Network Meta-Analysis of Treatments for Patients With Type 2 Diabetes Mellitus and Obesity

Lisa M. Neff, MD, MS[1]; Michael S. Broder, MD, MSHS[2]; David Beenhouwer, MD[2]; Eunice Chang, PhD[2]; Zhixiao Wang, PhD[3]

[1]*Department of Medicine, Feinberg School of Medicine, Northwestern University, Chicago, IL, USA;* [2]*Partnership for Health Analytic Research, LLC, Beverly Hills, CA, USA;* [3]*Eisai, Inc, Woodcliff Lake, NJ, USA*

Scan this code or visit obesityposters15.com/15091.pdf to view a PDF file of this poster.

T-P 3151

BACKGROUND

- When lifestyle changes and metformin fail to produce glycemic control in type 2 diabetes mellitus (T2DM), other antidiabetic medications (ADMs) are used
 - ADMs reduce glycated hemoglobin but their effect on weight varies
- Guidelines published by the American Association of Clinical Endocrinologists (AACE) in 2015 highlight the benefits of weight loss and minimizing risk of weight gain as an integral part of the management of T2DM[1]
- Lorcaserin (LOR), approved in 2012 in the US, is a serotonin-2C agonist indicated as an adjunct to diet and exercise for weight reduction in adults with a[2]:
 - Body Mass Index (BMI) greater than 30; OR
 - BMI greater than 27 plus at least one weight-related comorbidity including T2DM
- Clinical trials of LOR showed improvements in glycemic control

OBJECTIVE

- To compare the clinical effectiveness of adding LOR versus a second non-insulin ADM to metformin on weight and glycemic control

METHODS

- Systematic review using a combination of MeSH and keyword searching for relevant randomized controlled trials (RCTs) published from January 1, 1990 through December 16, 2014 in:
 - MEDLINE (PubMed)
 - EMBASE
 - ISI Web of Science
 - Cochrane CENTRAL
 - Conference abstracts and proceedings (2012-2014):
 - American Diabetes Association (ADA)
 - American Association of Clinical Endocrinologists (AACE)
- ADM categories:
 - Alpha-glucosidase inhibitor
 - Acarbose was the only alpha-glucosidase inhibitor that met the inclusion criteria
 - Sulfonylureas (SUs)
 - Thiazolidinediones (TZD) or glitazones
 - Glucagon-like peptide-1 (GLP-1) agonists
 - Exenatide was the only GLP-1 agonist that met the inclusion criteria
 - Dipeptidyl peptidase-4 (DPP-4) inhibitors
 - Meglitinides
 - Sodium-glucose cotransporter-2 (SGLT-2) inhibitors
- Studies had to include data for at least one of the primary study outcomes:
 - Change in hemoglobin A1c (HbA1c)
 - Achievement of HbA1c <7%
 - Change in baseline body weight
 - Number of episodes of hypoglycemia
- For the direct meta-analysis, separate analyses were conducted for each outcome and each pair of drug classes using DerSimonian and Laird random effects model
- For the network meta-analysis, a Bayesian Markov-chain Monte Carlo random effects model was conducted using the Bayesian software WinBUGS with weakly informative priors
- Pooled estimates from the posterior distribution and 95% credible intervals were reported

RESULTS

- 6,552 articles were screened; 191 underwent full review and 41 were included (**Figure 1**)

