Biochemical Control during Long Term Follow up of 230 Patients with Cushing's Disease: A Multi-Center Retrospective Study

Eliza B. Geer[1*], Ismat Shafiq[2], Murray B. Gordon[3*], Vivien Bonert[4], Alejandro Ayala[5], Ronald S. Swerdloff[6], Laurence Katznelson[7*], Yelena Lalazar[1], Ekaterina Manuylova[3], Karen J. Pulaski-Liebert[8], John D. Carmichael[9], Zeina Hannoush[5], Vijaya Surampudi[6], Michael S. Broder[10], Dasha Cherepanov[10], Marianne Eagan[10], Jackie Lee[10], William H. Ludlam[11], Maureen P. Neary[12], Beverly MK Biller[8*]

[1] Icahn School of Medicine at Mount Sinai, New York, NY; [2] University of Rochester School of Medicine and Dentistry, Rochester, NY; [3] Allegheny Neuroendocrinology Center, Allegheny General Hospital, Pittsburgh, PA; [4] Cedars-Sinai Medical Center, Los Angeles, CA; [5] University of Miami and Jackson Memorial Hospital, Miami, FL; [6] Harbor-UCLA Medical Center, Los Angeles, CA; [7] Stanford University School of Medicine, Stanford, CA; [8] Massachusetts General Hospital, Boston, MA; [9] University of Southern California Pituitary Center, Los Angeles, CA; [10] Partnership for Health Analytic Research, LLC, Beverly Hills, CA; [11] Chiasma, Inc., Newton, MA, previously of Novartis Pharmaceuticals Corporation, East Hanover, NJ; [12] Novartis Pharmaceuticals Corporation, East Hanover, NJ

* Potential conflict of interest may exist for several co-authors. Please refer to the abstract

SAT 550

BACKGROUND

- Cushing's disease (CD) results from excessive exposure to glucocorticoids caused by an adrenocorticotropic hormone-secreting pituitary tumor.
- Primary treatment usually involves pituitary surgery to remove the tumor.
- For recurrent or persistent disease, repeat operation, radiotherapy, medical treatment, or bilateral adrenalectomy (BLA) may be used, but little is known about outcomes of these treatments in multi-center routine clinical practice.

OBJECTIVE

- To describe treatment outcomes in a diverse, multi-center cohort of patients receiving clinical care for CD.

METHODS

Study Design

- Retrospective data were collected from patients' medical records at 8 US pituitary/endocrine centers, including major referral centers and regional/local centers, selected based on adequate number of CD patients treated, geographic location, and diversity of patient populations, across 4 US regions.
- The study was approved by the Institutional Review Boards at each site.

Patient Selection

- Patients with initial CD diagnosis or recurrence during the past 20 years and who were ≥18 years old at the time of diagnosis were included in the study.

Data Collection

- Each site identified eligible patients, and trained site abstractors entered data via a secure eCRF.
- Data were collected from the time of presentation (i.e., the date patient first sought care for symptoms, signs, or comorbidities that were later ascribed to CD) for each patient through end of follow-up (i.e., by study end, 5/2015).

Measures

- Patient demographics and baseline comorbidities were captured from available data recorded during the pretreatment period (on or before the first CD therapy).
- Pituitary adenoma details were recorded based on pituitary MRI before pituitary surgery.
- The dates and types of CD treatments were captured. The first treatment given was classified as primary therapy.
- Biochemical control was defined: 1) following pituitary surgery, any value of 5-9AM serum cortisol <5 mcg/dl, or ≤1.8 mcg/dl after 1 mg dex (or, in patients with no serum cortisol or dex suppression tests, a 24 hour UFC<lower limit of normal); 2) on pharmacotherapy or post radiotherapy, any value of 24 hour UFC ≤upper limit of normal or 11PM-1AM salivary cortisol in Nl range; 3) all patients following BLA.
- Biochemical control was assessed with cortisol levels after initial surgery and at end of follow-up. If no values were available, control was considered, "indeterminate."

