High- or Moderate-Efficacy Treatment Sequence and Patterns Among People with Relapsing-Remitting Multiple Sclerosis in the United States: A Retrospective Cohort Study

DMT35

Lita Araujo[1], Jaja Sangbana[1], Nupur Greene[1], Marian H. Tarbox[2], Michelle Baglia[1], Roopali Gandhi[1], Ashis K. Das[2]

[1]Sanofi, Cambridge, MA, USA; [2]ADVI Health, Washington, DC, USA

BACKGROUND

- High-efficacy (HE) and moderate-efficacy (ME) disease-modifying therapies (DMTs) are widely used for relapsing-remitting multiple sclerosis (RRMS)[1]
- Despite the proven efficacy of DMTs, treatment persistence in real-world settings is suboptimal, with considerable discontinuation and switching[2]
- These challenges were likely compounded during the coronavirus disease-19 (COVID-19) pandemic, where concerns related to certain multiple sclerosis (MS) DMTs that were associated with an increased risk of severe COVID-19 may have contributed to disruptions in DMT initiation and continuation[3]

OBJECTIVE

To describe treatment sequencing and patterns of DMT use among people with RRMS (pwRRMS), to characterize unmet needs related to DMT discontinuation and switching, and to evaluate safety-related factors associated with the therapies in the United States (US)

METHODS

Study design and population

- A retrospective cohort study was conducted using Merative™ MarketScan®, which is a large, US-administrative claims database, from January 01, 2018, to June 30, 2023 (**Figure 1**)
- PwRRMS who were newly treated with HE-DMTs (anti-cluster of differentiation 20 [anti-CD20] monoclonal antibodies and natalizumab) or ME-DMTs (sphingosine-1-phosphate [S1P] receptor modulators) during the study period were characterized based on specific inclusion and exclusion criteria (**Figure 2**)
- The first HE- or ME-DMT initiated was defined as the index class, and the initiation date of these DMTs as the index date
- PwRRMS were naïve to the index class, but not necessarily DMT-naïve overall during the baseline period (12-months prior to the index date)
- All pwRRMS were followed up for 2-years after the index date

