Budget Impact of Somatostatin Analogs (SSAs) as Treatment for Metastatic Gastroenteropancreatic Neuroendocrine Tumors (mGEP-NETs) in US Hospitals

Jesse D. Ortendahl[1]; Sonia J. Pulgar[2]; David Cox[2]; Tanya G.K. Bentley[1]; Alexandria T. Phan[3]

[1]Partnership for Health Analytic Research; [2]Ipsen Biopharmaceuticals; [3]Houston Methodist Hospital

BACKGROUND

- Gastroenteropancreatic neuroendocrine tumors (GEP-NETs) are rare neoplasms that originate in the secretory cells of the neuroendocrine system. Many of these tumors produce peptides and neuroamines, causing characteristic hormonal syndromes such as carcinoid syndrome.[1,2]
- Diagnosed NETs incidence is increasing, with US prevalence likely exceeding 100,000.[3,4]
- Initial systemic therapy for NETs often consists of treatment with a somatostatin analog (SSA) such as lanreotide depot (Somatuline) and octreotide LAR (Sandostatin).
- In the United States, only lanreotide depot is indicated in patients with GEP-NETs to improve progression free survival; octreotide LAR is approved for the treatment of severe diarrhea/ flushing episodes associated with metastatic carcinoid tumors. Both products are included in guidelines and are used frequently in practice.
- Trials have shown improved progression-free survival with both products when compared with placebo.[5,6]
- Real world costs of SSAs can be influenced by how they are used in clinical practice. Budget impact analysis (BIA) can be useful because it considers how changes in the mix of drugs and factors such as dosing, efficiency, and injection success can impact the trajectory of healthcare spending within a specific healthcare setting. This BIA is based on cost and dosing within the therapeutic class and does not evaluate efficacy or safety outcomes.[7]

OBJECTIVE

This model-based budget impact analysis estimates the financial impact on US hospitals of adopting and shifting the mix of somatostatin analogs (SSAs) utilized to treat GEP-NETs.

METHODS

Model Overview

Structure: Deterministic cohort model
Population: Patients with metastatic GEP-NETs
Perspective: US hospital
Time horizon: 1 year
Model inputs: Patients eligible for SSA treatment, product acquisition costs, preparation and mixing costs, product utilization
Outcome measures: Annual costs, costs per treated patient

Table 1. Eligible Patient Population

Parameter	Value	Source
GEP-NET Patients (N)	500	Assumption
Metastatic/Inoperable	80.0%	8
Treated with an SSA	78.2%	9
Final Model Patient Population (N)	**313**	

GEP-NET, gastroenteropancreatic neuroendocrine tumor; SSA, somatostatin analogue.

Table 2. Product Acquisition Costs[a]

Product	Acquisition Cost (per syringe)[10]
Lanreotide Depot	
60mg	$3,328
90mg	$4,434
120mg	$5,494
Octreotide LAR	
10mg	$2,380
20mg	$3,118
30mg	$4,670

[a] Prices reported as wholesale acquisition costs (WAC).

Model Structure

- Patients entered the model and received either lanreotide depot or octreotide LAR.
- The model assessed two scenarios:
 - Current Utilization, in which the market share reflects current treatment patterns;
 - Comparator Scenario, which reflects a hypothetical shift in utilization from octreotide LAR to lanreotide depot.
- The model population was based on a hypothetical hospital with 500 GEP-NET patients, assuming 80% had metastatic disease or were inoperable, and 78% were treated with an SSA (**Table 1**).
- Patients were assumed to be treated for the entirety of the 1-year period with 100% medication adherence.
- Dosing patterns and injection frequency of each product remain constant for the two scenarios.
- Patients incurred costs associated with product acquisition and administration (**Tables 2-3**).

Table 3. Product Preparation and Administration Costs

Product	Mixing/ Preparation Time (seconds)[11]	Wage Rate[12]	Cost (per syringe)
Lanreotide Depot	66	$82.27	$1.51
Octreotide LAR	329		$7.52

Table 4: Market Share by Product[a]

Product	Current Utilization	Comparison Scenario
Lanreotide Depot	5%	30%
Octreotide LAR	95%	70%

[a] Current utilization scenario defined as current market share based on market research data. Comparison scenario defined as a hypothetical shift in market share.

Table 5: Distribution of Dosing by Product[a]

Product	1x per 4 weeks	1x per 3 weeks	1x per 2 weeks
Lanreotide Depot			
60mg	0%	0%	0%
90mg	0%	0%	0%
120mg	100%	0%	0%
Octreotide LAR[13]			
10mg	0%	0%	0%
20mg	0%	0%	0%
30mg	64%	3%	7%
40mg	14%	6%	0%
60mg	4%	2%	0%

[a] Default estimates for lanreotide depot based on the labeled dosing regimen of 120 mg every four weeks. For octreotide LAR, proportion receiving dosing above indication based on a real-world study. All others assumed to get highest indicated dose.

Analyses

- **Base case:** Total hospital and per patient costs were estimated for the current utilization and the comparison scenario.
- **One-way sensitivity analyses:** All model parameters were independently varied by +/- 20%.