Figure 1. Systematic Review Flowchart

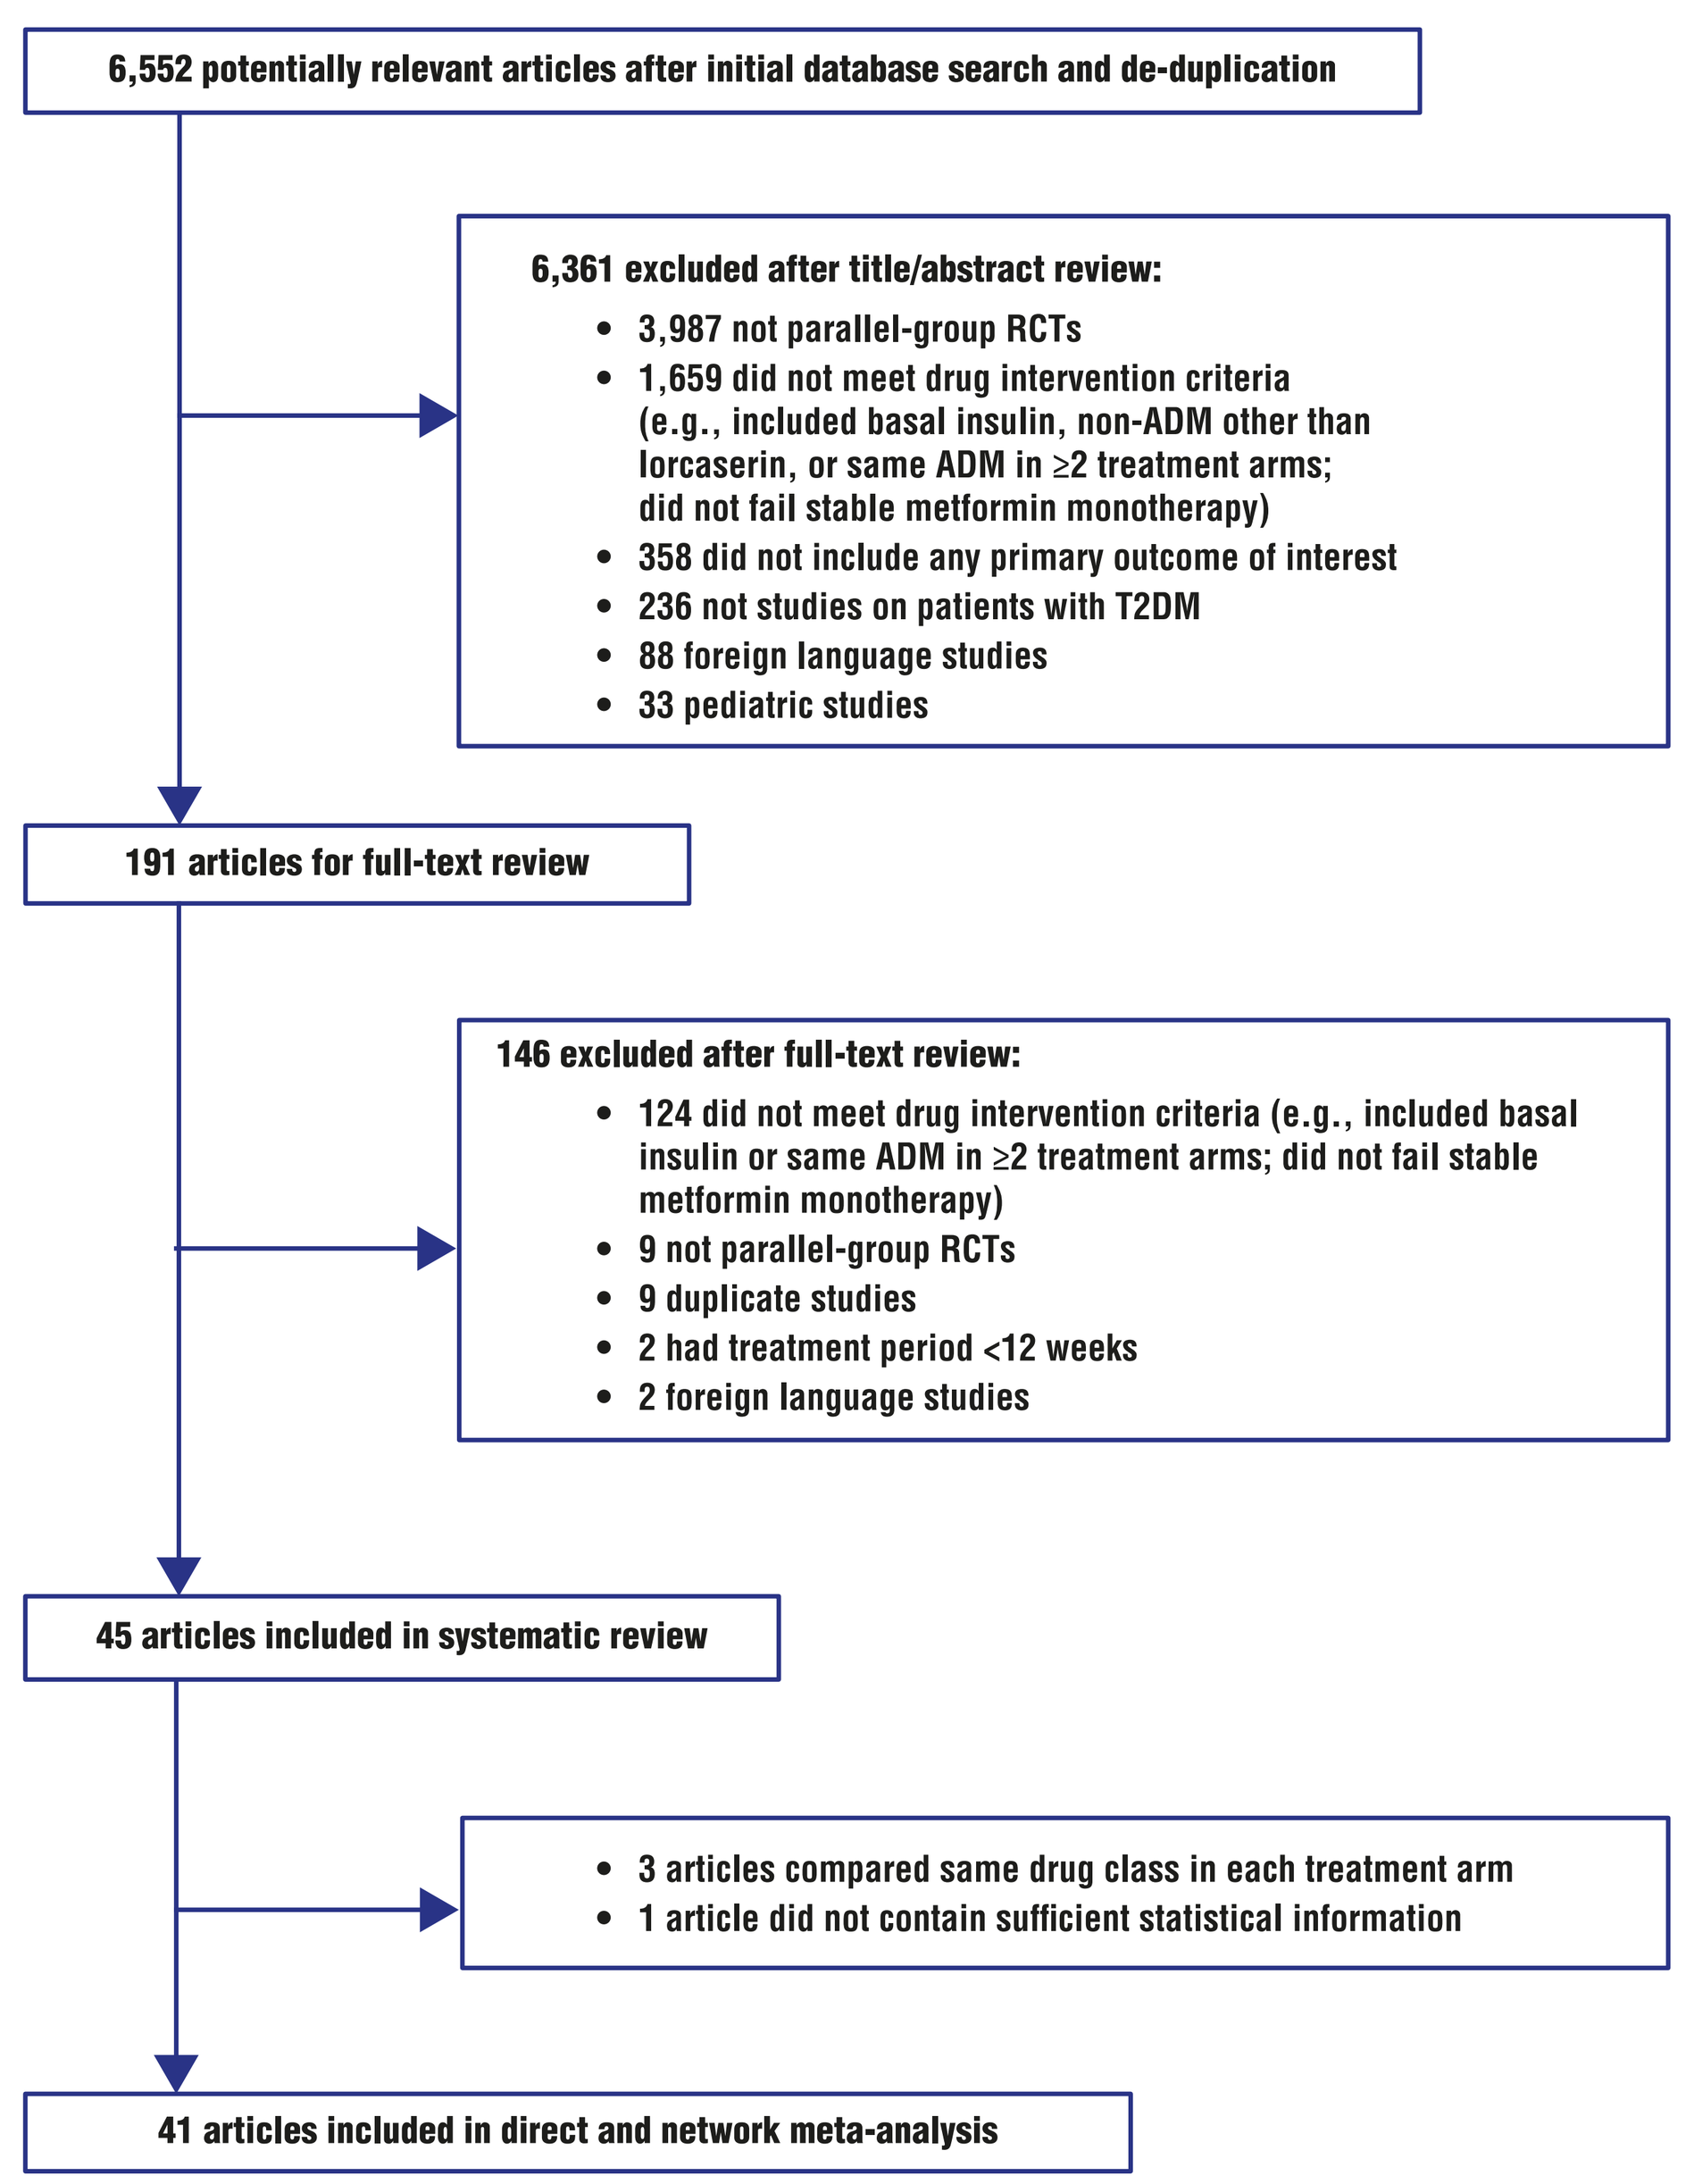
6,552 potentially relevant articles after initial database search and de-duplication
6,361 excluded after title/abstract review:
• 3,987 not parallel-group RCTs
• 1,659 did not meet drug intervention criteria (e.g., included basal insulin, non-ADM other than lorcaserin, or same ADM in ≥2 treatment arms; did not fail stable metformin monotherapy)
• 358 did not include any primary outcome of interest
• 236 not studies on patients with T2DM
• 88 foreign language studies
• 33 pediatric studies
191 articles for full-text review
146 excluded after full-text review:
• 124 did not meet drug intervention criteria (e.g., included basal insulin or same ADM in ≥2 treatment arms; did not fail stable metformin monotherapy)
• 9 not parallel-group RCTs
• 9 duplicate studies
• 2 had treatment period <12 weeks
• 2 foreign language studies
45 articles included in systematic review
• 3 articles compared same drug class in each treatment arm
• 1 article did not contain sufficient statistical information
41 articles included in direct and network meta-analysis

