Scientific Content On-demand
To receive a copy of this poster
Scan QR code via a barcode reader application
By requesting this content, you agree to receive a one-time communication using automated technology. Messaging & data rates may apply. Links are valid for 30 days after the congress presentation date.
Copies of this poster obtained through Quick Response (QR) code or text message are for personal use only and may not be reproduced without permission from EULAR and the authors of this poster.
If you need further information regarding this poster or other BMS presentations at EULAR 2019, please inquire at the BMS Medical Information Booth (#24) in Exhibition Hall 10.

Real-World Evaluation of Persistence with Early-Line Abatacept versus Tumor Necrosis Factor-Inhibitors for Rheumatoid Arthritis Complicated by Poor Prognostic Factors

SAT0157

D Paul,[1] I Yermilov,[2] SN Gibbs,[2] MS Broder [2]

[1]Bristol-Myers Squibb, Lawrenceville, NJ, USA;
[2]Partnership for Health Analytic Research (PHAR), LLC, Beverly Hills, CA, USA

Introduction

- Treatment of active rheumatoid arthritis (RA) usually includes a conventional disease-modifying anti-rheumatic disease (DMARD), such as methotrexate, sulfasalazine, or leflunomide. Patients who are intolerant or show an inadequate response to conventional DMARDs are often treated with a targeted DMARD.
- There are multiple classes of targeted DMARDS including tumor necrosis factor inhibitors (TNFi), interleukin-6 inhibitors, CD20 inhibitors, Janus kinase inhibitors, and T-cell co-stimulators such as abatacept.
- In patients with moderately to severely active RA, abatacept inhibits the progression of structural damage, reduces symptoms, and improves physical function.[1] However, the real-world data on abatacept's use as an early-line biologic agent are limited.
- This study assessed the 12-month treatment persistence in early-line abatacept versus TNFi treated patients with RA complicated by poor prognostic factors.

Methods

- We performed a multicentre retrospective medical record review of adult RA patients with poor prognostic factors treated at 5 United States clinics located in the West, Midwest, and Southeast.
- Patients were treated with abatacept or TNFi as the first biologic treatment at the clinic (defined as early line).
- Poor prognostic factors included:[2]
 - Positive anti-cyclic citrullinated peptide antibodies
 - Positive rheumatoid factor antibodies
 - Increased C-reactive protein levels
 - Elevated erythrocyte sedimentation rate levels
 - Presence of joint erosions
- TNFis included adalimumab, etanercept, infliximab (and their biosimilars), certolizumab pegol, or golimumab.
- Chart data were abstracted into an electronic case report form. Demographic, disease, and treatment information (start, stop, reason for discontinuation) was abstracted. Data were collected from biologic treatment initiation for ≥1 year. See Figure 1.
- Treatment persistence (continuation of index treatment with gap ≤60 days) at 12 months and time to discontinuation were reported.
- Multivariate logistic and Cox regressions were used to compare 12-month persistence and overall discontinuation rates between abatacept and TNFi, controlling for demographic and clinical characteristics (age at index, gender, Charlson comorbidity index, time from RA diagnosis to index), baseline utilizations (number of physician office visits, number of hospitalizations), and clinic.

Results

- Data on 209 patients (88 abatacept, 121 TNFi) were collected.
- Abatacept patients were older than TNFi patients, however there were no significant differences in either gender or duration of treatment at the clinic. See Table 1.
- At 12 months, abatacept patients had significantly higher persistence than TNFi patients. Median time to discontinuation was 1,672 days for abatacept versus 612 days for TNFi. See Figure 2.
- In the Cox model, the risk of discontinuation was 77% higher in TNFi patients (Hazard Ratio 1.767, 95% Confidence Interval (CI) 1.113-2.806, p=0.0158). See Table 2.
- In the logistic regression, the odds of TNFi patients being persistent at 12 months was 52% lower than abatacept, although this difference was not statistically significant (Odds Ratio 0.485, 95% CI 0.208-1.133, p=0.0947). See Table 2.
- Reasons for discontinuation differed between cohorts, with more TNFi patients discontinuing due to disease progression and adverse effects of medication. See Table 1.

