Real-world use of risdiplam for the treatment of SMA in infants under 2 months of age

N Goedeker,[1]* VJ Carson,[2] S Bernes,[3] CC Haws,[3] C Zaidman,[1] C Campos,[4] I Yermilov,[4] MS Broder,[4] E Chang,[4] S Edwards,[5] TL Dickendesher,[6] S Tam,[6] S Shapouri[6]

[1]Department of Neurology, Neuromuscular Division, Washington University School of Medicine, St. Louis, MO, USA; [2]Clinic for Special Children, Gordonville, PA, USA; [3]Phoenix Children's, Phoenix, AZ, USA; [4]ADVI Health, Washington, DC, USA; [5]F. Hoffmann-La Roche Ltd, Basel, Switzerland; [6]Genentech, Inc., South San Francisco, CA, USA.

*ngoedeker@wustl.edu

Genentech
A Member of the Roche Group

Background

- Early diagnosis of SMA has increased due to prenatal and newborn screening.[1]
- Recent consensus and best practice publications recommend that treatment should be administered as early as possible to preserve motor neurons.[1,2]
- Risdiplam (EVRYSDI®) is a once-daily, orally administered, *SMN2* pre-mRNA splicing modifier approved by the US FDA to treat pediatric and adult patients with SMA.[3]
- In May 2022, the US FDA approved a label extension for risdiplam to treat infants with SMA <2 months of age based on interim data from the RAINBOWFISH study (NCT03779334), an open-label, single-arm study of risdiplam in presymptomatic infants with genetically diagnosed SMA.[4,5]
- We present the interim analysis of a multicenter, retrospective chart review characterizing the early real-world use of risdiplam in infants <2 months of age with SMA in the US.

Methods

Study design

- This study is a descriptive, non-interventional, real-world study involving de-identified data collection based on retrospective chart reviews.
- Each infant's study period was from birth to the approval date of their local IRB.
- Data were collected from infants who had ≥6 months of medical records after risdiplam initiation.

Study objectives

- In infants with SMA who initiated risdiplam before 2 months of age and have ≥6 months of available medical records after risdiplam initiation, we aim to:
 - Describe demographics, clinical characteristics, and treatment patterns of infants with SMA before initiating risdiplam;
 - Characterize clinical outcomes, HCRU, and treatment patterns of infants with SMA after risdiplam initiation.

Patients

- Infants included in the study were newborn infants diagnosed with SMA who received ≥1 dose of risdiplam before 2 months of age.
 - At least 6 months of medical records after risdiplam initiation were available.
 - Infants were treated with risdiplam as indicated by standard practice and according to the US FDA-approved USPI for risdiplam. Infants participating in clinical trials were excluded.
- Data for this interim analysis were collected from three neurology clinics across the USA.
 - The Clinic for Special Children (Gordonville, PA), Phoenix Children's Hospital (Phoenix, AZ), and Washington University in St. Louis (St. Louis, MO).

Statistical methods

- Interim data were analyzed using the data cutoff date of March 6, 2025.
- Descriptive analyses and frequency summaries of the data collected were conducted.

Demographic and clinical characteristics

- There were 16 infants enrolled in this study as of the data cutoff; all were born full term (**Table 1**).
- Most infants were male (62.5%), White (81.3%), and had two copies of *SMN2* (68.8%).
- Risdiplam was initiated at a mean (SD) age of 17.4 (10.1) days, with 14 infants (88%) initiating risdiplam ≤30 days after birth.

Table 1. Demographic and clinical characteristics

	N=16
Male, n (%)	10 (62.5)
Ethnicity, n (%)	
White	13 (81.3)
Hispanic or Latino	1 (6.3)
Black or African American	1 (6.3)
Unknown	1 (6.3)
Health insurance, n (%)	
Medicaid	8 (50.0)
Private	5 (31.3)
Uninsured	3 (18.8)
Deceased prior to study period end	0
Full term	16 (100.0)
Diagnosis method, n (%)	
Newborn screening only	11 (68.8)
Newborn/prenatal screening and confirmatory testing	5 (31.3)
Mean (SD) age at risdiplam initiation, days	17.4 (10.1)
Mean (SD) age at last encounter, days	431.8 (147.9)
***SMN2* copy number, n (%)**	
2	11 (68.8)
3	4 (25.0)
4	1 (6.3)
Presymptomatic infants	13 (81.3)
SMA signs/symptoms pre-risdiplam initiation, n (%)*	3 (18.8)
Abnormal deep tendon reflexes	2 (12.5)
Decreased muscle tone	1 (6.3)
Muscle weakness	1 (6.3)

*An infant could have more than one of these symptoms.

Respiratory and bulbar support

- Overall, 93.8% of infants were able to feed orally without restrictions at the end of the study period (**Table 2**).
- One (6.3%) infant required a cough assist only when ill (e.g. with an upper respiratory infection).

Table 2. Respiratory and bulbar support

	N=16
Nutritional support, n (%)	
Total oral intake with no restrictions	15 (93.8)
Unknown	1 (6.3)
Respiratory support, n (%)	0
Cough assist, n (%)	
No	15 (93.8)
Yes, used only during illness	1 (6.3)

Treatment patterns

- Most infants (n=13/16, 81.3%) received risdiplam as their first DMT; nine infants (56.3%) received OA as their second DMT (**Figure 1**).
- One infant (6.3%) received a third DMT with the following sequence: risdiplam, nusinersen, OA.
- Overall, the mean (SD) duration on risdiplam was 10.3 (7.3) months.

