Consensus guidance for management of nausea/vomiting in patients treated with zolbetuximab + chemotherapy: A RAND/UCLA modified Delphi panel study

Samuel J. Klempner[1], Stephanie Braun[2], Sarah N. Gibbs[3], Rupali Fuldeore[2]

[1]Department of Medicine, Division of Hematology-Oncology, Massachusetts General Cancer Center, Boston, MA, USA; [2]Astellas Pharma Global Development, Inc., Northbrook, IL, USA; [3]Partnership for Health Analytic Research (PHAR), LLC, Beverly Hills, CA, USA

BACKGROUND

- The phase 3 SPOTLIGHT (NCT03504397) and GLOW (NCT03653507) studies reported statistically significant improvement in PFS and OS with first-line zolbetuximab + chemotherapy (SPOTLIGHT: mFOLFOX6; GLOW: CAPOX) in patients with HER2-negative, LA unresectable or mG/GEJ adenocarcinoma whose tumors were CLDN18.2-positive[1,2]
- In phase 1–3 clinical studies, the most common TEAEs with zolbetuximab treatment were nausea and vomiting[1–8]
- In SPOTLIGHT and GLOW, approximately three-quarters of patients experienced nausea, and two-thirds of patients experienced vomiting[1,2,9]
 - The occurrences of nausea or vomiting were most common during the first zolbetuximab infusion and decreased thereafter
 - The median time to the first occurrence of nausea and/or vomiting was less than 1 hour after starting the first zolbetuximab infusion (48 minutes in SPOTLIGHT and 57 minutes in GLOW)
 - In SPOTLIGHT, the median time to the first occurrence of vomiting was shorter than that of nausea
- Nausea and vomiting led to higher rates of discontinuations in the zolbetuximab arms versus the placebo arms in both SPOTLIGHT and GLOW[1,2,9]
- Guidance for the prevention and management of nausea and vomiting based on clinical studies of zolbetuximab is limited

OBJECTIVE

- This study aims to develop consensus-based guidelines for the prevention and management of nausea and vomiting in patients treated with zolbetuximab + chemotherapy

METHODS

- The international RAND/UCLA modified Delphi panel included 15 expert panelists (Table 1) who were key opinion leaders in G/GEJ cancers and were involved in phase 2 or 3 clinical studies of zolbetuximab + chemotherapy
- The study schematic for the modified Delphi panel on the prevention and management of nausea and vomiting in patients treated with zolbetuximab + chemotherapy is as follows:

Develop a rating survey (questionnaire)
- Based on a literature review and panelist interviews, a rating form survey consisting of hypothetical patient scenarios and potential interventions was developed

Round 1
- Panelists completed anonymized, individual ratings of appropriateness of each intervention in each scenario using a scale of 1 (highly inappropriate, risks outweigh benefits) to 9 (highly appropriate, benefits outweigh risks)

Panel meeting
- The panel reviewed results of the first-round survey and shared perspectives during the professionally moderated panel meeting

Round 2
- Panelists repeated ratings following the meeting

Develop guidance
- Second-round survey ratings were analyzed using the RAND/UCLA Appropriateness Method to determine consensus guidance
- The median and range of all panelist ratings were calculated, and ratings were categorized as appropriate, uncertain, or inappropriate (Table 2)

Table 1. Expert Panelists Who Participated in the Modified Delphi Panel

Name	Affiliation	City	Country
Samuel Klempner, MD	Massachusetts General Hospital	Boston	USA
Robert Pazo-Cid, MD	Miguel Servet University Hospital	Zaragoza	Spain
Sara Lonardi, MD	Veneto Institute of Oncology	Padua	Italy
Leslie Swanson, ARNP	Fred Hutch Cancer Center	Seattle	USA
Matthew Arango, PharmD	The Ohio State University James Comprehensive Cancer Center	Columbus	USA
Peter Enzinger, MD	Dana-Farber Cancer Institute	Boston	USA
Andrew Ko, MD	UCSF Helen Diller Family Comprehensive Cancer Center	San Francisco	USA
Gina Vaccaro, MD	Tennessee Oncology	Lebanon	USA
Kensei Yamaguchi, MD	Cancer Institute Hospital of Japanese Foundation for Cancer Research	Tokyo	Japan
Anwaar Saeed, MD	University of Pittsburgh Medical Center	Pittsburgh	USA
Keun-Wook Lee, MD	Seoul National University Bundang Hospital	Seongnam	South Korea
Kohei Shitara, MD	National Cancer Center Hospital East	Kashiwa	Japan
David Ilson, MD, PhD	Memorial Sloan Kettering Cancer Center	New York	USA
Jaffer Ajani, MD	MD Anderson Cancer Center	Houston	USA
Manish Shah, MD	Weill Cornell Medicine	New York	USA

