Better medication adherence to atypical antipsychotics is associated with lower psychiatric hospitalization rates and costs in bipolar I disorder

Mallik Greene, BPharm, PhD, DBA[1]; Jessie Tingjian Yan, PhD[2]; Eunice Chang, PhD[2]; Ann Hartry, PhD[3]; Irina Yermilov, MD, MPH, MS[2]

[1] Otsuka Pharmaceutical Development & Commercialization, Inc., Princeton, NJ, USA; [2] Partnership for Health Analytic Research, LLC, Beverly Hills, CA, USA; [3] Lundbeck, Deerfield, IL, USA

Background

- Bipolar I disorder (BD-I) is a chronic, relapsing mood disorder affecting 2.8% of the US adult population.[1]
- Medication nonadherence is a significant driver of healthcare utilization, such as hospitalization and ED visits.[2]
 - The direct healthcare cost of BD is more than $46 billion per year in the US, and the indirect costs are estimated to be over $146 billion.[3]
 - Hospitalizations account for up to two-thirds of the overall costs.[3]
- Previous literature has focused on comparing complete adherence (usually a threshold of 80%).[2,4]

Objective

The objective of the study was to examine association between different levels of medication adherence to atypical oral antipsychotics (AP) and psychiatric hospitalization and costs in real-world patients with BD-I.

Methods

- Retrospective cohort study using the IBM Health MarketScan® Medicaid, Commercial, and Medicare Supplemental Databases
- Patient identification **(Figure 1)**:
 - ≥1 inpatient or 2 outpatient claims for existing or newly diagnosed BD-I (ICD-9-CM: 290.0x, 296.1x, 296.4x-296.8x, excluding 296.82; ICD-10-CM: F30.x-F31.x, excluding F31.81) during the study period (1/1/2015-12/31/2016 Medicaid; 1/1/2015-9/30/2016 Commercial and Medicare Supplemental)
 - ≥1 pharmacy claim for any atypical oral AP (aripirazole, asenapine, brexpiprazole, cariprazine*, iloperidone*, lurasidone, olanzapine, paliperidone*, quetiapine, risperidone, and ziprasidone; not all indicated for treatment of BD-I) during ID period (7/1/2015-6/30/2016 for Medicaid; 7/1/2015-3/31/2016 for Commercial and Medicare Supplemental)
 - Index date: first date of starting atypical oral AP; index therapy: atypical oral AP used on the index date
 - To ensure new treatment initiation, no index therapy 6 months prior to the index date (baseline period) was allowed; non-index therapy in the baseline was allowed
 - Continuous health plan enrollment during the baseline period and 6 months after the index date (follow-up period)
 - Exclusion criteria
 - <18 years old on index date
 - Use of >1 antipsychotic medication (e.g., typical AP and long-acting injectables) on the index date
 - ≥1 diagnosis of schizophrenia (ICD-9-CM: 295.xx, except 295.4x and 295.7x; or ICD-10-CMs: F20x, except F20.81)

* Excluded from final sample due to small sample size.

Methods (continued)

- Medication adherence reported as proportion of days covered (PDC) during follow-up
 - PDC = number of days when index medication was available / 180 days
 - Three levels of medication adherence
 - Non-adherence: 0 ≤ PDC <40%
 - Partial adherence: 40% ≤ PDC <80%
 - Full adherence: 80% ≤ PDC ≤100%
- Outcome measures
 - Psychiatric hospitalization and costs (having a primary diagnosis for mental health [ICD-9-CM: 290.xx-311.xx; ICD-10-CM:F01.xx-F99.xx])
- Statistical analysis:
 - Logistic regression model used to estimate association between levels of medication adherence and risk of hospitalization
 - General linear regression model used to estimate association between levels of medication adherence and hospitalization costs
 - All models adjusted for patient demographics (e.g., clinical characteristics, baseline medication use, and baseline hospitalization)

Results

- The final sample consisted of 18,288 patients: 5,892 (32.0%) fully adherent, 4,246 (23.1%) partially adherent, and 8,250 (44.9%) non-adherent patients **(Table 1)**.
 - Non-adherent patients were younger than the partially and fully adherent groups [38.4 (14.0) vs 40. 0 (14.0) and 43.2 (15.0) years, respectively; p<0.001], and a greater proportion of them had psychiatric comorbidities compared with the partially and fully adherent patients [69.7% vs 68.4% and 66.2%, respectively; p<0.001] **(Table 1)**.

