HEALTHCARE UTILIZATION AND COST AMONG PATIENTS WITH HEREDITARY TRANSTHYRETIN AMYLOIDOSIS (hATTR)

Spencer Guthrie, MBA[1]; Sheila R. Reddy, PhD, RPh[2]; Ryan Tieu, MS[2]; Jennifer Munday, PhD, MPH[2]; Marian Tarbox, MPP[2]; Michael S. Broder, MD, MSHS[2]; Michael R. Pollock[1]

[1] Akcea Therapeutics, Cambridge, MA, USA; [2] Partnership for Health Analytic Research, LLC, Beverly Hills, CA, USA

OBJECTIVE

- hATTR is a rare genetic, progressive and fatal disease caused by build-up of misfolded transthyretin protein (amyloid) in organs and tissues.[1]
- Diagnosis and treatment of hATTR remains challenging.[2]
 - FDA approved therapies do not currently exist.
 - Organ transplantation, primarily liver transplantation, has limited use.
- The study's objective was to add to the limited data on the economic burden of amyloidosis by estimating healthcare resource utilization (HCRU) and costs for patients with hATTR.

METHODS

- Retrospective study using Truven Health Analytics MarketScan® Commercial and Medicare Supplemental databases and the IQVIA™ Real-World Data Adjudicated Claims - US databases from 1/1/2012-12/31/2016 (study period).
- Patient identification **(Figure 1)**
 - Patients ≥18 years at index who were newly diagnosed with hATTR
 - ≥1 medical claim with a relevant diagnosis code (ICD-9-CM 277.30-31, 277.39; ICD-10-CM E85.0-4, E85.82-89, E85.9) between 7/1/2012-9/30/2016 (ID period) AND ≥1 additional qualifying criteria occurring any time during study period: ≥15 days diflunisal use without >30-day gap, liver transplant, or claim with ICD-10-CM codes E85.1 or E85.2.
 - Study index: date of first claim in the ID period with a diagnosis code for amyloidosis.
 - To ensure a new diagnosis, patients with diagnosis code for any amyloidosis in 6 months pre-index (baseline period) were excluded.
- Patients enrolled during baseline period and followed ≥3 months post-index until enrollment or study end, whichever occurred first.
- Study measures
 - Baseline: age, gender, comorbidities, and year of diagnosis.
 - Outcome: HCRU and costs reported quarterly (every 3 months; Q1-Q4) during the first-year post-index.
 - Hospitalization, outpatient services (e.g., emergency department [ED] and physician office visits), therapeutic procedures and devices, and pharmacy utilization.
 - Total, inpatient, outpatient medical (ED and non-ED services), organ transplant-related, and outpatient pharmacy costs.
- Statistical analysis
 - Descriptive statistics, including means, standard deviations (SD), and relative frequencies and percentages for continuous and categorical data, respectively, were reported.
 - All data transformations and statistical analyses were performed using SAS© version 9.4.

