A Retrospective Cohort Study to Assess the Clinical and Economic Burden in Treated versus Untreated People with Relapsing-Remitting Multiple Sclerosis in the United States

001

Nupur Greene[1], Ashis K. Das[2], Lita Araujo[1], Eunice Chang[2], Jaja Sangbana[1], Marian H. Tarbox[2], Melinda Rossi[2], Michael S. Broder[2]

[1]Sanofi, Cambridge, MA, USA; [2]ADVI Health, Washington, DC, USA

BACKGROUND

- In the United States (US), approximately one million people are affected by multiple sclerosis (MS). Of these, 85% have the relapsing-remitting multiple sclerosis (RRMS) phenotype, which is characterized by events of relapses followed by partial or complete remission of symptoms[1,2,3]
- Disease-modifying therapies (DMTs) are central to managing RRMS and improving outcomes; however, despite the availability of multiple treatment options, many people with RRMS (pwRRMS) remain untreated[4]
- Currently, real-world evidence comparing treated versus untreated in pwRRMS is limited[5]

OBJECTIVE

To evaluate the real-world clinical and economic burden, and identify risks-factors associated with not receiving treatment in untreated versus treated pwRRMS in the US

METHODS

Study design and population

- A retrospective cohort study was conducted using a large, integrated, US-based administrative claims database from July 01, 2020 to June 30, 2023 (**Figure 1**)
- PwRRMS were identified using a validated algorithm[6] and were evaluated over a 1-year baseline period before the index date and were subsequently followed for a year during the follow-up period
- The index date was defined as a randomly selected medical claim with an MS diagnosis during the identification period
- PwRRMS were defined based on specific inclusion and exclusion criteria (**Figure 2**); eligible patients were categorized into two cohorts based on their prior DMT usage:
 - Untreated cohort: People who did not receive a DMT either during the baseline or follow-up periods
 - Treated cohort: People who received a DMT during the baseline and follow-up periods

Figure 1: Study time frame

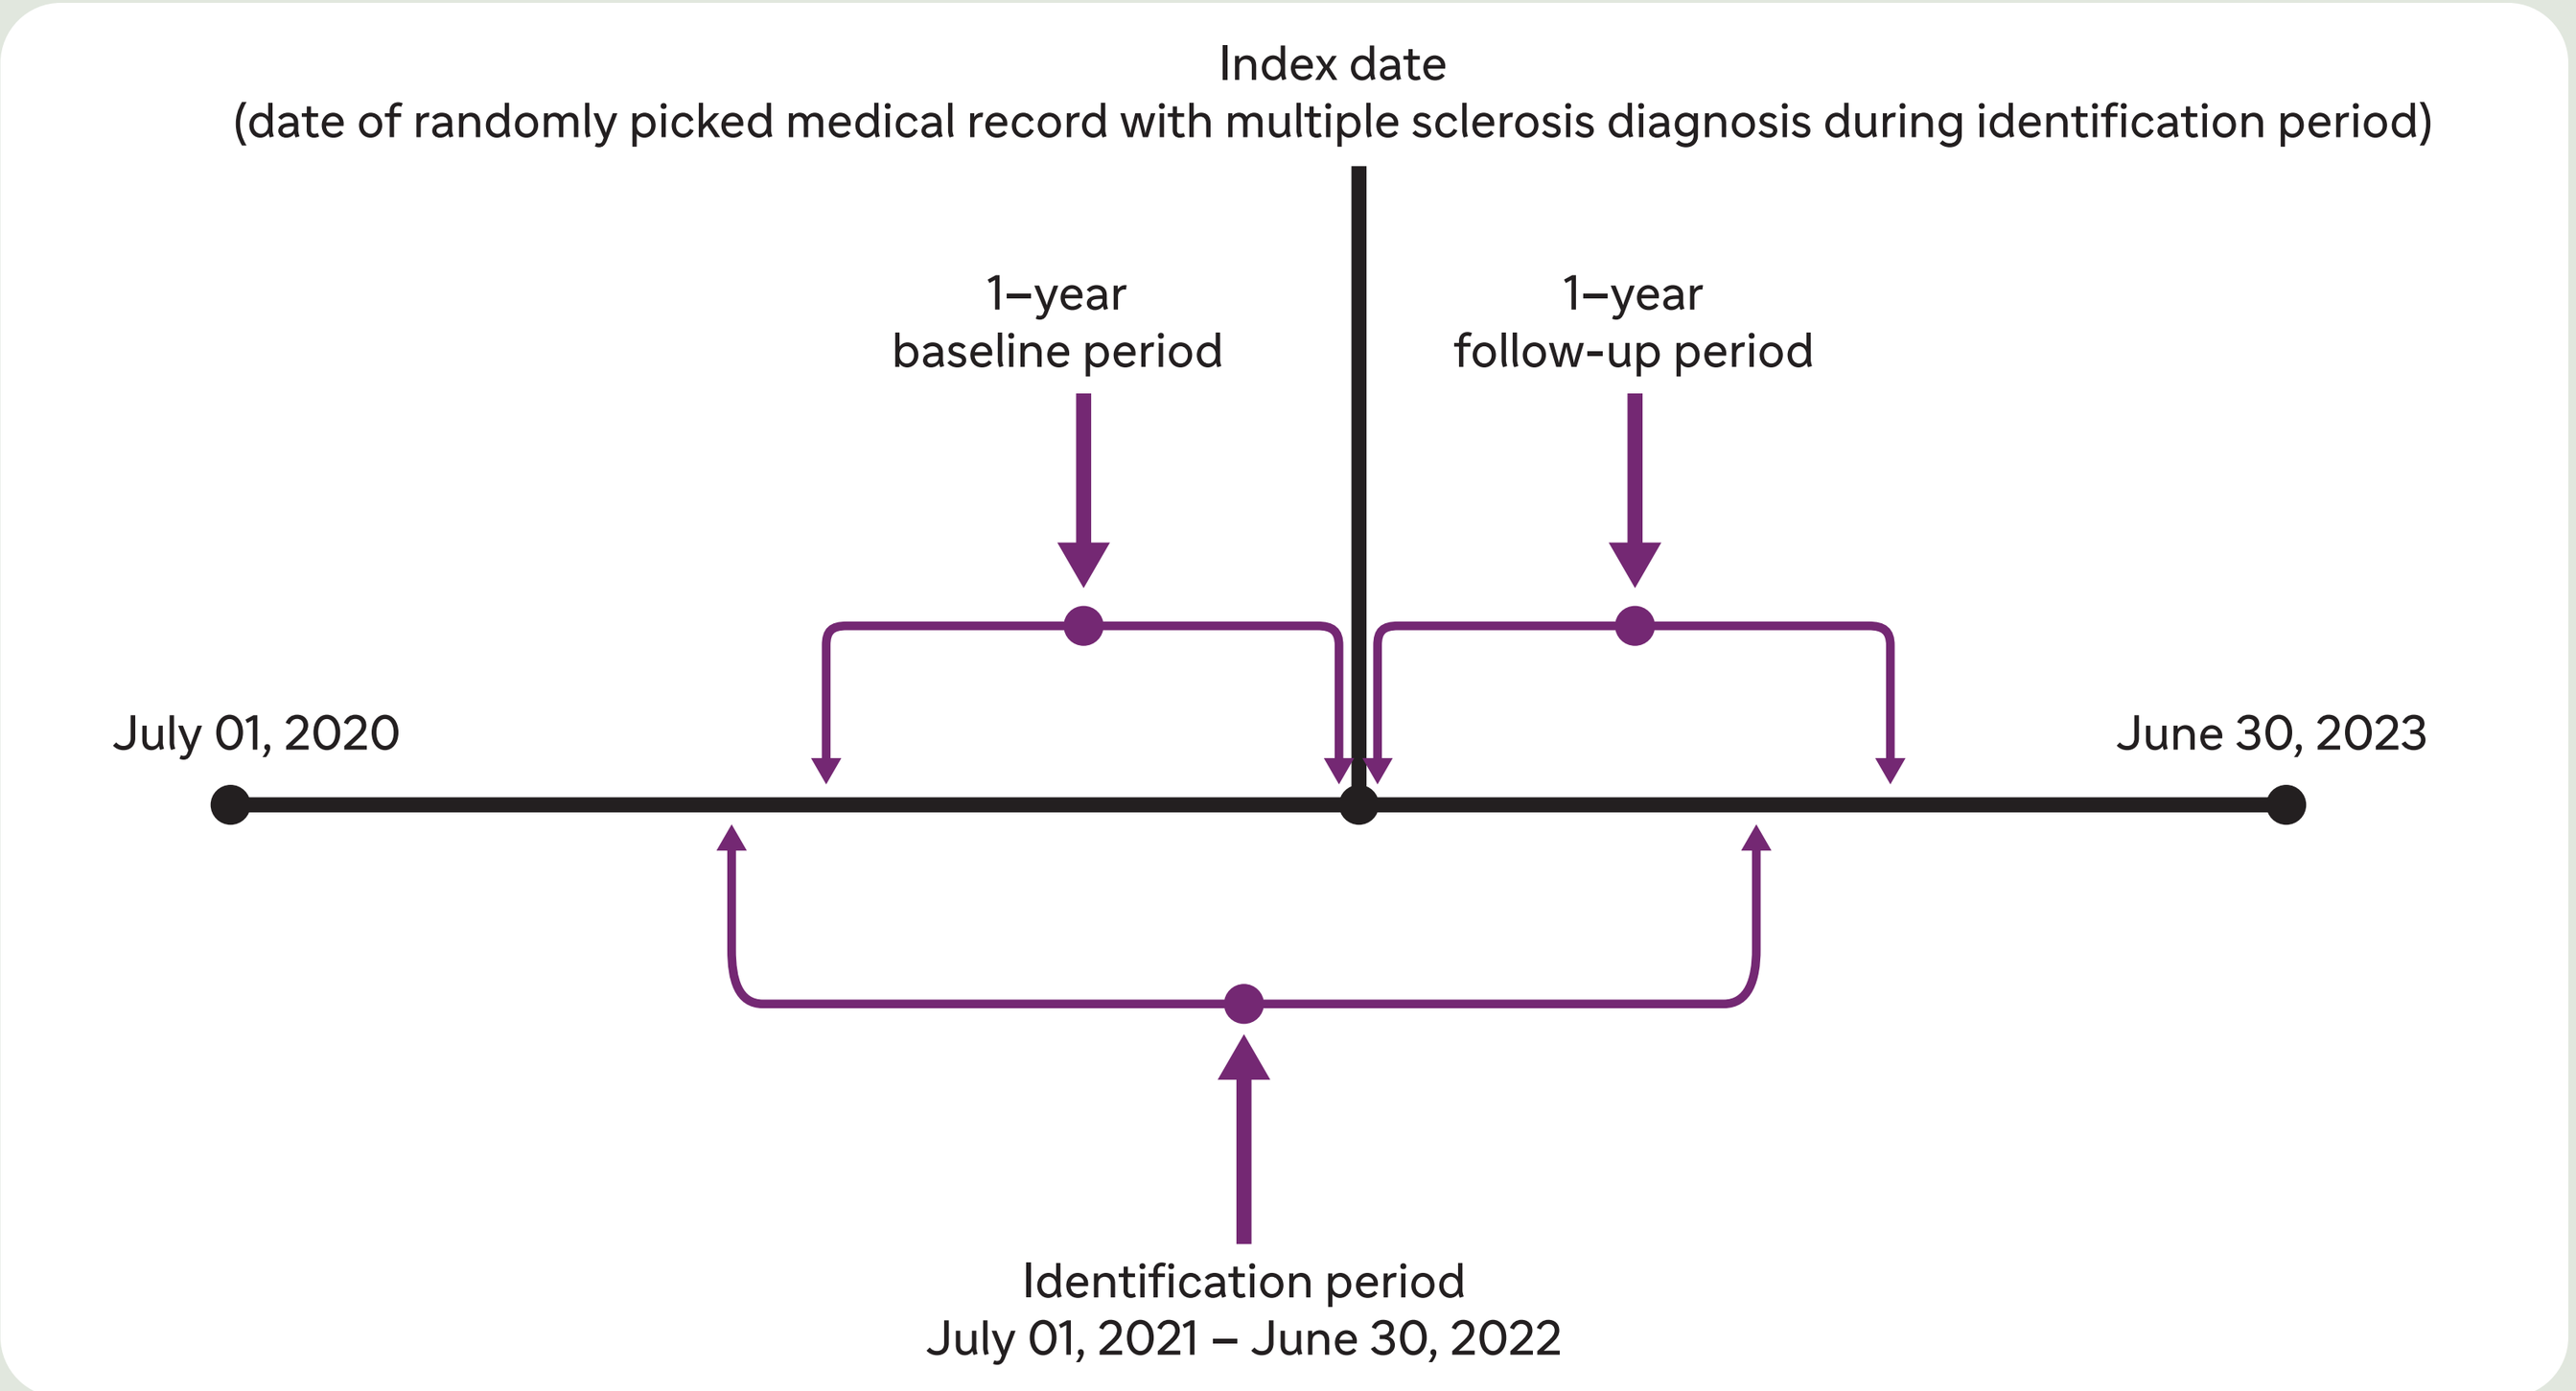
Index date
(date of randomly picked medical record with multiple sclerosis diagnosis during identification period)
1–year baseline period
1–year follow-up period
July 01, 2020
June 30, 2023
Identification period
July 01, 2021 – June 30, 2022