Figure 1. Sankey plot of treatment patterns

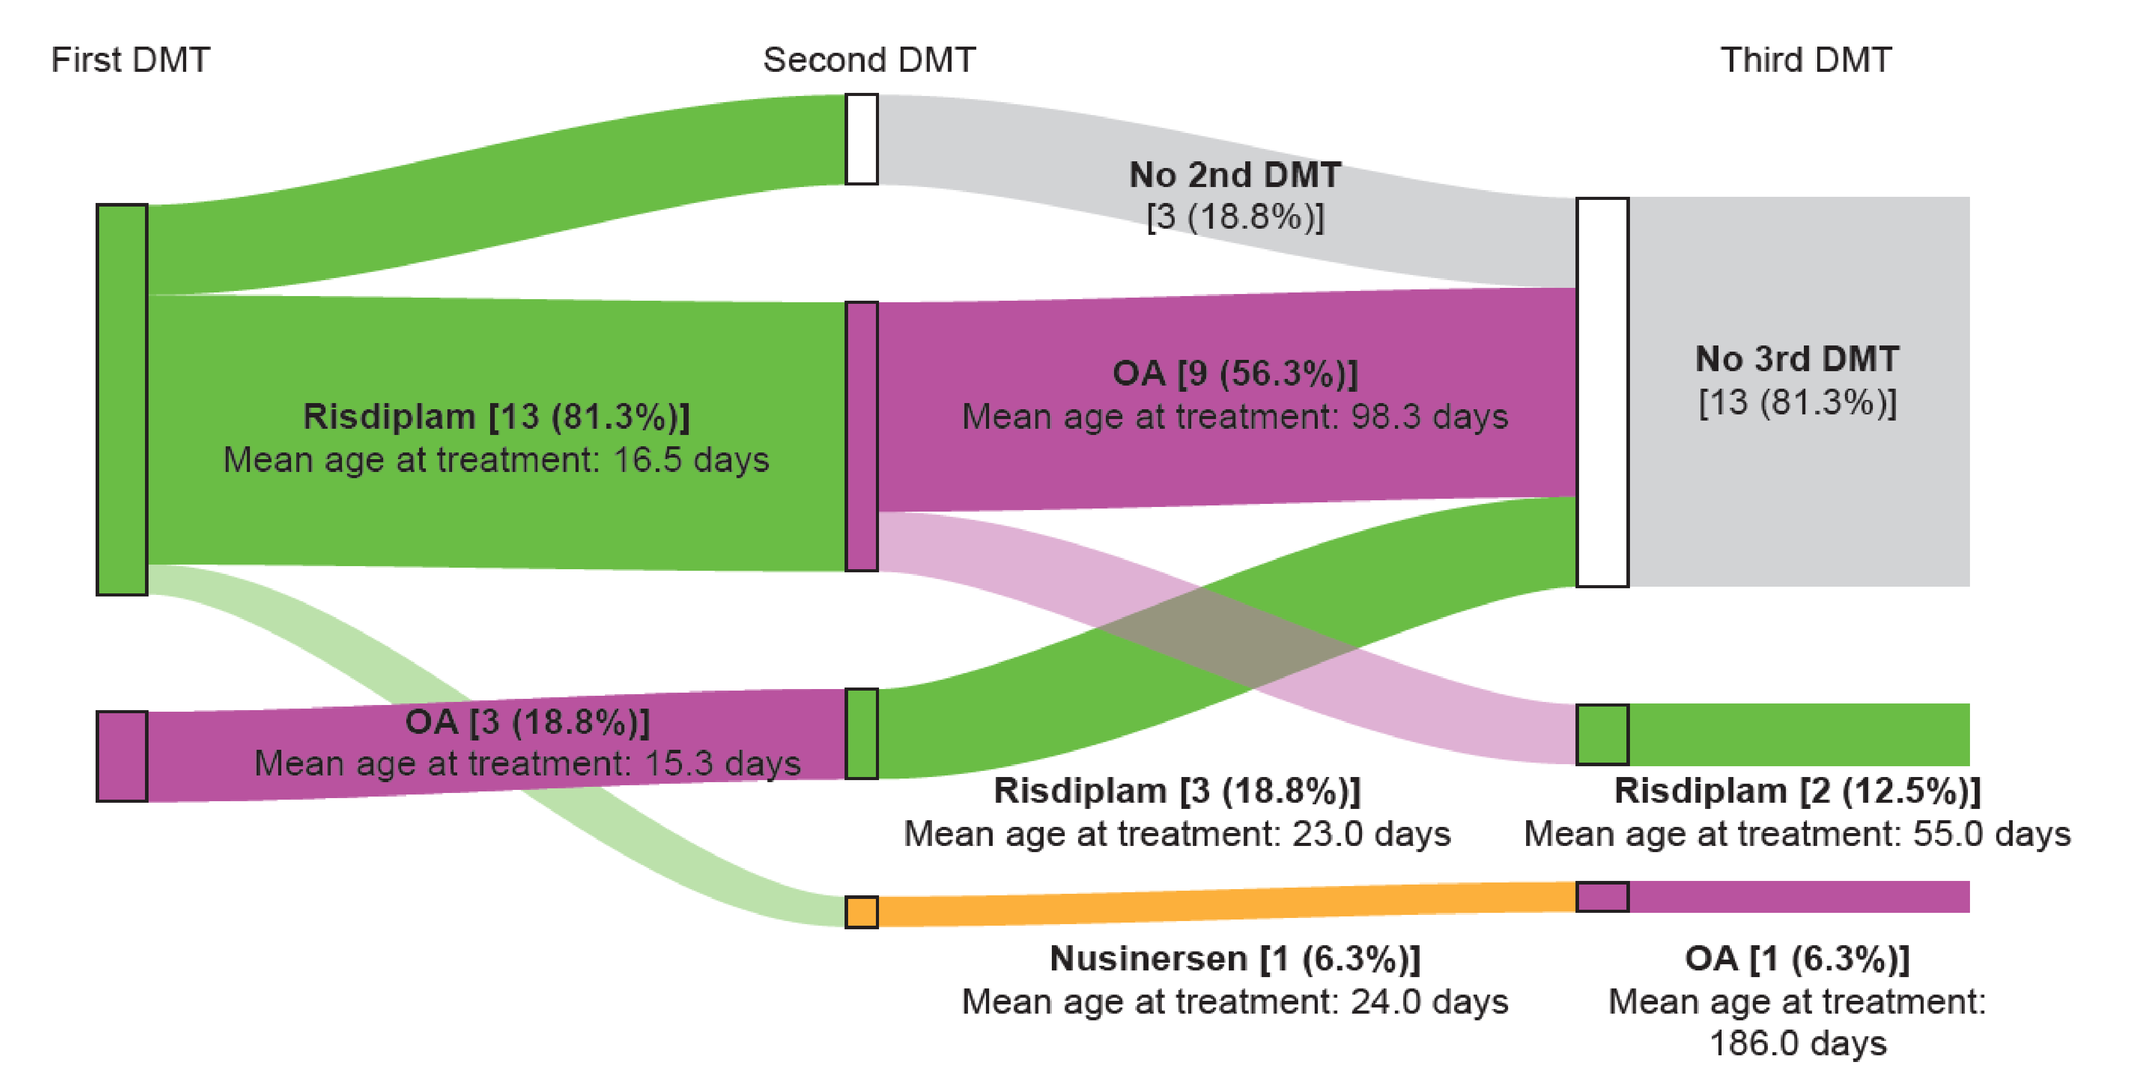
First DMT
Second DMT
Third DMT
No 2nd DMT [3 (18.8%)]
Risdiplam [13 (81.3%)] Mean age at treatment: 16.5 days
OA [9 (56.3%)] Mean age at treatment: 98.3 days
No 3rd DMT [13 (81.3%)]
OA [3 (18.8%)] Mean age at treatment: 15.3 days
Risdiplam [3 (18.8%)] Mean age at treatment: 23.0 days
Risdiplam [2 (12.5%)] Mean age at treatment: 55.0 days
Nusinersen [1 (6.3%)] Mean age at treatment: 24.0 days
OA [1 (6.3%)] Mean age at treatment: 186.0 days

HCRU

- Overall, hospitalizations were low (**Table 3**).
- Over half of infants (n=9, 56.3%) had an ED visit.
 - Among these nine infants, the mean (SD) number of visits was 1.6 (1.3).