Figure 1. Study Design

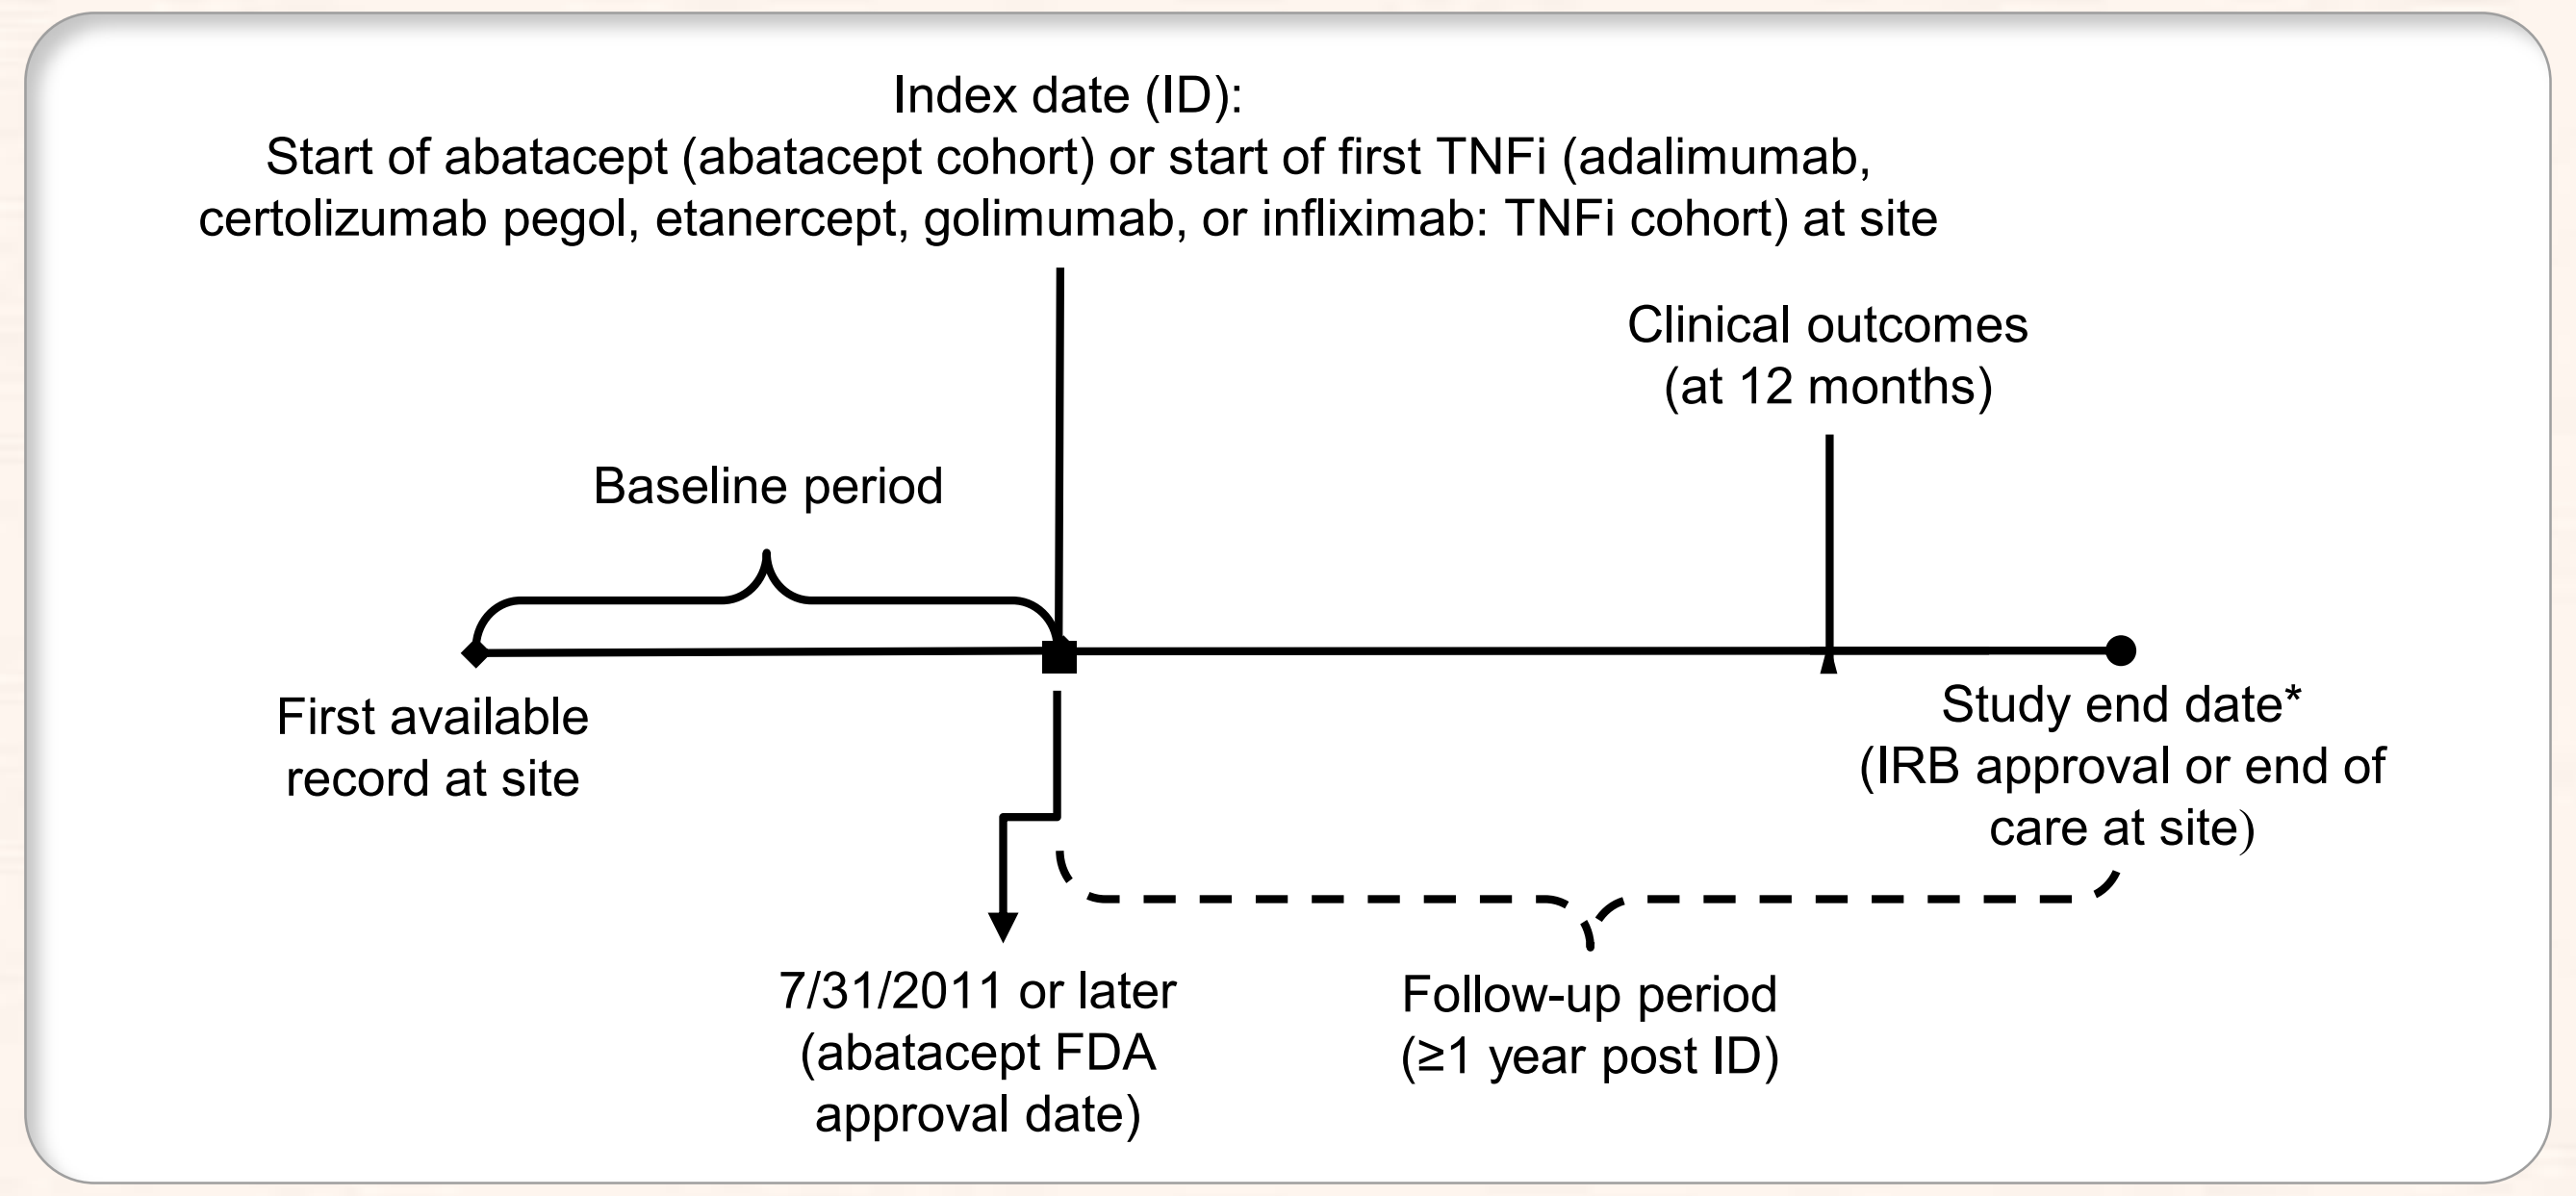
Index date (ID):
Start of abatacept (abatacept cohort) or start of first TNFi (adalimumab, certolizumab pegol, etanercept, golimumab, or infliximab: TNFi cohort) at site
Clinical outcomes (at 12 months)
Baseline period
First available record at site
Study end date* (IRB approval or end of care at site)
7/31/2011 or later (abatacept FDA approval date)
Follow-up period (≥1 year post ID)