Table 2. Analysis of Rating Scores for the RAND/UCLA Modified Delphi Panel

Agreement	Median score of 7–9	Experts agree that the approach is **appropriate**
	Median score of 4–6	Experts agree that the approach **may or may not be appropriate**
	Median score of 1–3	Experts agree that the approach is **inappropriate**
Disagreement	≥ 3 ratings of 1–3 and ≥ 3 ratings of 7–9	**No conclusions** can be made

RESULTS

- Experts reviewed 382 scenarios, reaching agreement in 85% (n = 324) of the scenarios for Round 2
- The flow chart for prevention and management of nausea and vomiting is shown in Figure 1

Figure 1. Consensus Guidance on the Prevention and Management of Nausea and Vomiting in Patients Treated With Zolbetuximab + Chemotherapy

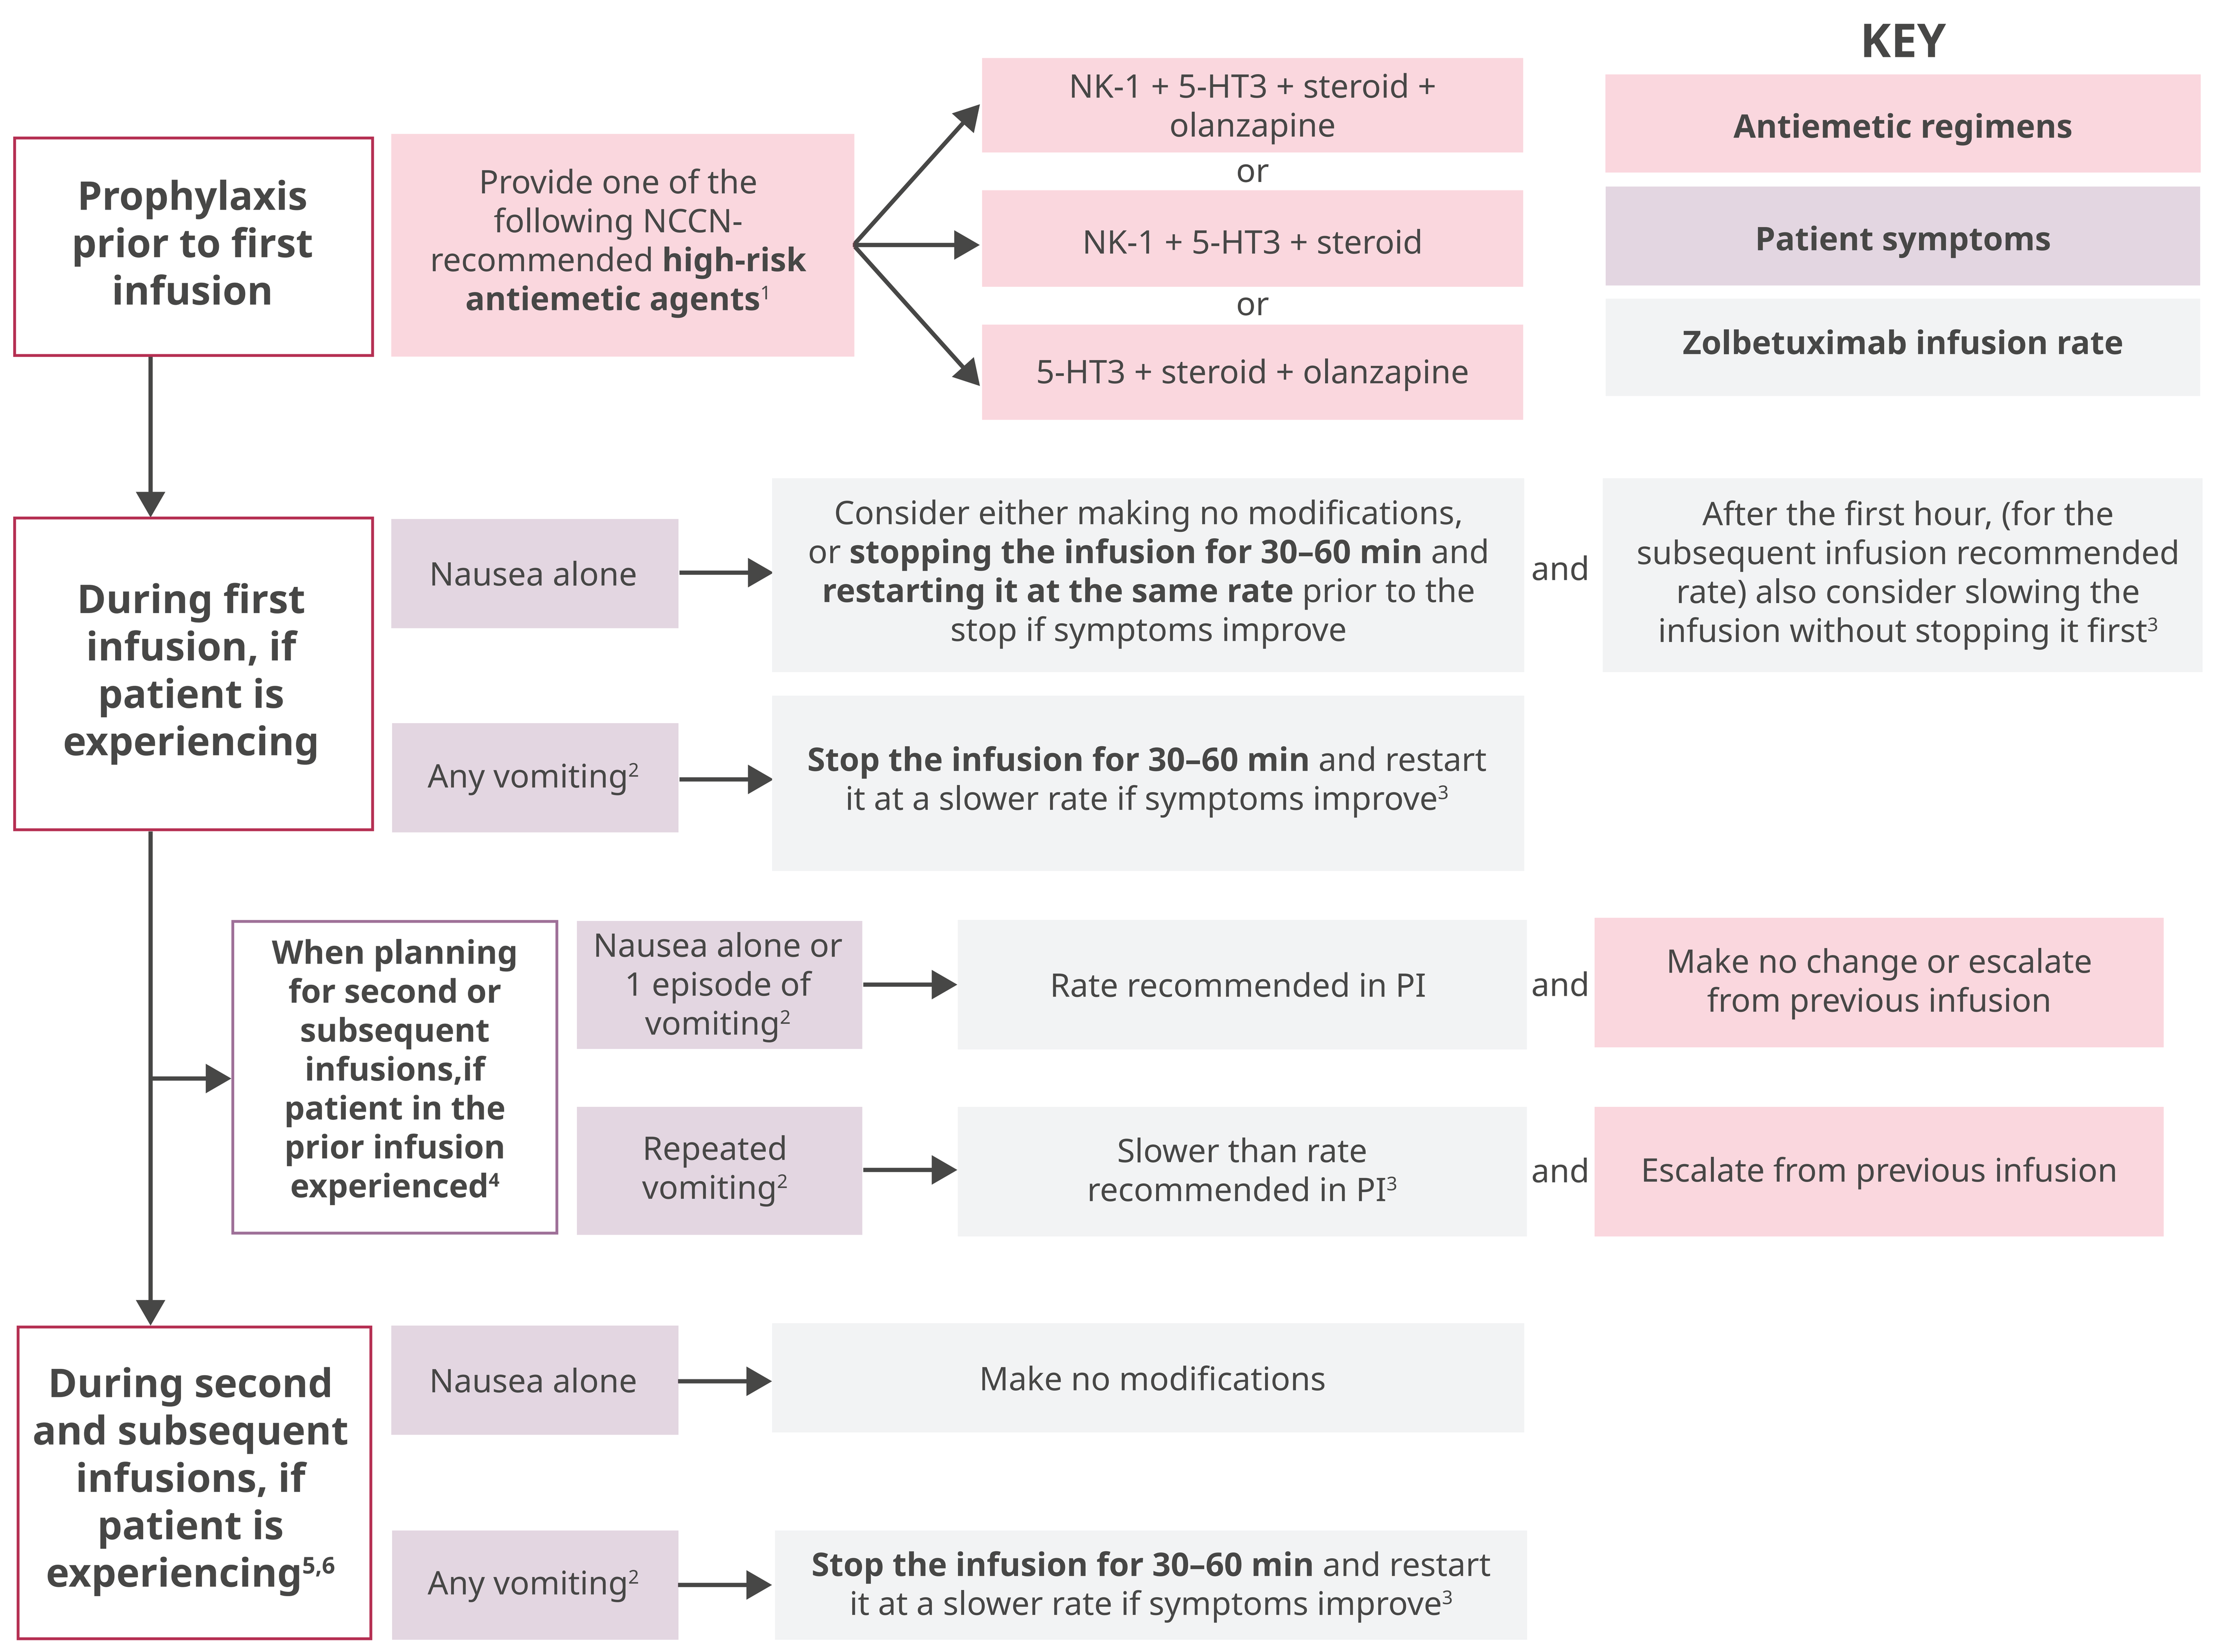
KEY
Antiemetic regimens
Patient symptoms
Zolbetuximab infusion rate
Prophylaxis prior to first infusion
Provide one of the following NCCN-recommended high-risk antiemetic agents[1]
NK-1 + 5-HT3 + steroid + olanzapine
or
NK-1 + 5-HT3 + steroid
or
5-HT3 + steroid + olanzapine
During first infusion, if patient is experiencing
Nausea alone
Consider either making no modifications, or stopping the infusion for 30–60 min and restarting it at the same rate prior to the stop if symptoms improve
and
After the first hour, (for the subsequent infusion recommended rate) also consider slowing the infusion without stopping it first[3]
Any vomiting[2]
Stop the infusion for 30–60 min and restart it at a slower rate if symptoms improve[3]
When planning for second or subsequent infusions,if patient in the prior infusion experienced[4]
Nausea alone or 1 episode of vomiting[2]
Rate recommended in PI
and
Make no change or escalate from previous infusion
Repeated vomiting[2]
Slower than rate recommended in PI[3]
and
Escalate from previous infusion
During second and subsequent infusions, if patient is experiencing[5,6]
Nausea alone
Make no modifications
Any vomiting[2]
Stop the infusion for 30–60 min and restart it at a slower rate if symptoms improve[3]

