REDUCING HEALTHCARE COSTS IN ANTIDEPRESSANT NON-RESPONDERS: DOES TIMING OF ANTIPSYCHOTIC AUGMENTATION MATTER?

Mallik Greene, BPharm, PhD, DBA[1]; Jessie Tingjian Yan, PhD[2]; Eunice Chang, PhD[2]; Ann Hartry, PhD[3]; Irina Yermilov, MD, MPH, MS[2]

[1] Otsuka Pharmaceutical Development & Commercialization, Inc., Princeton, NJ, USA;
[2] Partnership for Health Analytic Research, LLC, Beverly Hills, CA, USA; [3] Lundbeck, Deerfield, IL, USA

Objective

- Augmentation of conventional antidepressants (AD) with antipsychotics (AP) is one of the main treatment strategies recommended for patients with major depressive disorder (MDD) who inadequately respond to first-line AD.[1]
- There is little evidence available regarding the most effective timing of this augmentation.
- The objective of this study was to understand the association between timing of augmentation of AD with oral AP and overall healthcare costs in patients with MDD inadequately responding to first-line AD (inadequate responders).

Methods

- Study design and data source
 - Retrospective cohort study using the Truven Health Analytics MarketScan® Medicaid, Commercial, and Medicare Supplemental databases covering the study period [7/1/2009-12/31/2016 for Medicaid (MC), 7/1/2009-9/30/2016 for Commercial or Medicare Supplemental (C/SUP)]
- Patient identification
 - Adult (≥18 years old) inadequate responders were identified if they had 1 of the following qualifying events indicating incomplete response to their initial AD treatment in the ID period [10/1/2009-12/31/2014 (MC) or 10/1/2009-9/30/2014 (C/SUP)]:
 - Switch to or addition of another AD
 - Initiation of psychotherapy
 - Psychiatric hospitalization or psychiatric emergency department (ED) visit
 - Inclusion criteria:
 - ≥1 inpatient or ≥2 outpatient claims for MDD [International Classification of Diseases, 9th Revision, Clinical Modification (ICD-9-CM) codes: 296.2x, 296.3x; ICD-10-CM: F32.0-F32.5, F32.9, F33.0x-F33.4x, F33.9x] during study period among MC, C, and SUP databases
 - ≥1 oral AP within 1 year since the qualifying event and on mono oral AP on index date (Index date was defined as the first date of augmentation with an AP; the AP used on the index date was defined as index therapy)
 - No AP use prior to index date, and on index therapy for ≥60 days within 6 months following the index date
 - Having ≥1 AD pharmacy claim in the 90 days before and the 90 days after the index date, with ≥15 days overlap of AD supply with the first index oral AP
 - Having MDD diagnosis prior to or on index date
 - Being continuously enrolled for 1 year before (baseline) and 1 year after index date (follow-up)
 - Exclusion criteria:
 - Patients with a diagnosis of schizophrenia or bipolar I disorder
- Two mutually exclusive cohorts were identified based on time from first qualifying event date to index date (**Figure 1**):
 - ≤6 months (early add-on)
 - 7-12 months (late add-on)
- Statistical analysis
 - General linear regression used to estimate adjusted healthcare costs in the early vs. late add-on cohorts
 - Controlled for baseline patient demographics [age, gender, race, insurance type (MC/C/SUP)] and clinical characteristics [qualifying event type, Charlson Comorbidity Index (CCI), number of chronic conditions, psychiatric comorbidities (anxiety, personality disorder, substance abuse disorder), psychiatric (antidepressants, antianxiety medications, sedatives or hypnotics, mood stabilizers) and non-psychiatric (antidiabetic medications, lipid-lowering medications, antihypertensive medications) medication use, ED visits, and hospitalizations
 - All costs adjusted to Y2016 USD
 - All data transformations and statistical analyses performed using SAS© version 9.4