*In a retrospective study, data usually cannot be collected after the IRB approval date.

References
1. Westhovens R, et al. *Ann Rheum Dis*. 2009;**68**(12):1870–7
2. Emery P, et al. *Rheumatol Oxf Engl*. 2008;**47**(4):392–8.

Table 1. Baseline Characteristics and Persistence

	Abatacept N=88	TNFi N=121	P-Value
Age in years, mean (SD)	64.36 (13.01)	57.23 (13.61)	<0.001
Female, n (%)	70 (79.55)	88 (72.73)	0.257
Charlson comorbidity index, mean (SD)	0.80 (1.07)	0.61 (0.92)	0.178
Duration of treatment at site (years), mean (SD)	5.79 (4.43)	3.67 (4.40)	0.226
Time from index (years), mean (SD)	3.19 (1.52)	3.14 (1.49)	0.808
Index drug with 12 months of persistence, n (%)	76 (86.36)	79 (65.29)	<0.001
Patients who discontinued index treatment any time post-index, n (%)	36 (40.91)	78 (64.46)	<0.001
Reason for discontinuation (among patients who discontinued index treatment), n (%)			
Disease progression (uncontrolled symptoms or on laboratory testing)	10 (27.78)	42 (53.85)	<0.001
Adverse effects of medication	1 (2.78)	9 (11.54)	
Insurance coverage	7 (19.44)	10 (12.82)	
Adherence issues	1 (2.78)	0 (0.0)	
Physician preference	1 (2.78)	0 (0.0)	
Patient preference	2 (5.56)	4 (5.13)	
Unknown/not specified	4 (11.11)	0 (0.0)	