RESULTS

Base Case Results

- In the base case, lanreotide depot reduced per-patient costs compared with octreotide LAR ($71,442 vs. $75,508).
- For a hypothetical hospital with 500 GEP-NET patients, the annual cost of shifting from 5% to 30% lanreotide depot use resulted in cost savings of $317,977 ($23,555,246 vs. $23,237,269).
- In one-way sensitivity analyses, results were shown to be driven by product acquisition costs and proportion of octreotide LAR patients receiving above indicated dosing.

Table 6. Base Case Results[a,b]

	Annual Hospital/Institution Costs		Cost per Treated Patient		
	Current Utilization	Comparison Scenario	Product Acquisition	Preparation and Mixing	Total
Lanreotide depot	$1,117,347	$6,704,080	$71,422	$20	$71,442
Octreotide LAR	$22,437,899	$16,533,189	$75,400	$108	$75,508
Total	**$23,555,246**	**$23,237,269**	-	-	-
Difference[c]	-	**-$317,977**	-	-	-

[a] Current utilization defined as market share today, comparison scenario defined as a hypothetical change in market share.
[b] Costs include medications, administration, and mixing for initial injection.
[c] Difference reflects the change in total costs between the baseline and comparator year. A negative number denotes a cost savings in comparator year.

Figure 1. Total Hospital Costs

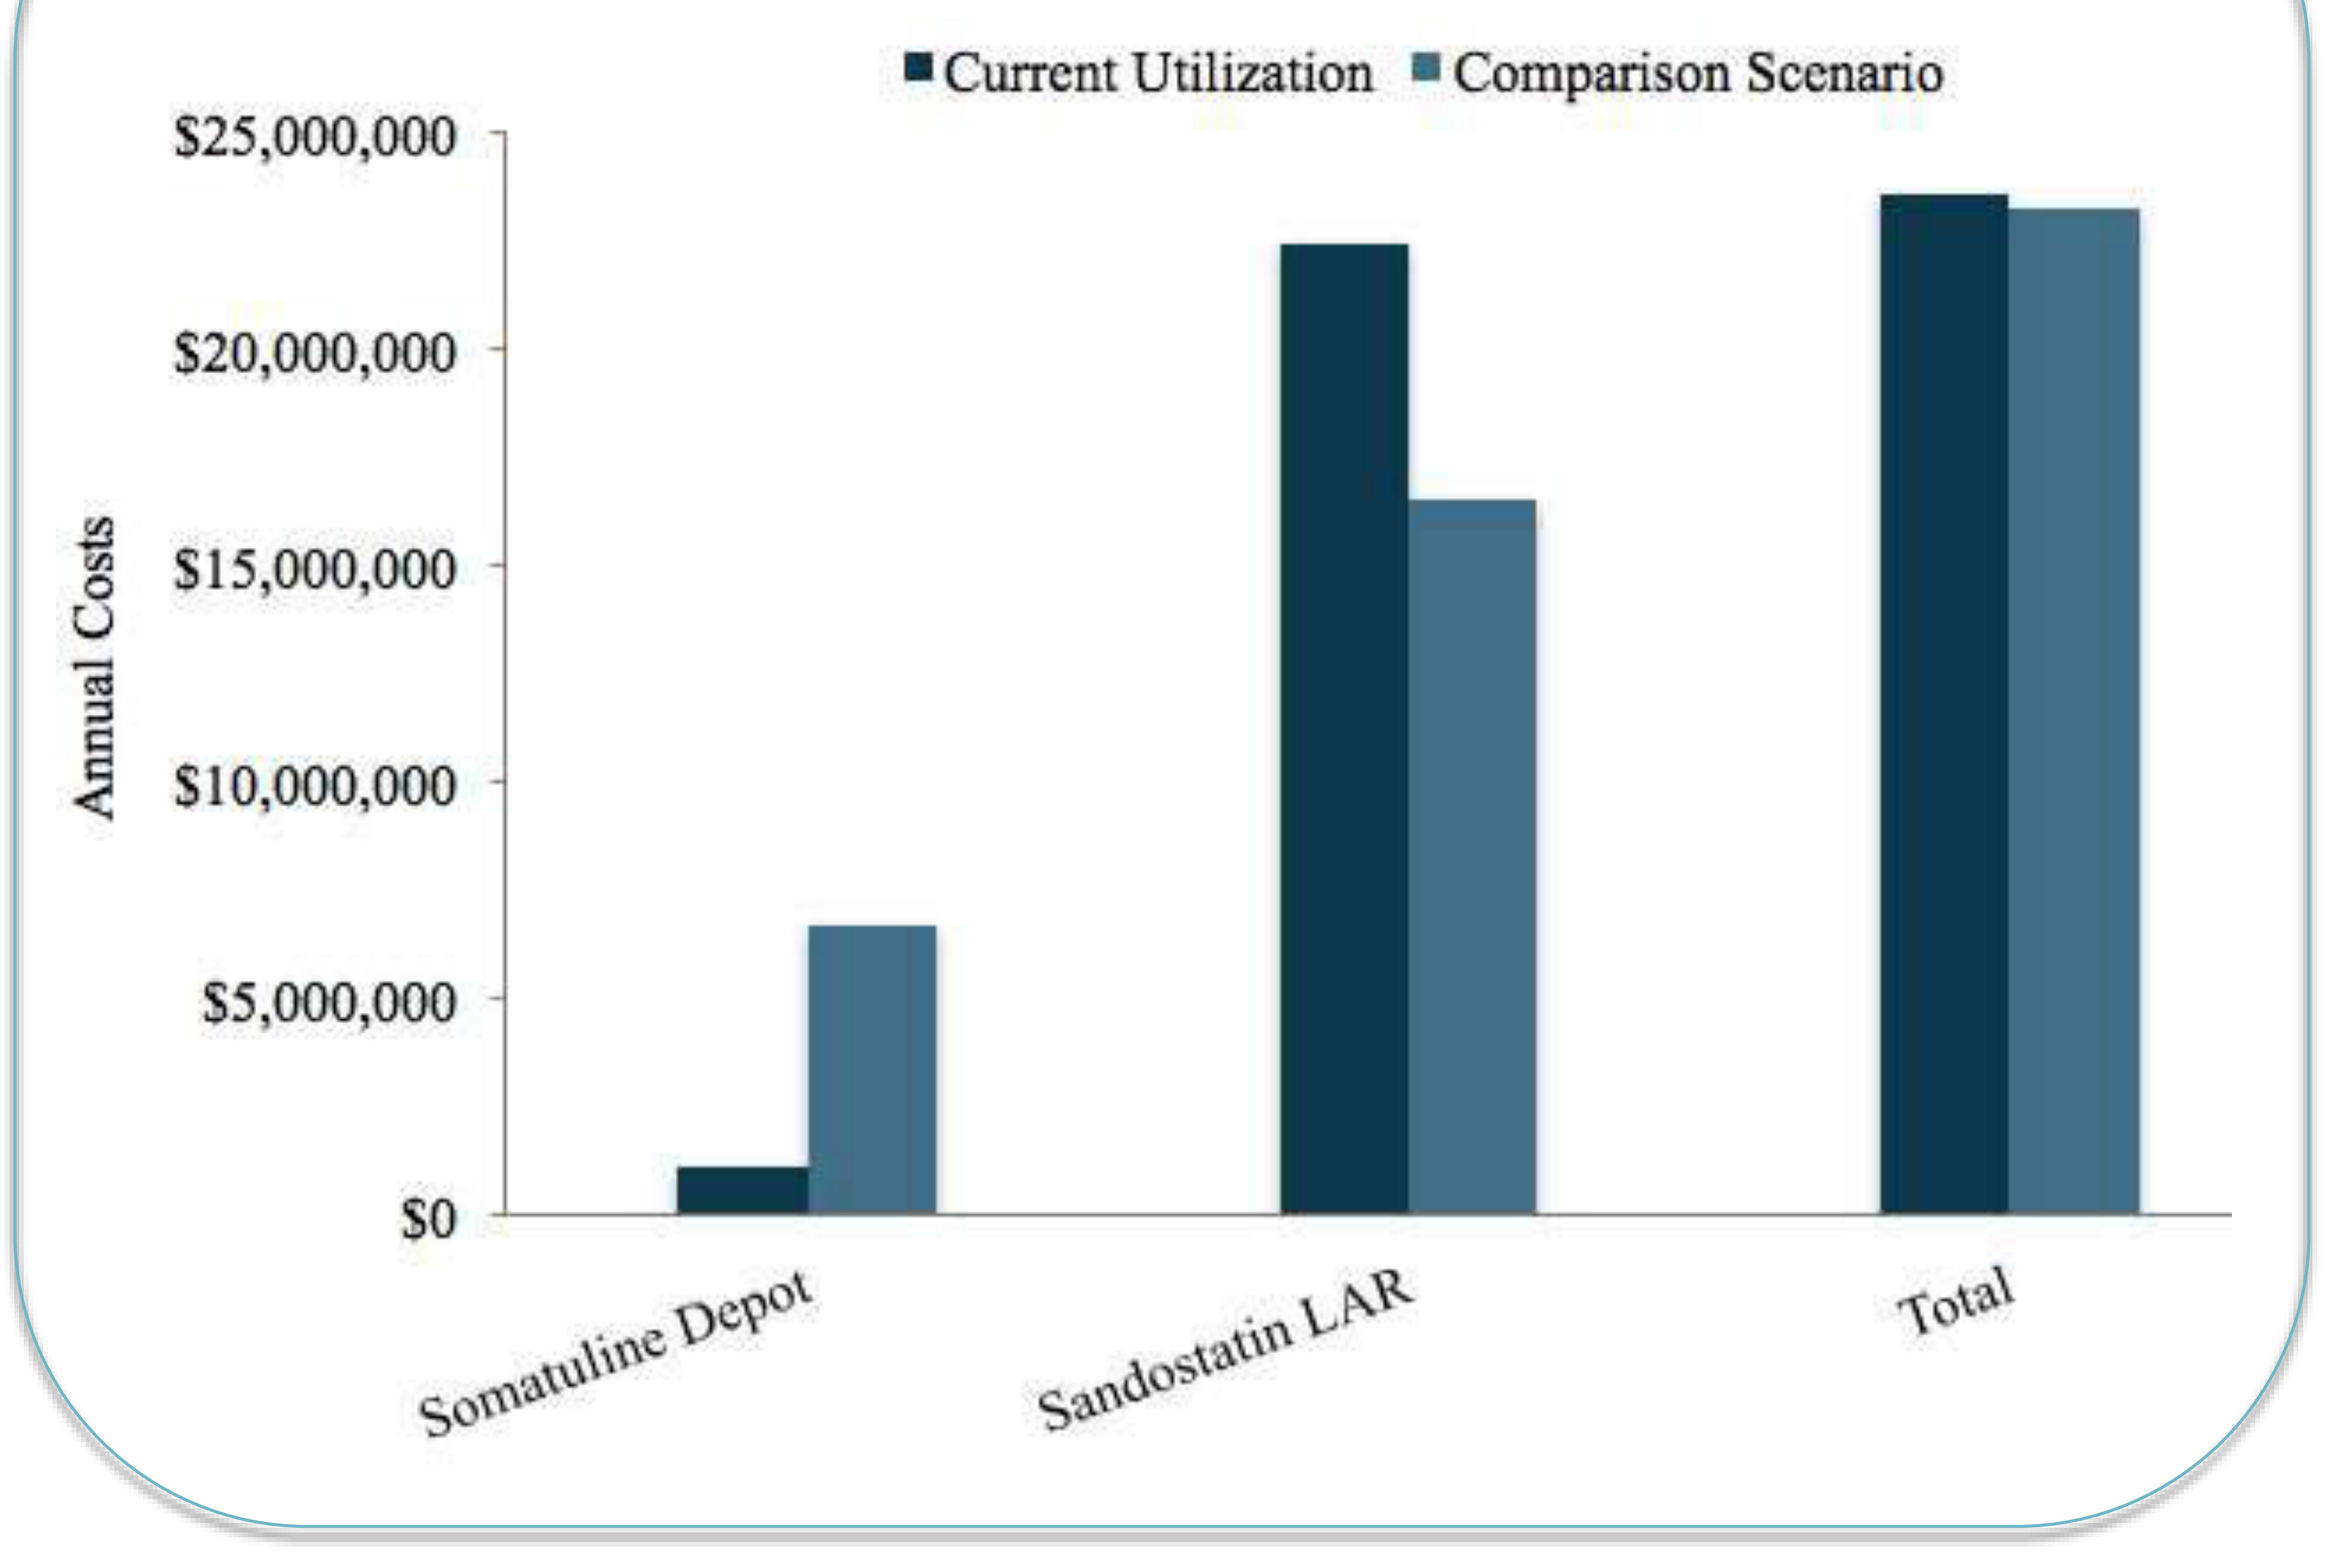
Current Utilization
Comparison Scenario
Annual Costs
$25,000,000
$20,000,000
$15,000,000
$10,000,000
$5,000,000
$0
Somatuline Depot
Sandostatin LAR
Total