Table 3. HCRU across the study period

	N=16
Hospitalizations	
Infants with any hospitalization, n (%)	1 (6.3)
Total number of hospitalizations among infants with a hospitalization	1
Mean (SD) number of hospitalization among infants with a hospitalization	1.0 (n/a)
Mean (SD) age at hospitalization, days	29.0 (n/a)
Mean (SD) length of stay, days	1.0 (n/a)
Primary diagnosis for hospitalization	
Fever and vomiting, n (%)	1 (100)
ED visits*	
Infants with any ED visit, n (%)	9 (56.3)
Total number of ED visits among infants with an ED visit	14
Mean (SD) number of ED visits among infants with an ED visit	1.6 (1.3)
Mean (SD) age at ED visit, days	245.4 (160.4)
Primary diagnosis at ED visit†	
Fever, n (%)	3 (21.4)
Fever and cold symptoms, n (%)	2 (14.2)

*ED visits did not lead to inpatient admissions. †Diagnoses reported in >1 participant. Additional diagnoses occurred in one infant each and were the following: acute otitis media; congestion and cough; cough, runny nose, ear pulling and decreased appetite; diaper rash; overdose; right femur fracture; fever, cold symptoms, and vomiting; vomiting; and vomiting and fatigue.

Motor function

CHOP-INTEND

- All infants (N=16; 100%) were assessed on the CHOP-INTEND scale (**Figure 2**).
- At the last reported assessment, the mean (SD) score was 57.8 (10.2), and the mean (SD) age was 6.3 (2.5) months.
- Twelve (75%) infants achieved a near maximum CHOP-INTEND score ≥60, including all infants with ≥3 *SMN2* copies (n=5/5; 100%).