Figure 1. Patient Identification

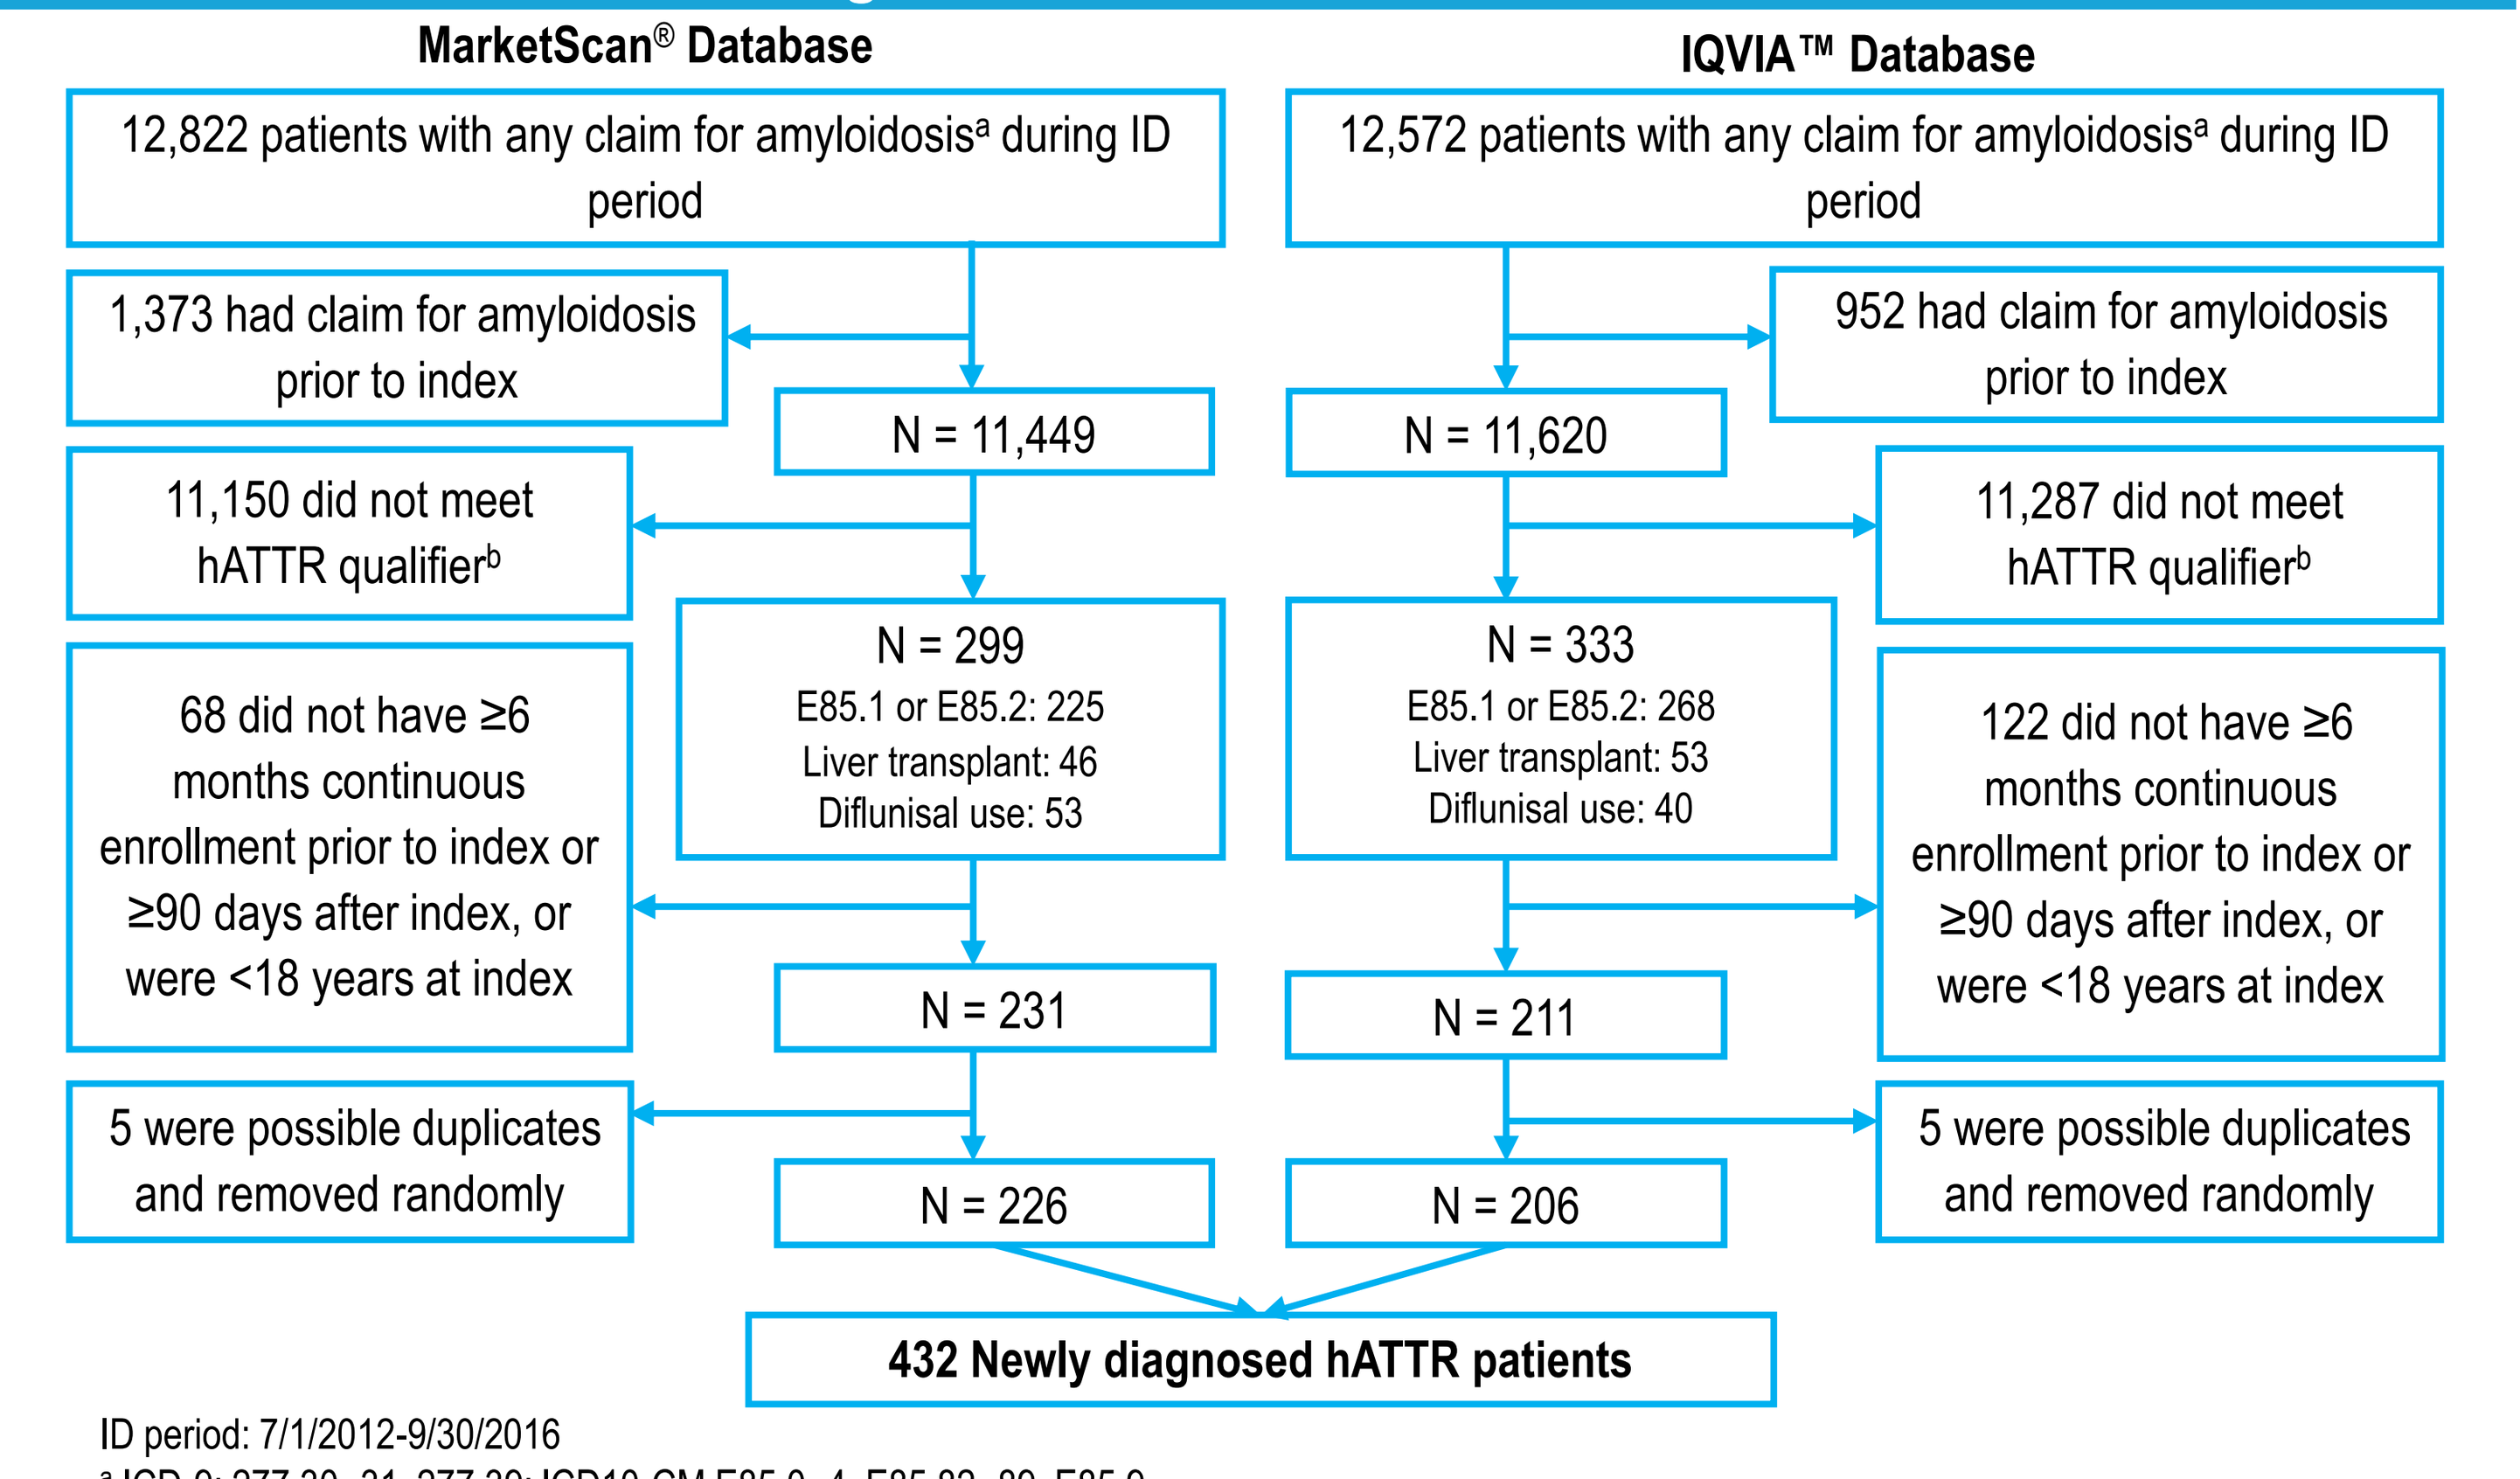
MarketScan® Database
IQVIA™ Database
12,822 patients with any claim for amyloidosis[a] during ID period
12,572 patients with any claim for amyloidosis[a] during ID period
1,373 had claim for amyloidosis prior to index
952 had claim for amyloidosis prior to index
N = 11,449
N = 11,620
11,150 did not meet hATTR qualifier[b]
11,287 did not meet hATTR qualifier[b]
N = 299
E85.1 or E85.2: 225
Liver transplant: 46
Diflunisal use: 53
N = 333
E85.1 or E85.2: 268
Liver transplant: 53
Diflunisal use: 40
68 did not have ≥6 months continuous enrollment prior to index or ≥90 days after index, or were <18 years at index
122 did not have ≥6 months continuous enrollment prior to index or ≥90 days after index, or were <18 years at index
N = 231
N = 211
5 were possible duplicates and removed randomly
5 were possible duplicates and removed randomly
N = 226
N = 206
432 Newly diagnosed hATTR patients