Figure 1: Study time frame

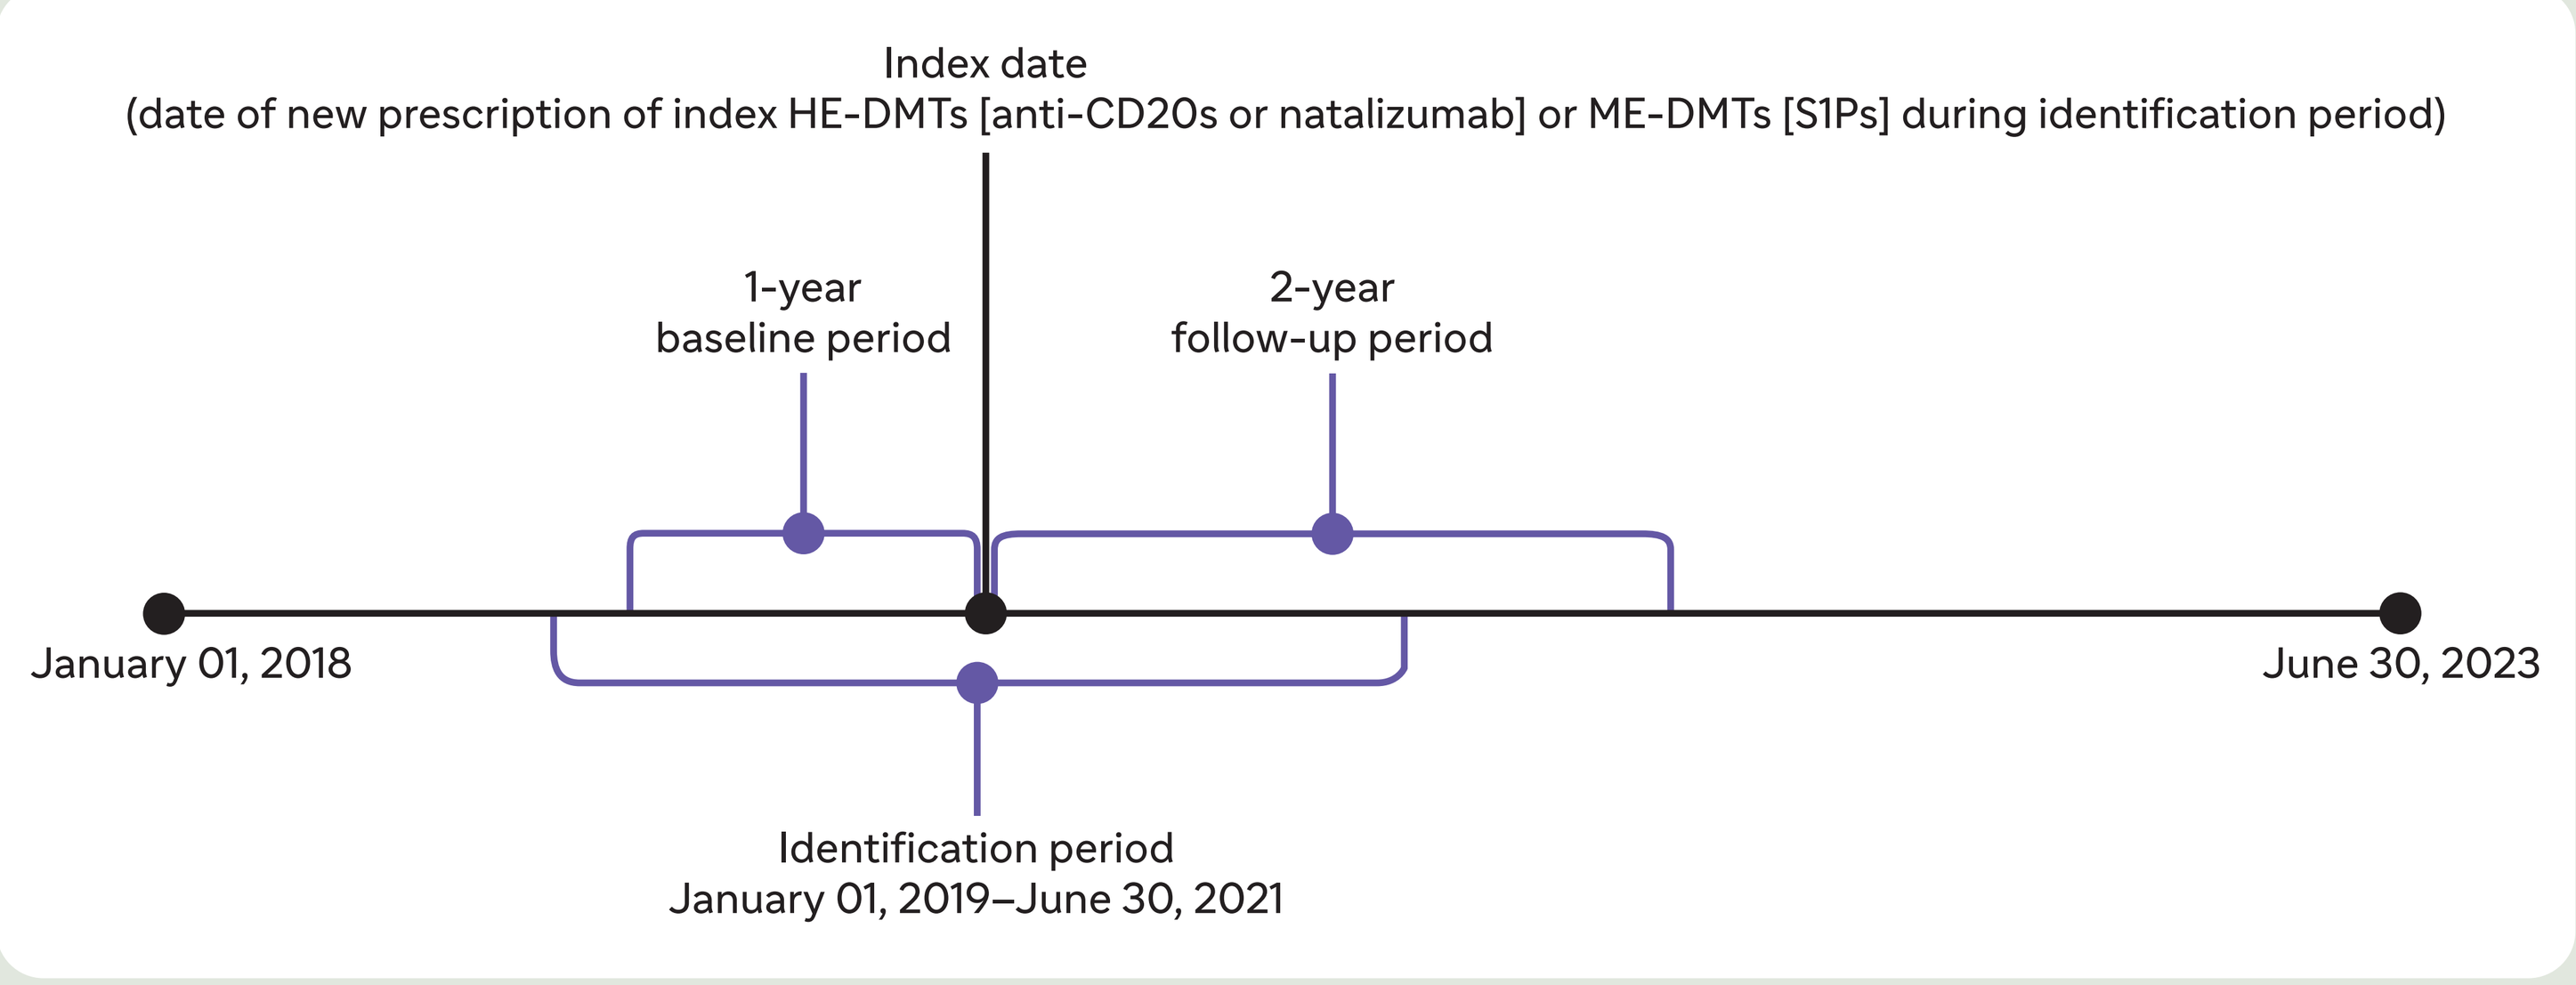
Index date
(date of new prescription of index HE-DMTs [anti-CD20s or natalizumab] or ME-DMTs [S1Ps] during identification period)
1-year
baseline period
2-year
follow-up period
January 01, 2018
June 30, 2023
Identification period
January 01, 2019–June 30, 2021