Figure 1. Study Timeline

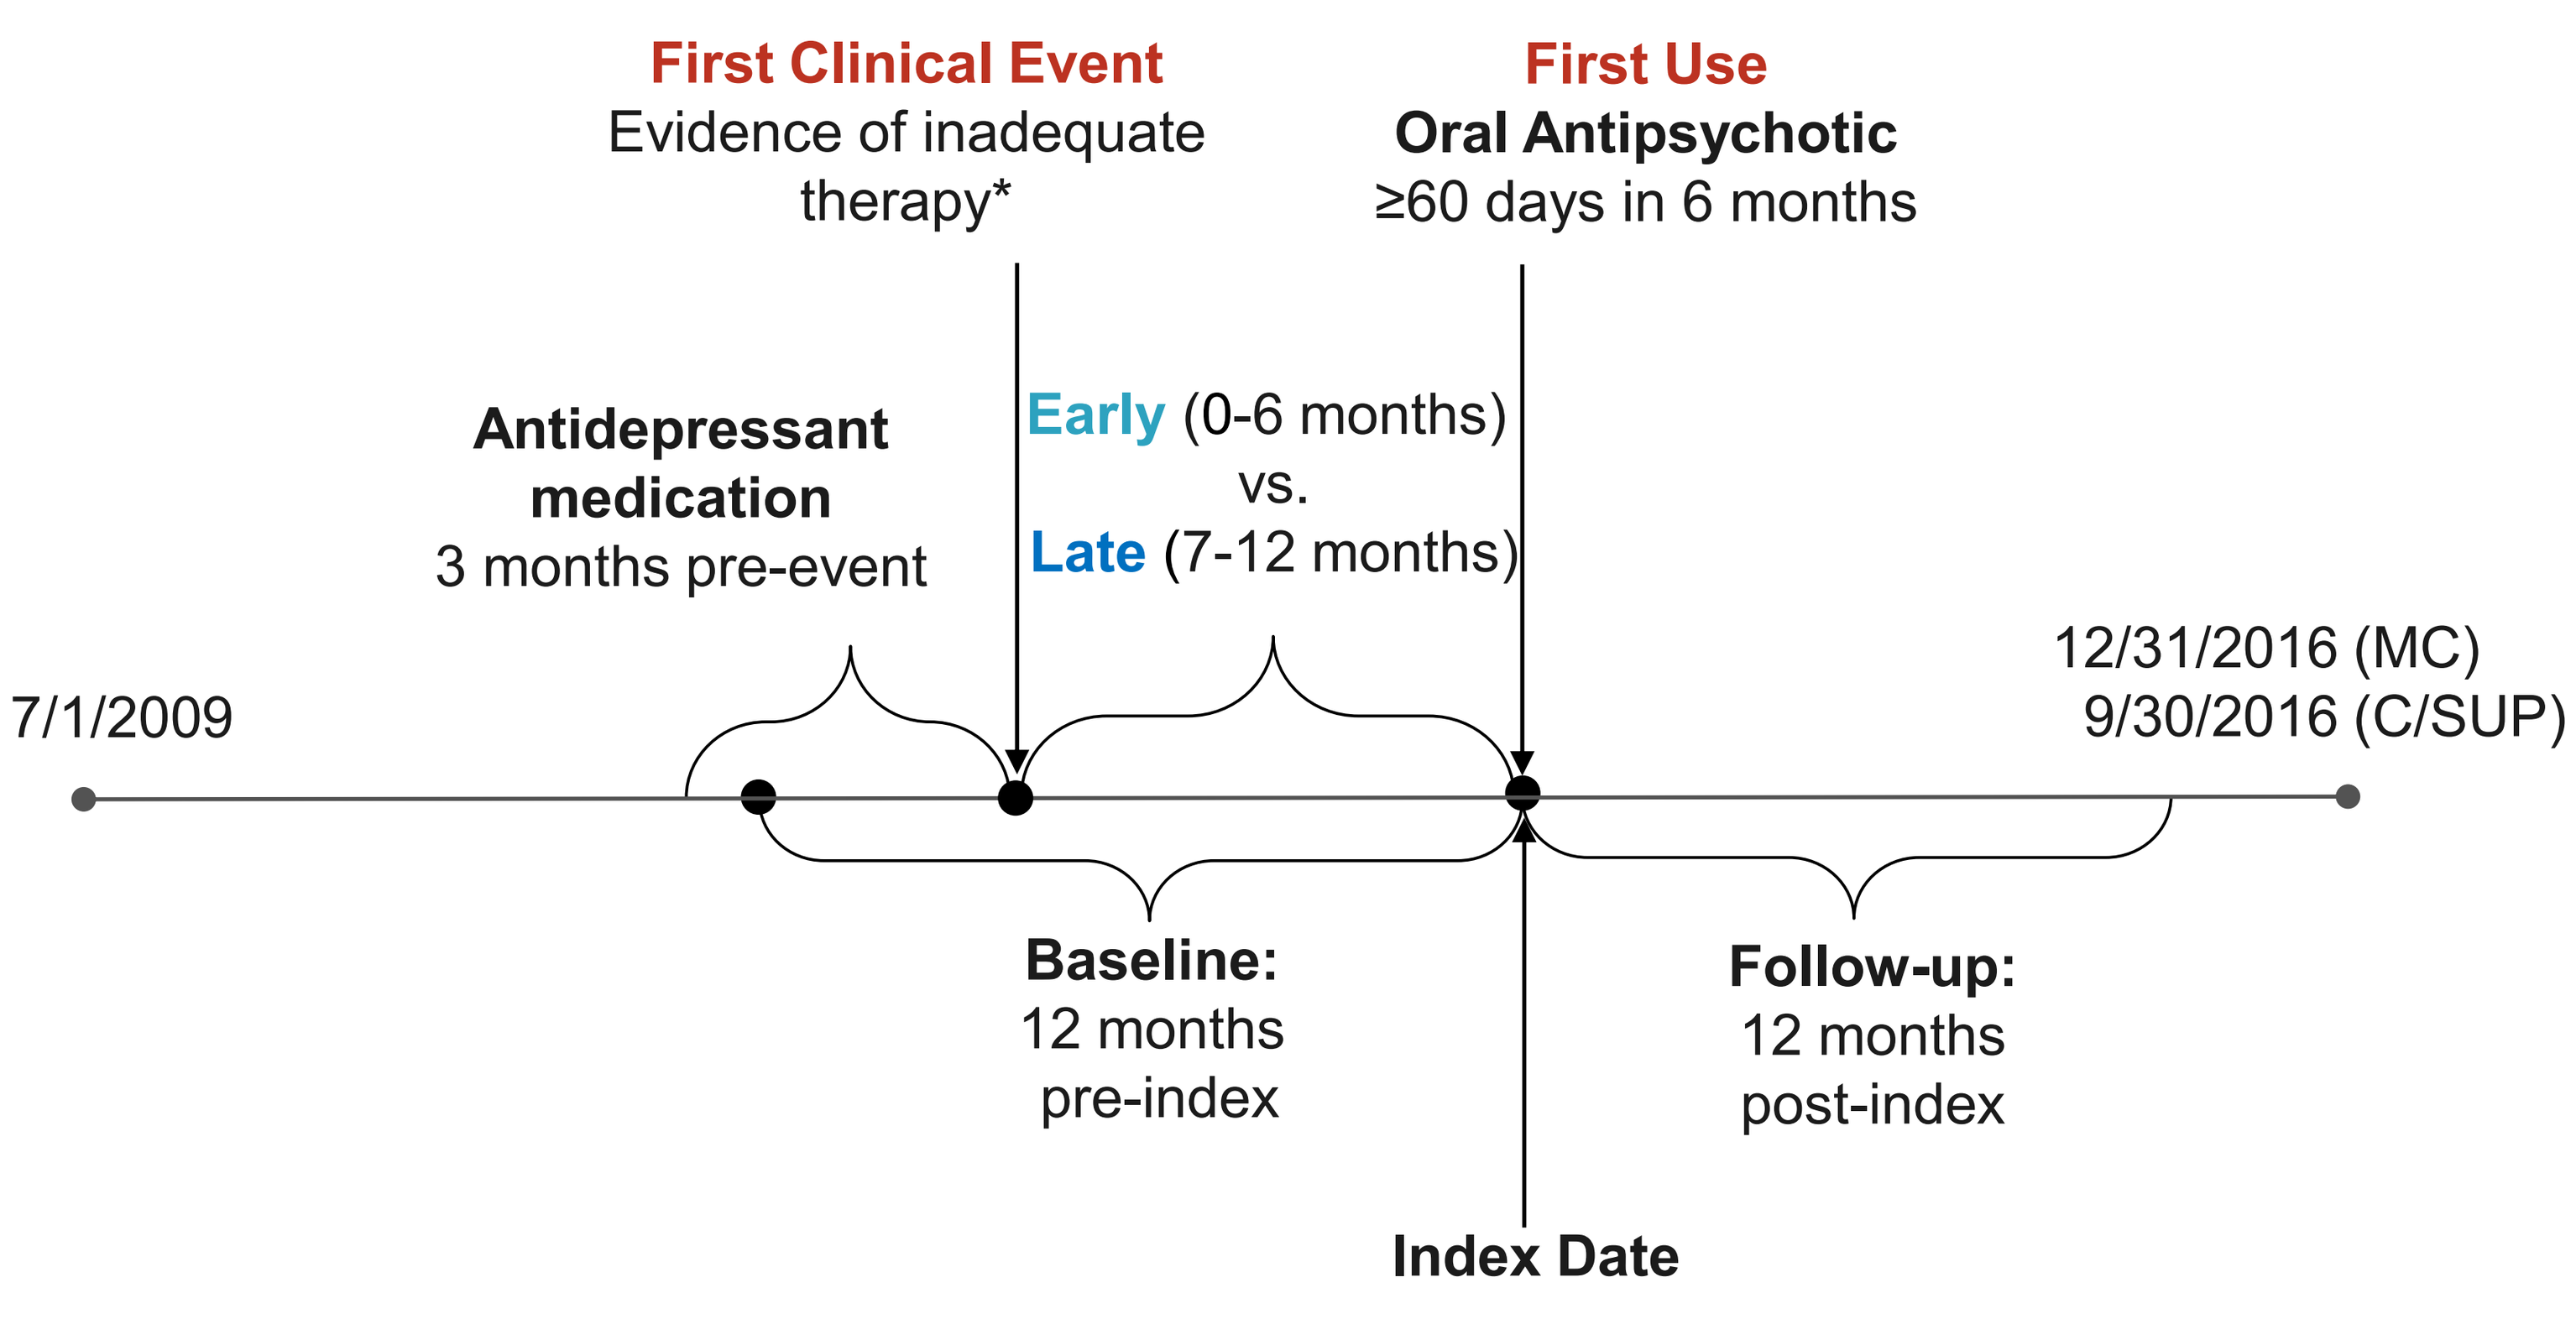
First Clinical Event
Evidence of inadequate therapy*
First Use
Oral Antipsychotic
≥60 days in 6 months
Antidepressant medication
3 months pre-event
Early (0-6 months)
vs.
Late (7-12 months)
7/1/2009
12/31/2016 (MC)
9/30/2016 (C/SUP)
Baseline:
12 months
pre-index
Follow-up:
12 months
post-index
Index Date