Figure 2. Cost per Treated Patient

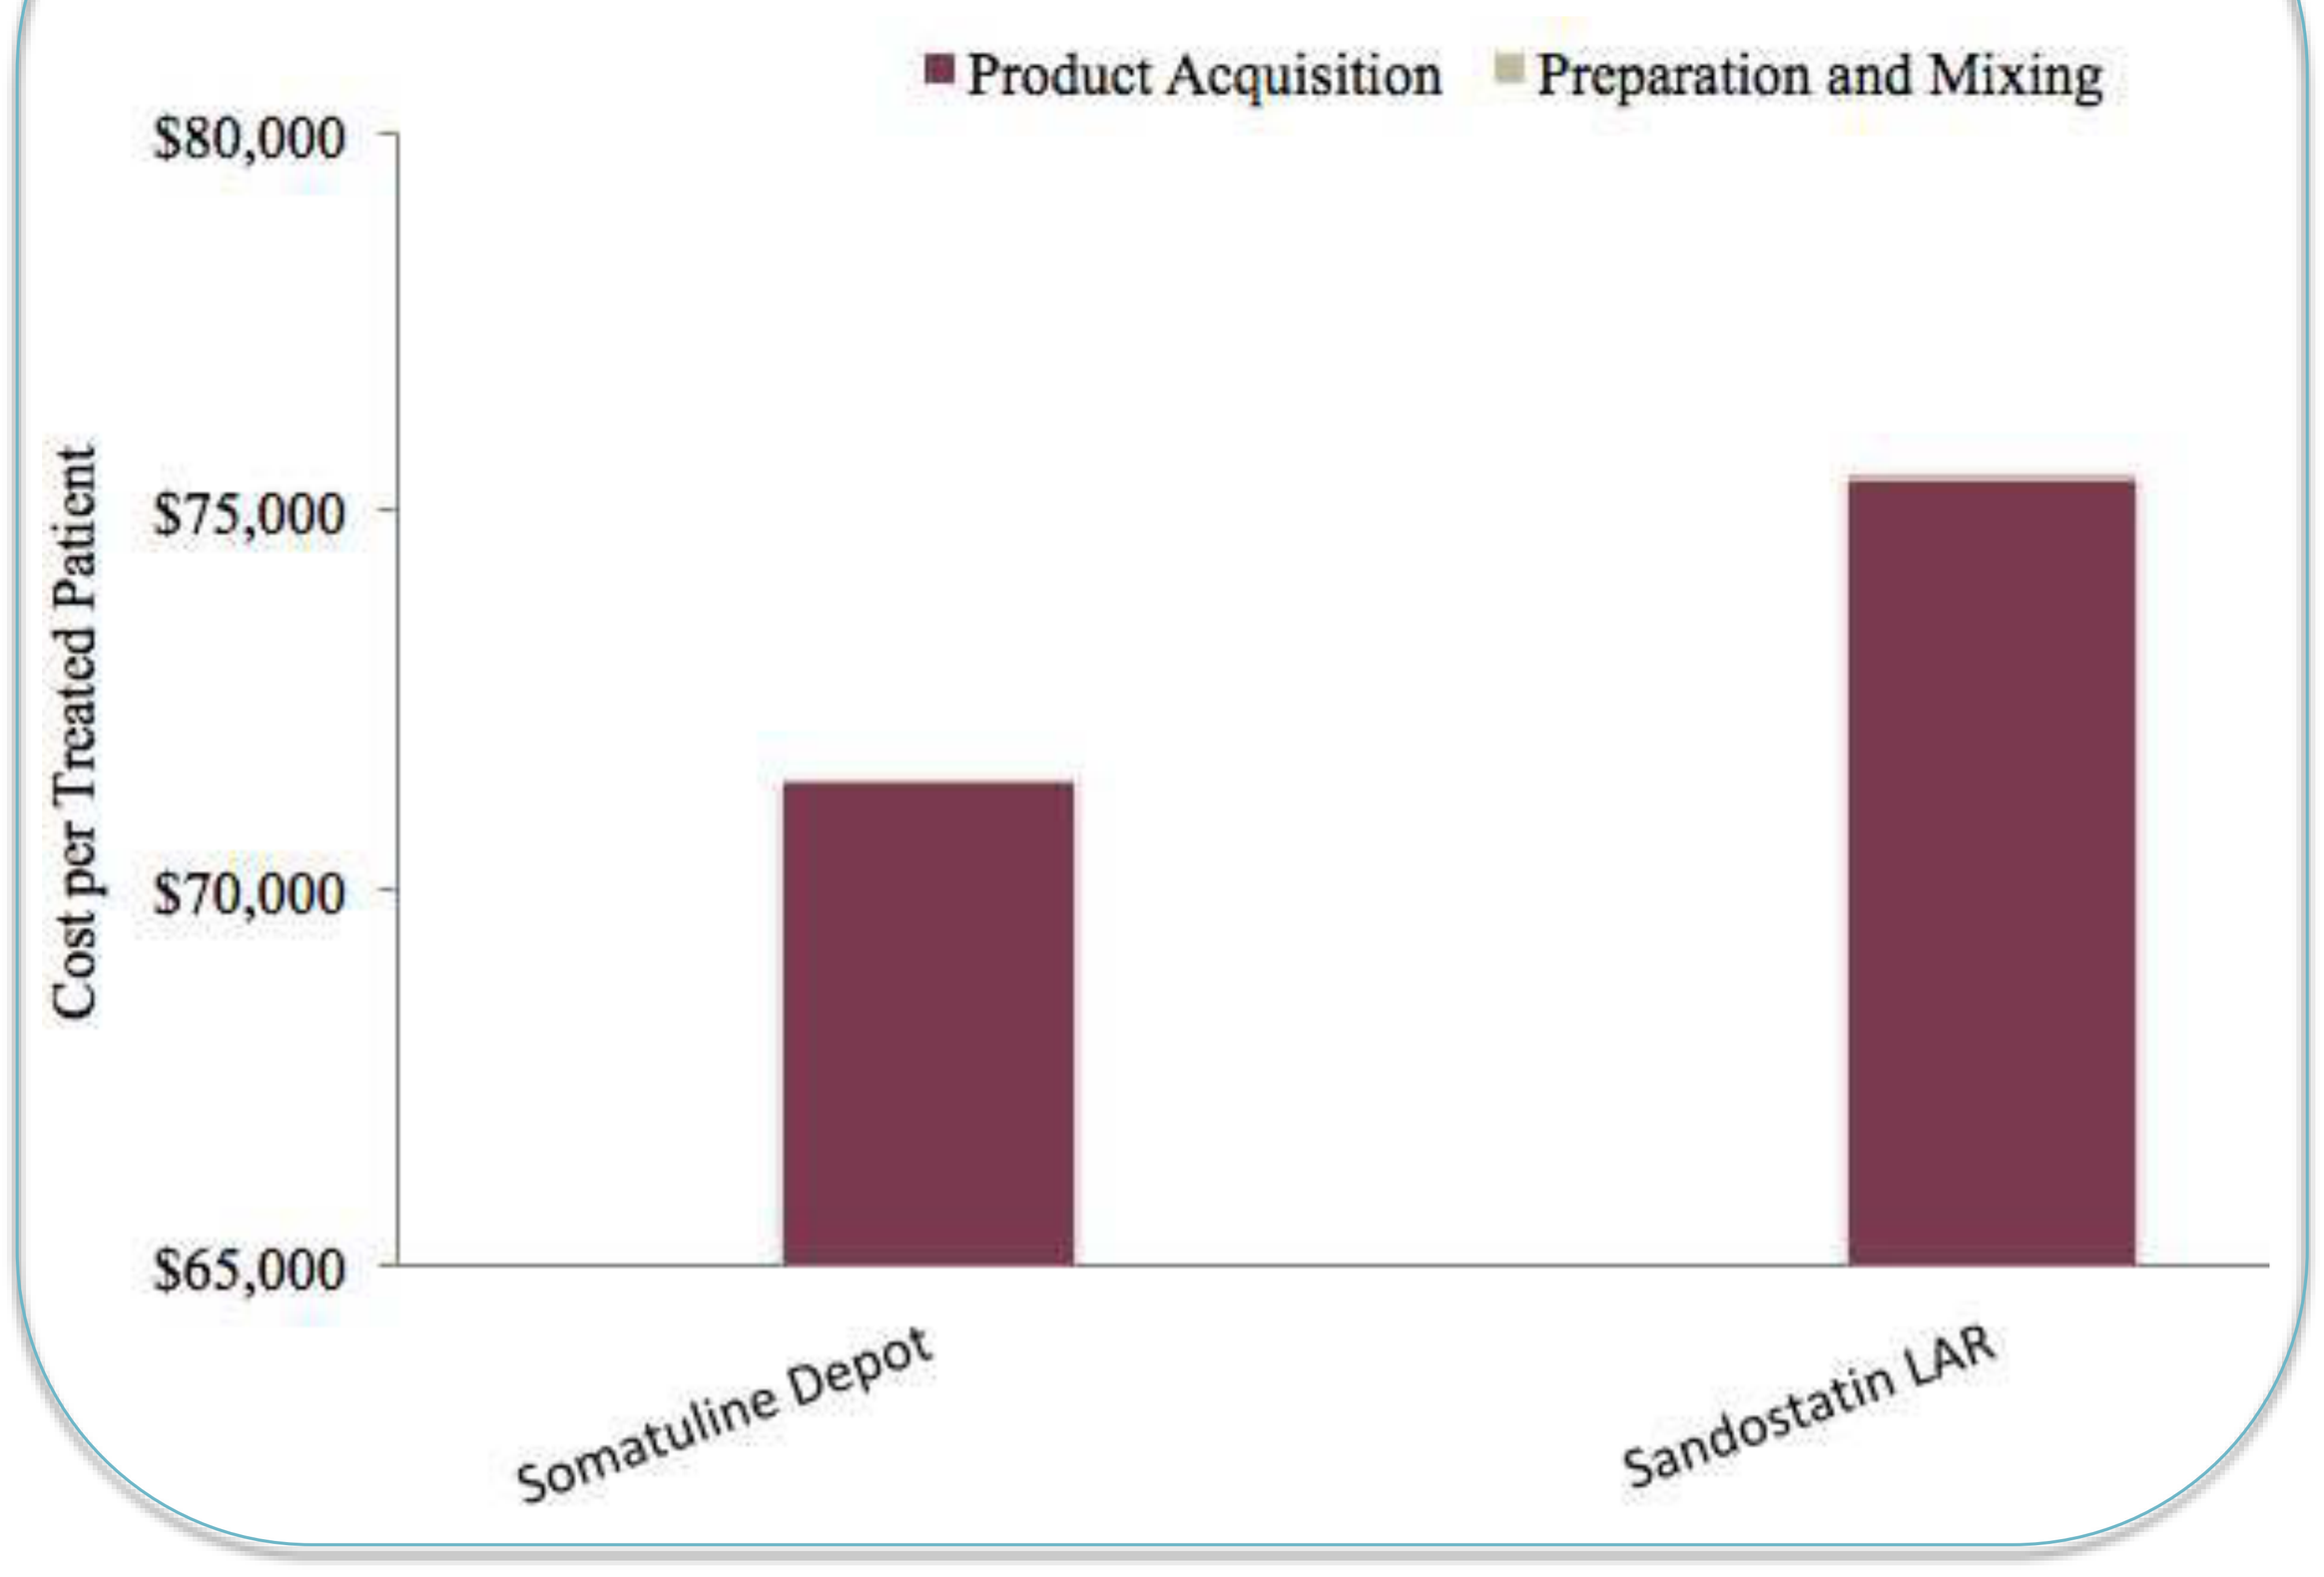
Product Acquisition
Preparation and Mixing
Cost per Treated Patient
$80,000
$75,000
$70,000
$65,000
Somatuline Depot
Sandostatin LAR