ID period: 7/1/2012-9/30/2016
[a] ICD-9: 277.30-.31, 277.39; ICD10-CM E85.0-.4, E85.82-.89, E85.9
[b] ICD-10-CM E85.1 or E85.2, ≥15 days of diflunisal use, OR had liver transplant

RESULTS

Baseline

- Among 432 qualifying newly diagnosed patients **(Figure 1)**, mean age was 57.5 (SD: 14.1), 52.5% were female, and baseline Charlson comorbidity index was 1.9 (2.5) **(Table 1).**
- By one year post-index, enrollment decreased to 179.

Table 1. Patient Characteristics

N	432
Age, mean (SD)	57.5 (14.1)
Sex (Female), n (%)	227 (52.5)
Year of diagnosis, n (%)	
2012	24 (5.6)
2013	33 (7.6)
2014	41 (9.5)
2015	129 (29.9)
2016	205 (47.5)
Charlson comorbidity index[a], mean (SD)	1.9 (2.5)

[a] Charlson comorbidity index was calculated during the 6 months prior to the hATTR diagnosis (baseline period).

- Selected baseline comorbidities are shown in **Figure 2.**
 - Cardiovascular-related comorbidities varied: dyspnea (27.3%), edema (17.1%), congestive heart failure (15.7%), ventricular hypertrophy (11.6%), and restrictive cardiomyopathy (10.0%); ischemic stroke, aortic stenosis, and hypertrophic cardiomyopathy occurred in <10% of patients.
 - Nearly a quarter of patients experienced diabetes (24.5%).
 - In nervous system-related comorbidities, 15.0% experienced neuropathy (4.6% peripheral), while fewer patients experienced cognitive impairment or incontinence (6.3% and 3.7%, respectively).
 - For gastrointestinal-related comorbidities, 11.1% experienced diarrhea and 10.4% constipation.
 - Fibromyalgia was the most common musculoskeletal-related comorbidity (8.3%), followed by spinal stenosis (7.2%), low bone mineral density (5.8%), and carpal tunnel syndrome (4.2%).
 - Less than 5% experienced ocular-related comorbidities (4.9% glaucoma, 3.9% blindness/loss of vision, and 2.5% vitreous opacity).

Healthcare utilization and costs

- In each quarter after hATTR diagnosis **(Table 2)**:
 - Proportion of patients hospitalized ranged from 19.4% (Q1) to 12.3% (Q4).
 - ED visits showed a similar range from 19.9% (Q1) to 12.8% (Q4).
 - Mean number of physician office visits by quarter were 3.9 (SD: 3.8), 2.9 (3.4), 2.9 (3.5), and 3.2 (3.6).
 - Mean prescription fills by quarter were 10.6 (9.6), 10.3 (10.5), 9.8 (9.6), and 9.6 (9.8).