MC: Medicaid; **C**: Commercial; **SUP**: Medicare Supplemental

* Includes psychiatric ED visits, psychiatric hospitalization, antidepressant switches/add-ons and use of psychotherapy.

Results

Baseline characteristics

- A total of 6,935 MDD inadequate responders were included in the final study sample (**Table 1**).
- Patients entered the study due to different qualifying events.
 - Most common qualifying events were initiation of psychotherapy (43.3%) in early add-on cohort and a switch/add-on of AD (49.7%) in late add-on cohort (**Figure 2**).
- Baseline patient characteristics
 - Mean (SD) overall age was 49.5 (15.6) years, 68.3% were female, and 74.7% had Commercial insurance.
 - 45.8% had ≥1 psychiatric comorbidity (excluding bipolar I disorder or schizophrenia); anxiety (40.3%) was the most common.
 - 28.3% were hospitalized in the baseline period; 23.5% had ≥1 ED visit.
 - Early add-on cohort had higher psychiatric comorbid disease burden (47.3% vs. 42.5%; p<0.001) and higher inpatient utilization [mean (SD) 0.41 (0.72) vs. 0.27 (0.67); p<0.001] than late add-on (**Table 1**).
- All-cause healthcare costs
 - Early add-on cohort had numerically lower unadjusted inpatient and outpatient costs (**Figure 3**).
 - Pharmacy costs were significantly lower in early add-on cohort vs. late add-on cohort [mean (SD) $6,238 (6,185) vs. $7,091 (6,305); p<0.001].
 - Adjusted total all-cause healthcare costs were significantly lower in early vs. late add-on cohorts [$18,864 (95% CI: 18,004-19,725) vs. $20,452 (19,167-21,736); p=0.046] (**Figure 4**).