RA=rheumatoid arthritis; TNFi=tumor necrosis factor inhibitor; SD=standard deviation

Table 2. Model Results

	Persistence at 12 months: OR (95% CI)	P-Value	Time to discontinuation: HR (95% CI)	P-Value
Age at index		0.9317		0.6620
Male vs. female	0.538 (0.245-1.184)	0.1235	1.095 (0.709-1.693)	0.6820
Charlson comorbidity index	1.079 (0.751-1.550)	0.6825	0.996 (0.814-1.220)	
Time from RA diagnosis to index (days)	1.000 (1.000-1.000)	0.9779	1.000 (1.000-1.000)	
No. physician office visits (1 year pre-index)	1.006 (0.885-1.144)	0.9227	0.985 (0.915-1.060)	
No. hospitalizations (1year pre-index)	1.246 (0.464-3.348)	0.6622	1.017 (0.559-1.850)	
TNFi vs. Orencia	0.485 (0.208-1.133)	0.0947	1.767 (1.113-2.806)	0.0158
Site		0.0056		0.1296
100 vs. 104	1.771 (0.455-6.887)		0.794 (0.368-1.771)	
101 vs. 104	0.956 (0.284-3.222)		0.856 (0.410-1.786)	
102 vs. 104	8.959 (1.972-40.702)		0.467 (0.221-0.991)	
103 vs. 104	1.992 (0.462-8.583)		0.859 (0.365-2.023)	

RA=rheumatoid arthritis; TNFi=tumor necrosis factor inhibitor; OR=odds ratio; CI=confidence interval; HR=hazard ratio

Conclusions

Limitations

- There were differences in persistence when stratified by site, which could be related to differences in sample sizes of abatacept and TNFi patients at the sites.
- Some patients were treated for RA prior to being treated at the clinic sites. It is possible these patients initiated targeted DMARD therapy at their previous clinic, and the index abatacept or TNFi was not first line therapy.

Conclusions

- In a real-world setting, RA patients with poor prognostic factors are significantly less likely to discontinue abatacept than discontinue TNFi.
- This difference may be explained by the lower proportion of patients discontinuing abatacept due to disease progression or adverse effects of medication.