Figure 2. CHOP-INTEND total scores over time*

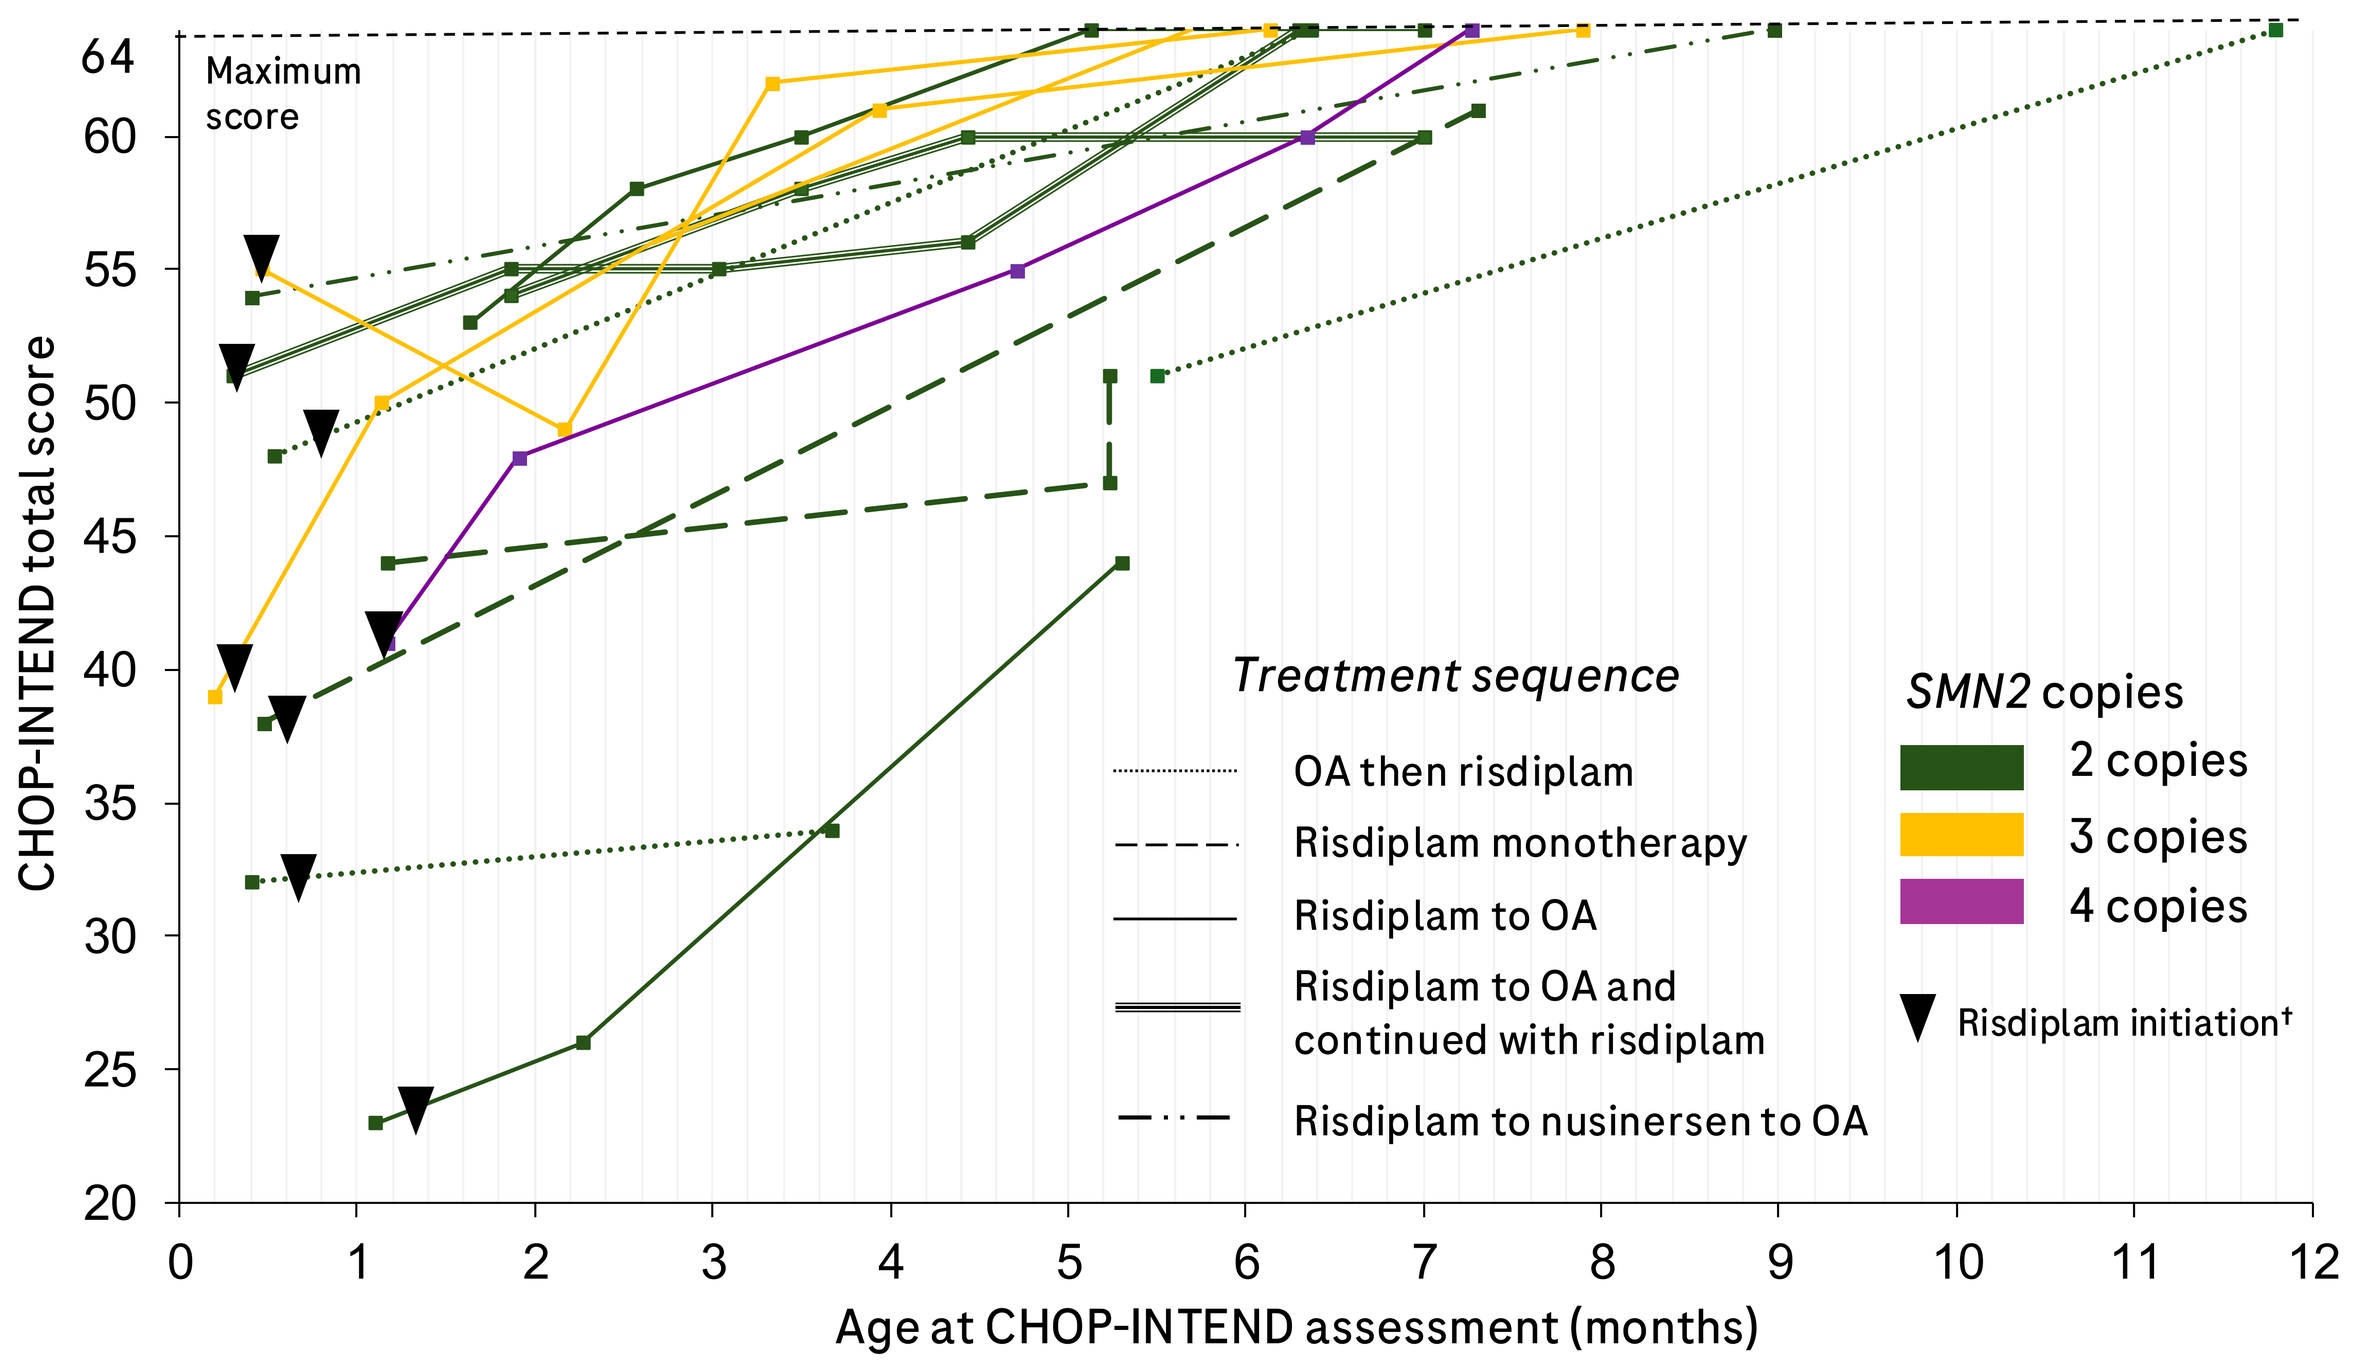
Maximum score
CHOP-INTEND total score
Age at CHOP-INTEND assessment (months)
Treatment sequence
OA then risdiplam
Risdiplam monotherapy
Risdiplam to OA
Risdiplam to OA and continued with risdiplam
Risdiplam to nusinersen to OA
SMN2 copies
2 copies
3 copies
4 copies
Risdiplam initiation†

81.8% (n=9/11) infants with 2 *SMN2* copies achieved a score of ≥40 points‡

In natural history, children with Type 1 SMA rarely reach a CHOP-INTEND total score of 40 points[6]

*Two infants had only one assessment. One infant with two *SMN2* copies was assessed at 8 days, treated with risdiplam at DOL 16, and had a score of 38. One infant with three *SMN2* copies was treated with risdiplam at DOL 15, assessed at 4 months and had a score of 60. †Risdiplam treatment was initiated before the first assessment among six of 14 infants. ‡Score at any point during the study period.