Figure 2. Qualifying Events for Adult Inadequate Responders

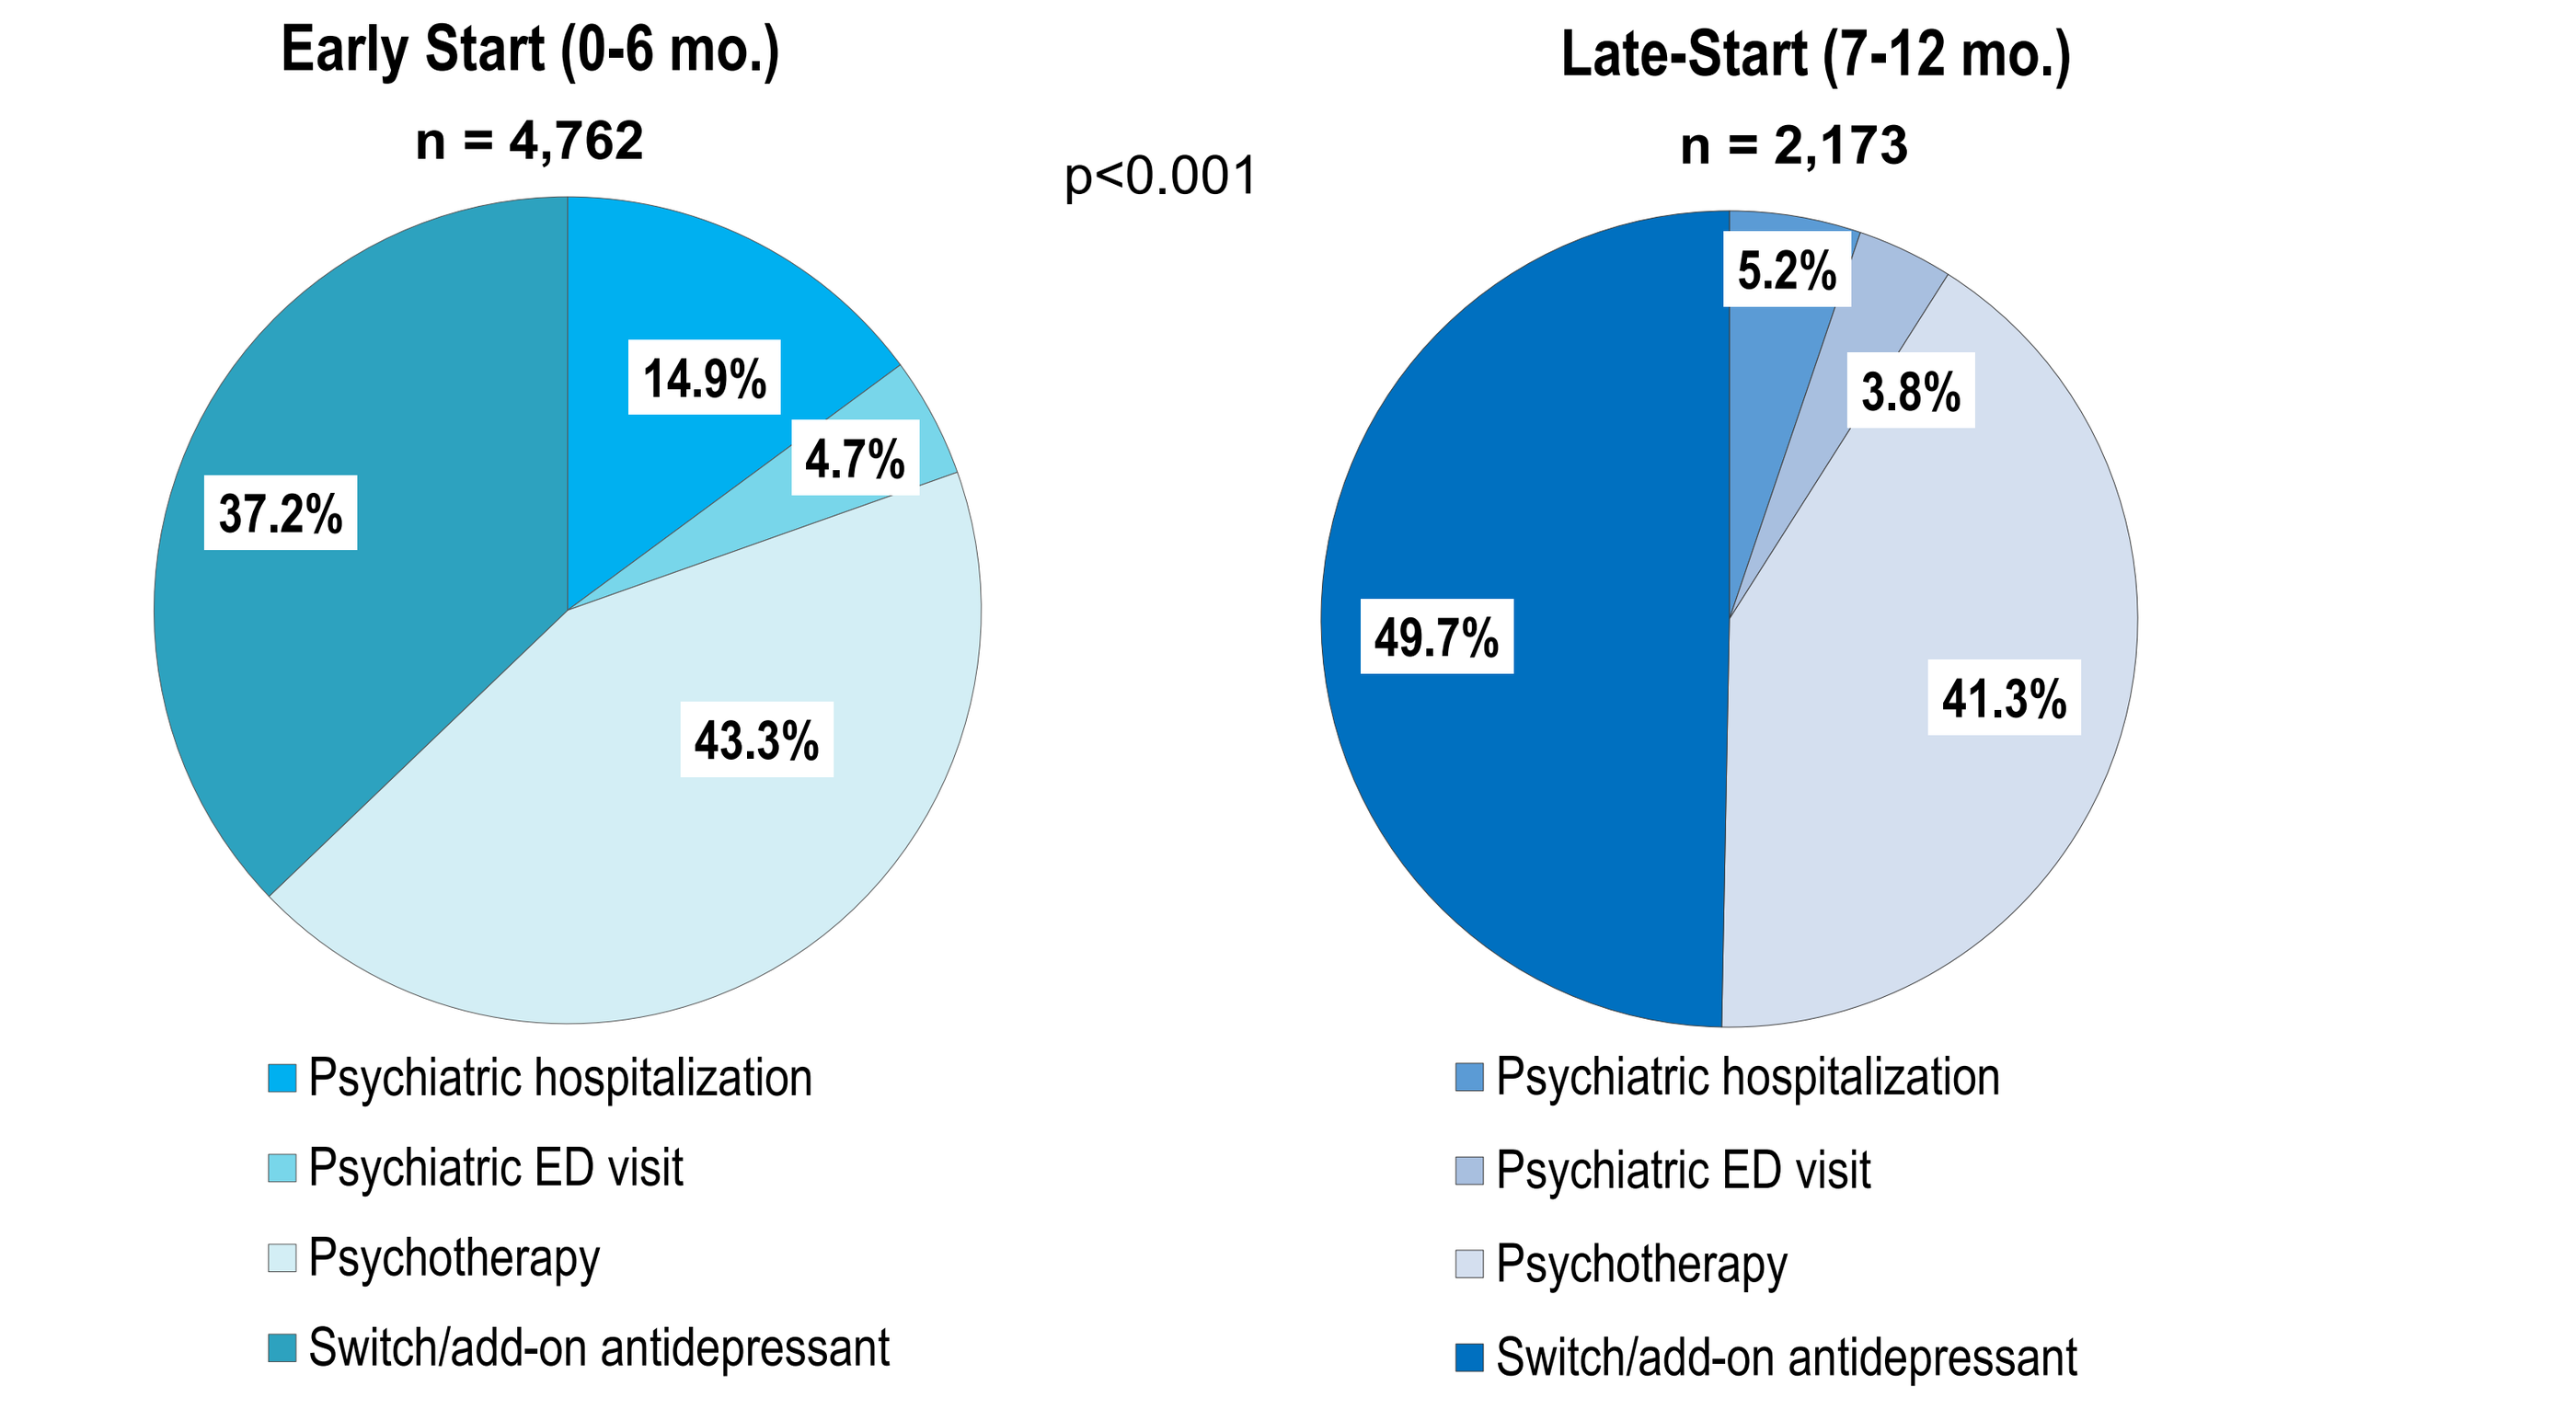
Early Start (0-6 mo.)
n = 4,762
p<0.001
Late-Start (7-12 mo.)
n = 2,173
14.9%
4.7%
37.2%
43.3%
5.2%
3.8%
49.7%
41.3%
Psychiatric hospitalization
Psychiatric ED visit
Psychotherapy
Switch/add-on antidepressant