- LOR vs. placebo in network meta-analysis (**Table 1 and Figures 2-3**)
 - LOR significantly reduced overall HbA1c (% difference: -0.55 [-0.84, -0.26]) and patients achieved an HbA1c goal <7% more often (Relative Risk [RR]: 2.38 [1.68, 3.30]) compared to placebo
- LOR vs. ADMs in network meta-analysis (**Table 1 and Figures 2-3**)
 - Compared with LOR, none of the other classes of ADMs produced a statistically significantly greater reduction in HbA1c or in the proportion of patients achieving HbA1c goal
 - The risk of hypoglycemia was not significantly different with LOR compared with any classes of ADMs or placebo except for SUs, which had a significantly higher risk of hypoglycemia than LOR (3.51 [1.12, 9.67])
 - Four classes of ADMs produced significantly poorer weight outcomes compared with LOR:
 - kg difference: TZDs (5.79 [3.99, 7.50]), Glinides (5.54 [3.58, 7.69]), SUs (5.38 [3.73, 7.10]), and DPP-4 inhibitors (3.20 [1.46, 4.86])
 - LOR was non-inferior to alpha-glucosidase inhibitors, GLP-1 agonists, and SGLT-2 inhibitors at lowering body weight (kg difference: alpha-glucosidase inhibitor [2.35 (-0.08, 4.78)], GLP-1 [0.41 (-1.72, 2.24)], SGLT-2 [1.06 (-0.85, 3.02)])