Figure 3. One-Way Sensitivity Analyses

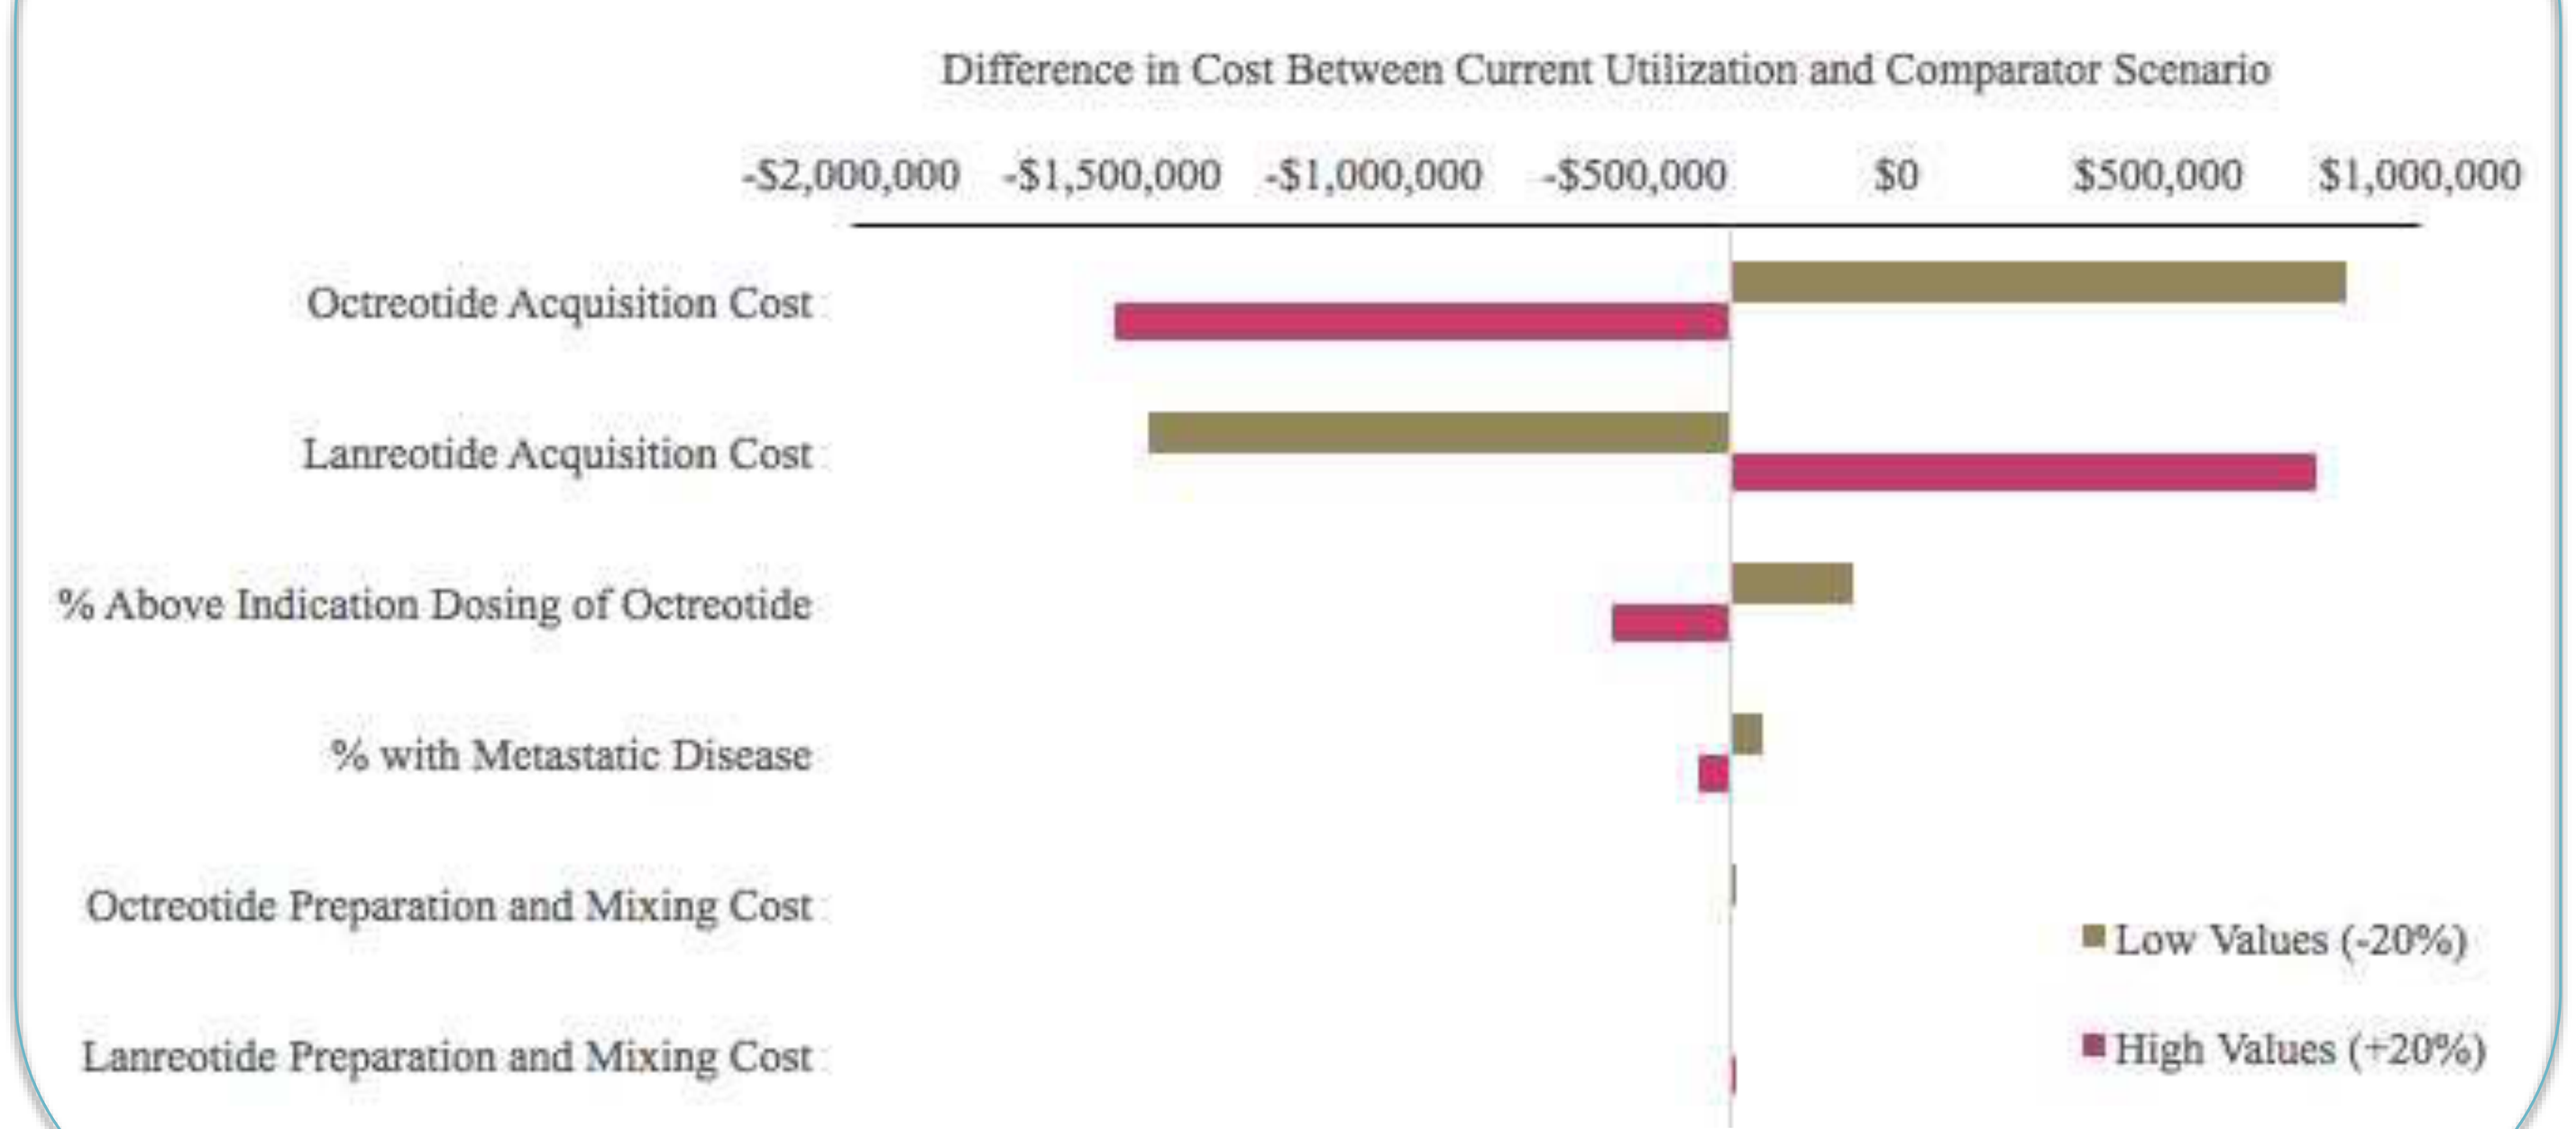
Difference in Cost Between Current Utilization and Comparator Scenario
-$2,000,000
-$1,500,000
-$1,000,000
-$500,000
$0
$500,000
$1,000,000
Octreotide Acquisition Cost
Lanreotide Acquisition Cost
% Above Indication Dosing of Octreotide
% with Metastatic Disease
Octreotide Preparation and Mixing Cost
Lanreotide Preparation and Mixing Cost
Low Values (-20%)
High Values (+20%)