Statistical Analysis

- Patient characteristics were reported in the full sample and by final biochemical control.
- Comorbidities were reported in the pretreatment subgroup (patients with information in the medical records on or before the first CD therapy).
- All statistical analyses were done in SAS® version 9.4 (SAS Institute, Cary, NC).

RESULTS

- There were 230 CD patients, median age 39 years at diagnosis; 67% were Caucasian; 6% were African American; 1% were Asian, and 26% were other/unknown; 79% were female.
- Median duration of follow-up was 1.9 years, ranging up to 27.5 years. Median time from presentation to diagnosis of CD was 0.8 years, ranging from 1 day to 23.6 years.

Figure 1. Time from Presentation to Diagnosis

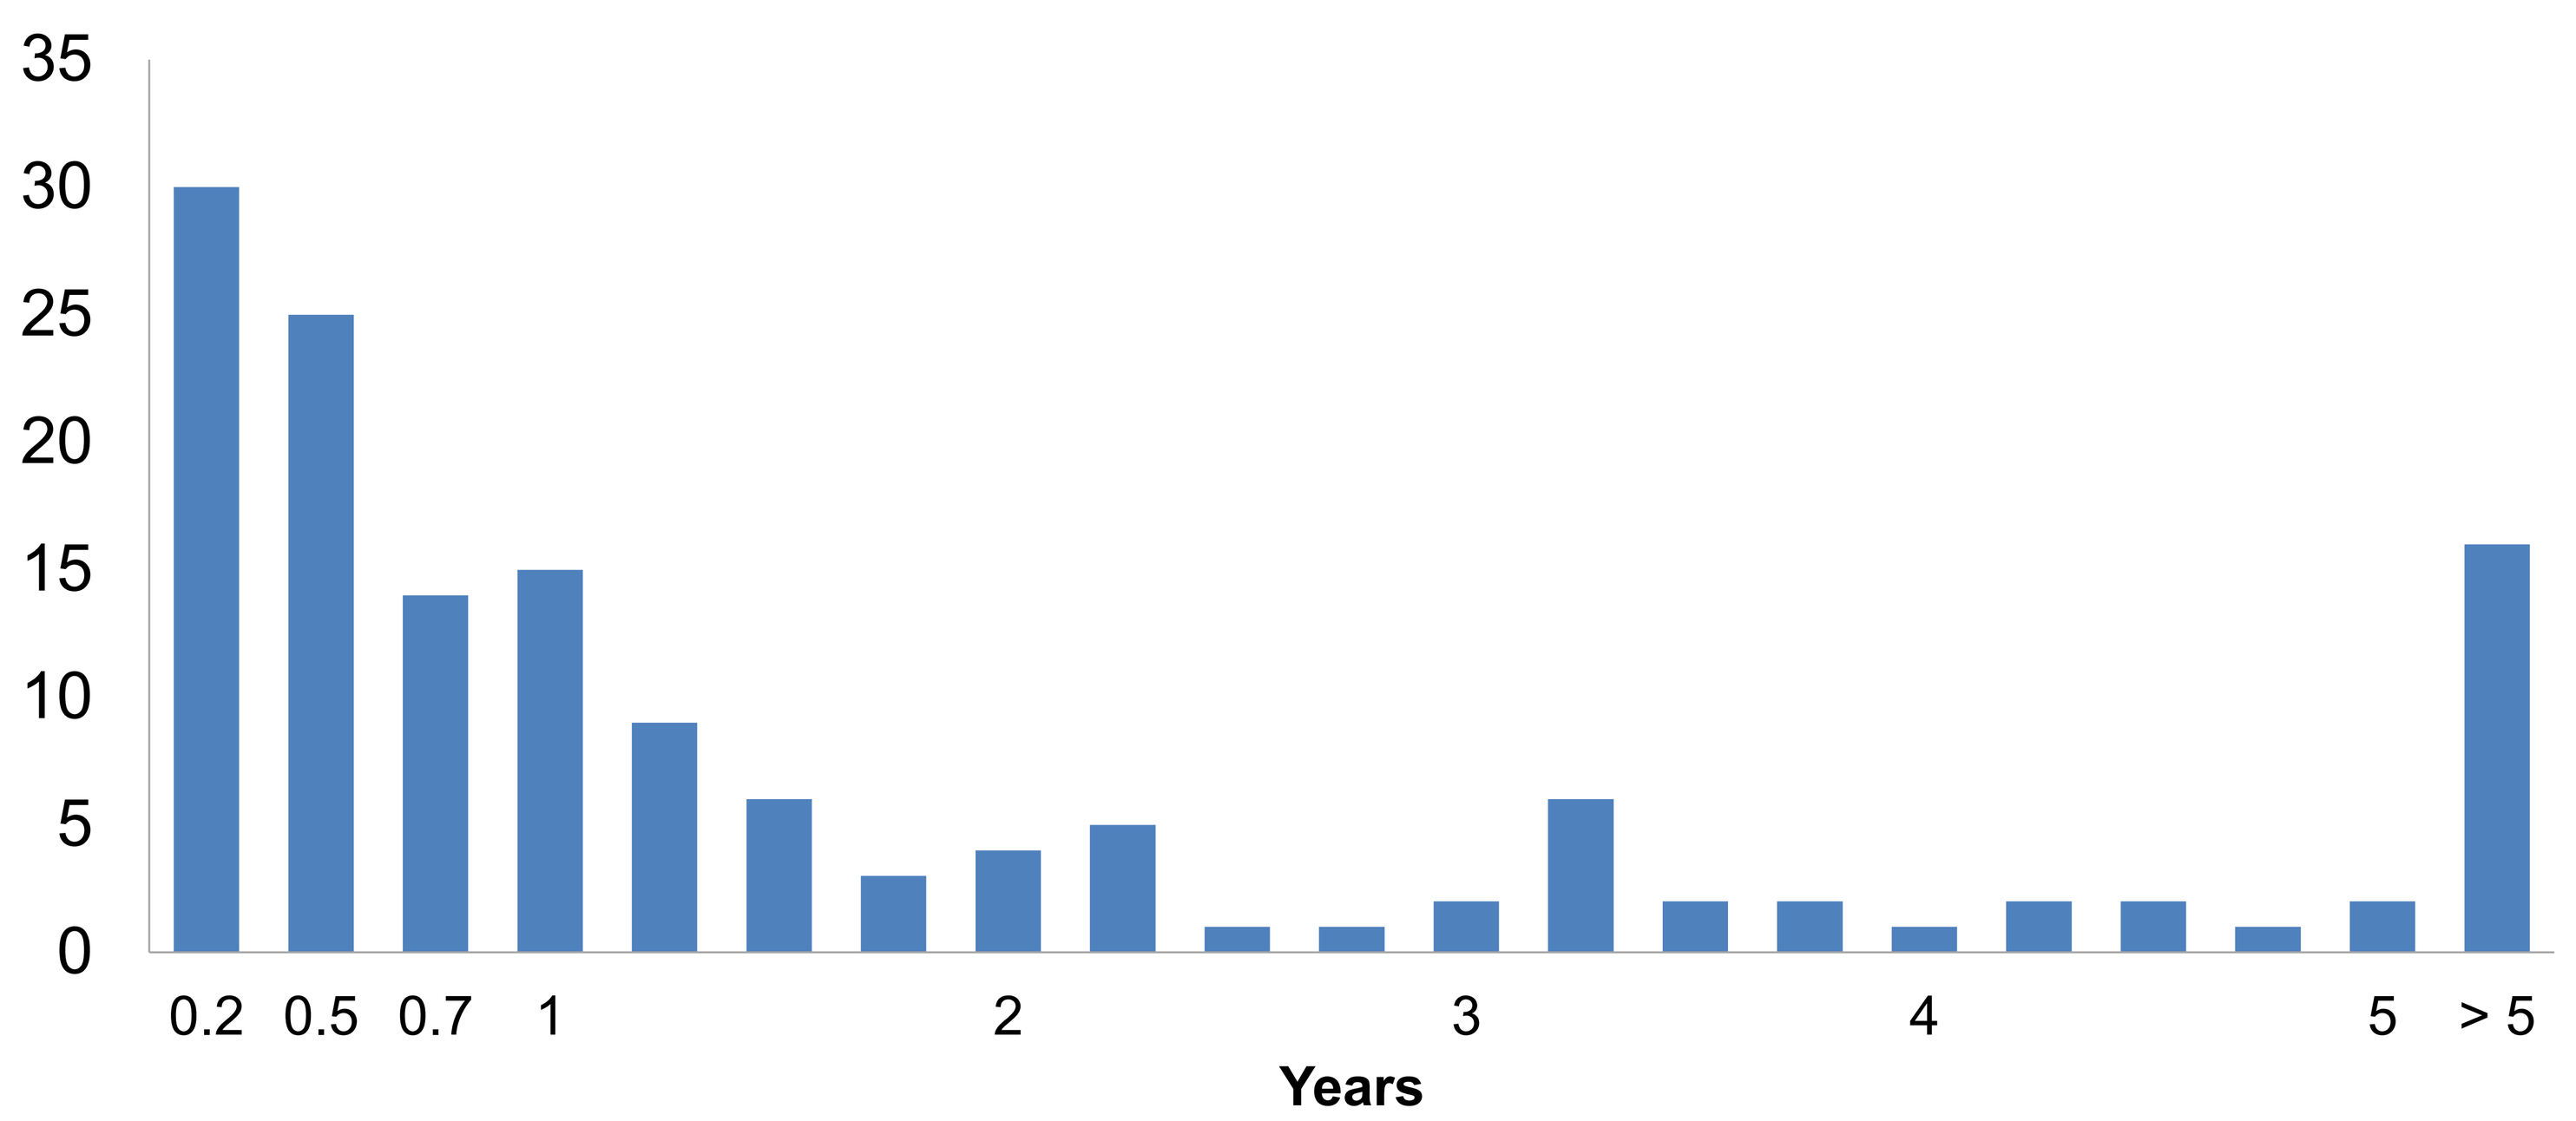
Number of Patients
0
5
10
15
20
25
30
35
0.2
0.5
0.7
1
2
3
4
5
> 5
Years