Study measures

- Baseline demographics, clinical characteristics (Kurtzke Functional Systems [KFSS]-adapted disability score, relapses, and infections), Charlson Comorbidity Index (CCI) score, specific comorbidities of interest, healthcare resource utilization (HCRU), and healthcare costs (HCCs) of pwRRMS were captured
- HCRU included the use of rehabilitation services, inpatient admissions, emergency department (ED) visits, and non-ED outpatient service visits; HCCs included costs of medical claims, pharmacy claims, and cost of infections
- Risk factors for not receiving DMTs were also identified using a logistic regression model

Statistical analysis

- Study measures were summarized using descriptive statistical methods and outcomes were compared between the treated and the untreated cohorts
- Costs were reported in US dollars (adjusted to Year 2023 values)
- All statistical tests were two-sided, with a significance threshold of $P<0.05$

RESULTS

Baseline demographics

- The final cohort comprised 14,236 pwRRMS, of whom 5,473 did not receive any DMT (untreated) and 8,763 received a DMT (treated) during the baseline and the follow-up period (**Figure 2**)

Figure 2: Attrition diagram

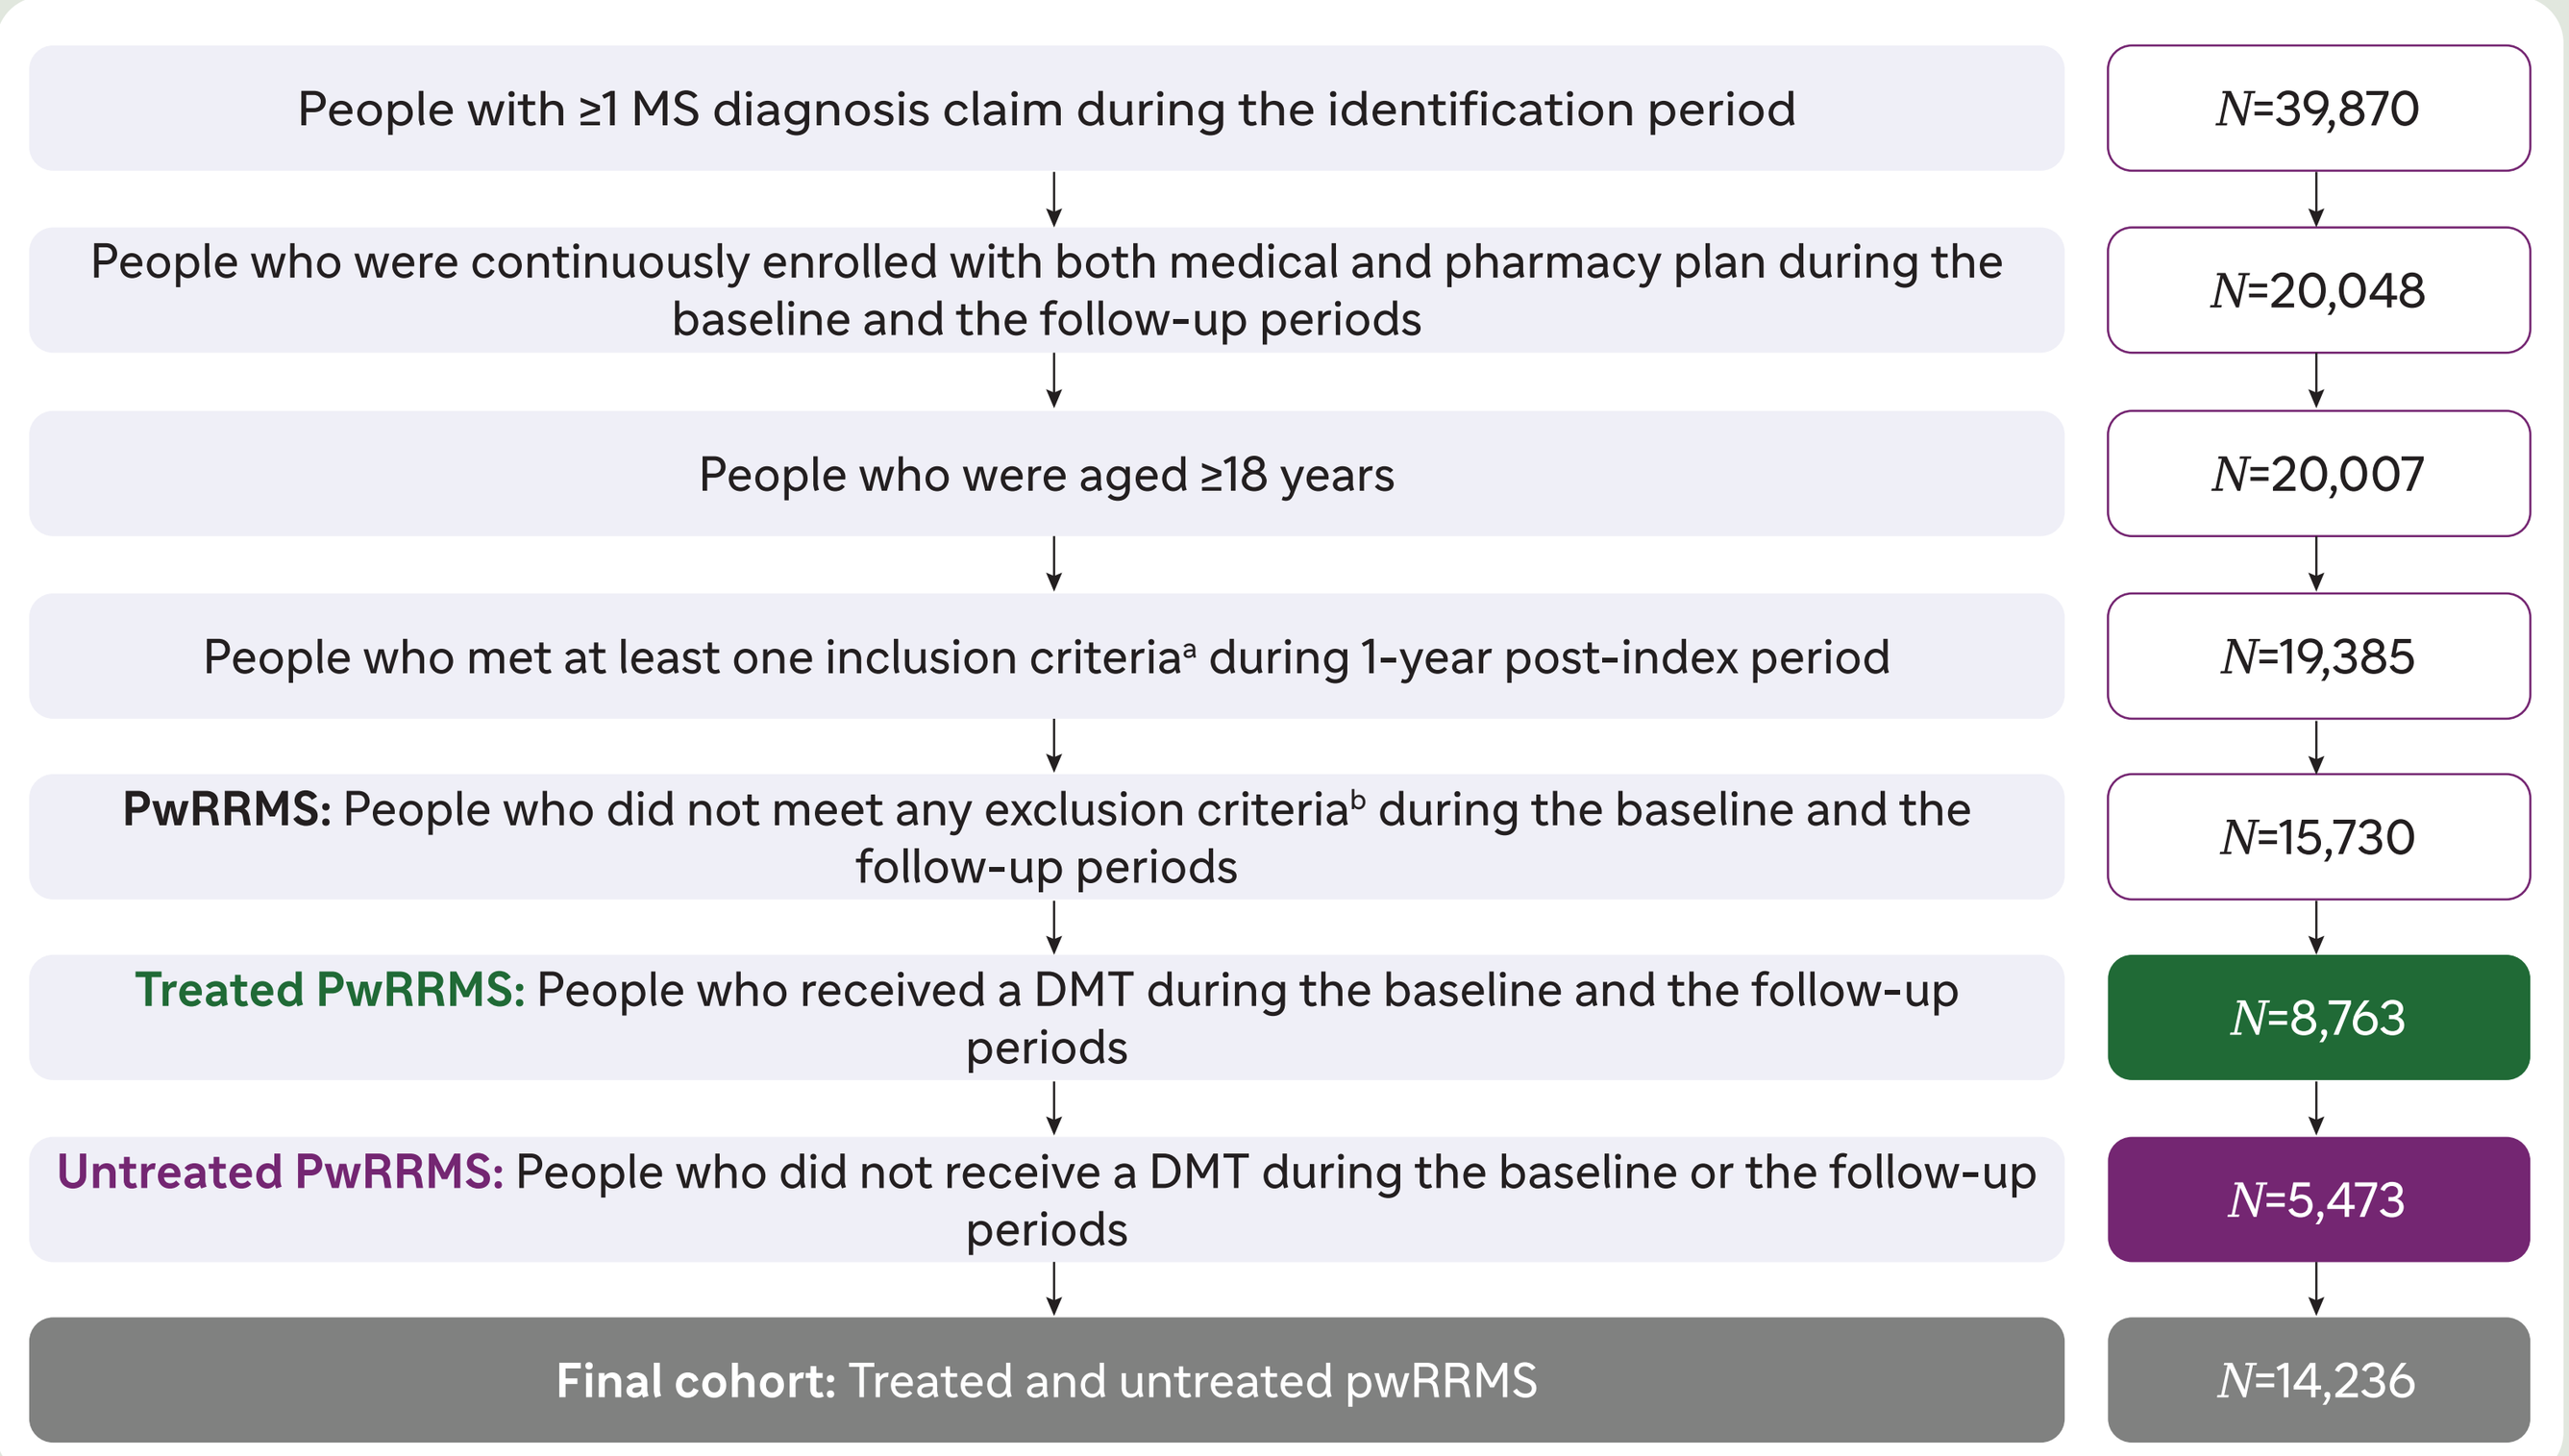