CD, cluster of differentiation; DMTs, disease-modifying therapies; HE, high-efficacy; ME, moderate-efficacy; S1P, sphingosine-1-phosphate.

Study measures

- Demographics of pwRRMS treated with either HE- or ME-DMT were reported at baseline
- Treatment patterns of pwRRMS were evaluated over a 2-year follow-up period and included:
 - Proportion of people who remained on their index DMT class at the end of follow-up
 - Discontinuation, defined as stopping the index DMT with no subsequent DMT initiated within follow-up
 - Switching, defined as initiation of a new DMT class within ≤60 days of the end of the prior DMT
 - Switch sequences, including first and second post index DMT classes
- Safety-related clinical outcomes within 60 days of DMT change (including infections, optic neuritis, autoimmune comorbidity and others) were also assessed in this population
 - The 60-day window reflects a clinically relevant decision horizon for treatment change; captured events may reflect disease activity, treatment-related adverse events, or unrelated comorbid conditions

Statistical analysis

- Study measures were summarized using descriptive statistical methods
- All data transformations and statistical analyses were performed using SAS© version 9.4

RESULTS

- The final sample comprised 1,713 pwRRMS (HE-DMTs: 81.6%, n=1,398; ME-DMTs: 18.4%, n=315) (**Figure 2**)

Figure 2: Attrition table for pwRRMS who were newly treated with anti-CD20s, S1Ps, or natalizumab

People with ≥1 MS diagnosis claim during the identification period (January 01, 2019–June 30, 2021)	N=61,533
People who were newly treated with either anti-CD20 or S1P, or natalizumab during the identification period (January 01, 2019–June 30, 2021)	N=9,480
People who were continuously enrolled with both medical and pharmacy plan during baseline (1-year pre-index) and follow-up (2-years post-index) periods	N=2,308
People who were aged ≥18 years	N=2,287
People who met at least one of the inclusion[a] criteria during the baseline or follow-up periods	N=2,287
People who did not meet any of the exclusion[b] criteria during the baseline or follow-up periods	N=1,713

N is the total study population.
[a]≥1 of the following inclusion criteria: DMT claim; brain/spinal MRI; internuclear ophthalmoplegia ≥30 days post-MS diagnosis; MS symptom therapy; MS symptoms ≥30 days post-MS diagnosis.
[b]Exclusion criteria: progressive MS therapies (mitoxantrone, cyclophosphamide, methotrexate); EDSS worsening; exacerbations.
CD, cluster of differentiation; DMT, disease-modifying therapy; EDSS, Expanded Disability Status Scale; MRI, magnetic resonance imaging; MS, multiple sclerosis; pwRRMS; people with relapsing-remitting multiple sclerosis; S1P, sphingosine-1-phosphate.

Baseline demographics

- The majority of pwRRMS who received HE-DMTs and ME-DMTs were aged 40–49 years (34.5% and 34.3%) and were females (71.9% and 73.7%), respectively (**Table 1**)

Table 1: Demographics of pwRRMS receiving HE-DMTs and ME-DMTs

	Index HE-DMTs (Anti-CD20s or Natalizumab; N=1,398)	Index ME-DMTs (S1Ps; N=315)
Age, year, n (%)		
18–29	147 (10.5%)	28 (8.9%)
30–39	343 (24.5%)	80 (25.4%)
40–49	482 (34.5%)	108 (34.3%)
50–59	328 (23.5%)	74 (23.5%)
60–69	91 (6.5%)	25 (7.9%)
70+	7 (0.5%)	0 (0.0%)
Female, n (%)	1,005 (71.9%)	232 (73.7%)
Region, n (%)		
Midwest	367 (26.3%)	82 (26.0%)
Northeast	240 (17.2%)	42 (13.3%)
South	610 (43.6%)	164 (52.1%)
West	181 (12.9%)	27 (8.6%)
Insurance type, n (%)		
Commercial	1,358 (97.1%)	309 (98.1%)
Medicare	38 (2.7%)	0 (0%)
Medicaid	2 (0.1%)	6 (1.9%)
Year of index date, n (%)		
2019	568 (40.6%)	157 (49.8%)
2020	481 (34.3%)	94 (29.8%)
2021	349 (25.0%)	64 (20.3%)