- There were many comorbidities reported before the primary treatment as shown in Figure 2.

Figure 2. Top 10 Most Prevalent Comorbidities

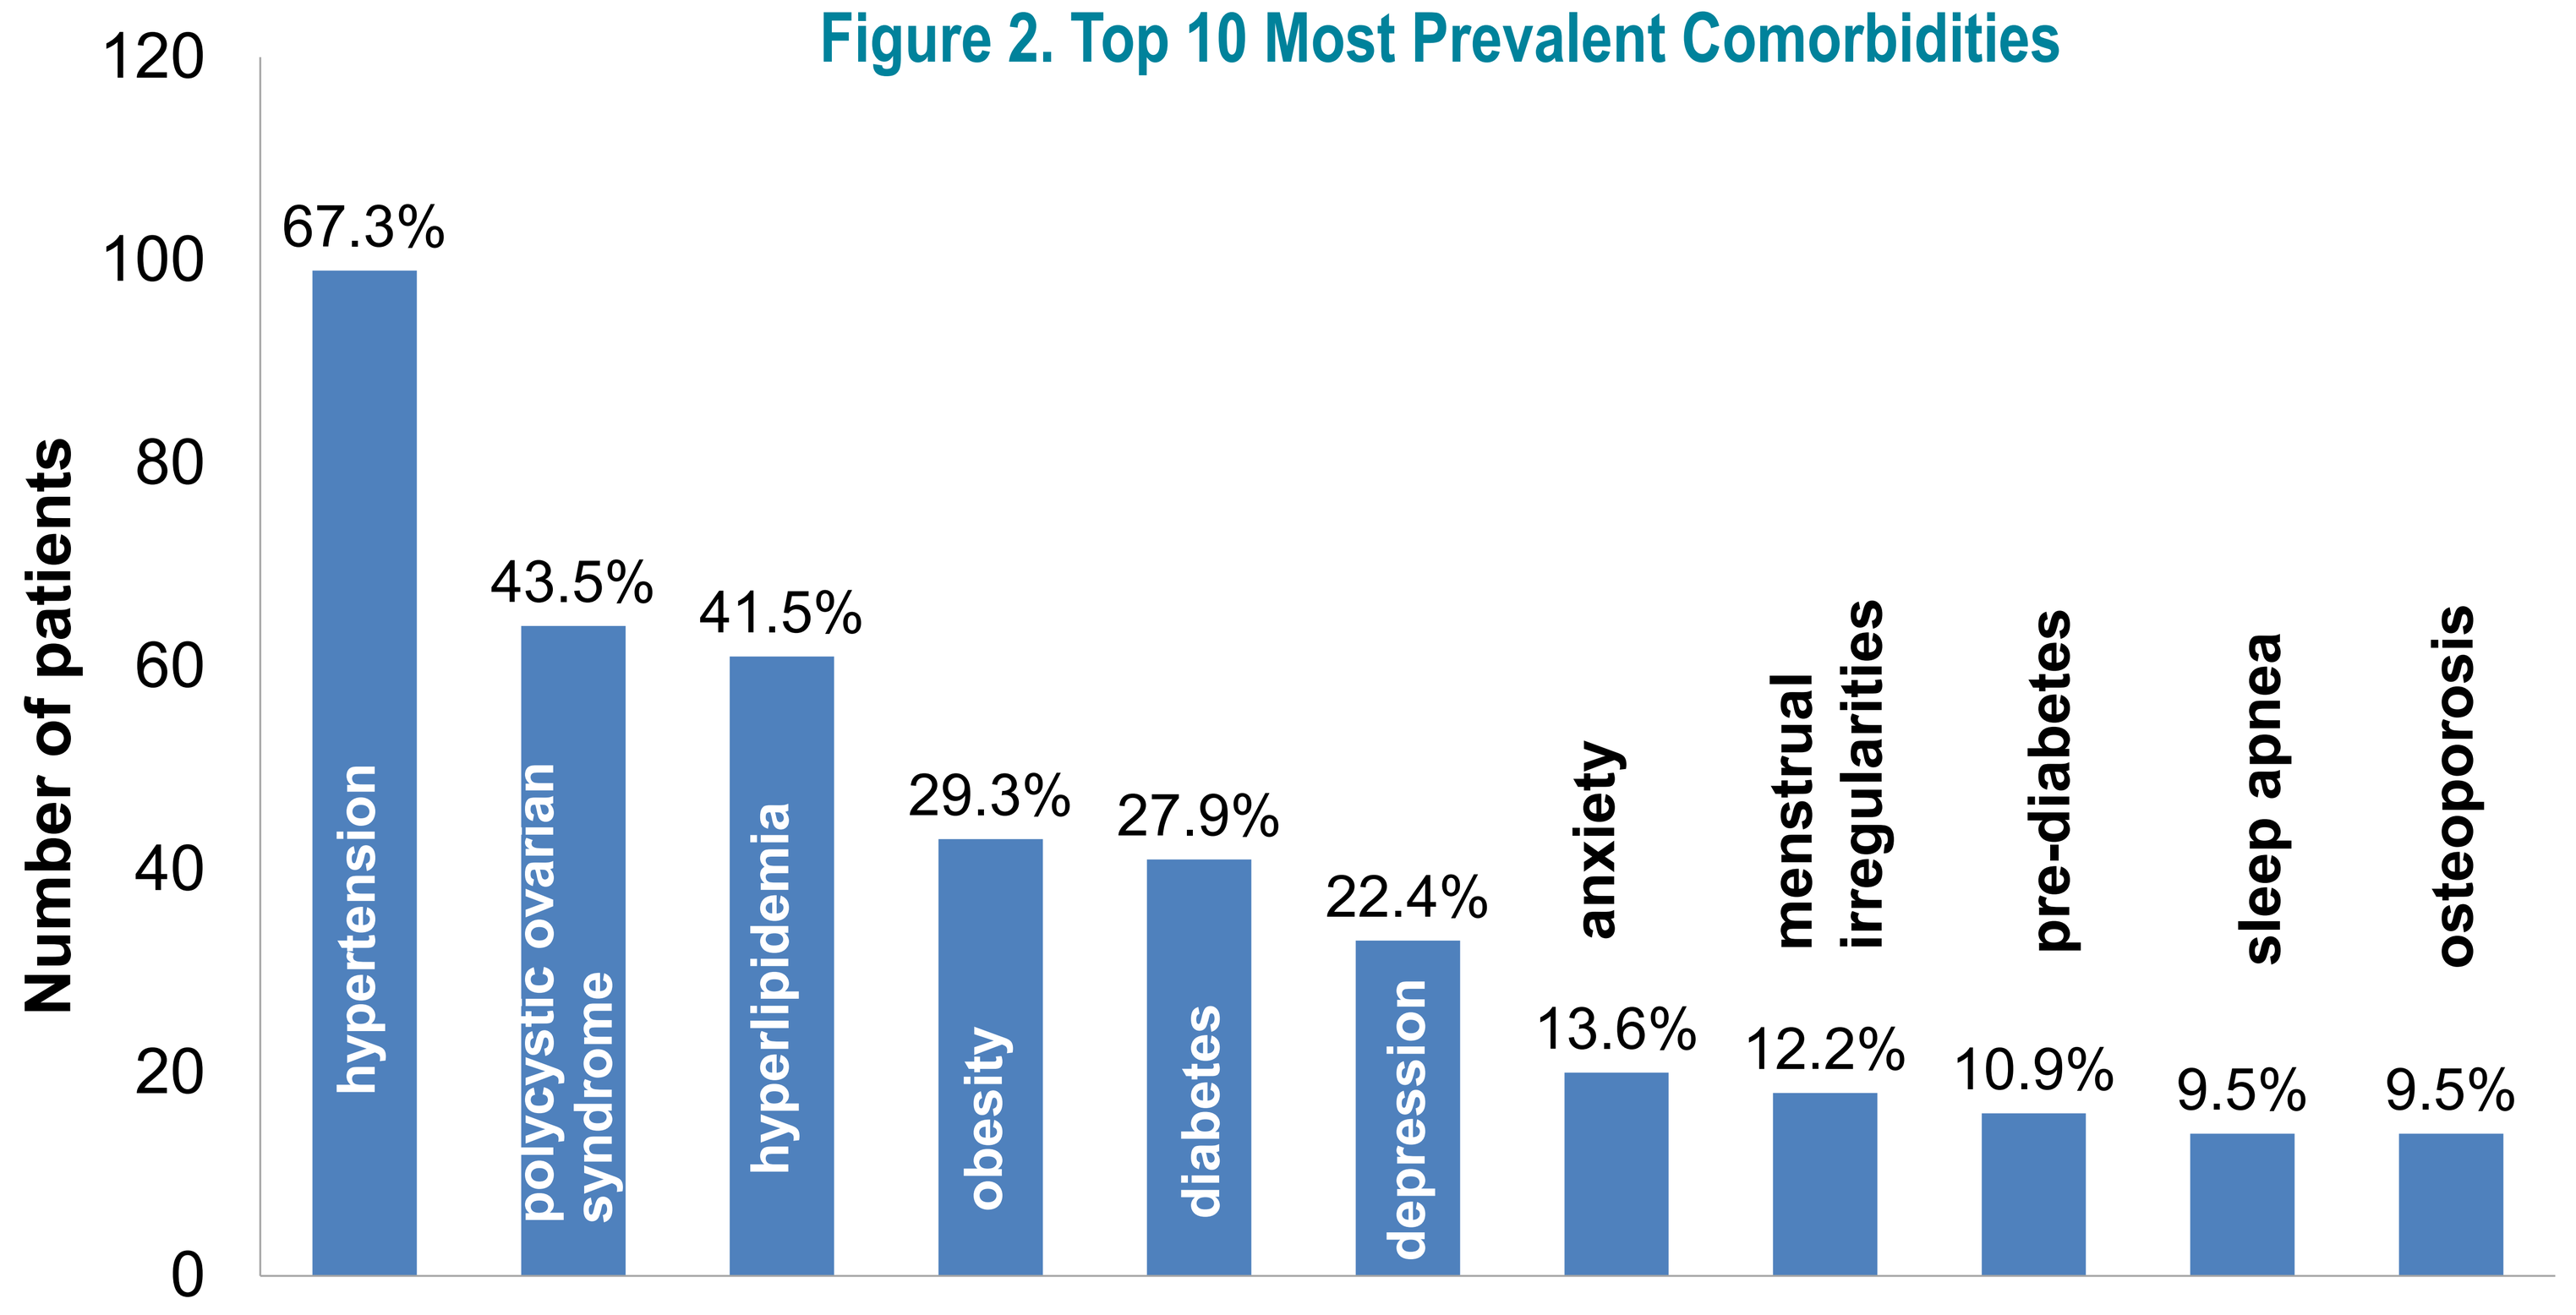
Number of patients
0
20
40
60
80
100
120
67.3%
hypertension
43.5%
polycystic ovarian syndrome
41.5%
hyperlipidemia
29.3%
obesity
27.9%
diabetes
22.4%
depression
13.6%
anxiety
12.2%
menstrual irregularities
10.9%
pre-diabetes
9.5%
sleep apnea
9.5%
osteoporosis