Table 1. Network Meta-Analysis

Comparison Group	Change in HbA1c, % Difference (95% Crl)	Achieved HbA1c Goal <7% RR (95%Crl)	Change in Weight, kg Difference (95% Crl)	Change in BMI, kg/m² Difference (95% Crl)	Overall Hypoglycemia RR (95%Crl)
Reference: Placebo					
Lorcaserin	**-0.55 (-0.84, -0.26)**	**2.38 (1.68, 3.30)**	**-3.24 (-4.66, -1.81)**	-1.06 (-2.76, 0.64)	1.46 (0.45, 3.28)
Alpha-glucosidase inhibitor	**-0.81 (-1.20, -0.43)**	**2.81 (1.43, 4.89)**	-0.89 (-2.87, 1.10)	n/a	0.82 (0.01, 3.94)
Sulfonylurea	**-0.78 (-0.94, -0.63)**	**2.33 (1.82, 3.01)**	**2.14 (1.29, 3.07)**	0.56 (-0.74, 1.86)	**4.00 (2.47, 6.30)**
Thiazolidinedione	**-0.79 (-0.96, -0.63)**	**2.06 (1.50, 2.83)**	**2.55 (1.50, 3.54)**	0.63 (-0.80, 2.12)	0.69 (0.29, 1.32)
GLP-1 agonist	**-0.83 (-1.07, -0.60)**	**3.44 (2.35, 4.98)**	**-2.83 (-4.31, -1.57)**	**-2.33 (-4.25, -0.38)**	1.35 (0.44, 2.86)
DPP-4 inhibitor	**-0.65 (-0.80, -0.51)**	**2.12 (1.69, 2.66)**	-0.04 (-0.97, 0.86)	0.004 (-1.42, 1.45)	1.05 (0.55, 1.83)
Glinide	**-0.90 (-1.18, -0.64)**	**1.90 (1.09, 3.08)**	**2.30 (0.87, 3.84)**	n/a	**4.02 (1.96, 7.32)**
SGLT-2 inhibitor	**-0.89 (-1.27, -0.52)**	**2.57 (1.82, 3.64)**	**-2.18 (-3.45, -0.85)**	n/a	0.57 (0.16, 1.46)
Reference: Lorcaserin					
Alpha-glucosidase inhibitor	-0.26 (-0.75, 0.21)	1.21 (0.58, 2.14)	2.35 (-0.08, 4.78)	n/a	0.73 (0.01, 3.88)
Sulfonylurea	-0.23 (-0.57, 0.09)	1.00 (0.69, 1.45)	**5.38 (3.73, 7.10)**	1.61 (-0.51, 3.77)	**3.51 (1.12, 9.67)**
Thiazolidinedione	-0.24 (-0.58, 0.09)	0.89 (0.57, 1.34)	**5.79 (3.99, 7.50)**	1.68 (-0.54, 3.96)	0.60 (0.14, 1.78)
GLP-1 agonist	-0.28 (-0.65, 0.09)	1.48 (0.97, 2.27)	0.41 (-1.72, 2.24)	-1.27 (-3.83, 1.29)	1.19 (0.23, 3.64)
DPP-4 inhibitor	-0.10 (-0.42, 0.22)	0.91 (0.63, 1.30)	**3.20 (1.46, 4.86)**	1.06 (-1.19, 3.30)	0.93 (0.26, 2.67)
Glinide	-0.35 (-0.75, 0.04)	0.82 (0.42, 1.41)	**5.54 (3.58, 7.69)**	n/a	3.54 (0.98, 10.25)
SGLT-2 inhibitor	-0.34 (-0.82, 0.13)	1.11 (0.71, 1.70)	1.06 (-0.85, 3.02)	n/a	0.51 (0.09, 1.78)

BMI=Body Mass Index; CrI=credible interval; DPP-4=dipeptidyl peptidase-4; GLP-1=glucagon-like peptide-1; HbA1c=hemoglobin A1c; n/a=non-applicable; SGLT-2=sodium-glucose cotransporter-2; RR=relative risk.

Bold: RR≠1 or difference≠0 with significance level=0.05

Figure 2.