N is the total study population of the cohort/group; n is the subset of total study population of the cohort/group.
CD, cluster of differentiation; DMTs, disease-modifying therapies; HE, high-efficacy; ME, moderate-efficacy; pwRRMS; people with relapsing-remitting multiple sclerosis; S1P, sphingosine-1-phosphate.

- Among pwRRMS treated with HE-DMTs (N=1,398) at the index date, 84.9% of people received anti-CD20s (n=1,187) and 15.1% of people received natalizumab (n=211)
- Among pwRRMS treated with HE-DMTs at the index date, the majority (74.5%; n=1,041) were treated with ocrelizumab, an anti-CD20; whereas, among those who were treated with ME-DMTs, majority (65.7%; n=207) were treated with fingolimod, an S1P

Treatment discontinuation and switching

- During the 2-year follow-up, discontinuation occurred in 35.2% of pwRRMS on HE-DMTs and 22.5% on ME-DMTs, while switching was more frequent with ME-DMTs (21.3%) than with HE-DMTs (6.2%) (**Figure 3**)

Figure 3: Treatment pattern during the 2-year follow-up period

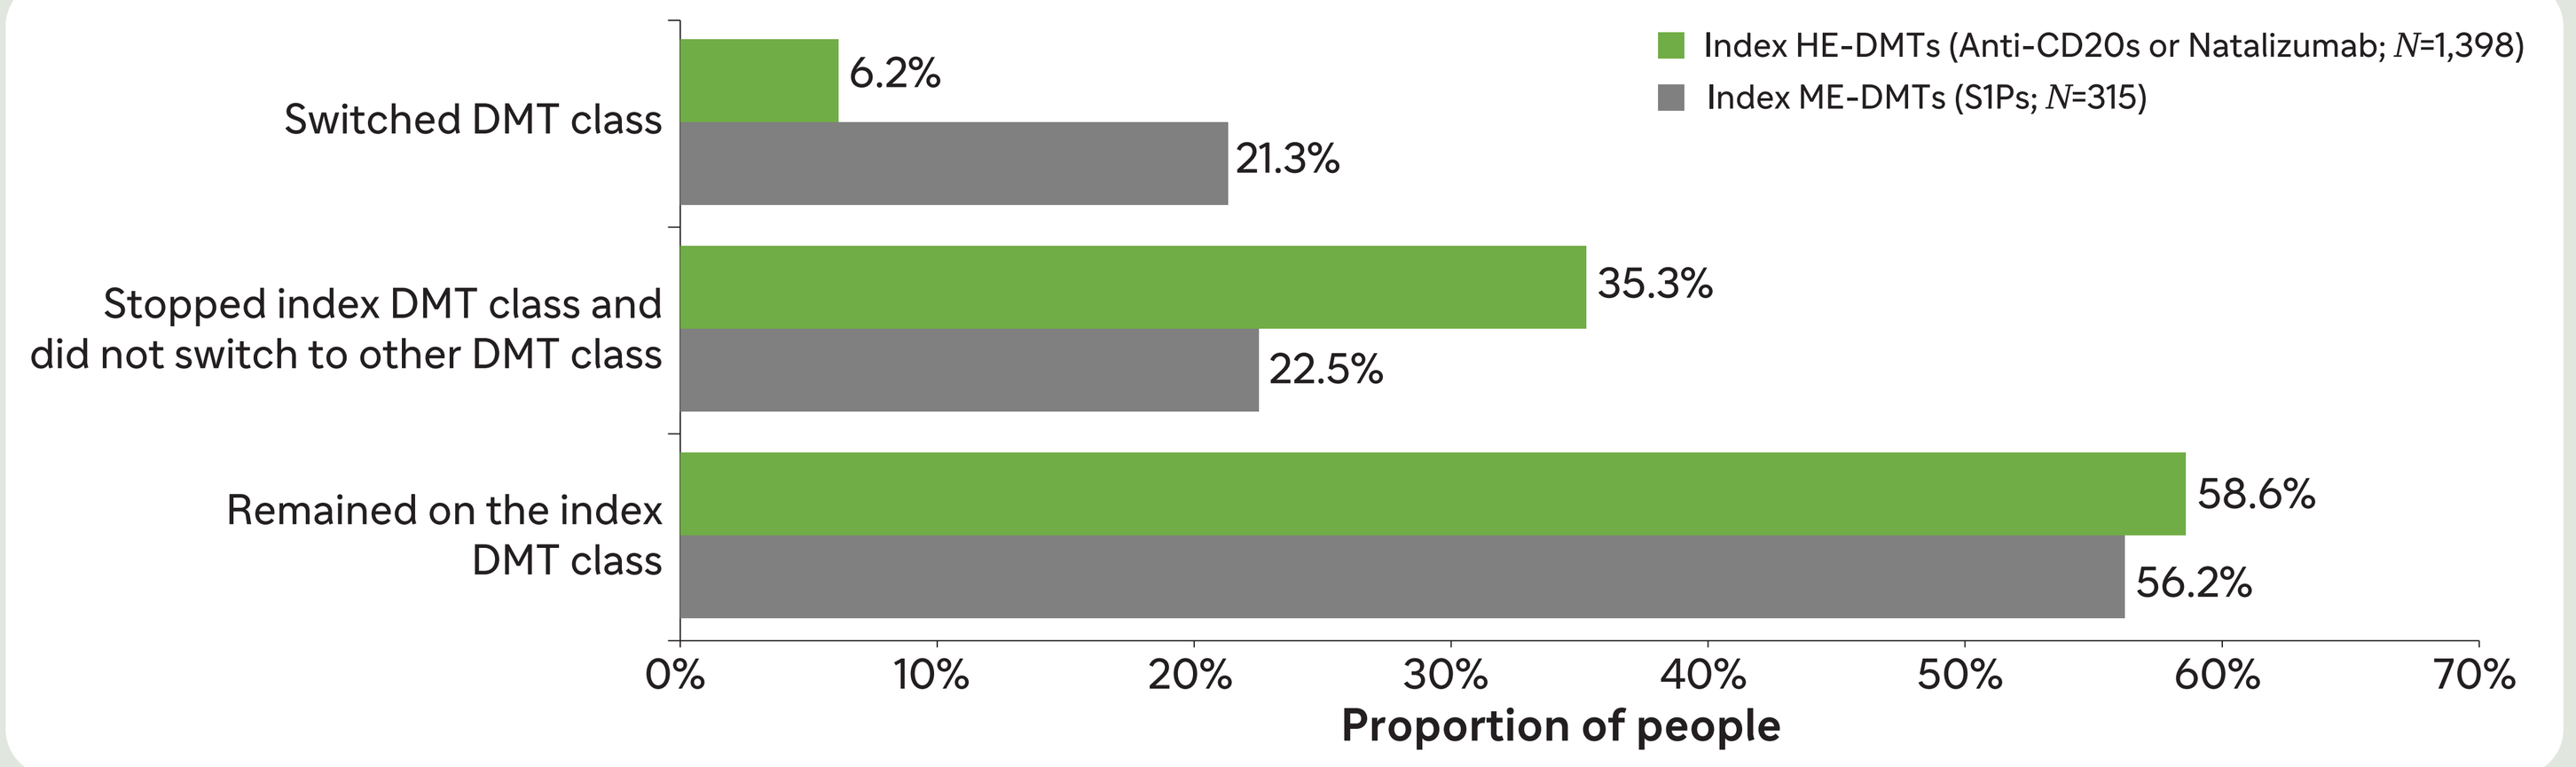
Index HE-DMTs (Anti-CD20s or Natalizumab; N=1,398)
Index ME-DMTs (S1Ps; N=315)
Switched DMT class
6.2%
21.3%
Stopped index DMT class and did not switch to other DMT class
35.3%
22.5%
Remained on the index DMT class
