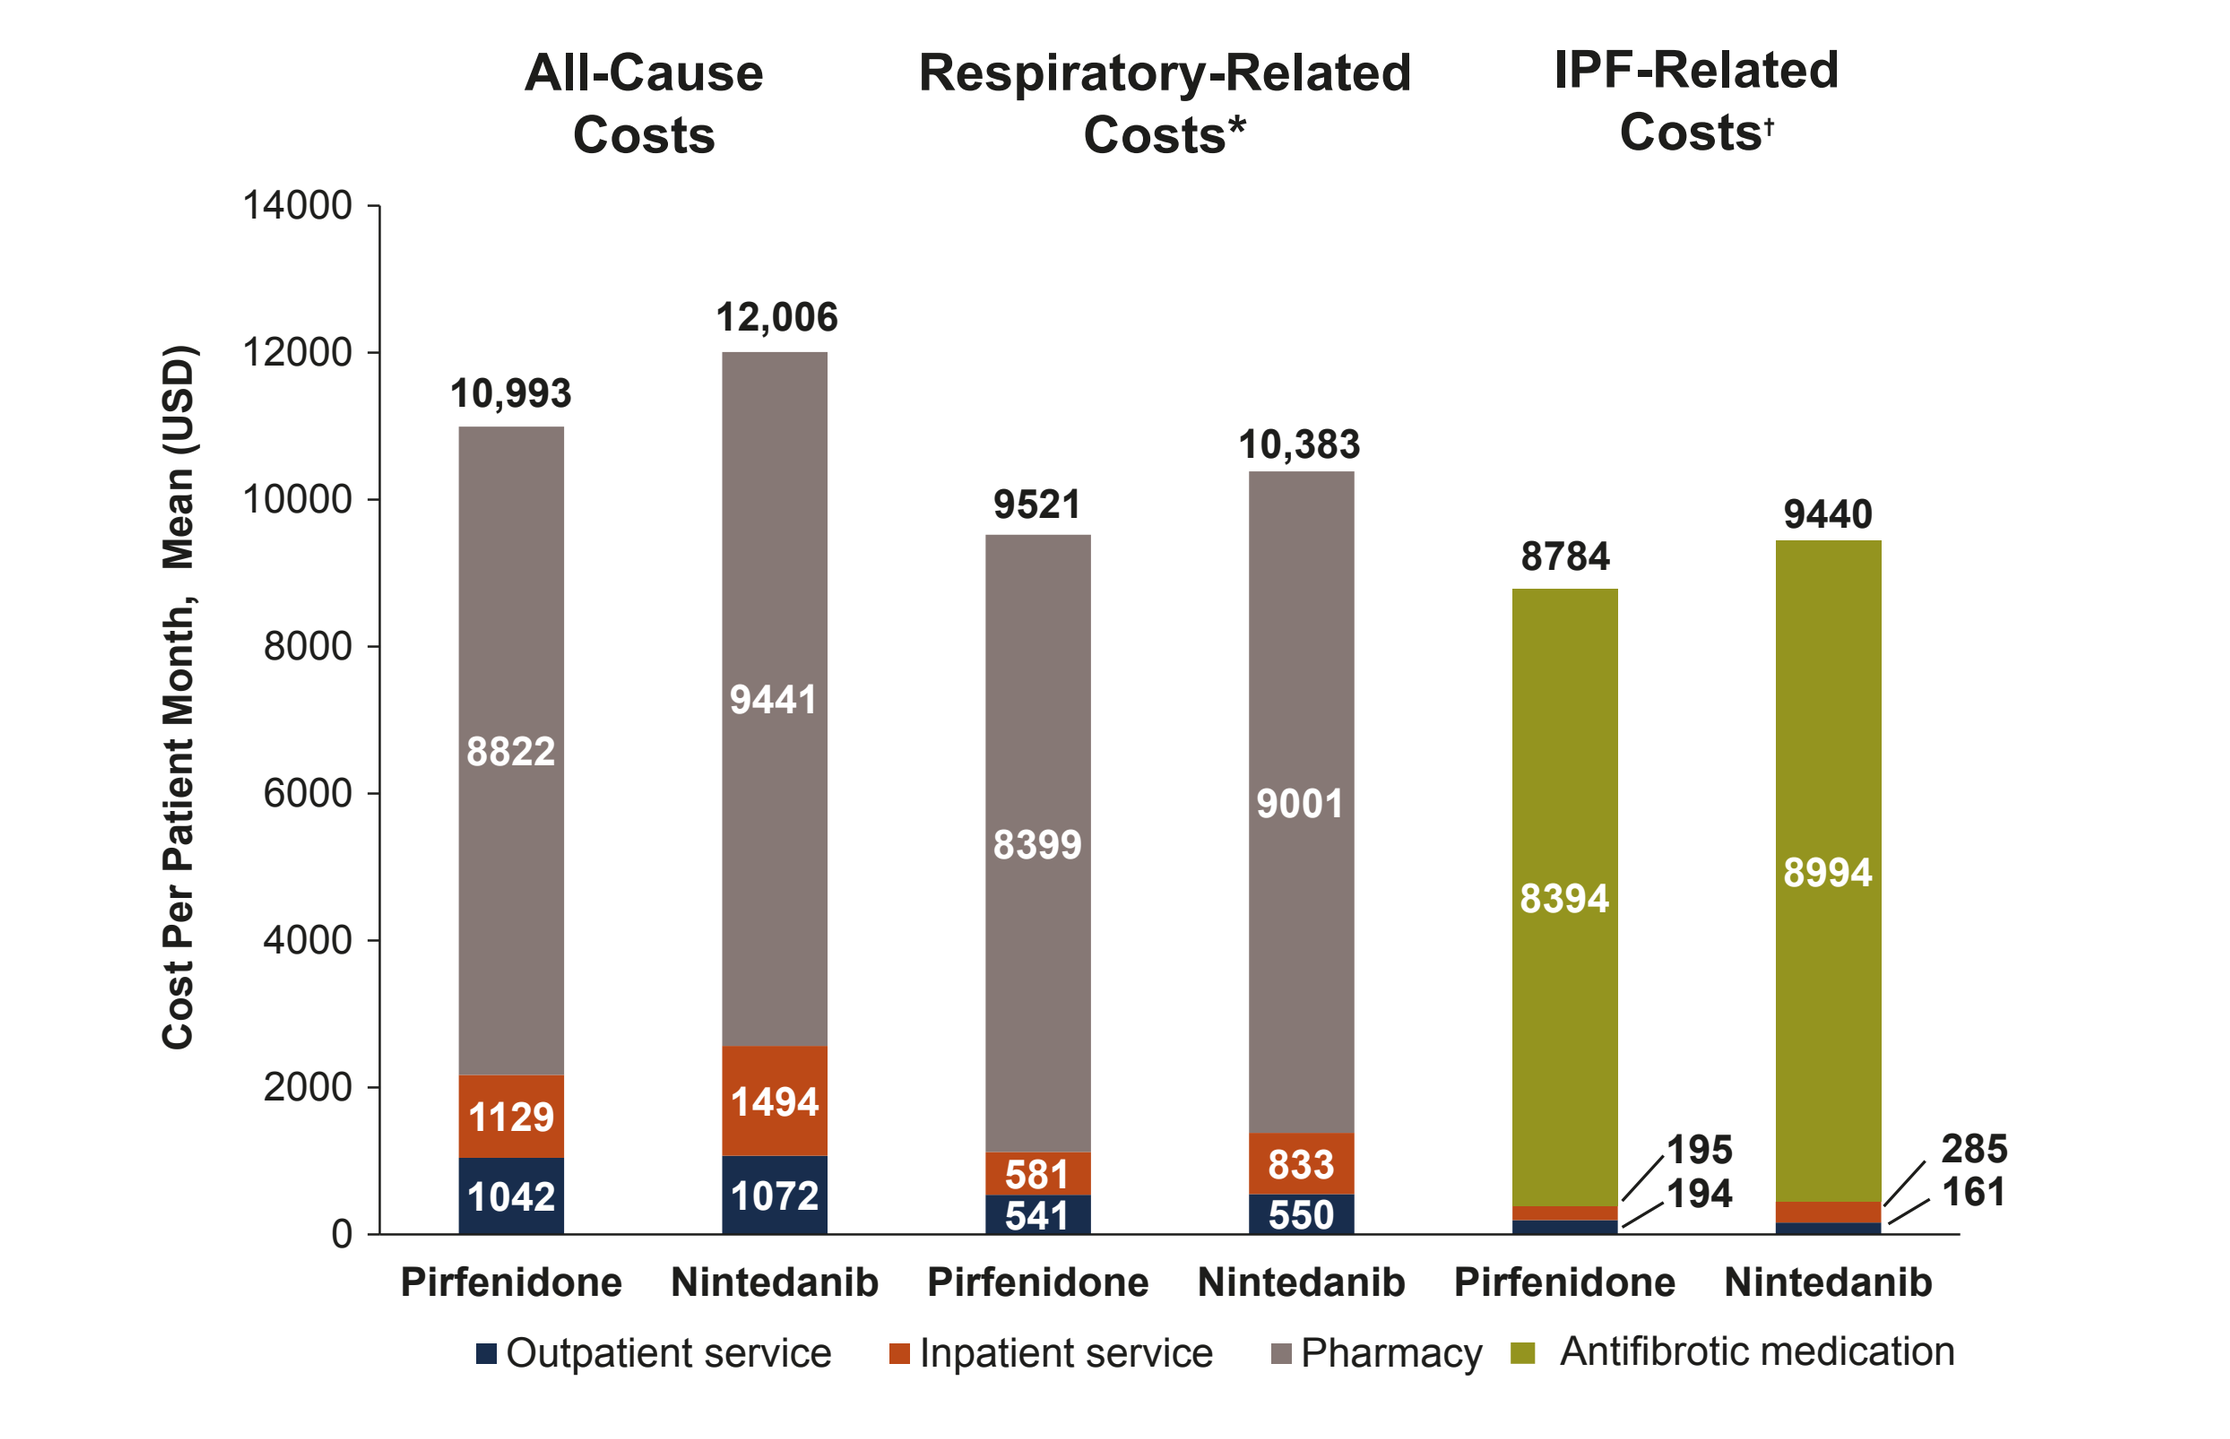

All-Cause Costs
Respiratory-Related Costs*
IPF-Related Costs†
Cost Per Patient Month, Mean (USD)
14000
12000
10000
8000
6000
4000
2000
0
10,993
12,006
9521
10,383
8784
9440
8822
9441
8399
9001
8394
8994
1129
1494
581
833
1042
1072
541
550
195
194
285
161
Pirfenidone
Nintedanib
Pirfenidone
Nintedanib
Pirfenidone
Nintedanib
Outpatient service
Inpatient service
Pharmacy
Antifibrotic medication

IPF, idiopathic pulmonary fibrosis.
* Inpatient claims with primary diagnosis of respiratory disease (ICD-9-CM: 460.xx–519.xx; ICD-10-CM: J00.xx–J99.xx), outpatient claims with any diagnosis of respiratory disease or pharmacy claims for an antifibrotic medication, inhaled corticosteroids, azathioprine, n-acetylcysteine and mycophenolate mofetil, as well as antibiotics filled ± 15 days of a claim with a selected pneumonia diagnosis (that can be treated with antibiotics) as a primary diagnosis in inpatient services or any diagnosis in outpatient services.