BSID-III

- Eight (50.0%) of 16 infants were assessed on the BSID-III motor scale and had available data (**Figure 3**).
- At the last reported assessment, all infants were within the range (±2 SD; grey shading) for normally developing children of a similar age.
- Mean (SD) change from baseline (earliest reported score) to last reported score was 30.1 (9.2).

Figure 3. BSID-III gross motor raw scores after risdiplam treatment over time*

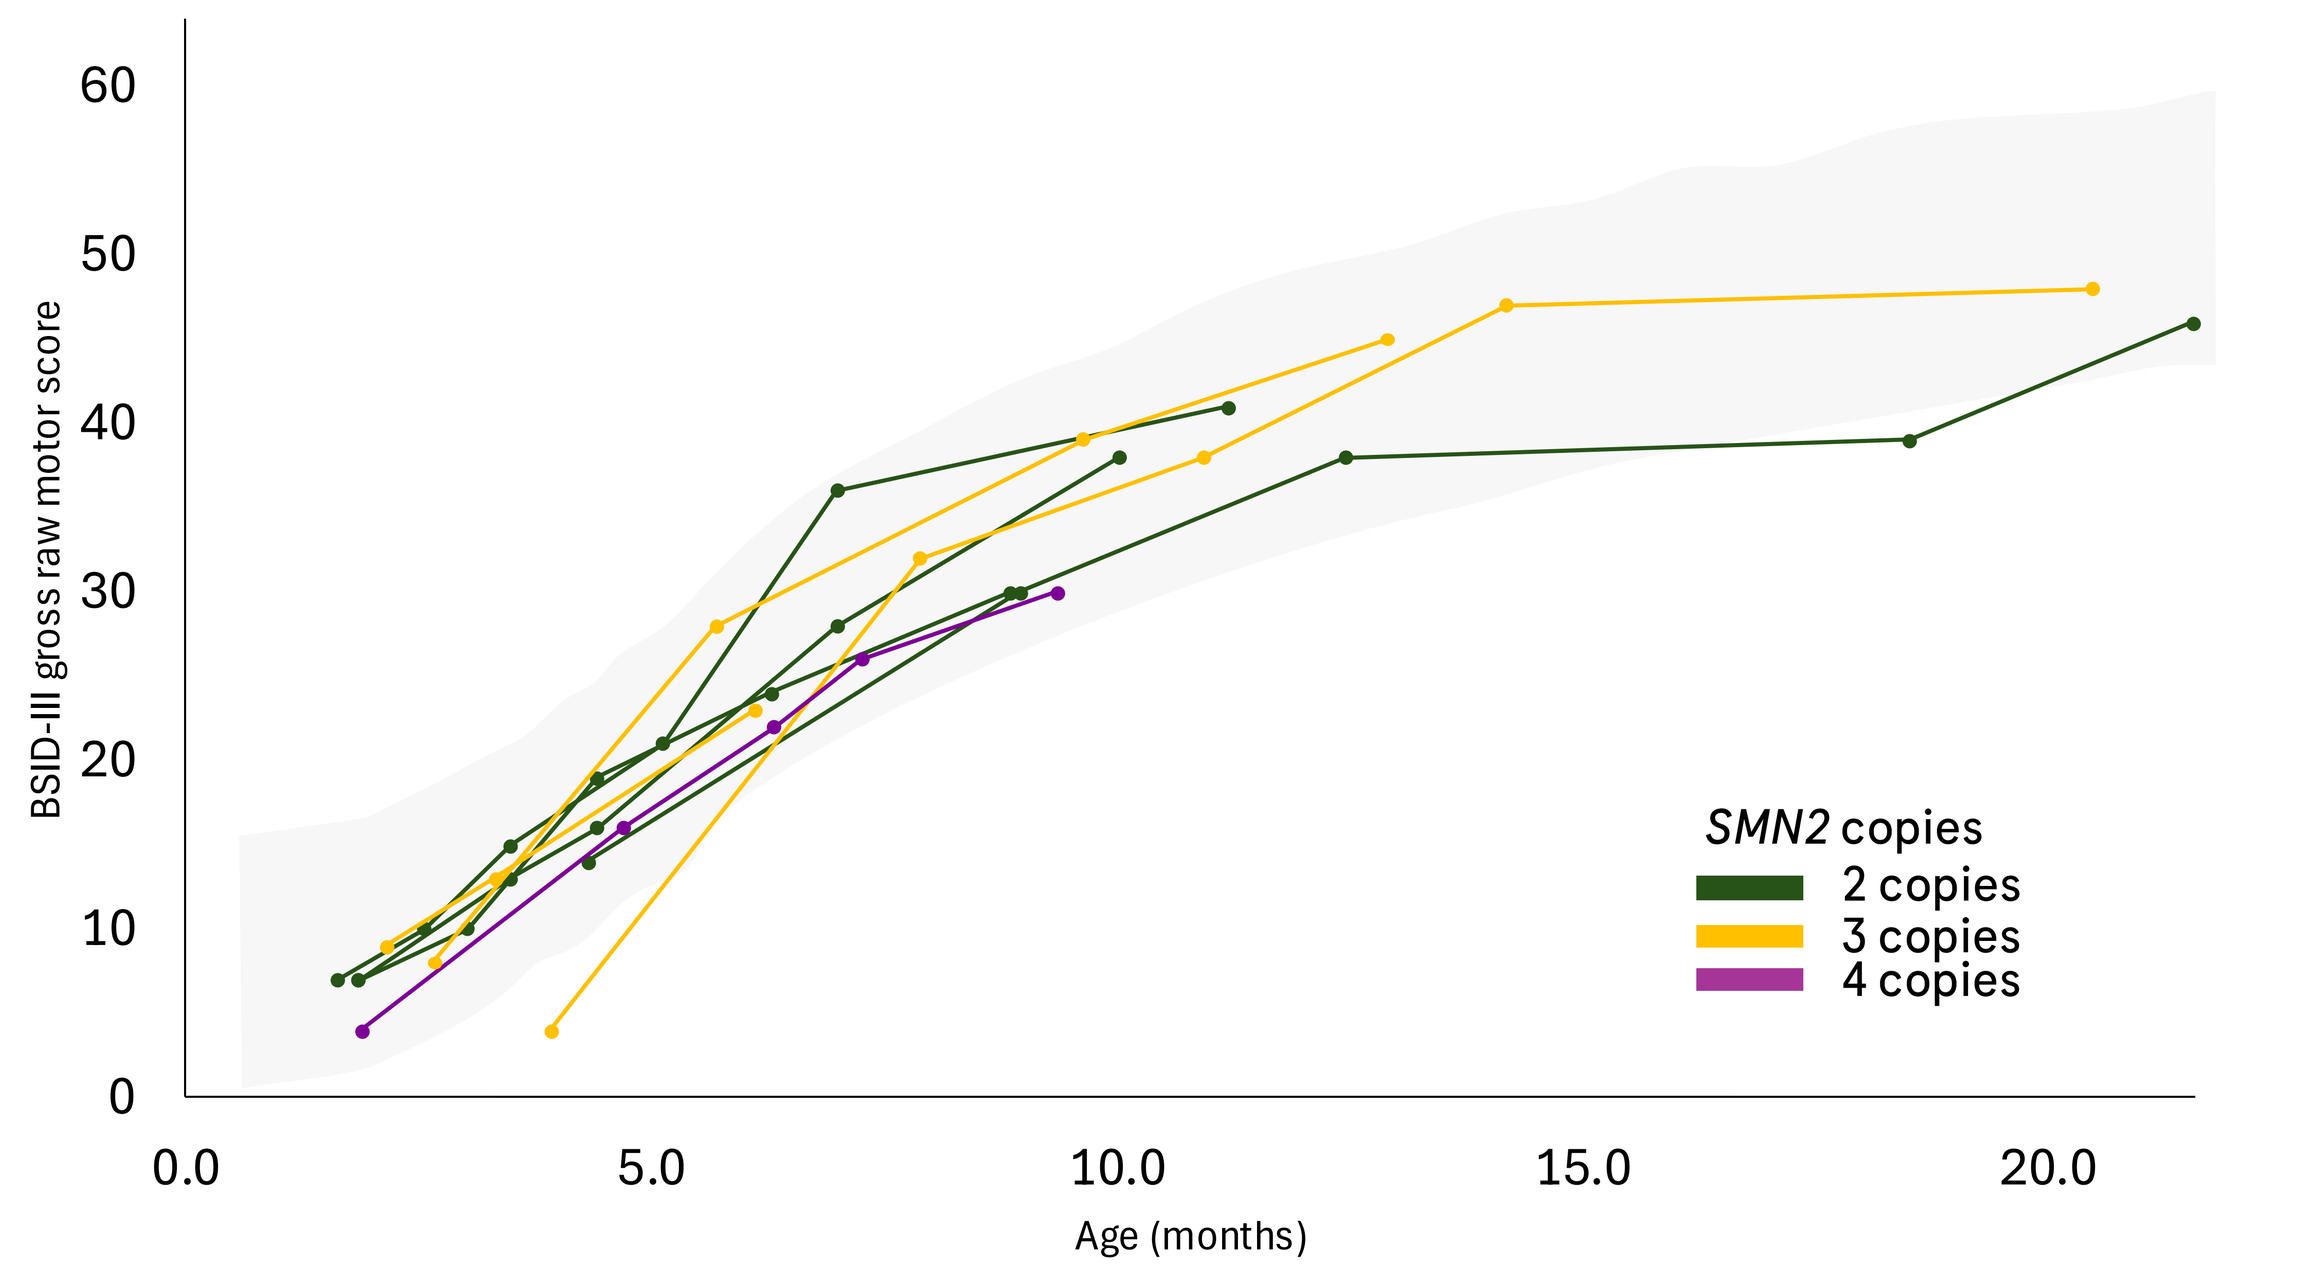
BSID-III gross raw motor score
Age (months)
SMN2 copies
2 copies
3 copies
4 copies