People with ≥1 MS diagnosis claim during the identification period — N=39,870
People who were continuously enrolled with both medical and pharmacy plan during the baseline and the follow-up periods — N=20,048
People who were aged ≥18 years — N=20,007
People who met at least one inclusion criteria[a] during 1-year post-index period — N=19,385
PwRRMS: People who did not meet any exclusion criteria[b] during the baseline and the follow-up periods — N=15,730
Treated PwRRMS: People who received a DMT during the baseline and the follow-up periods — N=8,763
Untreated PwRRMS: People who did not receive a DMT during the baseline or the follow-up periods — N=5,473
Final cohort: Treated and untreated pwRRMS — N=14,236

N is the total study population. [a]Inclusion criteria comprised of ≥1 claims of: DMT use; brain/spinal MRI; internuclear ophthalmoplegia ≥30 days post-MS diagnosis; MS symptom therapy; MS symptom ≥30 days post-MS diagnosis. [b]Exclusion criteria comprised of: use of mitoxantrone, cyclophosphamide and methotrexate; worsening of EDSS and evidence of exacerbations.
DMT, disease modifying therapy; EDSS, Expanded Disability Status Scale; MRI, magnetic resonance imaging; MS, multiple sclerosis; pwRRMS, people with relapsing-remitting multiple sclerosis; RRMS, relapsing-remitting multiple sclerosis.