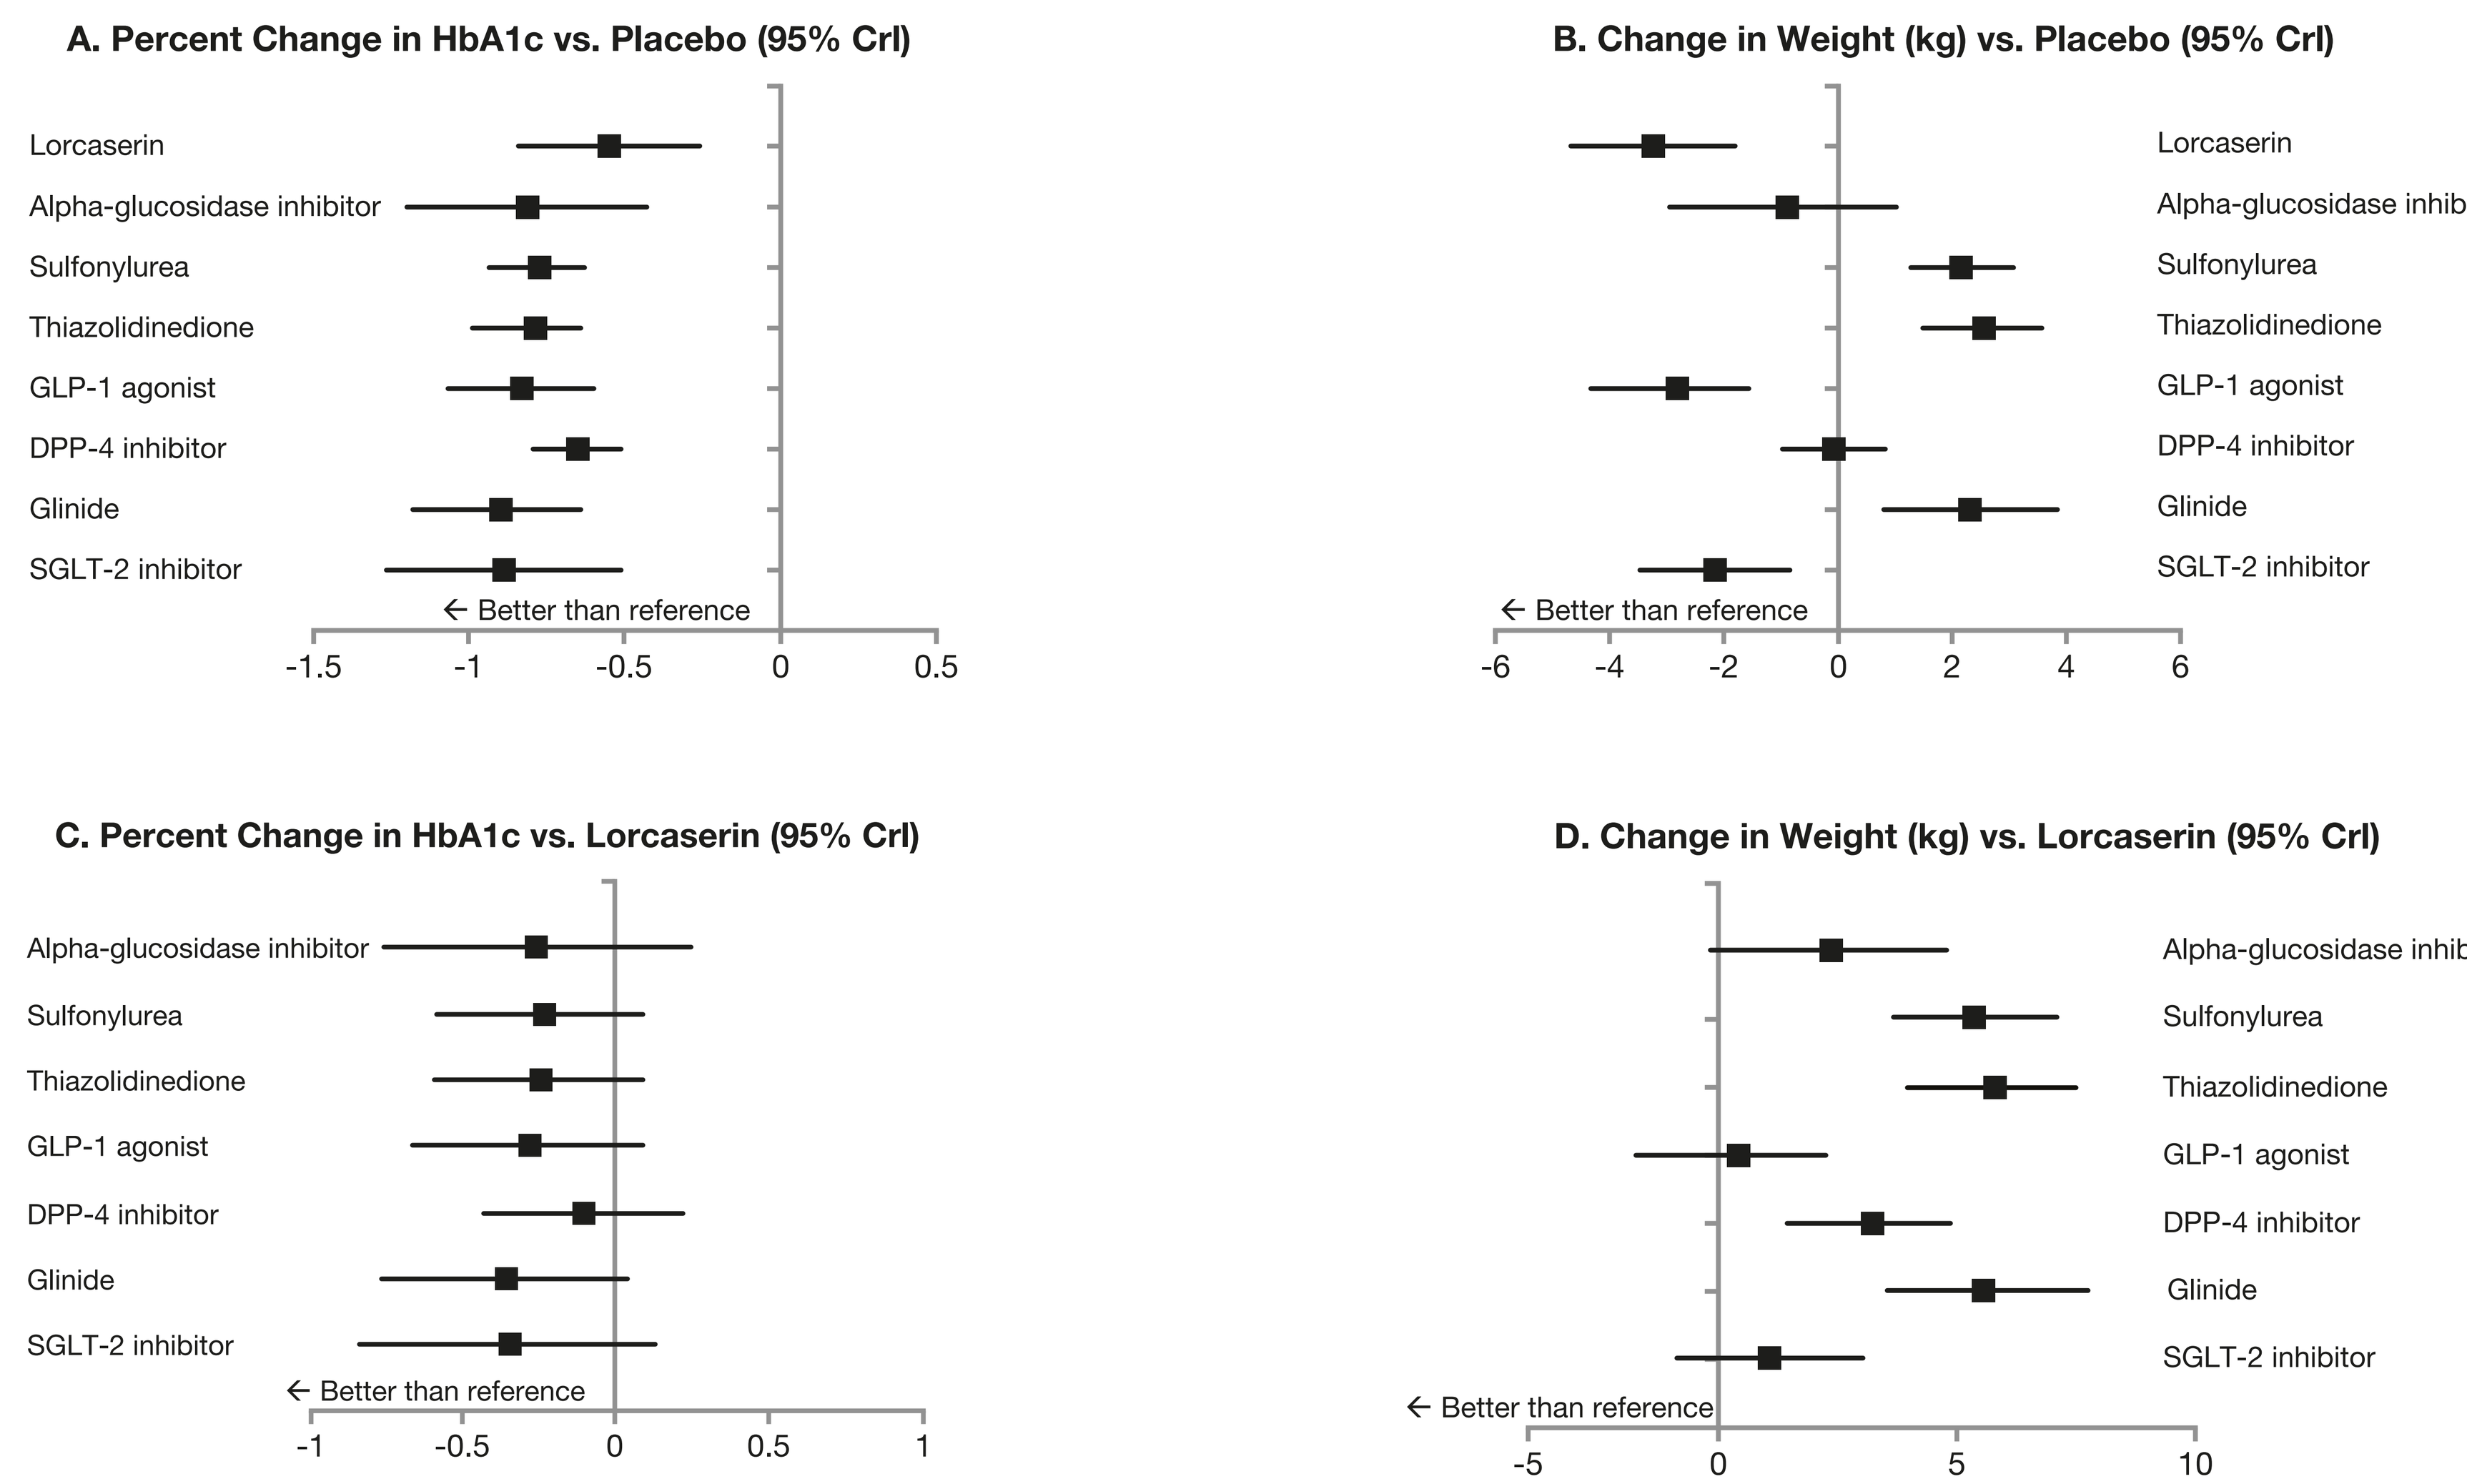
A. Percent Change in HbA1c vs. Placebo (95% CrI)
Lorcaserin
Alpha-glucosidase inhibitor
Sulfonylurea
Thiazolidinedione
GLP-1 agonist
DPP-4 inhibitor
Glinide
SGLT-2 inhibitor
← Better than reference
-1.5
-1
-0.5
0
0.5
B. Change in Weight (kg) vs. Placebo (95% CrI)
Lorcaserin
Alpha-glucosidase inhibitor
Sulfonylurea
Thiazolidinedione
GLP-1 agonist
DPP-4 inhibitor
Glinide
SGLT-2 inhibitor
← Better than reference
-6
-4
-2
0
2
4
6
C. Percent Change in HbA1c vs. Lorcaserin (95% CrI)
Alpha-glucosidase inhibitor
Sulfonylurea
Thiazolidinedione
GLP-1 agonist
DPP-4 inhibitor
Glinide
SGLT-2 inhibitor
← Better than reference
-1
-0.5
0
0.5
1
D. Change in Weight (kg) vs. Lorcaserin (95% CrI)
Alpha-glucosidase inhibitor
Sulfonylurea
Thiazolidinedione
GLP-1 agonist
DPP-4 inhibitor
Glinide
SGLT-2 inhibitor
← Better than reference
-5
0
5
10