RESULTS

Figure 2. Baseline Comorbidities

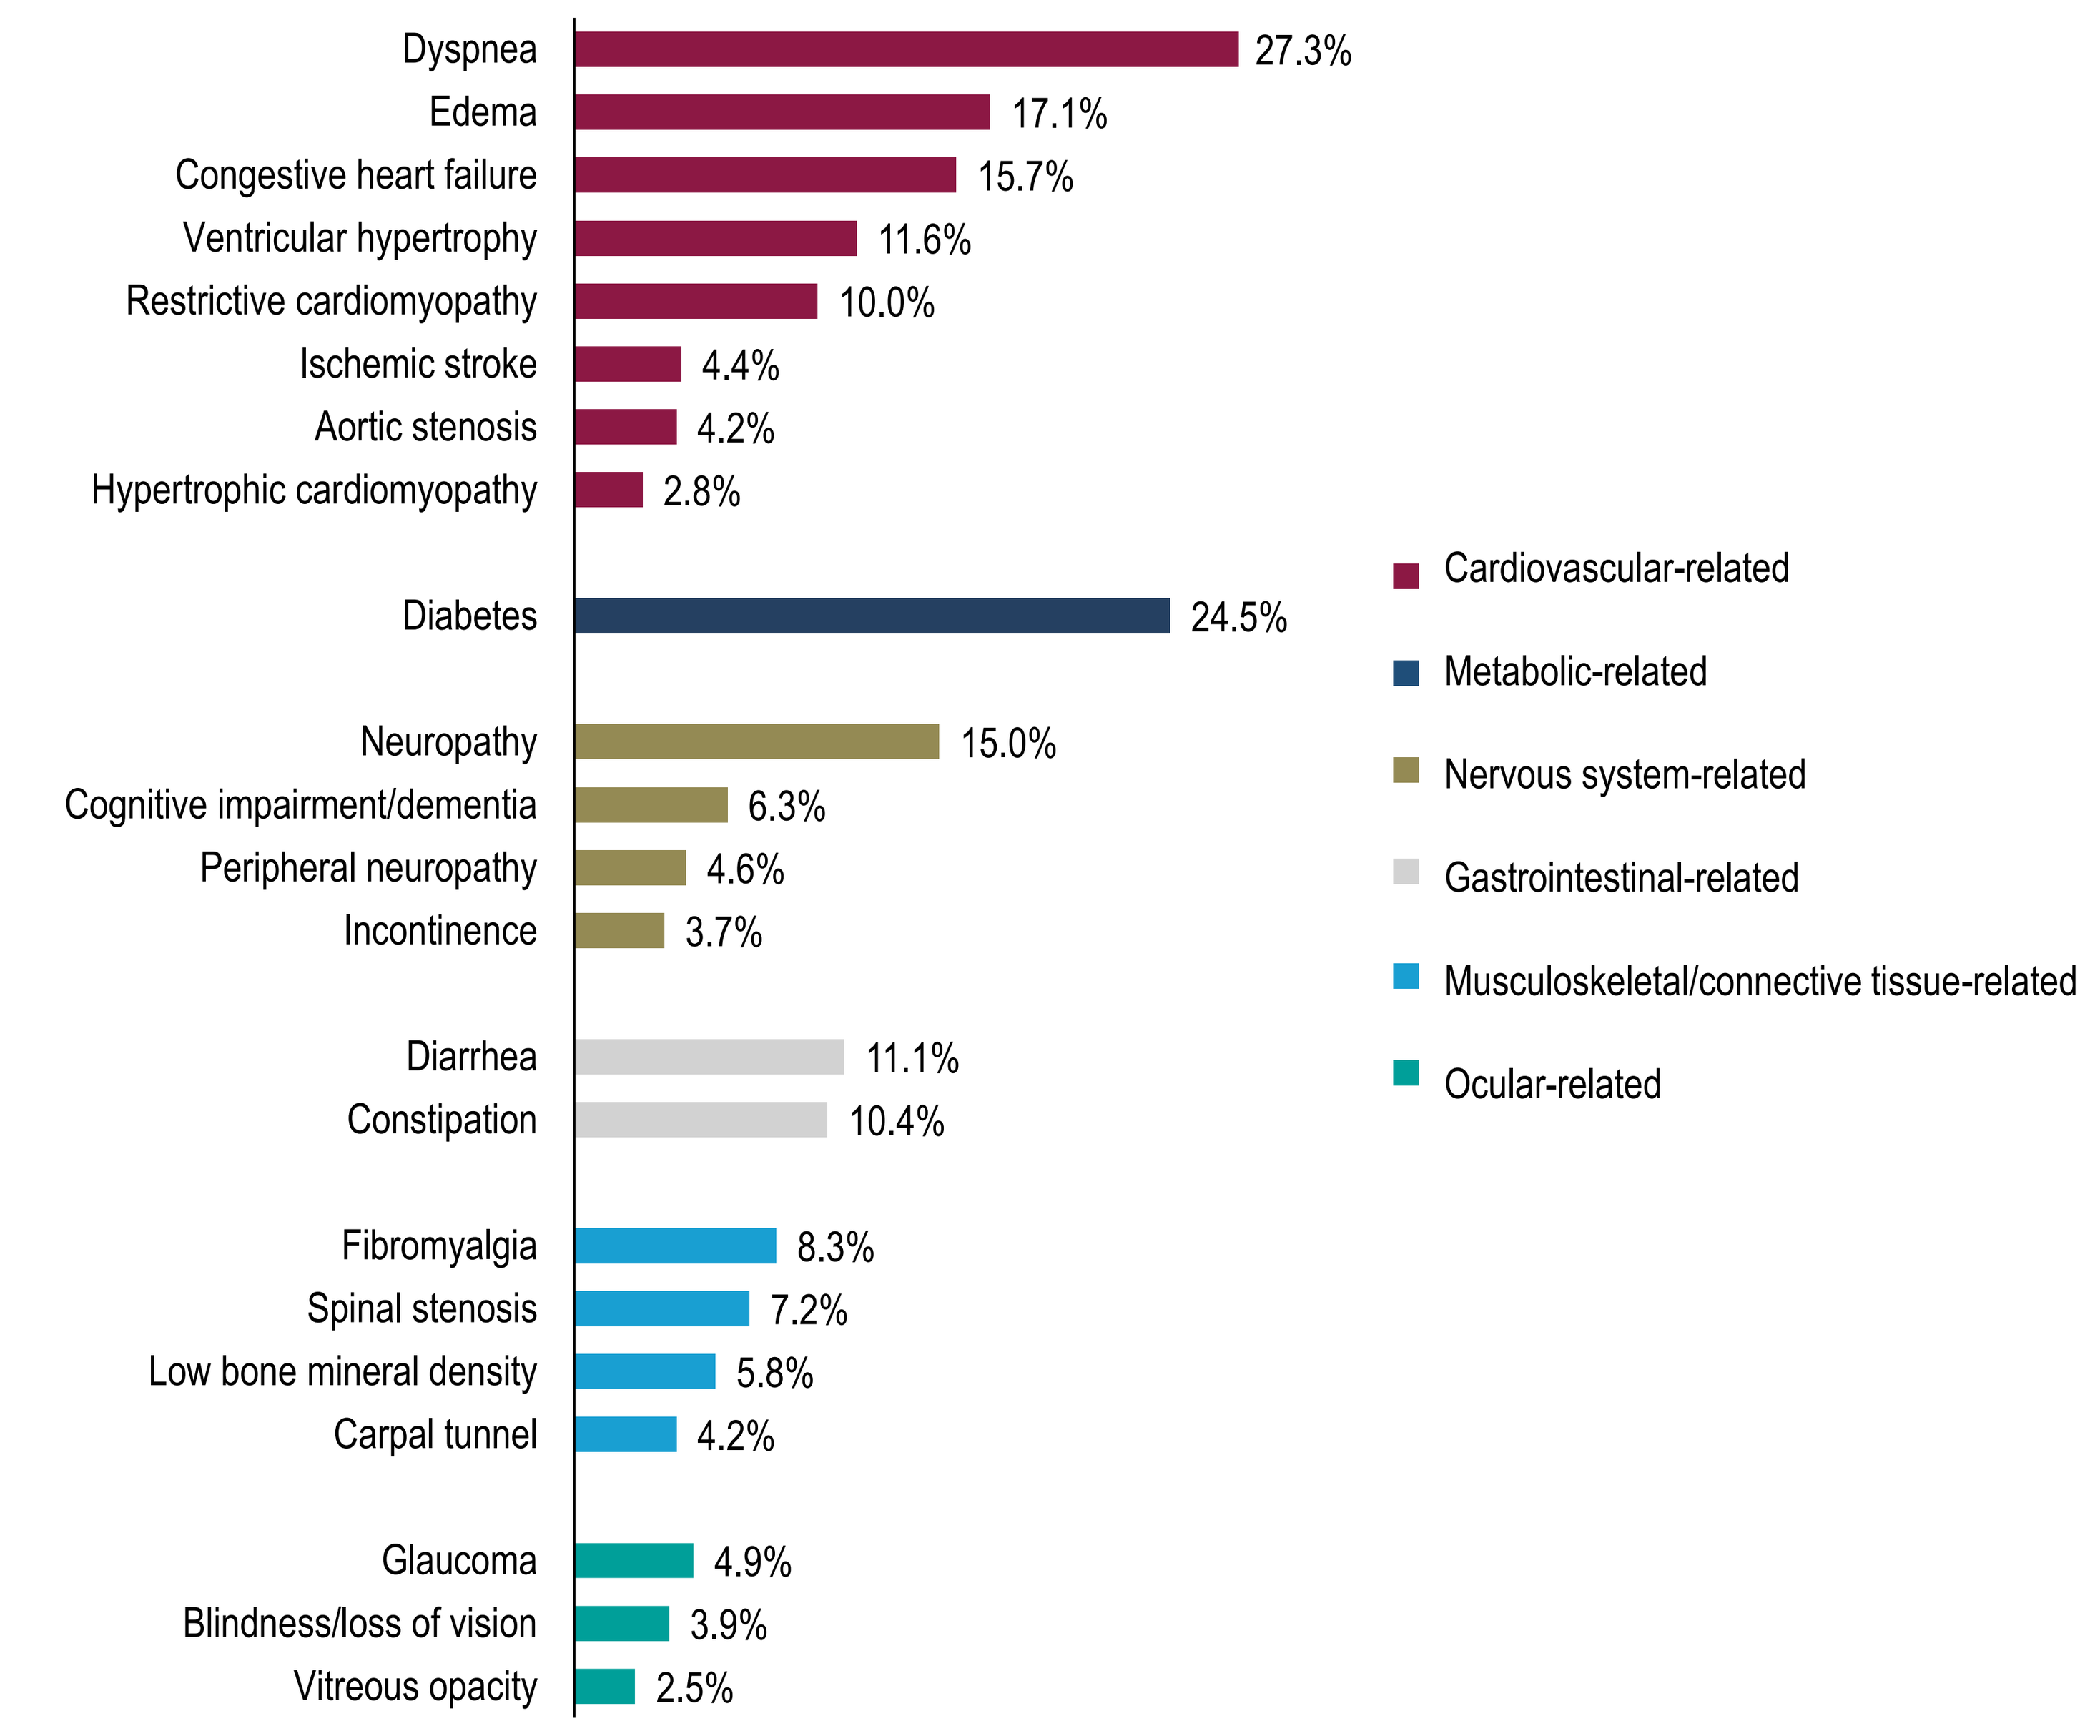
Dyspnea 27.3%
Edema 17.1%
Congestive heart failure 15.7%
Ventricular hypertrophy 11.6%
Restrictive cardiomyopathy 10.0%
Ischemic stroke 4.4%
Aortic stenosis 4.2%
Hypertrophic cardiomyopathy 2.8%
Diabetes 24.5%
Neuropathy 15.0%
Cognitive impairment/dementia 6.3%
Peripheral neuropathy 4.6%
Incontinence 3.7%
Diarrhea 11.1%
Constipation 10.4%
Fibromyalgia 8.3%
Spinal stenosis 7.2%
Low bone mineral density 5.8%
Carpal tunnel 4.2%
Glaucoma 4.9%
Blindness/loss of vision 3.9%
Vitreous opacity 2.5%
Cardiovascular-related
Metabolic-related
Nervous system-related
Gastrointestinal-related
Musculoskeletal/connective tissue-related
Ocular-related