- The pwRRMS in the untreated cohort were significantly older than those in the treated cohort (mean±standard deviation [SD] age: 53.5±13.9 vs 48.9±10.7 years; $P<0.001$). The majority were female (78.3% vs 73.6%; $P<0.001$) and commercially insured (80.3% vs 95.5%; $P<0.001$) in both groups (**Table 1**)

Table 1: Demographics of the untreated versus treated RRMS cohort

	Untreated pwRRMS (*N*=5,473)	Treated pwRRMS (*N*=8,763)	*P*-value
Mean age, years, mean±SD	53.5±13.9	48.9±10.7	<0.001
Categories, *n* (%)			<0.001
18–29	258 (4.7)	334 (3.8)	
30–39	621 (11.3)	1,357 (15.5)	
40–49	1,178 (21.5)	2,694 (30.7)	
50–59	1,568 (28.6)	2,985 (34.1)	
60–69	1,097 (20.0)	1,205 (13.8)	
70+	751 (13.7)	188 (2.1)	
Female, *n* (%)	4,287 (78.3)	6,452 (73.6)	<0.001
Region, *n* (%)			<0.001
Midwest	1,775 (32.4)	2,266 (25.9)	
Northeast	813 (14.9)	1,563 (17.8)	
South	2,214 (40.5)	3,712 (42.4)	
West	671 (12.3)	1,222 (13.9)	
Insurance type, *n* (%)			<0.001
Commercial	4,394 (80.3)	8,365 (95.5)	
Medicare	1,079 (19.7)	398 (4.5)	
Year of index date, *n* (%)			0.140
2021	2,636 (48.2)	4,332 (49.4)	
2022	2,837 (51.8)	4,431 (50.6)	

N is the total study population; *n* is the subset of pwRRMS.
pwRRMS, people with relapsing-remitting multiple sclerosis; RRMS, relapsing-remitting multiple sclerosis; SD, standard deviation.

Clinical characteristics

- The mean±SD KFSS-adapted disability scores were significantly higher in the untreated versus treated cohorts at baseline (2.1±2.5 vs 1.6±2.3; $P<0.001$) and follow-up (1.9±2.4 vs 1.3±2.1; $P<0.001$)
 - The use of ambulatory devices (e.g., cane, walker, wheelchair) was also significantly higher in the untreated versus treated cohorts at baseline (14.1% vs 12.0%; $P<0.001$) and follow-up (14.6% vs 11.6%; $P<0.001$)
- During the follow-up period, the overall relapse rates for the untreated and the treated cohorts were 15.3% and 16.9%, respectively ($P=0.012$)
- The untreated cohort reported significantly higher rate of infections (54.3% vs 50.6%; $P<0.001$) and infections during inpatient hospitalization (3.7% vs 2.0%; $P<0.001$) during the follow-up period when compared with the treated cohort

Specific comorbidities of interest

- The mean±SD CCI score at baseline was significantly higher for the untreated versus treated cohorts (1.0±1.7 vs 0.5±1.1; $P<0.001$)
- The top five most common MS-related comorbidities among the treated versus untreated cohorts at baseline were malaise/fatigue, depression, burning/numbness/tingling, anxiety and abnormal gait (**Figure 3**)

Figure 3: Most frequent MS-related comorbidities in the treated versus untreated RRMS cohorts at baseline

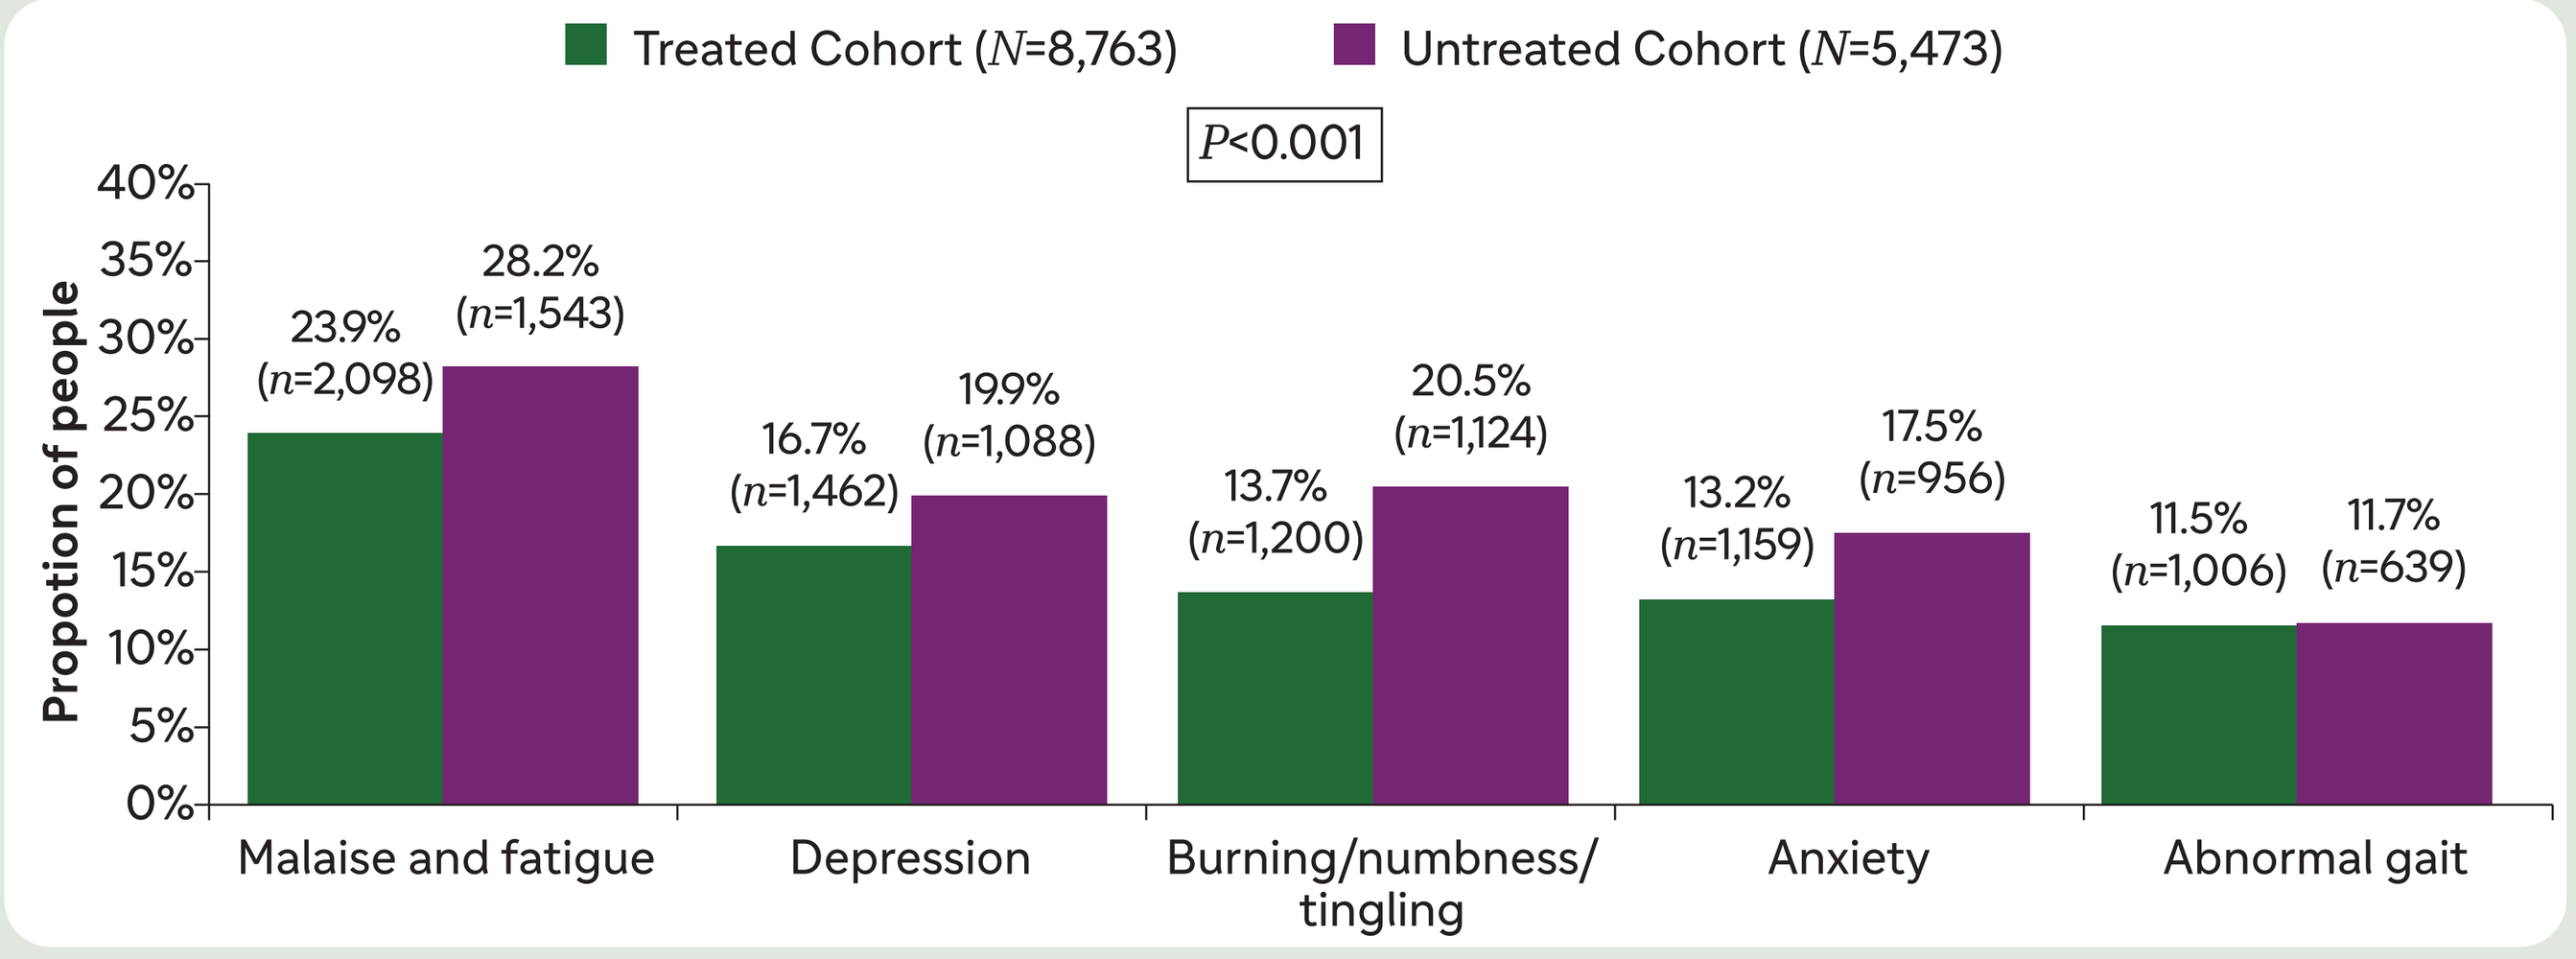
Treated Cohort (N=8,763)
Untreated Cohort (N=5,473)
P<0.001
Propotion of people
40%
35%
30%
25%
20%
15%
10%
5%
0%
23.9% (n=2,098)
28.2% (n=1,543)
16.7% (n=1,462)
19.9% (n=1,088)
13.7% (n=1,200)
20.5% (n=1,124)
13.2% (n=1,159)
17.5% (n=956)
11.5% (n=1,006)
11.7% (n=639)
Malaise and fatigue
Depression
Burning/numbness/ tingling
Anxiety
Abnormal gait