Figure 3.

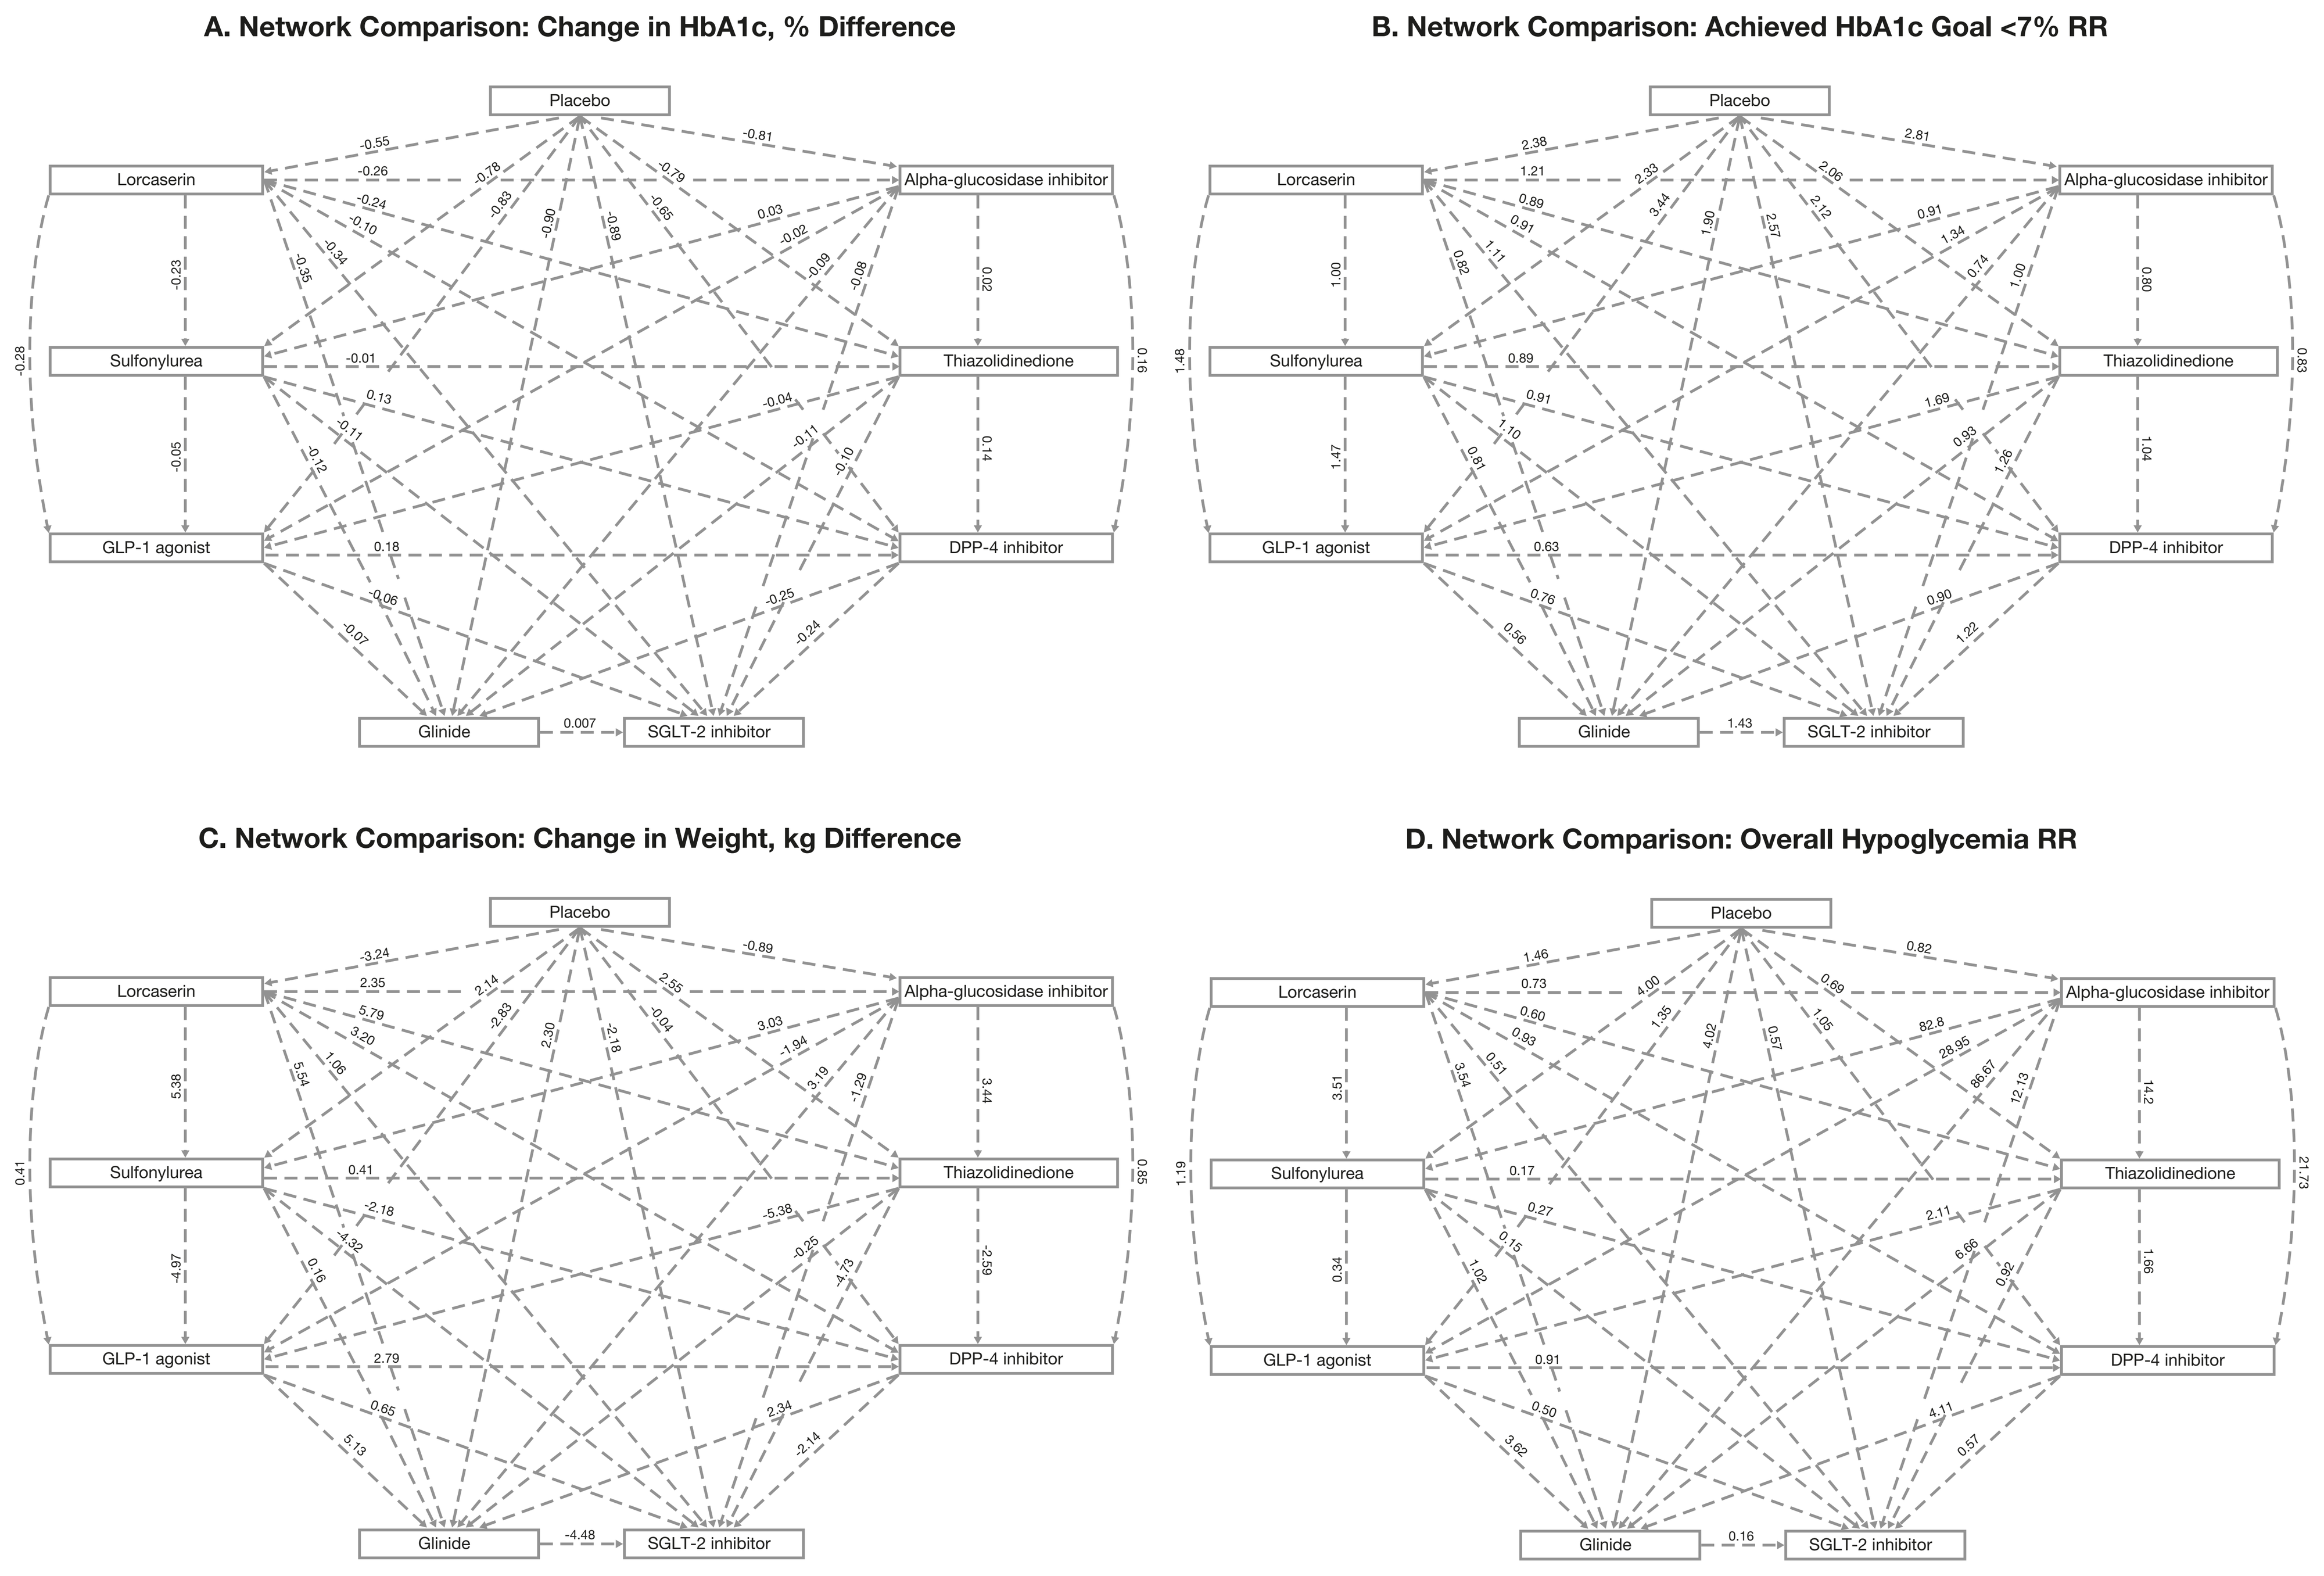
A. Network Comparison: Change in HbA1c, % Difference
B. Network Comparison: Achieved HbA1c Goal <7% RR
C. Network Comparison: Change in Weight, kg Difference
D. Network Comparison: Overall Hypoglycemia RR
Placebo
Lorcaserin
Alpha-glucosidase inhibitor
Sulfonylurea
Thiazolidinedione
GLP-1 agonist
DPP-4 inhibitor
Glinide
SGLT-2 inhibitor