Figure 1. Patient Identification

≥1 inpatient or 2 outpatient claims for BD-1 during study period[a] among MC, C, and SUP databases
N = 222,498

↓

Atypical oral AP medication in the ID period[a]
N = 101,677 (45.7%)

↓

New atypical oral AP (index date: first date of the new atypical AP)
N = 47,251 (21.2%)

↓

Antipsychotic monotherapy on index date
N = 45,035 (20.2%)

↓

BD-I prior to index date, and no schizophrenia diagnosis during study period[a]
N = 30,567 (13.7%)

↓

6-month before- and after- continuous enrollment and ≥18 years; no paliperidone, cariprazine, and iloperidone due to small sample size
N = 18,388 (8.3%)

BD-I: bipolar I disorder; **MC:** Medicaid; **C**: Commercial; **SUP**: Medicare supplemental; **AP:** antipsychotic

Results (continued)

Table 1. Patient Demographics

	Proportion of Days Covered (PDC)[a] of Index Medication			
	Non-adherence n = 8,250; 44.9%	**Partial adherence n = 4,246; 23.1%**	**Full adherence n = 5,892; 32.0%**	***P* Value**
Age in years, mean (SD)	38.4 (14.0)	40.0 (14.0)	43.2 (15.0)	<0.001
Female, n (%)	5,756 (69.8)	2,949 (69.5)	4,011 (68.1)	0.088
Insurance Type, n (%)				<0.001
Medicaid	3,577 (43.4)	1,837 (43.3)	2,066 (35.1)	
Commercial	4,401 (53.3)	2,239 (52.7)	3,448 (58.5)	
Medicare supplemental	272 (3.3)	170 (4.0)	378 (6.4)	
Comorbidities				
Charlson comorbidity index, mean (SD)	0.7 (1.4)	0.8 (1.4)	0.8 (1.5)	<0.001
No. chronic conditions, mean (SD)	3.1 (2.0)	3.3 (2.1)	3.4 (2.1)	<0.001
Psychiatric comorbidities[b], n (%)	5,751 (69.7)	2,904 (68.4)	3,900 (66.2)	<0.001
Non-index Antipsychotic Use, n (%)	2,325 (28.2)	1,285 (30.3)	2,123 (36.0)	<0.001
Any use of selected psychiatric medications[c], n (%)	6,307 (76.4)	3,399 (80.1)	4,975 (84.4)	<0.001
Non-psychiatric medications[d], n (%)	3,097 (37.5)	1,748 (41.2)	2,817 (47.8)	<0.001
Any baseline hospitalization, n (%)	2,389 (29.0)	1,150 (27.1)	1,571 (26.7)	0.006

[a] Non-adherent: 0 ≤ PDC <40%; partially adherent: 40% ≤ PDC< 80%; fully adherent: 80% ≤ PDC ≤100%.
[b] Major depressive disorder, anxiety, personality disorders, substance abuse disorders.
[c] Mood stabilizers, antidepressants, anti-anxiety medications, sedatives or hypnotics.
[d] Anti-diabetic medications, lipid-lowering medications, and anti-hypertensive medications.

- The unadjusted psychiatric hospitalization rates were lowest in the fully adherent (7.5%) group compared to the partially (10.0%) and non- (11.0%) adherent groups (p<0.001).
- Adjusting for baseline differences, the adjusted rate of psychiatric hospitalization during the 6-month follow-up period remained lower in the fully adherent (6.0%) than in the partially (8.3%) or non-adherent (8.8%) groups (p<0.001) **(Figure 2)**.
 - Using the fully adherent cohort as the reference group, the odds of psychiatric hospitalization were significantly higher for the non-adherent [OR (95% CI): 1.51 (1.33-1.71) and partially adherent (1.42 (1.23-1.64)] cohorts.
- Among hospitalized patients (n = 1,767), the mean adjusted psychiatric hospitalization cost was lower for the fully adherent cohort ($11,748), than the partially adherent ($15,051, p=0.002), or non-adherent cohorts ($13,170, not statistically significant) **(Figure 2)**.

Disclosures: Greene is an employee of Otsuka Pharmaceutical Development & Commercialization, Inc., Princeton, NJ. Chang, Yan, and Yermilov are employees of Partnership for Health Analytic Research, LLC, Beverly Hills, CA. Hartry is an employee of Lundbeck, Deerfield, IL. Funding for the study and this poster was received from Otsuka Pharmaceutical Development & Commercialization, Inc. and Lundbeck.