*The BSID-III uses a scaled score of 10 as the normative mean. Scaled scores of 4 and 16 represent two SDs from the normative mean and capture the 3rd to 97th percentile range of normally developing children. The scaled scores of 4, 10 and 16 were converted to gross motor raw scores and plotted by age (grey shading).[7,8] All infants started risdiplam treatment before their first assessment on the BSID-III motor scale.

Motor milestone achievement

- Overall, most infants achieved sitting (n=15/16 [93.8%]) and over half achieved walking (n=9/16 [56.3%]). All infants who achieved these milestones did so within the WHO time windows of typical development (**Figure 4**).
 - Some infants had study periods that were shorter than the WHO windows of typical development and may not have achieved a milestone yet.
- Among the 11 infants with two *SMN2* copies, eight (72.7%) achieved head control, ten (90.9%) achieved sitting independently, and six (54.5%) achieved walking.
- All infants (n=5/5 [100%]) with ≥3 *SMN2* copies achieved head control and sitting independently, while three (60.0%) infants achieved walking.

Figure 4. Age at motor milestones achievement by each infant across the study period

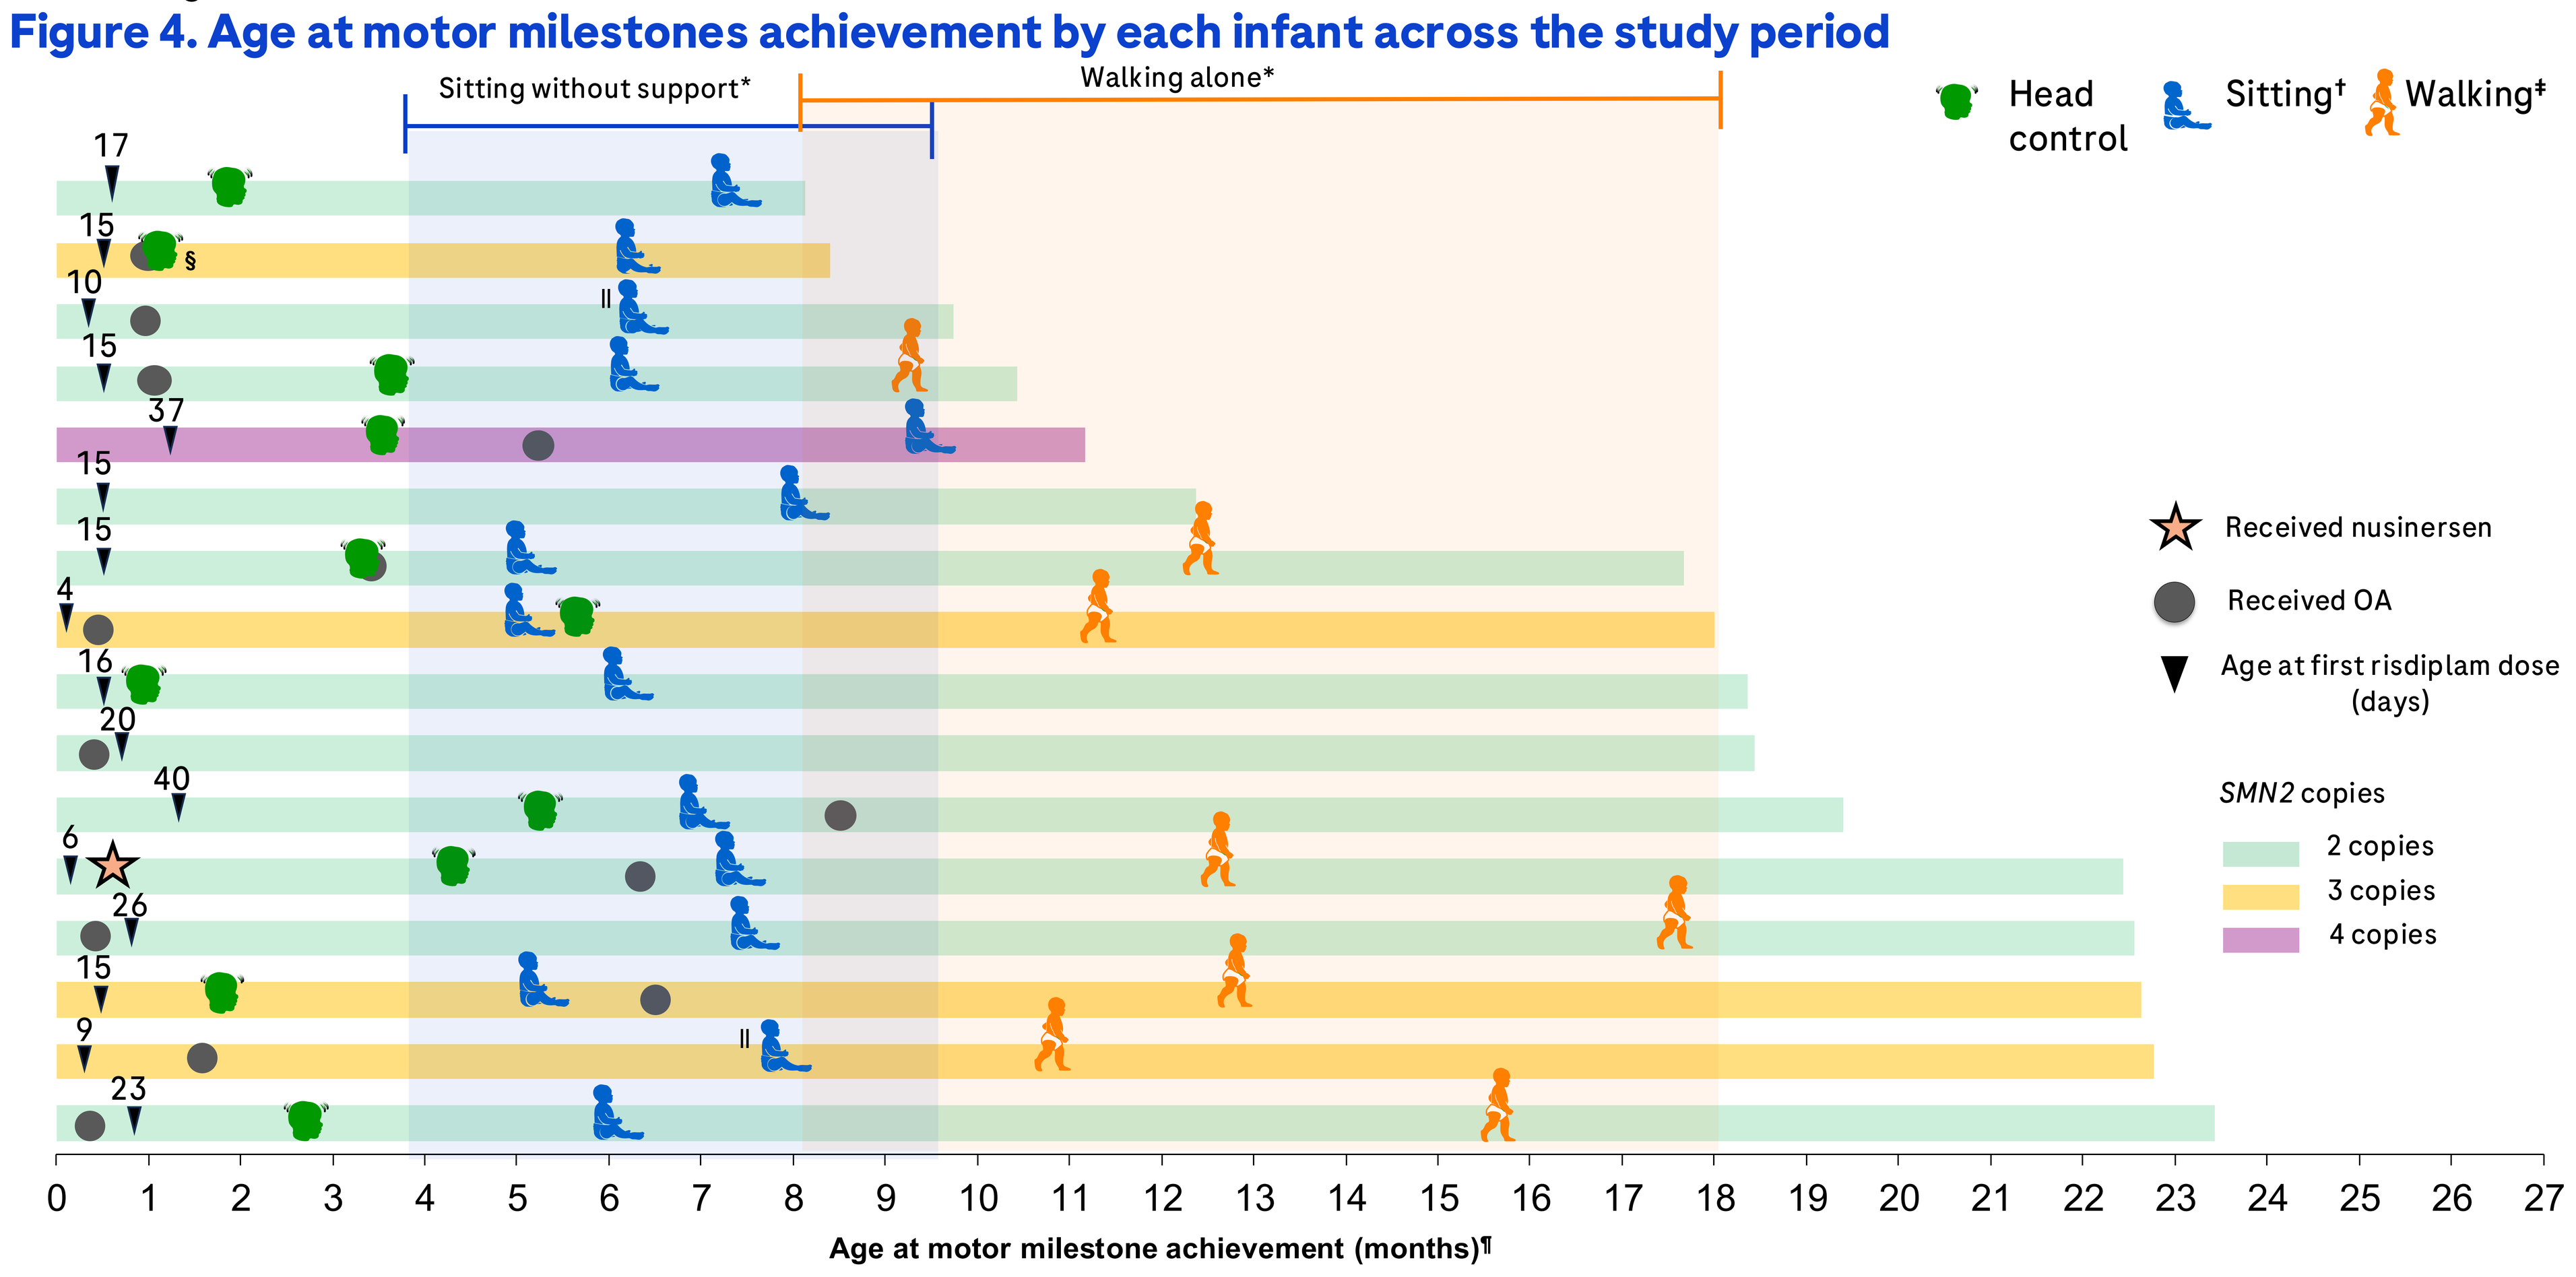
Sitting without support*
Walking alone*
Head control
Sitting†
Walking‡
Received nusinersen
Received OA
Age at first risdiplam dose (days)
SMN2 copies
2 copies
3 copies
4 copies
Age at motor milestone achievement (months)¶