CONCLUSIONS

- LOR is non-inferior to all studied classes of ADMs at lowering overall HbA1c and at achieving a goal of <7%
- Four of the studied ADMs produced significantly poorer weight outcomes as compared with LOR while none of the studied classes of ADMs reduced weight more than LOR
- LOR results in less hypoglycemia than SU and is non-inferior to other ADMs and to placebo with regard to hypoglycemia
- Although additional studies are needed, these analyses suggest that LOR may be added as an alternative to an add-on ADM in patients with BMI ≥ 27 who do not achieve glycemic control on a single agent
 - LOR may reduce HbA1c and achieve weight loss with a single intervention

LIMITATIONS, DISCLOSURES AND REFERENCES

- Studies with Jadad quality scores less than 3 (out of 5) were included
- Selection criteria were less stringent for the LOR study, as LOR is not an ADM
- There were only 15 direct connections among the 9 comparator regimens (including placebo). As a result, the network meta-analysis required many indirect comparisons, which could have compromised the confidence intervals of the point estimates

Disclosures

L Neff has consulted for Eisai Inc., received research grants from GI Dynamics, and is on the international speaker bureau of Pfizer (India). M Broder, D Beenhouwer, and E Chang are employees of PHAR, LLC, which was paid by Eisai to conduct the analysis described in this poster. Z Wang is an employee of Eisai Inc. Editorial support was provided by Imprint Publication Science, New York, NY, USA, with funding from Eisai Inc. This study was funded by Eisai Inc.

References

1. Handelsman Y et al. *Endocr Pract*. 2015;21(Suppl 1):1-87. **2.** BELVIQ Prescribing Information. Woodcliff Lake, NJ: Eisai Inc.; 2014.

Poster presented at Obesity Week 2015: November 2–7 in Los Angeles, CA, USA.