[1]NCCN-recommended high emetic risk regimens: NK-1 + 5-HT3 + steroid + olanzapine, or NK-1 + 5-HT3 + steroid, or 5-HT3 + steroid + olanzapine. Either oral or IV antiemetics may be appropriate based on individual patient circumstances.

[2]IV hydration may be appropriate depending on individual patient circumstances.

[3]If infusion was running at PI rate, slow rate by 50%; if infusion rate had already been slowed to 50%, slow by an additional 50% (ie, 25% of the initial rate).

[4]Adjust your plan for subsequent infusions based on the patient's symptoms during prior infusions.

[5]Begin second and subsequent infusions at the rate that was best tolerated during previous infusion (eg, if the prior infusion was tolerated at the PI rate, subsequent infusions should be given at the same rate; if infusion rate was slowed to 50% of PI rate and symptoms improved, start subsequent infusions at this rate).

[6]With second and/or subsequent infusions, the degree of nausea and vomiting is expected to diminish. In these cases, patients may tolerate titration of the infusion rate by increments of 25% (eg, if infusion rate was slowed to 50% and the patient remained asymptomatic for 30–60 min, consider increasing the rate to 75%) back to 100% or maximum tolerated infusion rate. Continue to closely monitor the patient for any recurrence of symptoms and administer additional antiemetic medications as needed to manage symptoms effectively.