- Proportions of patients receiving a liver transplant, dialysis, walkers/wheelchairs, and defibrillators/pacemakers were highest in Q1 after diagnosis and then varied in subsequent quarters. A very small percentage of patients received a stent in any quarter post-diagnosis **(Figure 3)**.
- Mean total costs by quarter were $19,387, $18,749, $12,785, and $14,623 **(Figure 4)**.
- Medical costs were highest in the first two quarters (Q1: $11,763 [inpatient] and $5,740 [outpatient]; Q2: $11,846 and $4,859), while pharmacy costs were highest in Q3 ($2,423)**.**

Table 2. Healthcare Utilization in the First Year after hATTR Diagnosis

	Follow-up period (days)			
	Q1 (1-90 days) N = 432	**Q2 (91-180 days) N = 349**	**Q3 (181-270 days) N = 272**	**Q4 (271-360 days) N = 179**
Patients with inpatient hospitalization	19.4%	12.3%	11.8%	12.3%
No. inpatient hospitalizations among hospitalized patients, mean (SD)	1.6 (1.0)	1.6 (1.0)	1.6 (1.1)	1.5 (0.6)
Length of stay among hospitalized patients, mean (SD)	10.6 (13.7)	16.1 (24.0)	13.5 (18.8)	16.1 (17.1)
Patients with ED visit	19.9%	14.9%	10.7%	12.8%
No. ED visits among ED patients, mean (SD)	1.5 (1.1)	1.6 (1.1)	1.6 (1.2)	1.3 (0.6)
Physician office visits, mean (SD)	3.9 (3.8)	2.9 (3.4)	2.9 (3.5)	3.2 (3.6)
Prescription fills, mean (SD)	10.6 (9.6)	10.3 (10.5)	9.8 (9.6)	9.6 (9.8)

Figure 3. Therapeutic Procedures and Devices in the First Year after hATTR Diagnosis

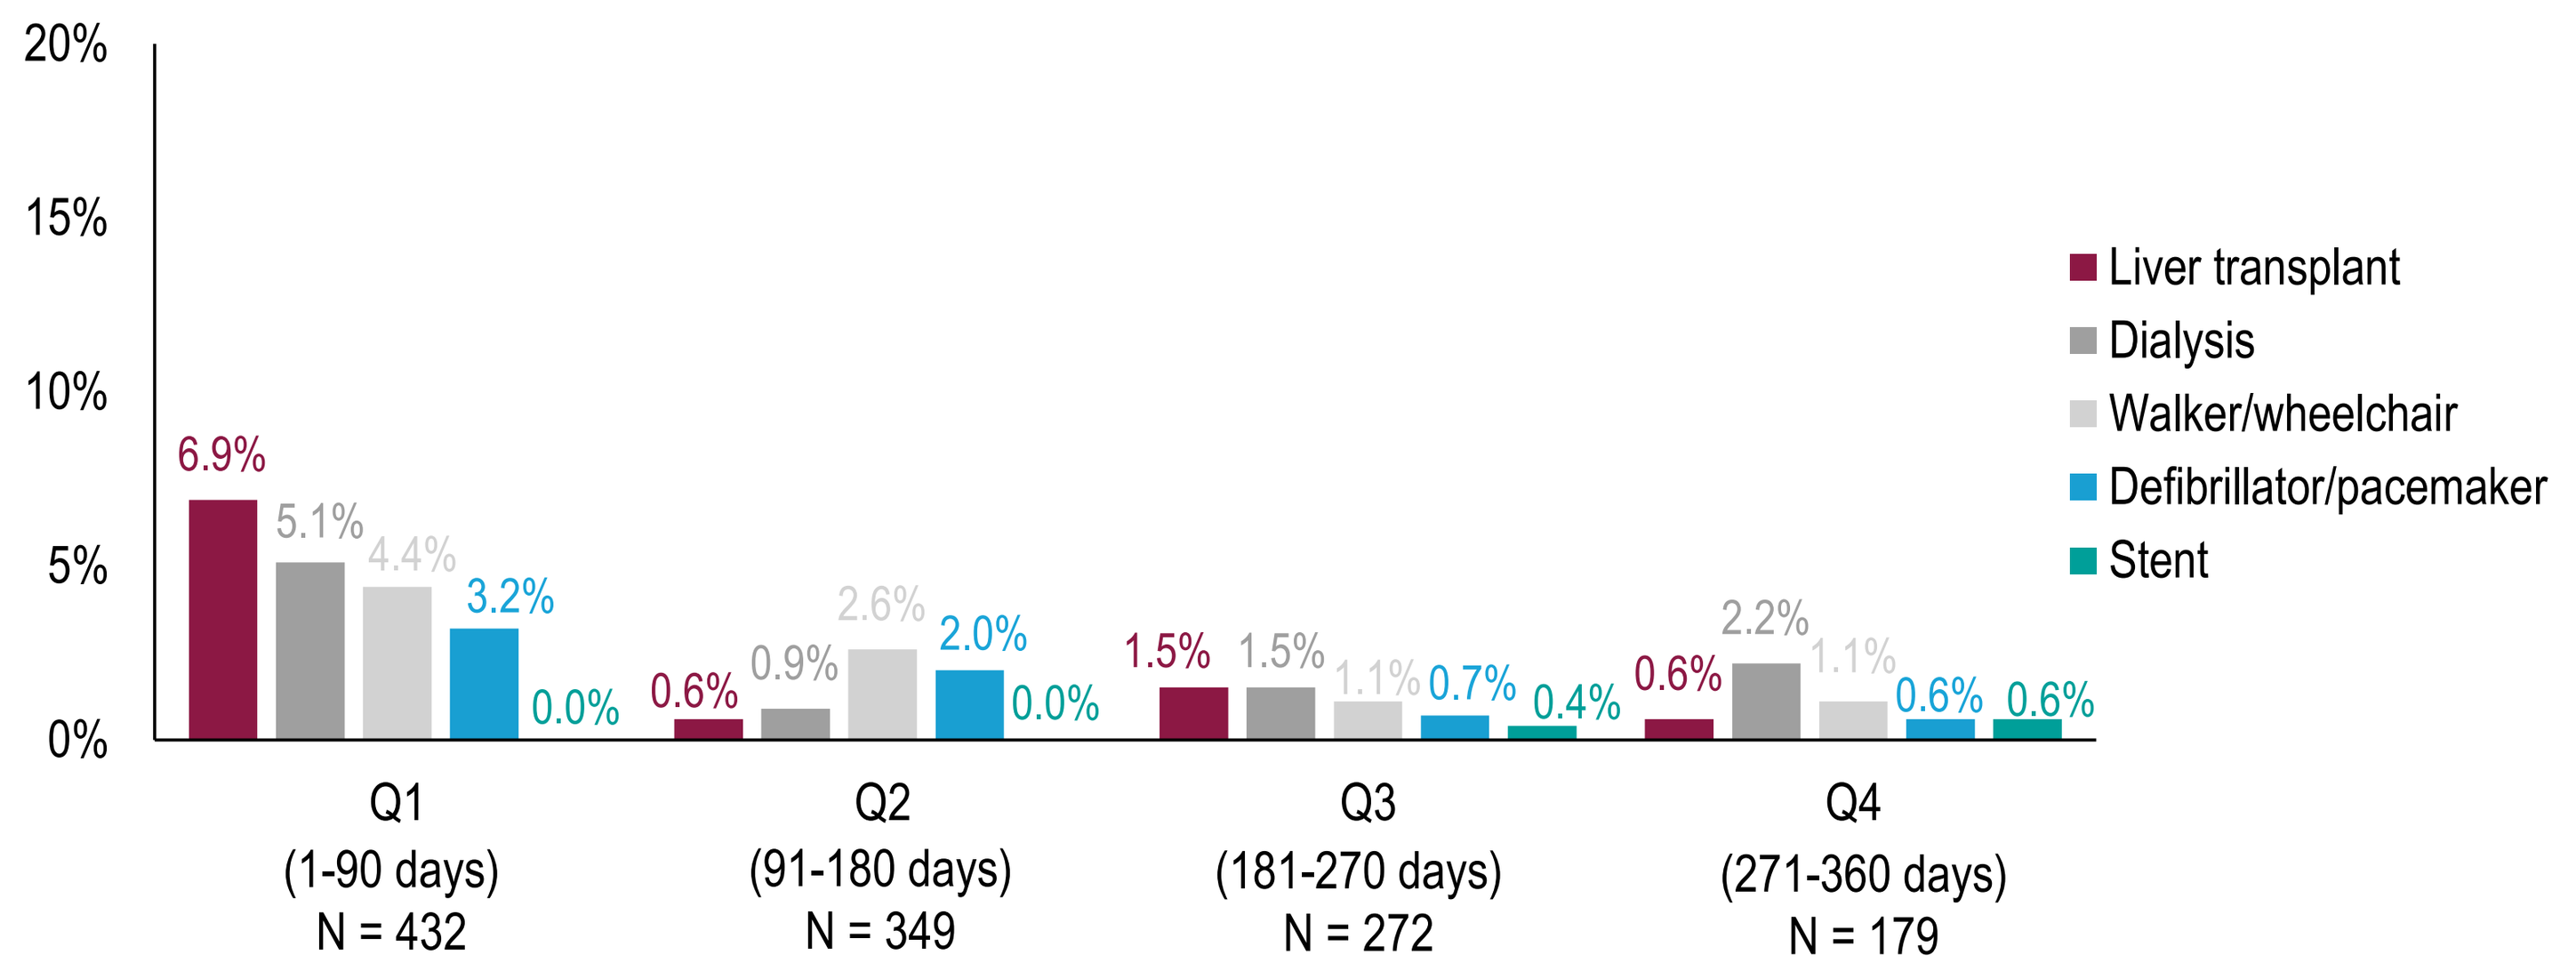
20%
15%
10%
5%
0%
6.9%
5.1%
4.4%
3.2%
0.0%
0.6%
0.9%
2.6%
2.0%
0.0%
1.5%
1.5%
1.1%
0.7%
0.4%
0.6%
2.2%
1.1%
0.6%
0.6%
Q1 (1-90 days) N = 432
Q2 (91-180 days) N = 349
Q3 (181-270 days) N = 272
Q4 (271-360 days) N = 179
Liver transplant
Dialysis
Walker/wheelchair
Defibrillator/pacemaker
Stent