‡ Inpatient claims with primary diagnosis of IPF (ICD-9-CM: 516.3, 516.30 and 516.31; ICD-10-CM: J84.111 and J84.112), outpatient claims with any diagnosis of IPF or pharmacy claims for antifibrotic medication.

LIMITATIONS

- The short time frame for measuring outcomes contributed to a high rate of administrative censoring
- Despite use of robust weighting procedures to balance observed characteristics across treatment groups, unobserved differences could not be fully controlled

CONCLUSIONS AND IMPLICATIONS

- Significantly lower respiratory-related utilization and costs were observed in patients with IPF receiving pirfenidone than in those receiving nintedanib in a US Medicare population
- Patients receiving pirfenidone may have overall lower all-cause inpatient costs and fewer respiratory-related hospital days than those receiving nintedanib
- After adjusting for baseline differences and treatment adherence, patients receiving pirfenidone had a significantly lower risk of all-cause and respiratory-related hospitalizations than those receiving nintedanib

REFERENCES

1. Raghu G, et al. *Am J Respir Crit Care Med.* 2015;192:e3–e19.
2. Ley B, et al. *Am J Respir Crit Care Med*. 2011;183:431–440.
3. Esbriet (pirfenidone) [package insert]. South San Francisco, CA: Genentech, Inc.; 2017.
4. Ofev (nintedanib) [package insert]. Ridgefield, CT: Boehringer Ingelheim Pharmaceuticals; 2018.
5. Noble PW, et al. *Lancet.* 2011;377:1760–1769.
6. King TE Jr, et al. *N Engl J Med.* 2014;370:2083–2092.
7. Richeldi L, et al. *N Eng J Med.* 2014;370:2071–2082.
8. Durheim MT, et al. *Lancet Respir Med.* 2015;3:388–396.
9. Raimundo K, et al. *BMC Pulm Med*. 2016;16:2.
10. du Bois, RM, et al. *Am J Respir Crit Care Med.* 2011;184:459–466.
11. Collard HR, et al. *Ann Am Thorac Soc.* 2015;12:981–987.

bitly.com/reddy2

Presented at the American College of Chest Physicians (CHEST) Annual Meeting 2019; October 19–23, 2019; New Orleans, LA.

This poster was sponsored by Genentech, Inc. and F. Hoffmann-La Roche, Ltd.
Third-party writing assistance was furnished by Health Interactions, Inc.