Results (continued)

Figure 2. Adjusted Psychiatric Hospitalization Rates and Costs in Follow-up

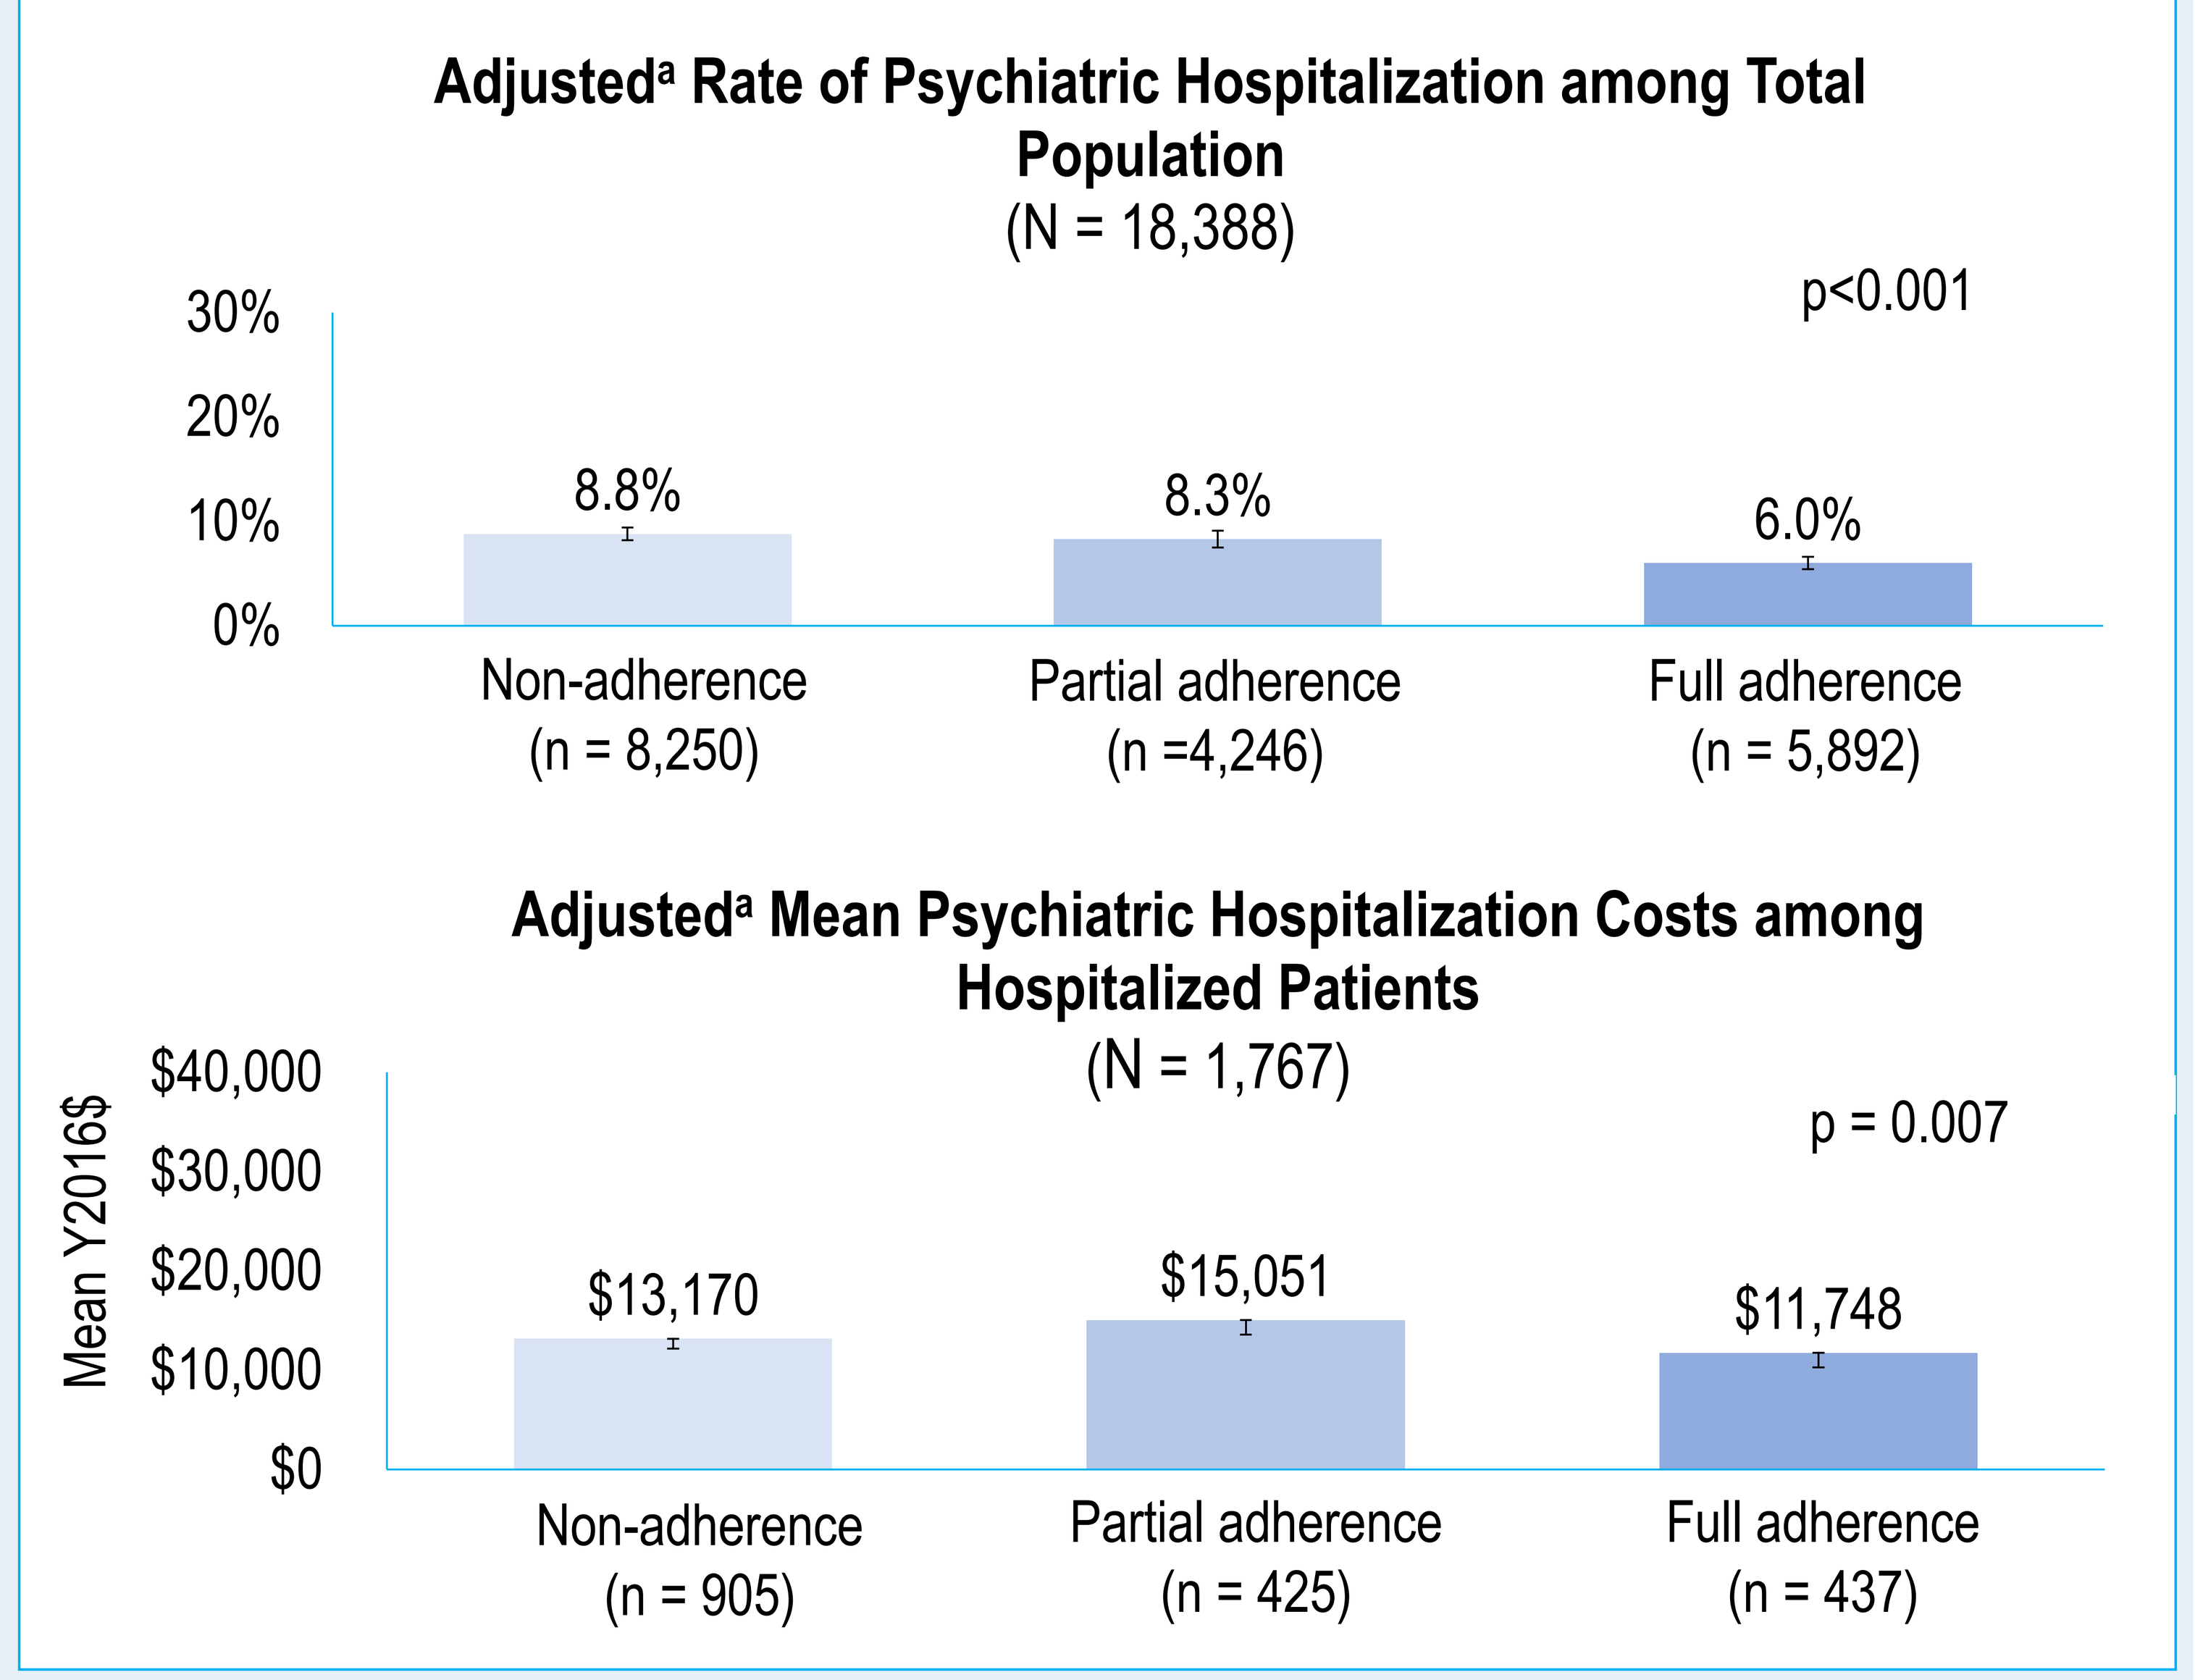
Adjusted[a] Rate of Psychiatric Hospitalization among Total Population
(N = 18,388)
30%
20%
10%
0%
p<0.001
8.8%
8.3%
6.0%
Non-adherence (n = 8,250)
Partial adherence (n =4,246)
Full adherence (n = 5,892)
Adjusted[a] Mean Psychiatric Hospitalization Costs among Hospitalized Patients
(N = 1,767)
Mean Y2016$
$40,000
$30,000
$20,000
$10,000
$0
p = 0.007
$13,170
$15,051
$11,748
Non-adherence (n = 905)
Partial adherence (n = 425)
Full adherence (n = 437)