58.6%
56.2%
0%
10%
20%
30%
40%
50%
60%
70%
Proportion of people

CD, cluster of differentiation; DMTs, disease-modifying therapies; HE, high-efficacy; ME, moderate-efficacy; S1P, sphingosine-1-phosphate.

- In the HE-DMTs group, anti-CD20 initiators switched to other DMTs (3.0%), while natalizumab initiators mostly switched to anti-CD20 therapies (8.0%) or other DMTs (7.0%) (**Table 2**)
- In the ME-DMTs group, the majority of pwRRMS switched to anti-CD20 therapies (11.0%), followed by other DMTs (6.0%) (**Table 2**)

Table 2: Switch sequence during the 2-year follow-up period

	Index HE-DMTs (Anti-CD20s or Natalizumab; N=1,398)				Index ME-DMTs	
	Anti-CD20s (N=1,187)		Natalizumab (N=211)		S1Ps (N=315)	
1st switch	anti-CD20 → Other DMT[a]	41 (3.0%)	Natalizumab → anti-CD20	16 (8.0%)	S1P → anti-CD20	34 (11.0%)
	anti-CD20 → S1P	6 (0.5%)	Natalizumab → Other DMT[a]	14 (7.0%)	S1P → Other DMT[a]	19 (6.0%)
	anti-CD20 → Natalizumab	5 (0.4%)			S1P → Natalizumab	7 (2.0%)
2nd switch	anti-CD20 → Other DMT[a] → Natalizumab	2 (0.2%)	Natalizumab → anti-CD20 → Other DMT[a]	1 (0.5%)	S1P → Other DMT[a] → anti-CD20	5 (2.0%)
	anti-CD20 → S1P → Other DMT[a]	1 (0.1%)			S1P → Natalizumab → Other DMT[a]	1 (0.3%)
					S1P → Other DMT[a] → Natalizumab	1 (0.3%)