CONCLUSIONS

- Consensus statements were developed using the modified Delphi method and can be utilized by clinicians to help guide the prevention and management of nausea and vomiting in patients treated with zolbetuximab + chemotherapy
 - The panel recommends using any of the NCCN's high-emetic risk regimens prophylactically, prior to the first zolbetuximab infusion
 - Based on patient symptoms of nausea and/or vomiting, recommendations include modifying the zolbetuximab infusion rate, interrupting zolbetuximab infusions for 30–60 min, administering antiemetic medications not used for prophylaxis, and/or providing IV hydrations
 - Zolbetuximab should not be discontinued without first attempting to modify or temporarily interrupt the infusion and/or without providing additional treatment for nausea and vomiting in the absence of hypersensitivity reactions or IRRs
 - If zolbetuximab and chemotherapy are given on the same day, zolbetuximab must be given first; if zolbetuximab infusion has been modified to the point where chemotherapy cannot be completed on the same day, chemotherapy can be administered on the following day

Abbreviations

5-HT3, 5-hydroxytryptamine 3 receptor antagonists; ARNP, Advanced Registered Nurse Practitioner; CLDN18.2, claudin 18 isoform 2; G/GEJ, gastric/gastroesophageal junction; HER2, human epidermal growth factor receptor-2; IRR, infusion-related reaction; IV, intravenous; LA, locally advanced; mG/GEJ, metastatic gastric/gastroesophageal junction; NCCN, National Comprehensive Cancer Network; NK-1, neurokinin-1; MD, Doctor of Medicine; OS, overall survival; PFS, progression-free survival; PharmD, Doctor of Pharmacy; PhD, Doctor of Philosophy; PI, prescribing information; TEAEs, treatment-emergent adverse events; UCSF, the University of California, San Francisco.

References

1. Shitara K et al. *Lancet*. 2023;401(10389):1655–1668.
2. Shah MA et al. *Nat Med*. 2023;29(8):2133–2141.
3. Sahin U et al. *Eur J Cancer*. 2018;100:17–26.
4. Shitara K et al. *Cancer Sci*. 2023;114(4):1606–1615.
5. Lordick F et al. *J Cancer Res Clin Oncol*. 2023;149(9):5937–5950.
6. Türeci O et al. *Ann Oncol*. 2019;30(9):1487–1495.
7. Klempner SJ et al. *Clin Cancer Res*. 2023;29(19):3882–3891.
8. Sahin U et al. *Ann Oncol*. 2021;32(5):609–619.
9. Shitara K et al. Presented at the American Society of Clinical Oncology (ASCO) Gastrointestinal Cancers Symposium 2024; San Francisco, CA, USA.

Acknowledgements

We thank all the panelists for their unique contribution to this study. We also thank Michael Broder and Hannah Dalglish from Partnership for Health Analytic Research (PHAR), LLC, for their valuable feedback.

Funding

This study was funded by Astellas Pharma Inc. Medical writing support, conducted in accordance with Good Publication Practice (GPP 2022) and the International Committee of Medical Journal Editors (ICMJE) guidelines, was provided by Jing Xu, PhD, of Oxford PharmaGenesis Inc., Newtown, PA, USA, and funded by Astellas Pharma Inc.

Conflicts of Interest

SJK reports receiving consulting fees from Astellas Pharma and Novartis; receiving honoraria from Merck Sharp & Dohme; serving a consulting or advisory role for Amgen, Astellas Pharma, Novartis, Pfizer, Sanofi-Aventis, Merck Sharp & Dohme, Bristol-Myers Squibb, IMAB, Mersana Therapeutics, Natera, AstraZeneca, Daiichi Sankyo, Laboratoires Servier, Coherus BioSciences; serving as a committee member for National Comprehensive Cancer Network; and had received stocks from Turning Point Therapeutics (by June 2022) and Nuvalent (by November 2022). **SNG** is a full-time employee of Partnership for Health Analytic Research. **SB** and **RF** are full-time employees of Astellas Pharma Inc.

Click or scan this quick response (QR) code to download this poster. Copies of this poster obtained through QR codes are for personal use only and may not be reproduced without permission of the authors.

For more information, please direct comments or inquiries to: Samuel J. Klempner,
email: sklempner@mgb.org

©2024 Authors. Contents not to be distributed, reproduced, presented, or otherwise shared without permission. All rights reserved by copyright holder(s).