Figure 2. Time to Discontinuation of Index Treatment

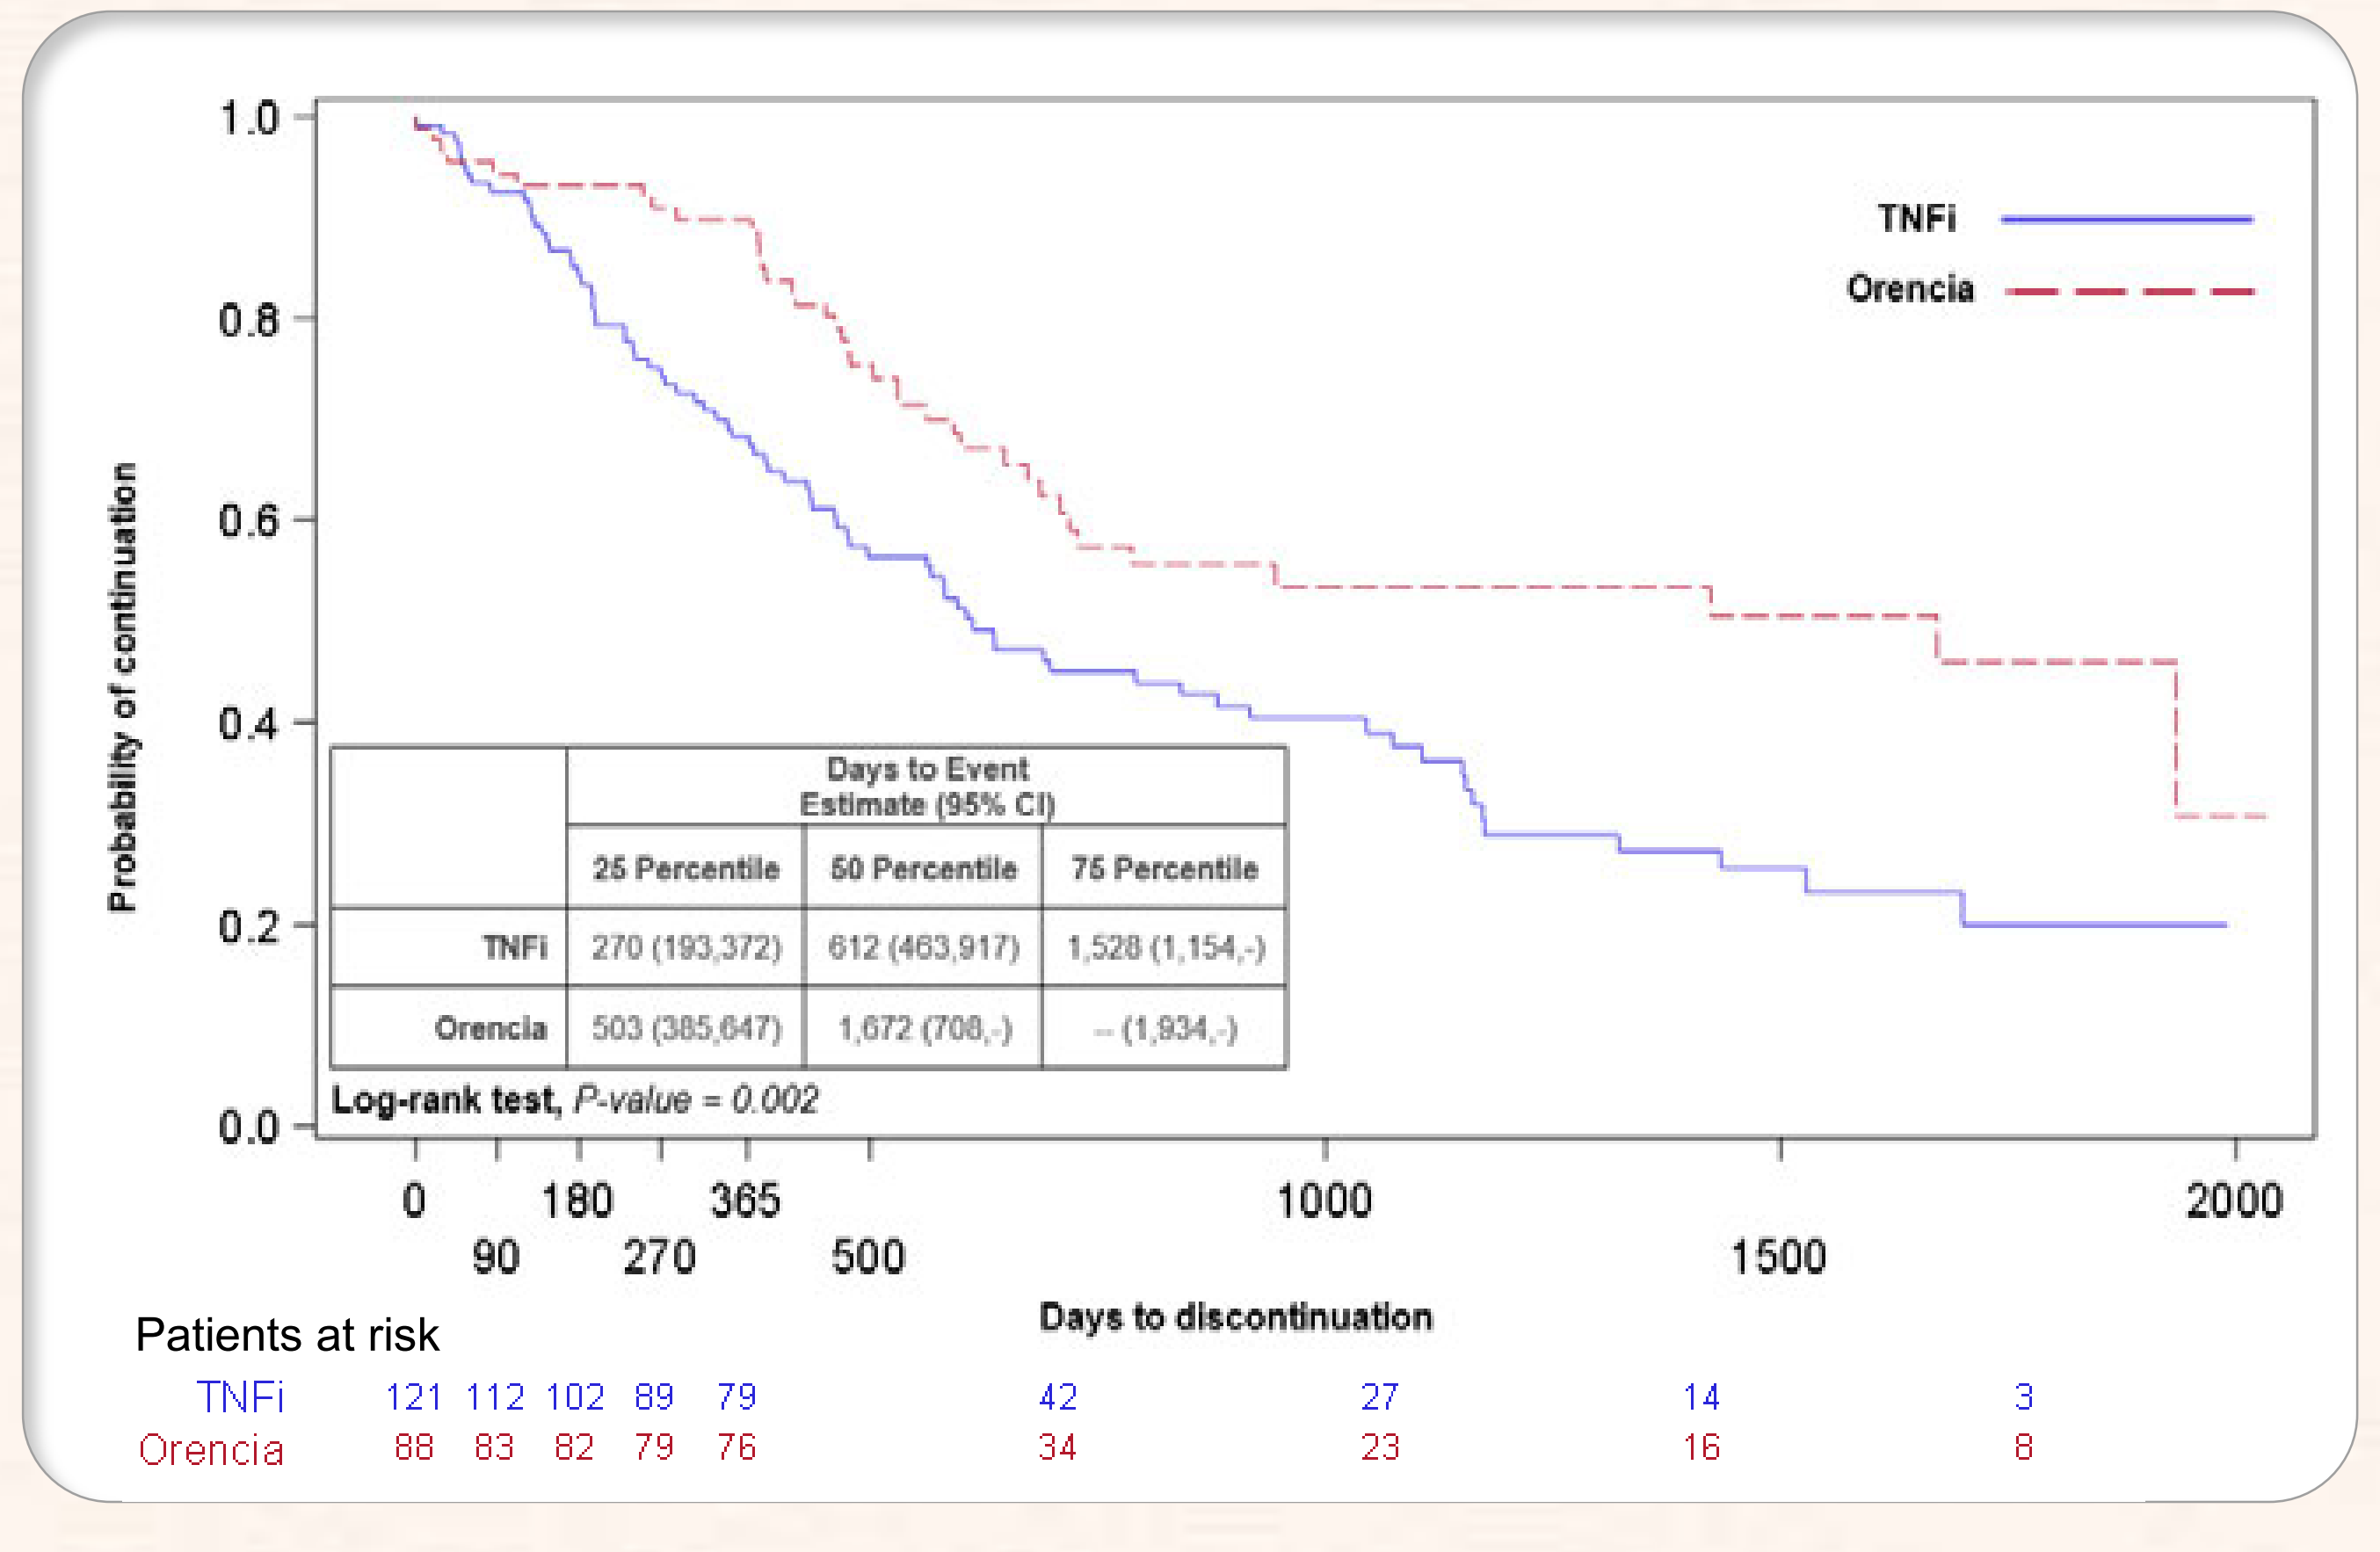
Probability of continuation
1.0
0.8
0.6
0.4
0.2
0.0
TNFi
Orencia
Days to Event Estimate (95% CI)
25 Percentile
50 Percentile
75 Percentile
TNFi 270 (193,372) 612 (463,917) 1,528 (1,154,-)
Orencia 503 (385,647) 1,672 (708,-) -- (1,934,-)
Log-rank test, P-value = 0.002
0 90 180 270 365 500 1000 1500 2000
Days to discontinuation
Patients at risk
TNFi 121 112 102 89 79 42 27 14 3
Orencia 88 83 82 79 76 34 23 16 8

Acknowledgements

This study was sponsored by Bristol-Myers Squibb.

Disclosures

D Paul is an employee of and shareholder in Bristol-Myers Squibb (BMS). **I Yermilov, SN Gibbs, and MS Broder** are employees of the Partnership for Health Analytic Research (PHAR) LLC, which was paid by Bristol-Myers Squibb (BMS) to conduct the research described in this poster.

EULAR Annual European Congress of Rheumatology • 12–15 June 2019 • Madrid, Spain