Table 1. Baseline Demographic and Clinical Characteristics

	Early Start (0-6 mo.) N = 4,762; 68.7%	Late Start (7-12 mo.) N = 2,173; 31.3%	P Value
Age in years, mean (SD)	49.4 (15.8)	49.6 (15.1)	0.612
Female, n (%)	3,170 (66.6)	1,564 (72.0)	<0.001
Race, n (%)			0.024
White	364 (7.6)	199 (9.2)	
African American	87 (1.8)	50 (2.3)	
Other	47 (1.0)	31 (1.4)	
Unknown (Commercial/Medicare supplemental)	4,264 (89.5)	1,893 (87.1)	
Insurance Type, n (%)			0.008
Medicaid	498 (10.5)	280 (12.9)	
Commercial	3,574 (75.1)	1,603 (73.8)	
Medicare supplemental	690 (14.5)	290 (13.3)	
Index oral antipsychotic, n (%)			0.887
Atypical antipsychotic	4,681 (98.3)	2,135 (98.3)	
Typical antipsychotic	81 (1.7)	38 (1.7)	
Comorbidities			
Charlson comorbidity index, mean (SD)	0.8 (1.4)	0.7 (1.5)	0.510
No. HCUP chronic conditions, mean (SD)	3.2 (1.9)	3.3 (2.0)	0.488
Psychiatric comorbidities[a], n (%)	2,252 (47.3)	924 (42.5)	<0.001
Somatic comorbidities[b], n (%)	2,251 (47.3)	1,014 (46.7)	0.639
Baseline[c] medication and healthcare service use			
Any use of selected psychiatric medications[d], n (%)	4,762 (100.0)	2,173 (100.0)	n/a
Somatic medications[e], n (%)	2,512 (52.8)	1,202 (55.3)	0.047
Baseline hospitalization, mean (SD)	0.41 (0.72)	0.27 (0.67)	<0.001[f]

[a] Anxiety, personality disorder, substance abuse disorders. [b] Obesity, diabetes mellitus type 2, hyperlipidemia, hypertension. [c] One year prior to the index date. [d] Antidepressant, antianxiety medications, sedatives or hypnotics, mood stabilizer. [e] Antidiabetic medications, lipid-lowering medications, antihypertensive medications. [f] Kruskal Wallis test.

Figure 3. Components of All-Cause Healthcare Costs (unadjusted) in 1 Year Post-Index Period