N is the total study population; *n* is the subset of pwRRMS.
MS, multiple sclerosis; pwRRMS, people with relapsing-remitting multiple sclerosis; RRMS, relapsing-remitting multiple sclerosis.

Healthcare resource utilization and healthcare costs

- During follow-up, rates of rehabilitation services, hospitalizations, ED visits (all $P<0.001$), and mean number of physician visits ($P=0.002$) remained significantly higher for untreated pwRRMS as compared to treated pwRRMS (**Figure 4**)

Figure 4: All-cause healthcare resource utilization in treated vs untreated RRMS cohorts at 1-year follow-up

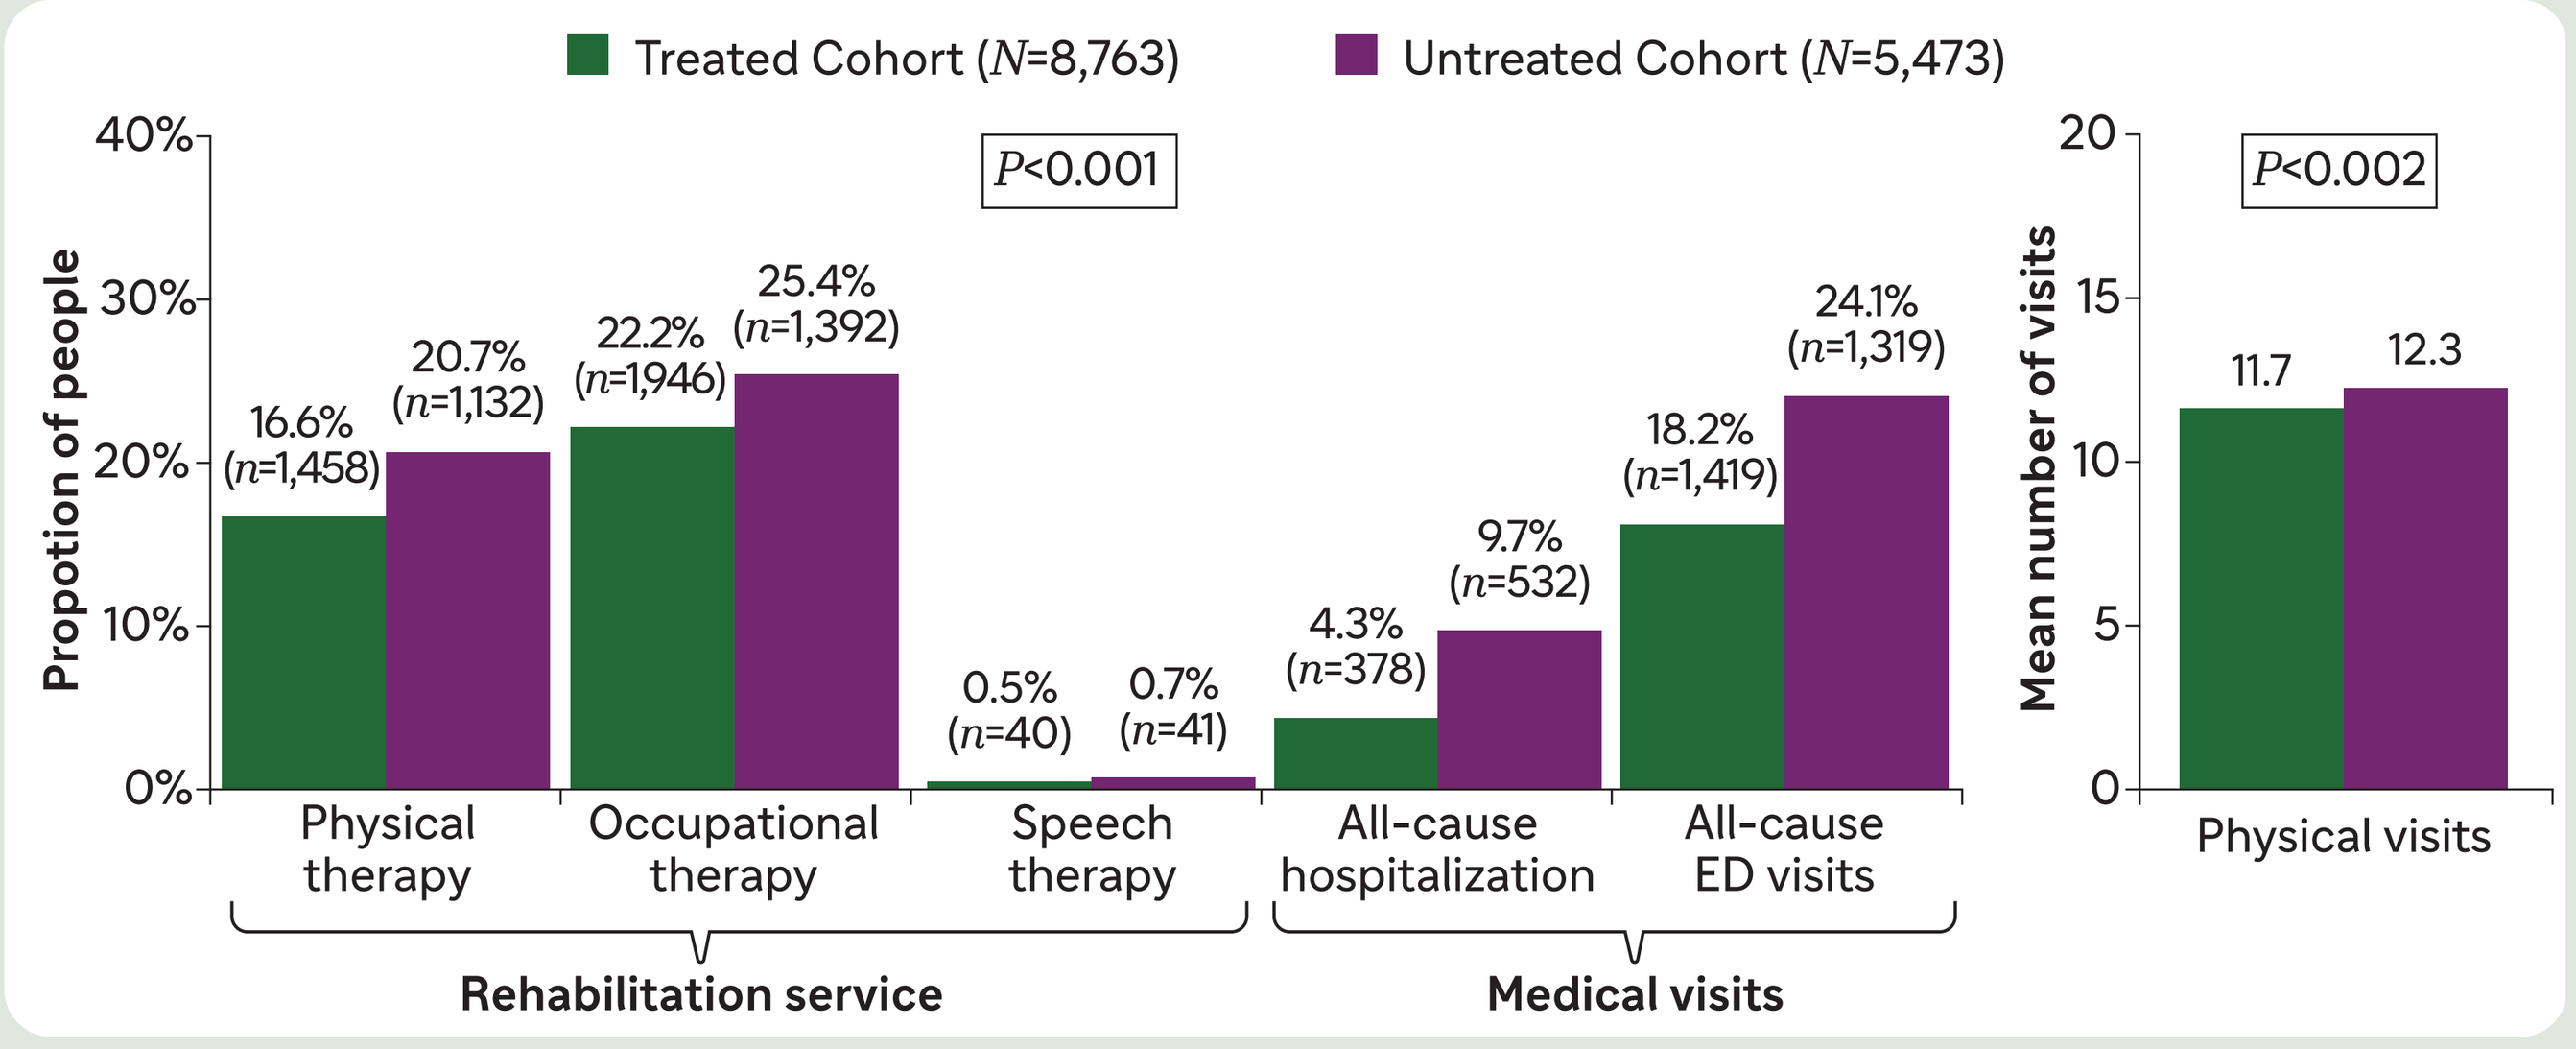
Treated Cohort (N=8,763)
Untreated Cohort (N=5,473)
P<0.001
Propotion of people
40%
30%
20%
10%
0%
16.6% (n=1,458)
20.7% (n=1,132)
22.2% (n=1,946)
25.4% (n=1,392)
0.5% (n=40)
0.7% (n=41)
4.3% (n=378)
9.7% (n=532)
18.2% (n=1,419)
24.1% (n=1,319)
Physical therapy
Occupational therapy
Speech therapy
All-cause hospitalization
All-cause ED visits
Rehabilitation service
Medical visits
P<0.002
Mean number of visits
20
15
10
5
0
11.7
12.3
Physical visits