[a] Adjusted by age group, gender, insurance type, Charlson comorbidity, no. of chronic conditions, baseline psychiatric comorbidity, baseline non-index anti-psychotic use, baseline psychiatric medication use, baseline non-psychiatric medication use, baseline hospitalization, and index medication.

Conclusion

- In a mixed population of Medicaid, Medicare, and Commercially insured patients with BD-I who initiated treatment with an AAP, better medication adherence was associated with lower psychiatric hospitalization rates and costs.
- These findings suggest that improving adherence in BD-I may be a valuable goal from both clinical and economic perspectives.
- Limitations of the study
 - Claims are meant for reimbursement, not research, so misclassification is possible
 - Claims for a medication indicate that a prescription was filled and not that it was actually taken as prescribed. A future study with longer follow-up is warranted.

References

1. NIMH. 2017. Available at: https://www.nimh.nih.gov/health/statistics/bipolar-disorder.shtml.
2. Rascati KL, et al. Psychiatr Serv. 2011;62(9):1032-40.
3. Cloutier M, et al. J Affec Disord. 2017;226(2018):45-51.
4. Velligan DI, et al. J Clin Psychiatry. 2009;70(Suppl 4):1-46.

AMCP Managed Care & Specialty Pharmacy Annual Meeting
April 23-26, 2018, Boston, MA

Research was conducted by Partnership for Health Analytic Research, LLC.