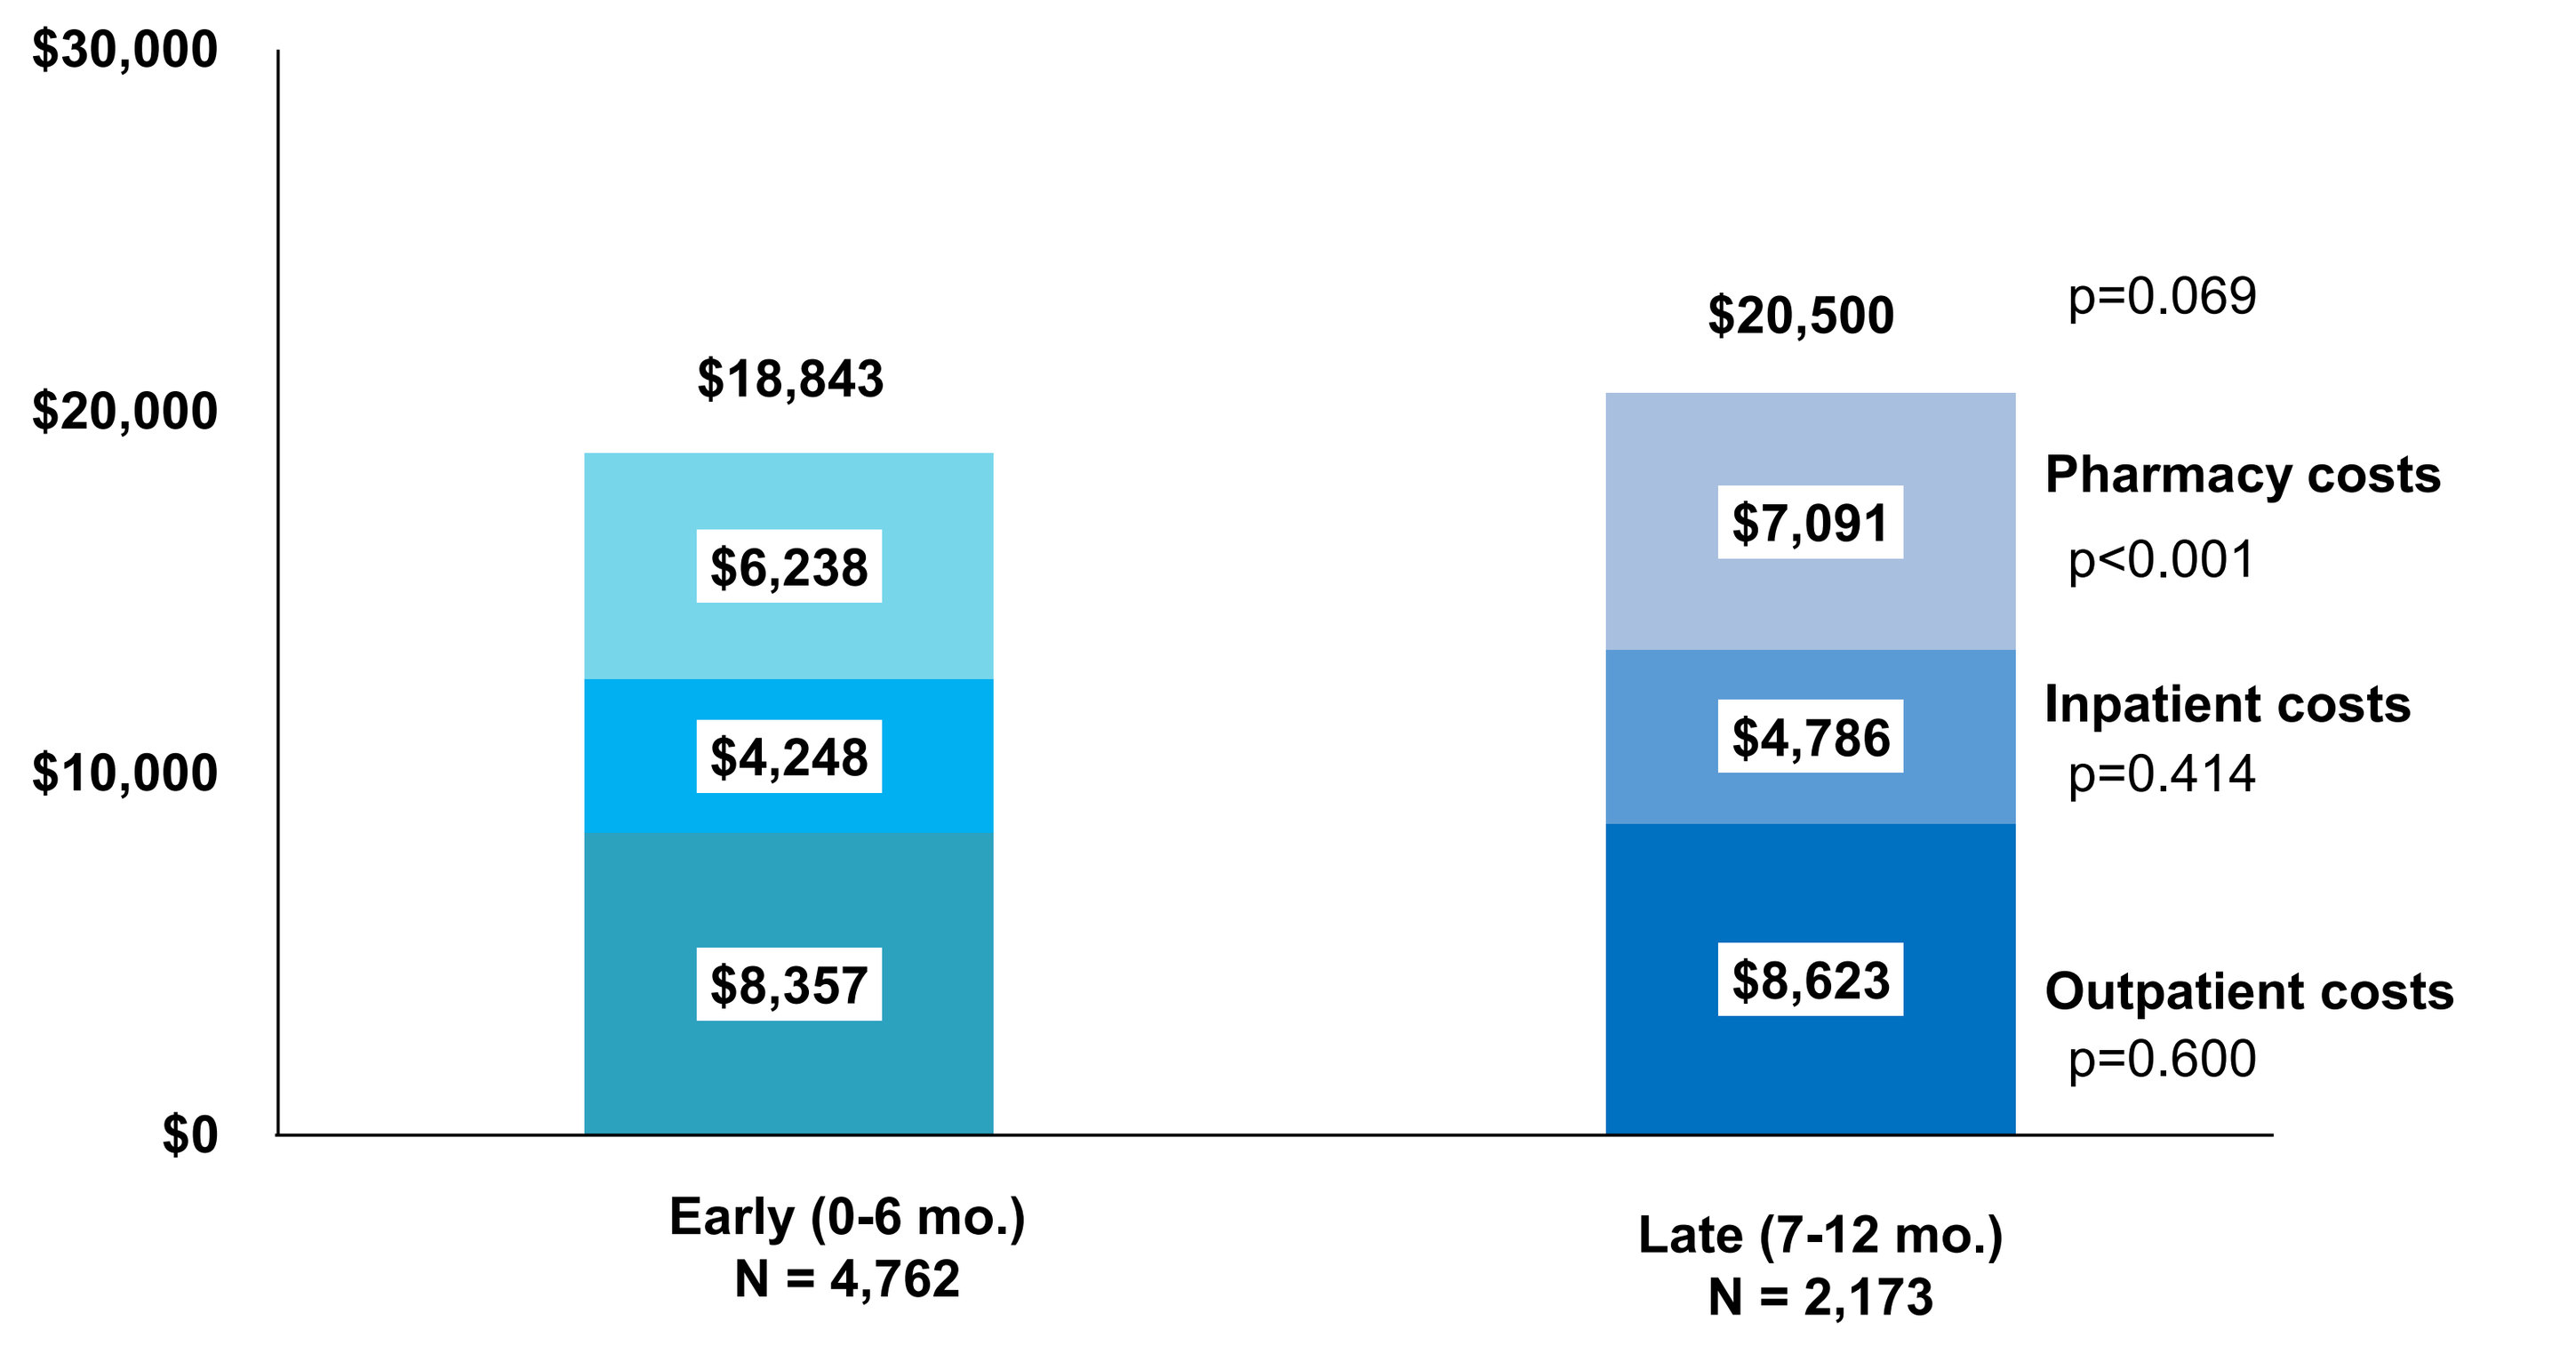
$30,000
$20,000
$10,000
$0
$18,843
$6,238
$4,248
$8,357
$20,500
$7,091
$4,786
$8,623
p=0.069
Pharmacy costs
p<0.001
Inpatient costs
p=0.414
Outpatient costs
p=0.600
Early (0-6 mo.)
N = 4,762
Late (7-12 mo.)
N = 2,173