One-way Sensitivity Analyses

Results were most sensitive to the acquisition costs for each product and the proportion of octreotide patients receiving above indicated dosing.

REFERENCES

1. Pearse J Histochem Cytochem 1969 **2.** Moertel J Clin Oncology 1987 **3.** Yao J Clin Oncology 2008 **4.** Halfdanarson Annals of Oncology 2008 **5.** Caplin New England J Medicine 2014 **6.** Rinke J Clin Oncology 2009 **7.** Mauskopf Value in Health 2007 **8.** Modlin Lancet Oncology 2008 **9.** Strosberg World J Gastroenterology 2013 **10**. PriceRx 2015 **11**. Adelman Med Devices Evidence Research 2012 **12**. US Bureau Labor Statistics 2015 **13**. Strosberg ASCO 2013 http://www.nccn.org/posters/2013/asco2.pdf.

Conclusions

- Results from this analysis suggest that factors beyond drug acquisition cost can influence the overall hospital budget impact with SSA treatment for GEP-NETs.
- Incorporating factors such as higher than indicated dosing of octreotide LAR observed in a real-world study and differential preparation and mixing costs, we found that increasing use of lanreotide depot at its indicated dosing results in cost-savings for the hospital.
- These results are based on the current treatment patterns of SSAs in the US; these results may change as more real world data becomes available, especially for lanreotide depot, which became available more recently in the US for management of NETs.

Presented at the 13th Annual ENETS Conference, March 9–11, 2016, Barcelona, Spain.

This study was funded by Ipsen and conducted by Partnership for Health Analytic Research, LLC.