N is the total study population; *n* is the subset of pwRRMS.
ED, emergency department; pwRRMS, people with relapsing-remitting multiple sclerosis; RRMS, relapsing-remitting multiple sclerosis.

- The mean total all-cause HCC was $82,985 for treated pwRRMS and $19,238 for untreated pwRRMS, with medical claims and outpatient pharmacy claims being the primary cost drivers in both cohorts (all $P<0.001$; **Figure 5**)
- The mean all-cause costs of ED visits ($617 vs $365) and inpatient hospitalizations ($3,473 vs $1,621) were significantly higher in the untreated cohort versus treated cohort (all $P<0.001$)

Figure 5: All-cause healthcare costs in treated vs untreated RRMS cohorts at 1-year follow-up (adjusted to Year 2023)

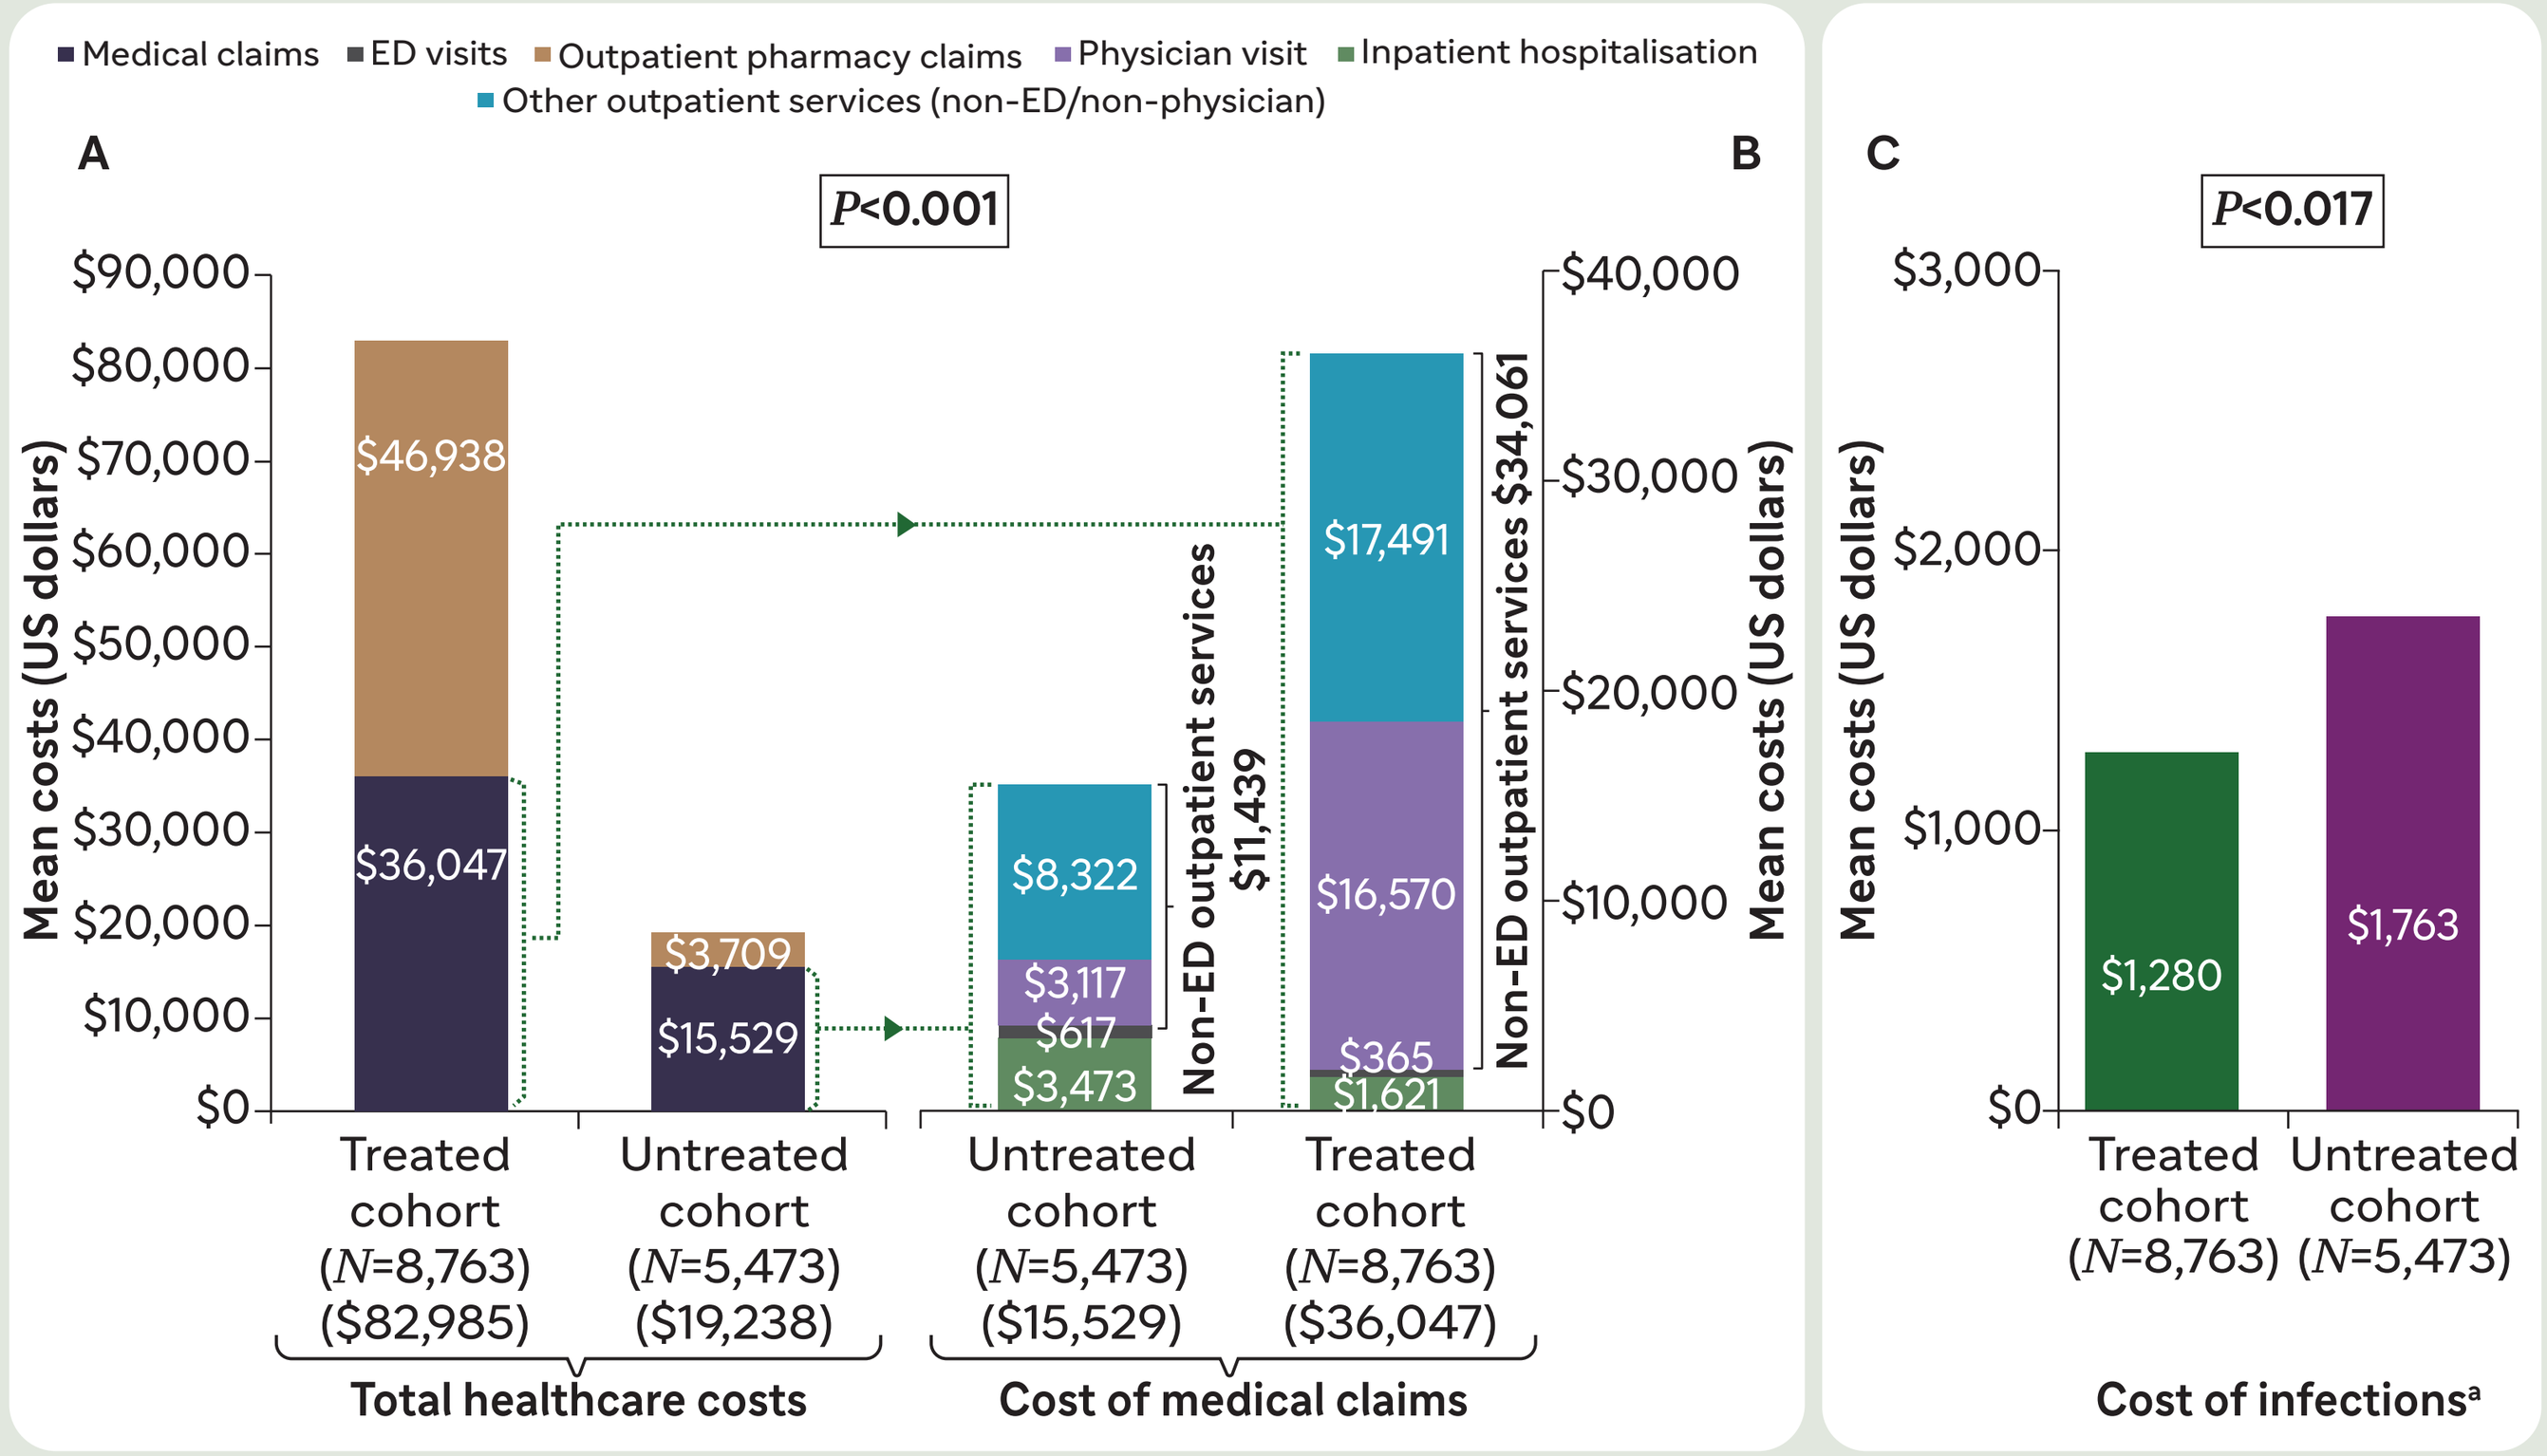
Medical claims
ED visits
Outpatient pharmacy claims
Physician visit
Inpatient hospitalisation
Other outpatient services (non-ED/non-physician)
A
B
P<0.001
Mean costs (US dollars)
$90,000
$80,000
$70,000
$60,000
$50,000
$40,000
$30,000
$20,000
$10,000
$0
$46,938
$36,047
$3,709
$15,529
$8,322
$3,117
$617
$3,473
Non-ED outpatient services $11,439
$17,491
$16,570
$365
$1,621
Non-ED outpatient services $34,061
$40,000
$30,000
$20,000
$10,000
$0
Treated cohort (N=8,763) ($82,985)
Untreated cohort (N=5,473) ($19,238)
Untreated cohort (N=5,473) ($15,529)
Treated cohort (N=8,763) ($36,047)
Total healthcare costs
Cost of medical claims
C
P<0.017
$3,000
$2,000
$1,000
$0
$1,280
$1,763
Treated cohort (N=8,763)
Untreated cohort (N=5,473)
Cost of infections[a]

N is the total study population. Data represented as mean costs.
[a]Costs of medical claims with a diagnosis of infections in any field plus the costs of antibiotics or antivirals pharmacy claims with days of supply <21 days filled within 7 days of an infection medical claim.
ED, emergency department; RRMS, relapsing-remitting multiple sclerosis; US, United States.

Risk factors associated with not receiving DMT among pwRRMS

- Among the factors significantly associated with not receiving DMT, the presence of stroke, and fibromyalgia and myositis had the largest impact with an 85% increase in odds (**Figure 6**)

Figure 6: Risk factors for not receiving DMT in untreated pwRRMS[a]