Figure 4. Adjusted Total All-Cause Healthcare Costs in 1-Year Post-Index Period

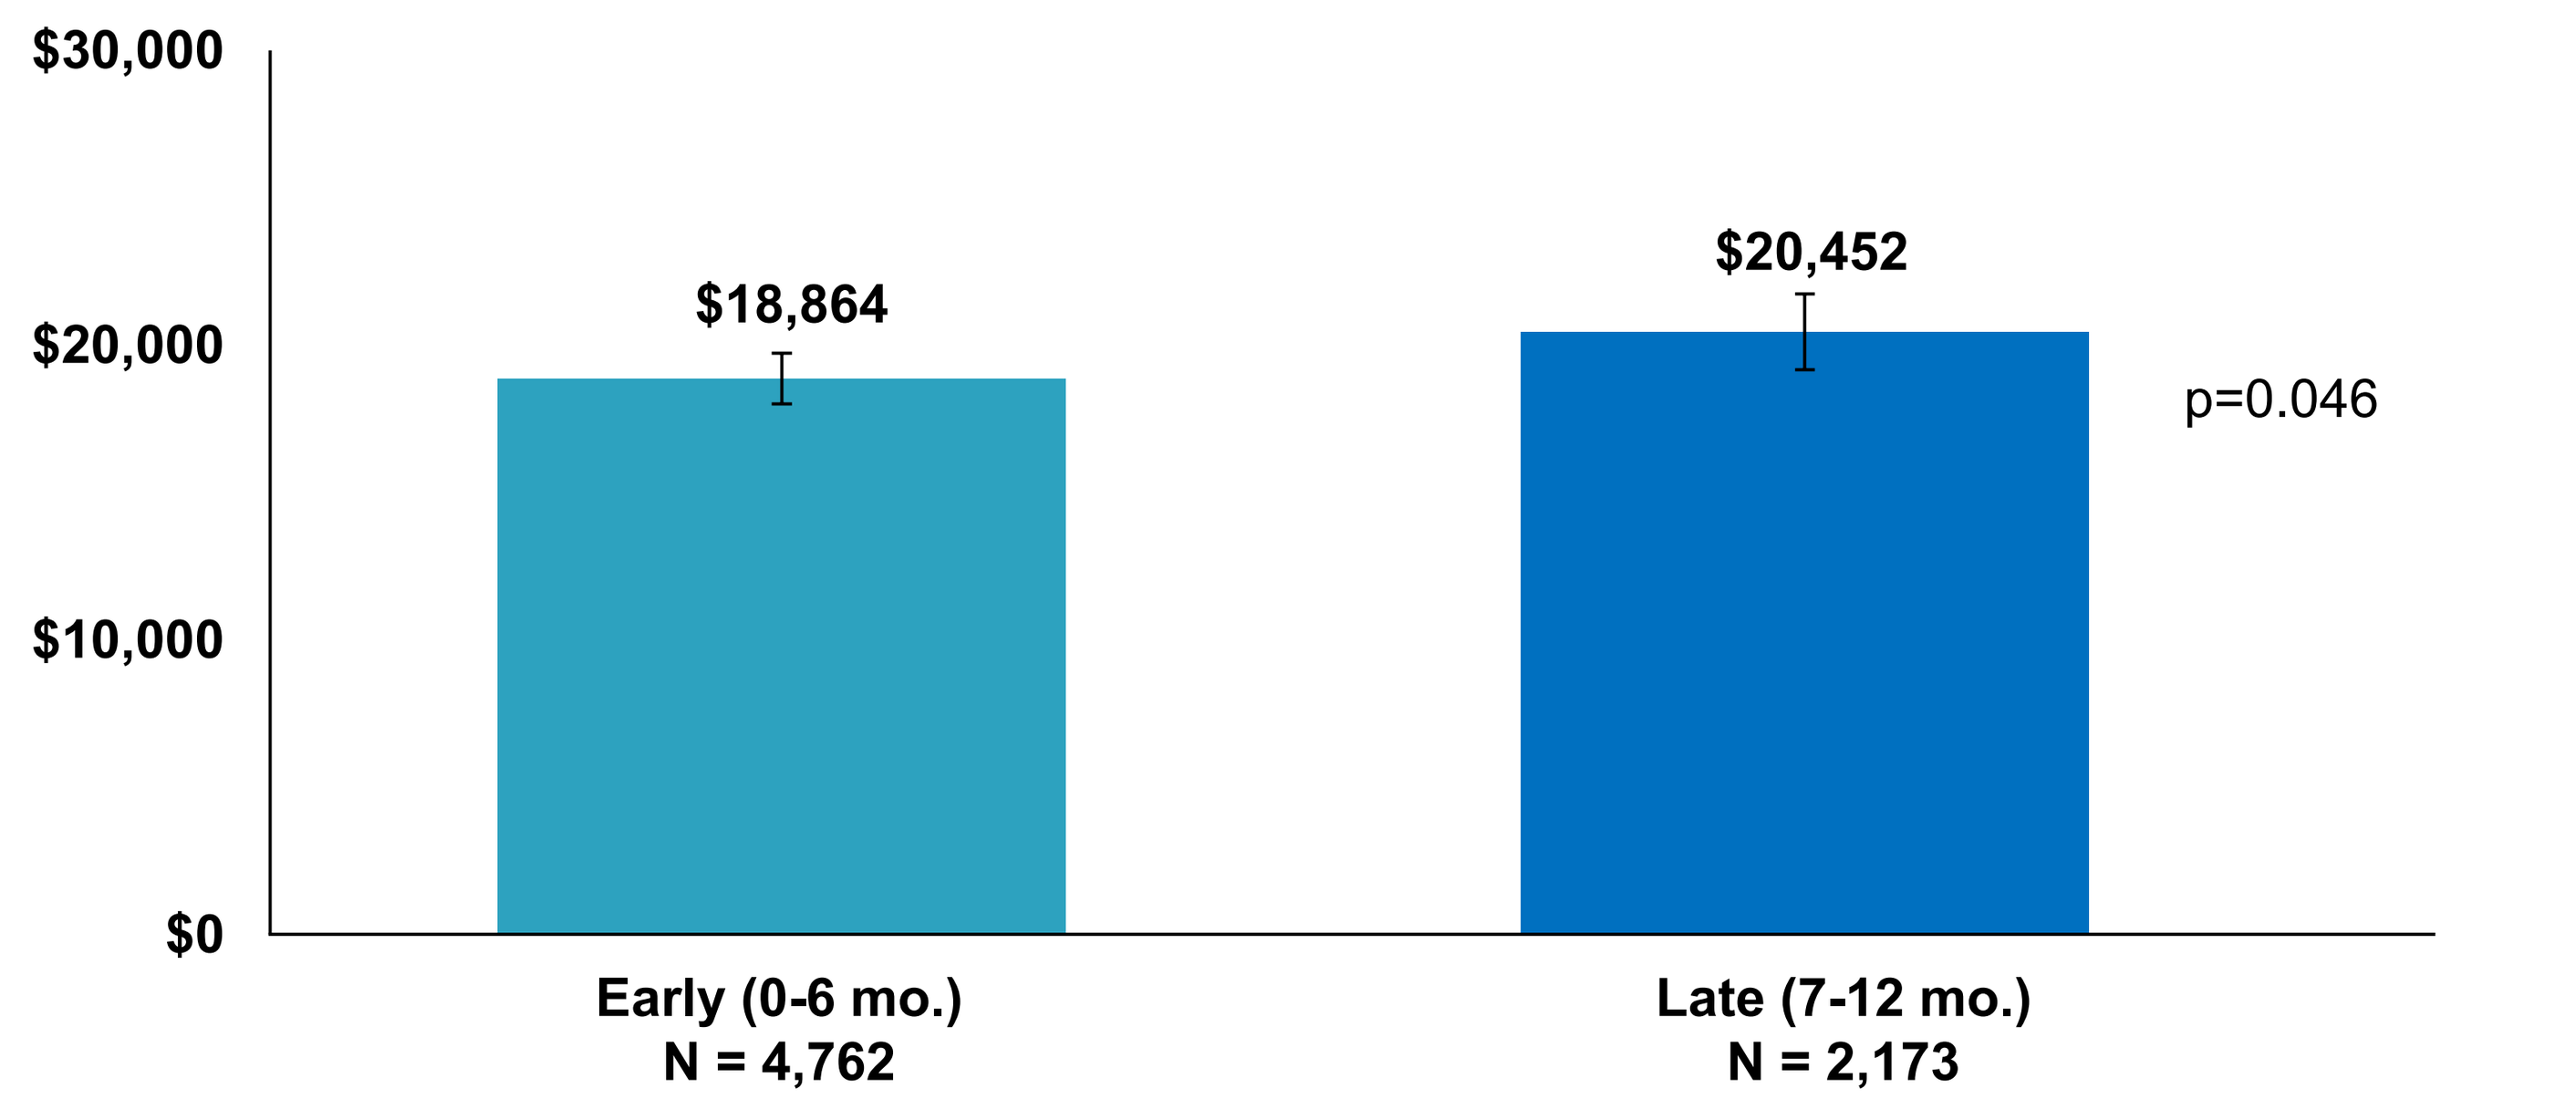
$30,000
$20,000
$10,000
$0
$18,864
$20,452
p=0.046
Early (0-6 mo.)
N = 4,762
Late (7-12 mo.)
N = 2,173

Conclusions

- Findings of this real-world study suggest that, in patients with MDD who inadequately responded to first-line AD treatment, adding an AP earlier may reduce overall healthcare costs.
- Limitations
 - Insurance claims are designed for reimbursement, not research. Disease severity and timing of remission were unavailable in this dataset, and we could not adjust for them in models. Psychiatric hospitalizations and ED visits were used as proxies.
 - Patients who discontinued an AP due to side effects or intolerability may have been excluded.
 - MDD diagnoses were identified through claims, where misclassification, diagnostic uncertainty, or coding errors are possible.
 - Baseline total costs were not adjusted for as this measure tends to be highly correlated with other confounders, such as ED visits or hospitalization.

References

1. American Psychiatric Association. Practice Guideline for the Treatment of Patients with Major Depressive Disorder. 2010.

Disclosures: M. Greene is an employee of Otsuka Pharmaceutical Development and Commercialization, Inc., Princeton, NJ. J. Yan, E. Chang, and I. Yermilov are employees of Partnership for Health Analytic Research, LLC, Beverly Hills, CA. A. Hartry is an employee of Lundbeck, Deerfield, IL. This study was sponsored by Otsuka Pharmaceutical Development and Commercialization, Inc. and Lundbeck; conducted by Partnership for Health Analytic Research, LLC.

ISPOR Asia Pacific,
September 8-11, 2018; Tokyo, Japan