*The WHO normal development time window for motor milestones which includes 95% CI.[9] †Includes sitting independently and sitting independently for 5 and 30 seconds with the highest motor milestone achieved represented. ‡Includes walking with and without support with the highest motor milestone achieved represented. §This infant received OA on the same day that head control was assessed. ‖This infant also achieved the milestone 'head control' which was assessed on the same DOL. ¶The length of the shaded bar represents the infant's study period (date of birth to local IRB approval date). The length of study period was variable between each infant.

Limitations

- The study was limited by the availability of data in a real-world setting.
- Medical chart records do not reflect all data as HCPs primarily documented information based on clinical care and not for research purposes.

Conclusions

- Most infants in this study had two *SMN2* copies and were diagnosed through newborn screening, with risdiplam initiated at a mean (SD) age of 17.4 (10.1) days.
- Most infants (81.3%) received risdiplam as their first DMT.
- There was a low rate (6.3%) of hospitalizations, and a little over half (56.3%) of infants had an ED visit.
- Among the infants who had available motor function assessments as measured by BSID-III and CHOP-INTEND, all infants showed improvements over time.
- Among the infants who had achieved sitting (n=15) and walking (n=9), all achieved these milestones within the WHO normal development time window.
- These interim results support the early use of risdiplam in infants with SMA <2 months of age.

Abbreviations

BSID-III, Bayley Scales of Infant and Toddler Development, third edition; CHOP-INTEND, Children's Hospital of Philadelphia Infant Test of Neuromuscular Disorders; CI, confidence intervals; DMT, disease-modifying therapy; DOL, day of life; ED, emergency department; FDA, US Food and Drug Administration; HCP, healthcare professional; HCRU, healthcare resource utilization; IRB, internal review board; N/A, not applicable; OA, onasemnogene abeparvovec; SD, standard deviation; SMA, spinal muscular atrophy; SMN, survival of motor neuron; USPI, US prescribing information; WHO, World Health Organization.

Acknowledgments

The authors thank the participants and families included in the study, their clinical care teams, and the participating centers and their staff members responsible for abstracting medical records. This study is sponsored by Genentech, Inc., South San Francisco, CA, USA. Writing and editorial assistance was provided by Michelle B. Kim, PhD, of Nucleus Global, in accordance with Good Publication Practice (GPP) 2022 guidelines (https://www.ismpp.org/gpp-2022) and funded by Genentech, Inc., South San Francisco, CA, USA.

References

1. Yeo CJJ, et al. *Lancet Neurol.* 2024; 23:205–218;
2. Dangouloff T and Servais L. *Ther Clin Risk Manag.* 2019; 15:1153–1161;
3. Genentech, Inc. EVRYSDI® (risdiplam) USPI: https://www.gene.com/download/pdf/evrysdi_prescribing.pdf (Accessed June 2025);
4. ClinicalTrials.gov. NCT03779334 (Accessed June 2025);
5. Finkel RS, et al. *N Engl J Med.* 2025. Accepted;
6. Finkel RS, et al. *Neurology.* 2014; 83:810–817;
7. Bayley, N. Bayley Scales of Infant and Toddler Development: Administration Manual 3rd edn (Pearson PsychCorp, 2006);
8. WHO Multicentre Growth Reference Study Group. *Acta Paediatr Suppl.* 2006; 450:86–95;
9. Strauss KA, et al. *Nat Med.* 2022; 28:1381–1389.

Disclosures

NG has no disclosures.
VJC has no disclosures.
SB and CCH have no disclosures.
CZ has no disclosures
CC, IY, MSB, and EC are employees of ADVI Health which received financial compensation from Genentech, Inc. to conduct the research described in this poster.
SE is an employee and shareholder of F. Hoffmann La Roche.
TLD, SS, and ST are employees of Genentech, Inc., and shareholders of F. Hoffmann-La Roche Ltd.

Please scan using your QR reader application to access the graphs and data presented in this poster. Alternatively, this can be accessed at https://ter.li/nzeis1

NB: There may be associated costs for downloading data. These costs may be high if you are using your smartphone abroad. Please check your mobile data tariff or contact your service provider for more details.

Presented at the 2025 Cure SMA Annual SMA Research & Clinical Care Meeting, Anaheim, CA, USA; June 25–27, 2025