Data represented as n (%); → indicates a switch.
N is the total study population of the cohort/group; n is the subset of total study population of the cohort/group.
[a]Includes alemtuzumab, beta-interferon, cladribine, daclizumab, dimethyl fumarate, diroximel fumarate, immunoglobulin, glatiramer acetate, monomethyl fumarate, and teriflunomide.
CD, cluster of differentiation; DMTs, disease-modifying therapies; HE, high-efficacy; ME, moderate-efficacy; S1P, sphingosine-1-phosphate.

- Among individuals who switched to other DMTs, HE-initiators mainly switched to cladribine (24.6%), whereas ME-initiators primarily switched to teriflunomide (36.0%) (**Figure 4**)

Figure 4: Switch patterns to "other" DMTs during the 2-year follow-up period

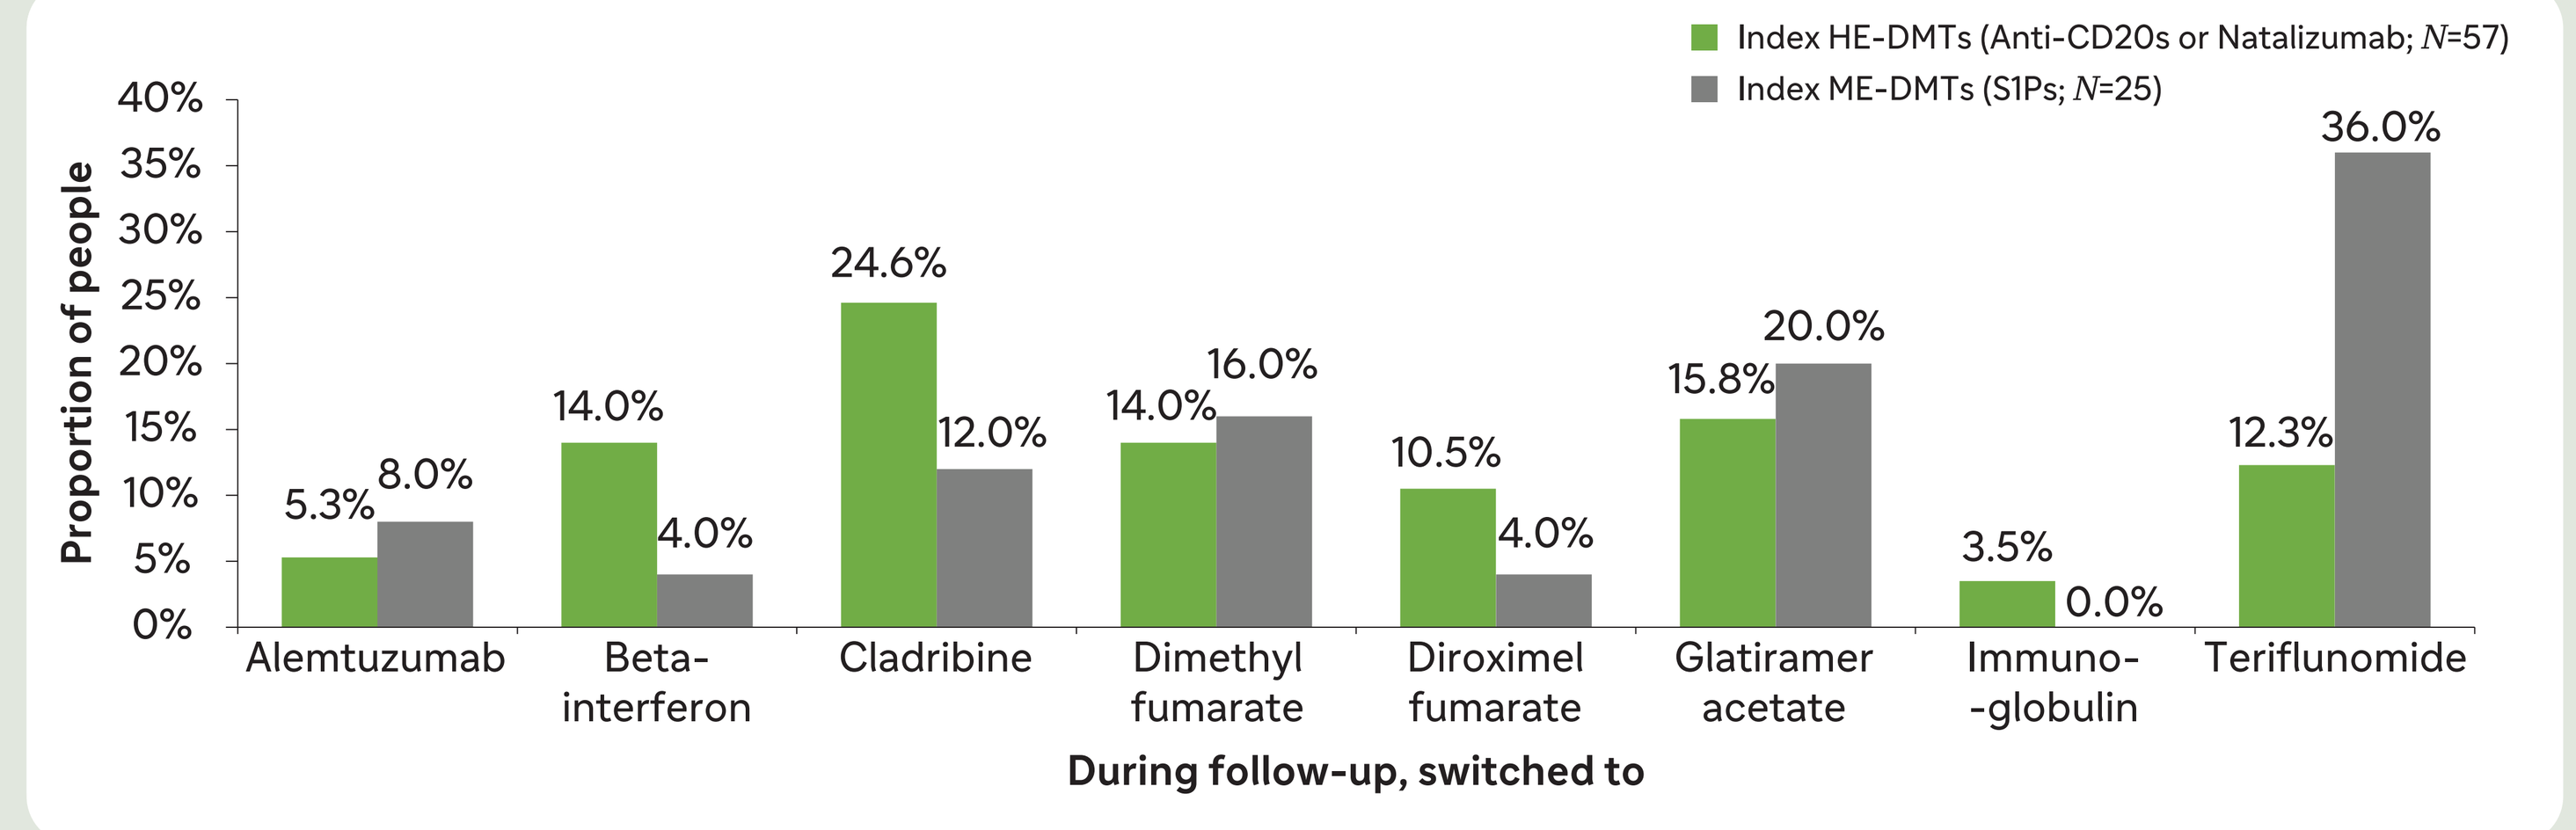
Index HE-DMTs (Anti-CD20s or Natalizumab; N=57)
Index ME-DMTs (S1Ps; N=25)
Proportion of people
40%
35%
30%
25%
20%
15%
10%
5%
0%
5.3%
8.0%
14.0%
4.0%
24.6%
12.0%
14.0%
16.0%
10.5%
4.0%
15.8%
20.0%
3.5%
0.0%
12.3%
36.0%
Alemtuzumab
Beta-interferon
Cladribine
Dimethyl fumarate
Diroximel fumarate
Glatiramer acetate
Immuno--globulin
Teriflunomide
During follow-up, switched to

N is the total study population of the cohort/group.
CD, cluster of differentiation; DMTs, disease-modifying therapies; HE, high-efficacy; ME, moderate-efficacy; S1P, sphingosine-1-phosphate.