- First-line treatment was pituitary surgery in 95.7% (n=220)
- Control after initial surgery was achieved in 41.4% (91/220), not achieved in 50.0% (110), and indeterminate in 8.6% (19).
- 27.3% had additional pituitary surgery, 25.2% had pharmacotherapy, 12.6% had radiotherapy, and 7.0% BLA over the period of observation.
- Among 58 patients that received pharmacotherapy, ketoconazole was commonly used (69%), followed by cabergoline (27.6%), pasireotide (12.1%), metyrapone (8.6%), mifepristone (3.4%), rosiglitazone (3.4%), bromocriptine (1.7%), and pioglitazone (1.7%).

RESULTS (CONT.)

- At the end of observation (which varied across patients from 1 to 27.5 years, median 1.9 years), control was achieved in 49.1% (n=110), not achieved in 29.9% (67), and indeterminate in 21.0% (47) (6 pts. had no treatment during the study observation period).
- Many variables were examined to see whether there were differences between the groups with final control versus lack of final control versus indeterminate final status and all were non-significant as shown in Table 1.

Table 1. Patient Characteristics by Final Biochemical Control

	Final Biochemical Control			
	Control n=110 (49.1%)	**No Control** n=67 (29.9%)	**Indeterminate** n=47 (21.0%)	**All** N=224
Female, n (%)	86 (78.2)	53 (79.1)	39 (83.0)	178 (79.5)
Age at CD diagnosis, years, mean (SD)	39.7 (11.9)	39.1 (13.6)	42.8 (14.0)	40.2 (12.9)
[median] (range)	[40] (19- 78)	[36] (18-71)	[41] (18-73)	[39] (18- 78)
Macroadenoma, n (%)	37 (33.6)	16 (23.9)	11 (23.4)	64 (28.6)
Microadenoma,[a] n (%)	38 (34.5)	24 (35.8)	14 (29.8)	76 (33.9)
No adenoma size,[b] n (%)	35 (31.8)	27 (40.3)	22 (46.8)	84 (37.5)
Time from presentation to CD diagnosis, years n, median (range)	70, 0.9 (0.0-17.3)	44, 0.7 (0.1-11.8)	30, 0.8 (0.0-23.6)	144, 0.8 (0.0-23.6)
Time from CD diagnosis to first CD treatment, years, n, median (range)	99, 0.1 (0.0-1.2)	57, 0.1 (0.0-2.0)	42, 0.1 (0.0-1.8)	198, 0.1 (0.0-2.0)
Initial intervention to end of observation, years, median (range)	3.7 (0.0-19.3)	2.3 (0.0-21.1)	2.3 (0.0-27.3)	2.5 (0.0-27.3)
Last intervention to end of observation, years median (range)	1.8 (0.0 -15.3)	1.1 (0.0-16.1)	0.1 (0.0-14.6)	1.2 (0.0-16.1)

[a] Microadenoma includes 9 patients with indeterminate MRI result and 35 with no visible adenoma on MRI.
[b] No MRI result was available.
All comparisons not statistically significant.

STRENGTHS

- Large sample size for a very rare condition.
- These results capture biochemical control and long-term follow-up of CD patients in routine clinical practice across multiple study sites representing a variety of geographic locations and institution types.
- Inclusion of large proportion of patients with macroadenoma allowed us to examine outcomes in CD patients who may have more aggressive tumors who often need adjuvant therapy.

LIMITATIONS

- Retrospective data collection.
- These results are based on a convenience (i.e., not random) sample of patients with different lengths of follow-up, including patients with long periods of follow-up, so that changes in clinical care standards may have affected CD management over time.
- Because patients were recruited from specific regional and tertiary care centers, subjects may not be representative of the general CD population.
- The study sample may have included clinical trial patients. Trial data were not collected, if these data were not part of study patients' medical records.
- Study centers have different practice and charting styles; some information could not be obtained from the available records.
- Patients presented at the study sites at different points in their treatment timeline, so that not all were initially diagnosed or treated at the study sites.

SUMMARY AND CONCLUSION

- **In this large, multi-center study with a median follow-up of 2 years, only 41% of patients achieved remission with initial pituitary surgery.**
- **Many CD patients therefore required additional treatment(s), including more pituitary surgery in 27%. More than 25% received pharmacotherapy, about 13% received radiation, and 7% underwent BLA.**
- **Despite these treatments, at the end of follow-up, biochemical control was achieved only in half of the patients. Nearly one-third of patients had still not achieved control, while 21% of patients had indeterminate status.**
- **This study has a much higher proportion of patients with macroadenoma than expected in the general CD population, which may reflect that the study sites are referral care centers seeing more challenging cases.**
- **These multi-center data demonstrate that long-term control is not achieved in many patients with CD, indicating the need for improvement in the care and long-term biochemical follow-up of this disorder.**

URL:http://novartis.medicalcongressposters.com/Default.aspx?doc=f5718
Text Code: **Qf5718** To: 8NOVA (86682) *US Only;* +18324604729 *North, Central and South Americas; Caribbean; China;* +447860024038 *UK, Europe & Russia;* +46737494608 *Sweden, Europe.*
Standard data or message rates may apply.

ENDO 2016, April 1-4, 2016, Boston, MA, USA.

Analysis was conducted by Partnership for Health Analytic Research, LLC.

This study was sponsored by Novartis Pharmaceuticals Corporation.