Figure 4. Healthcare Costs in the First Year after hATTR Diagnosis

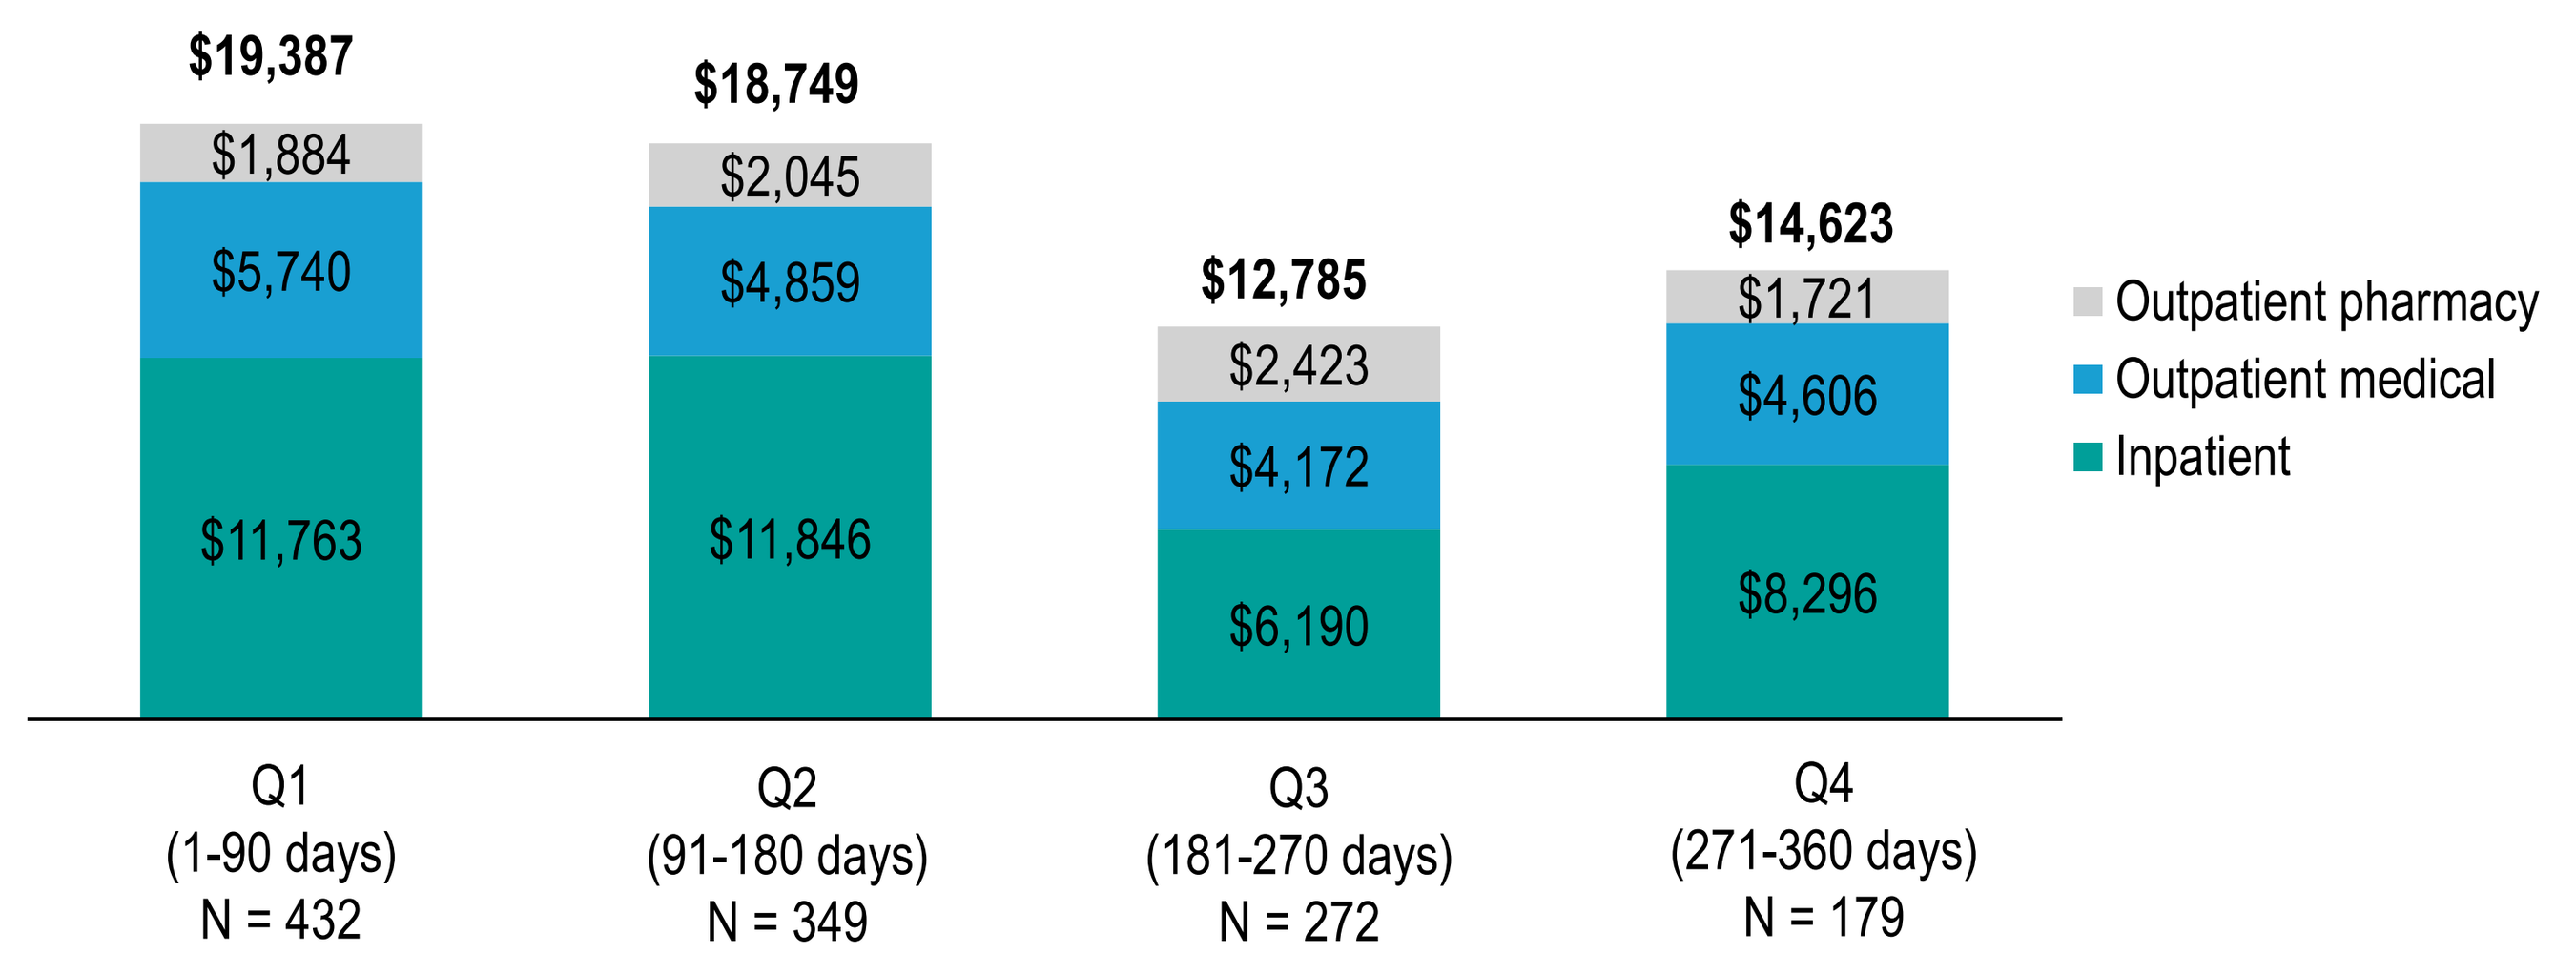
$19,387
$1,884
$5,740
$11,763
$18,749
$2,045
$4,859
$11,846
$12,785
$2,423
$4,172
$6,190
$14,623
$1,721
$4,606
$8,296
Q1 (1-90 days) N = 432
Q2 (91-180 days) N = 349
Q3 (181-270 days) N = 272
Q4 (271-360 days) N = 179
Outpatient pharmacy
Outpatient medical
Inpatient

CONCLUSION

- Patients newly diagnosed with hATTR have substantial HCRU and costs in the first year after diagnosis.
- As in common fatal diseases, progression in hATTR is associated with high costs. This may be particularly true in the terminal phase.
- This study has potential limitations:
 - Results for each quarter represent slightly different populations, however patient characteristics are similar in all samples.
 - Our study presents an incomplete assessment of the burden of hATTR, as we had no data on indirect costs (e.g., due to productivity losses or caregiver burden) or on quality of life.
 - Claims are meant for reimbursement, not research, so misclassification of hATTR is possible.

REFERENCES

1. Balhse E, Ybo A, Suhr O, et al. *J Pathol*. 2008;216(2):253–261.
2. Gertz M. *Am J Manag Care*. 2017;23:S107-S112.

Disclosure statement: S. Guthrie and M. Pollock are employees of Akcea Therapeutics. S.R. Reddy, R. Tieu, J. Munday, M. Tarbox, and M.S. Broder are employees of Partnership for Health Analytic Research, LLC.