- Switching and discontinuation patterns may not show a strong age-related impact, possibly due to the younger population captured in MarketScan® database

Safety-related clinical outcomes

- Safety-related outcomes were observed within the 60 days prior to switching (HE-DMTs: 43.9%; ME-DMTs: 56.0%), as well as prior to discontinuation (HE-DMTs: 25.4%; ME-DMTs: 23.9%), with infections being the most common in both groups (**Table 3**)
 - Among pwRRMS who stopped the index DMT, about a quarter of them reported safety-related clinical outcomes within 60 days before stopping treatment
 - Among pwRRMS who switched to other DMTs, about half of them reported safety-related clinical outcomes within 60 days before switching

Table 3: Safety-related clinical outcomes during 60 days before or on the stopping/switching date

	Index HE-DMTs (Anti-CD20s or Natalizumab)	Index ME-DMTs (S1Ps)
Stopped the index DMT and were not on any treatment by the end of follow-up (N=564), n (%)	**N=493**	**N=71**
Any safety-related clinical outcomes within 60 days before or on the stopping date	125 (25.4%)	17 (23.9%)
Infections	84 (17.0%)	13 (18.3%)
Optic neuritis	23 (4.7%)	2 (2.8%)
Autoimmune comorbidities[a]	23 (4.7%)	1 (1.4%)
Others[b]	7 (1.4%)	2 (2.8%)
Switched the index DMT to other DMT[c] during the follow-up period (N=82), n (%)	**N=57**	**N=25**
Any safety-related clinical outcomes within 60 days before or on the stopping date	25 (43.9%)	14 (56.0%)
Infections	14 (24.6%)	7 (28.0%)
Optic neuritis	1 (1.8%)	2 (8.0%)
Autoimmune comorbidities[a]	8 (14.0%)	1 (4.0%)
Others[b]	5 (8.8%)	7 (28.0%)

N is the total study population of the cohort/group; n is the subset of total study population of the cohort/group.
[a]Autoimmune comorbidities included anemia, nephropathy, thrombocytopenia, unspecified acquired hypothyroidism.
[b]Other safety outcomes include seizures, macular edema, neutropenia, nasopharyngitis, any malignancies.
[c]Other DMTS include alemtuzumab, beta-interferon, cladribine, daclizumab, dimethyl fumarate, diroximel fumarate, immunoglobulin, glatiramer acetate, monomethyl fumarate, and teriflunomide.
CD, cluster of differentiation; DMTs, disease-modifying therapies; HE, high-efficacy; ME, moderate-efficacy; S1P, sphingosine-1-phosphate.

Limitations

- As with any analysis using administrative claims data, the study is subject to possible coding inaccuracies, which may affect the precise identification of patients and outcomes
- The results may not be fully generalizable to populations underrepresented in commercial claims databases, such as uninsured individuals and adults older than 65 years
- The algorithm used to identify individuals with RRMS may have introduced misclassification across MS subtypes

CONCLUSION

- Overall, the real-world findings emphasize the need for RRMS therapies that support durable treatment persistence while balancing efficacy, safety, and tolerability across evolving clinical contexts
 - Despite the use of HE- or ME-DMTs, substantial treatment discontinuation and switching persisted over a 2-year follow-up period, highlighting ongoing unmet needs in long-term RRMS management
 - Switching was more frequently observed among individuals initiating ME-DMTs, whereas discontinuation occurred more commonly among HE-DMT initiators, underscoring distinct real-world treatment trajectories across efficacy classes
 - Safety-related clinical outcomes were frequently observed within 60 days before stopping or switching, with infections being the most common; however, these findings are descriptive, and a casual relationship between safety events and treatment changes cannot be inferred
 - The study period (2018–2023) encompassed the COVID-19 pandemic, which may have contributed to observed treatment interruptions and switching patterns, particularly in the context of therapies associated with infection risk

Disclosures

Lita Araujo, Jaja Sangbana, Nupur Greene, and Michelle Baglia: Employees of Sanofi and may hold stock or stock options in the company.

Roopali Gandhi: At the time of the study, she was an employee of Sanofi and may or may not have held stock or stock options in the company.

Marian H. Tarbox and **Ashis K. Das:** At the time of the study, they were employees of PHAR (now a part of ADVI Health), which was paid by Sanofi to conduct the research described in this abstract. PHAR also discloses financial relationships with the following commercial entities outside of the submitted work: Akcea, Amgen, Celgene, Delfi Diagnostics, Dompe, Exact Sciences Corporation, Genentech, Gilead, GRAIL, Greenwich Biosciences, Ionis, Nobelpharma, Novartis, Pardes, Prothena, Pfizer, Recordati, Regeneron, Sanofi US Services, and Sunovion.

Poster presented at the Consortium of Multiple Sclerosis Centers (CMSC) Annual Meeting 2026; Charlotte, NC, US; May 27–29, 2026.

Acknowledgments

Medical writing and editorial assistance were provided by Dhaval Srivastava and Avinash Kumar of Sanofi. Planning, review, and coordination assistance was provided by Leah Granby of Sanofi.

Funding

This study was sponsored by Sanofi.

References

1. Müller J et al. *Brain Behav.* 2024;14:e3498.
2. Freeman L et al. *Drugs Real World Outcomes*. 2021;8:497–508.
3. Zaheer R et al. *Mult Scler Relat Disord*. 2023;71:104575.

Copies of this poster obtained through Quick Response (QR) code are for personal use only.