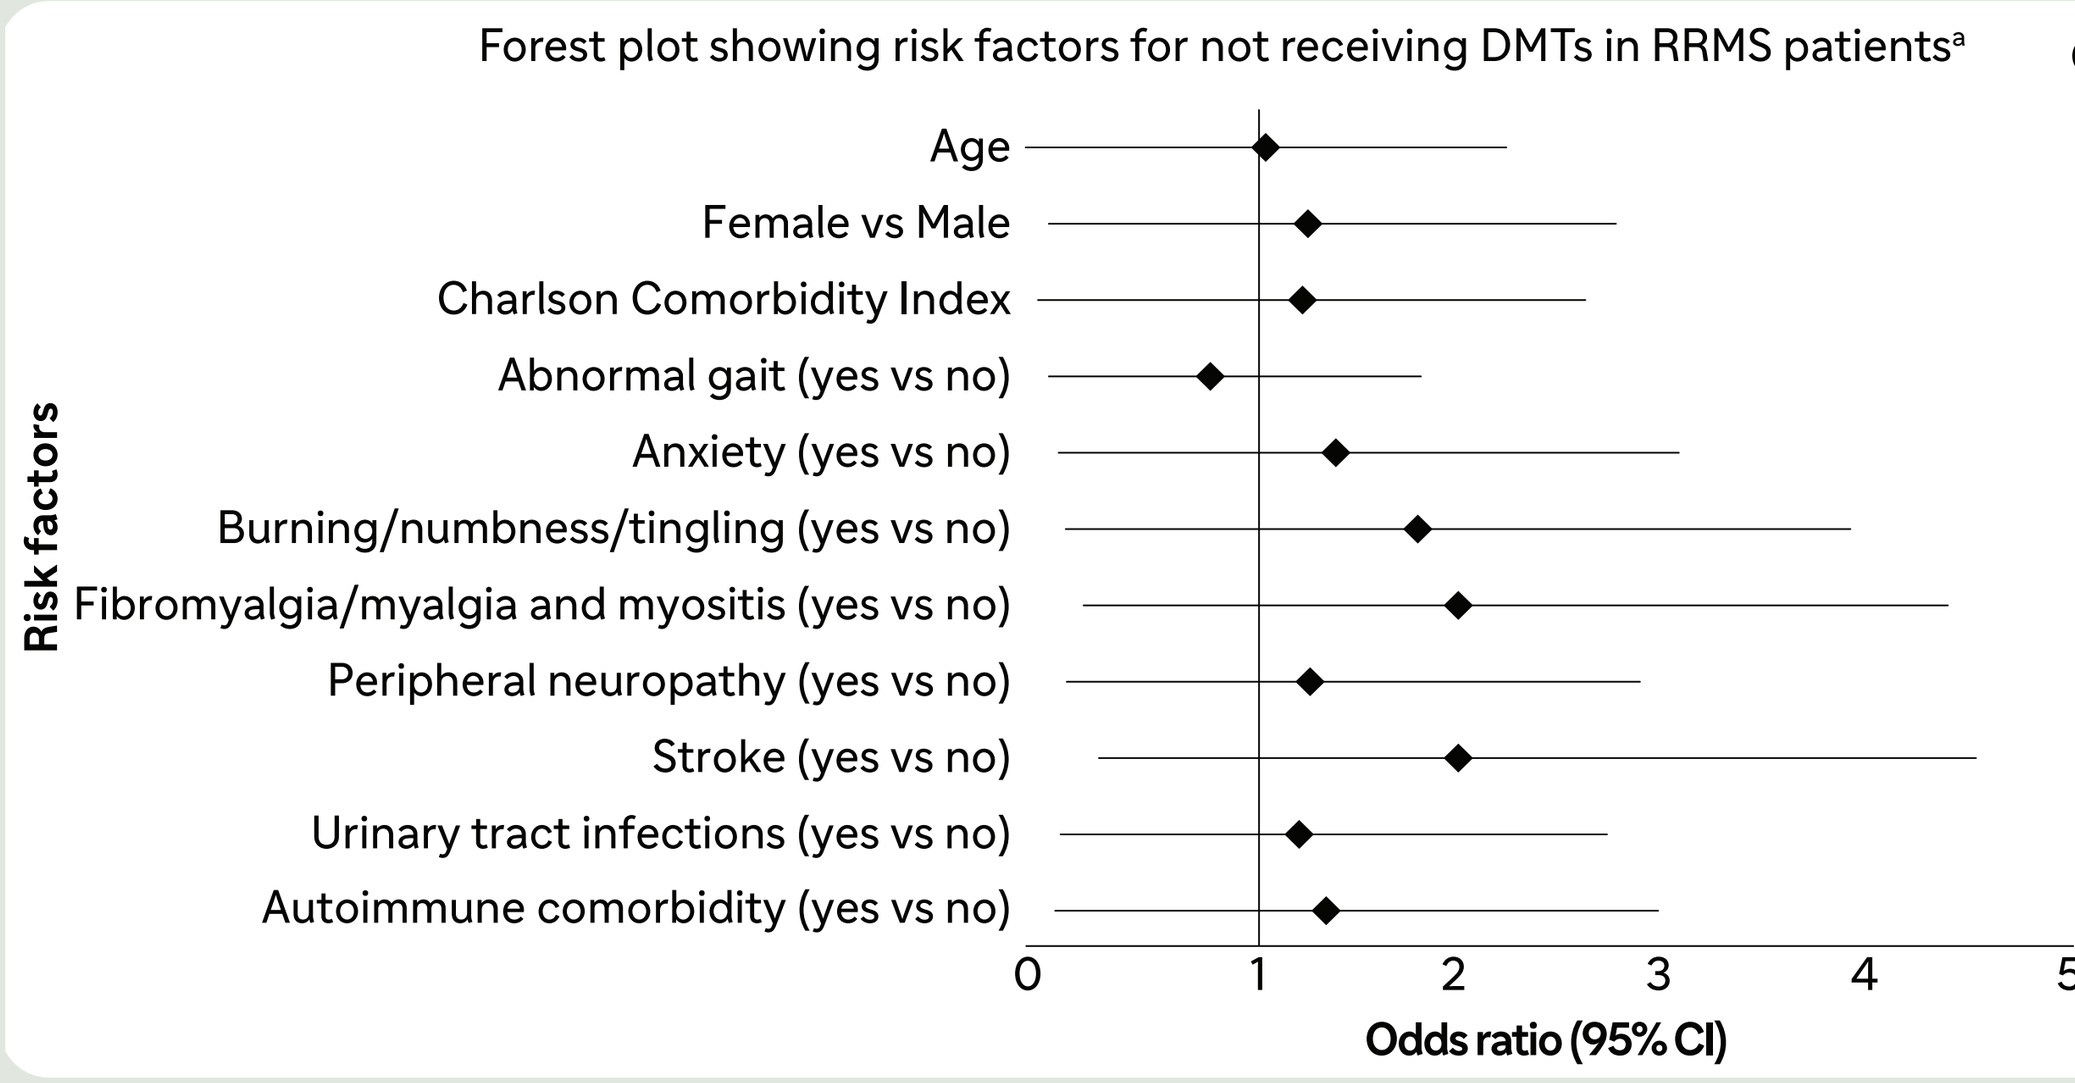
Forest plot showing risk factors for not receiving DMTs in RRMS patients[a]
Risk factors
Odds ratio (95% CI)
0 1 2 3 4 5

Risk factor	Odds ratio (95% CI)	*P*-value
Age	1.03 (1.03–1.03)	<0.001
Female vs Male	1.21 (1.12–1.32)	<0.001
Charlson Comorbidity Index	1.18 (1.14–1.22)	<0.001
Abnormal gait (yes vs no)	0.79 (0.70–0.90)	<0.001
Anxiety (yes vs no)	1.33 (1.20–1.47)	<0.001
Burning/numbness/tingling (yes vs no)	1.68 (1.52–1.85)	<0.001
Fibromyalgia/myalgia and myositis (yes vs no)	1.85 (1.61–2.11)	<0.001
Peripheral neuropathy (yes vs no)	1.22 (1.05–1.41)	0.008
Stroke (yes vs no)	1.85 (1.54–2.23)	<0.001
Urinary tract infections (yes vs no)	1.17 (1.03–1.32)	0.013
Autoimmune comorbidity (yes vs no)	1.29 (1.17–1.42)	<0.001

[a]Includes only significant variables using forward selection method.
CI, confidence interval; DMT, disease modifying therapy; pwRRMS, people with relapsing-remitting multiple sclerosis; RRMS, relapsing-remitting multiple sclerosis.

LIMITATIONS

- As with all administrative claims database analyses, our study is inherently subject to potential miscoding, which could influence accurate identification and outcome assessment
- The findings may not be generalizable to populations not well represented in commercial claims databases, including uninsured individuals and adults aged over 65 years
- The use of an algorithm to identify the RRMS population may have resulted in misclassification of patients across MS subtypes
- Reliance on claims-based proxy measures for relapse identification limits the capture of clinically confirmed disease activity and outcomes

CONCLUSIONS

- The untreated cohort had a higher comorbidity burden and medical costs, and greater HCRU, versus the treated cohort, highlighting the potential clinical and economic benefits of treatment whilst underscoring the continued unmet need in untreated pwRRMS
- Stroke and fibromyalgia/myositis were identified as key risk factors associated with not receiving DMT

Copies of this poster obtained through Quick Response (QR) code are for personal use only

Disclosures
Nupur Greene, Lita Araujo and **Jaja Sangbana:** Employees of Sanofi and may hold stock or stock options in the company.
Ashis K. Das, Eunice Chang, Marian H. Tarbox, Melinda Rossi and **Michael S. Broder:** At the time the study was conducted, employees of PHAR (now a part of ADVI Health), which was paid by Sanofi to conduct the research described in this abstract. PHAR also discloses financial relationships with the following commercial entities outside of the submitted work: Akcea, Amgen, Celgene, Delfi Diagnostics, Dompe, Exact Sciences Corporation, Genentech, Gilead, GRAIL, Greenwich Biosciences, Ionis, Nobelpharma, Novartis, Pardes, Prothena, Pfizer, Recordati, Regeneron, Sanofi US Services, and Sunovion.

Acknowledgments
Medical writing and editorial assistance were provided by Dhaval Srivastava and Shreya Dam of Sanofi. Planning, review, and coordination assistance was provided by Leah Granby of Sanofi.

Funding
This study was sponsored by Sanofi.

References
1. Atlas of MS Database. http://www.atlasofms.org/ (accessed 02 Feb 2026).
2. Wallin MT, et al. *Neurology*. 2019;92(10):e1029–40.
3. Vasanthaprasad V, et al. *BMC Neurol.* 2022;23:301.
4. Finkener S, et al. *Mult Scler Relat Disord*. 2022;68:104396.
5. Köhler M, et al. *BMC Neurol*. 2025;25:328.
6. Van Le H, et al. *Value Health*. 2019;22(1):77–84.

Poster presented at the American Academy of Neurology (AAN) Annual Meeting 2026; Chicago, IL, US; April 18–22, 2026.